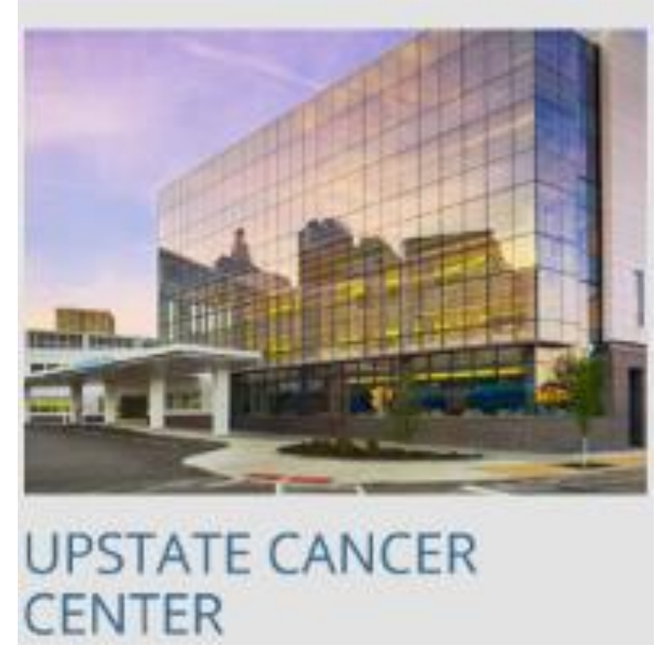

UPSTATE CANCER
CENTER

Immunotherapy Predictive Biomarkers in Metastatic Breast Cancer

A Sivapiragasam[1], ES Sokol[2], LA Albacker[2], JA Elvin[2], JA Vergilio[2], JK Killian[2], N Ngo[2], S Ramkissoon[2], EA Severson[2], A Hemmerich[2], DI Lin[2], SM Ali[2], AB Schrock[2], J Chung[2], P Reddy[2], BM Alexander[2], VA Miller[2], K McGregor[2], JS Ross[1,2]

1. Upstate Medical University, Syracuse, NY, USA; 2. Foundation Medicine, Cambridge, MA, USA

UPSTATE MEDICAL UNIVERSITY

ABSTRACT

Background: We queried whether comprehensive genomic profiling (CGP) of mBC subtypes could identify biomarkers that have been linked to responsiveness to immunotherapy (IO) treatments.

Methods: DNA was extracted from 3,871mBC: 1,388 ER+/HER2-, 1,969 *HER2* amplified (amp) and 643 TNBC. CGP was performed using a hybrid-capture assay. Tumor mutational burden (TMB) was determined on 0.83-1.14 Mbp of sequenced DNA and microsatellite instability (MSI) was determined on 114 loci in 564 cases. PD-L1 on TILs in subsets was determined by IHC (Ventana Sp142).

Results: Patient ages were similar. Genomic alterations (GA)/tumor were similar ranging from 5.9 to 7.3. MTOR pathway targets were common; lowest in TNBC. *CDH1* and *ESR1* GA most frequent in ER+/HER2- cases. *ERBB2* short variant (SV) mut were most frequent in ER+/HER2- and *HER2* amp and not seen in TNBC. Other kinase targets were uncommon except for *FGFR1* GA in ER+/HER2-. *BRCA1/2* GA least frequent in *HER2* amp. *AR* was amplified in 1% of all mBC. Markers of potential IO benefit: *CD274* (*PD-L1*) amp (1-3%), *BRAF* GA (1-4%), TMB of ≥10 mut/Mb (8-12%), TMB of ≥20 mut/Mb (2-3%), MSI-High (0.1-0.4%), *PBRM1* GA (1%) and low (1-10%) or high (0-3%) PD-L1 staining. Potential markers of resistance: inactivating GA in *STK11* (1-2%) and *MDM2* amplification (3-6%).

Conclusions: In addition to guiding targeted therapy selection, CGP shows potential to identify GA linked to response and resistance to IO in mBC. The demonstrations of clinical benefit of immunotherapy in mBC supports the need for the development of biomarkers used to guide the use of ICPI drugs for these patients.

MATERIALS AND METHODS

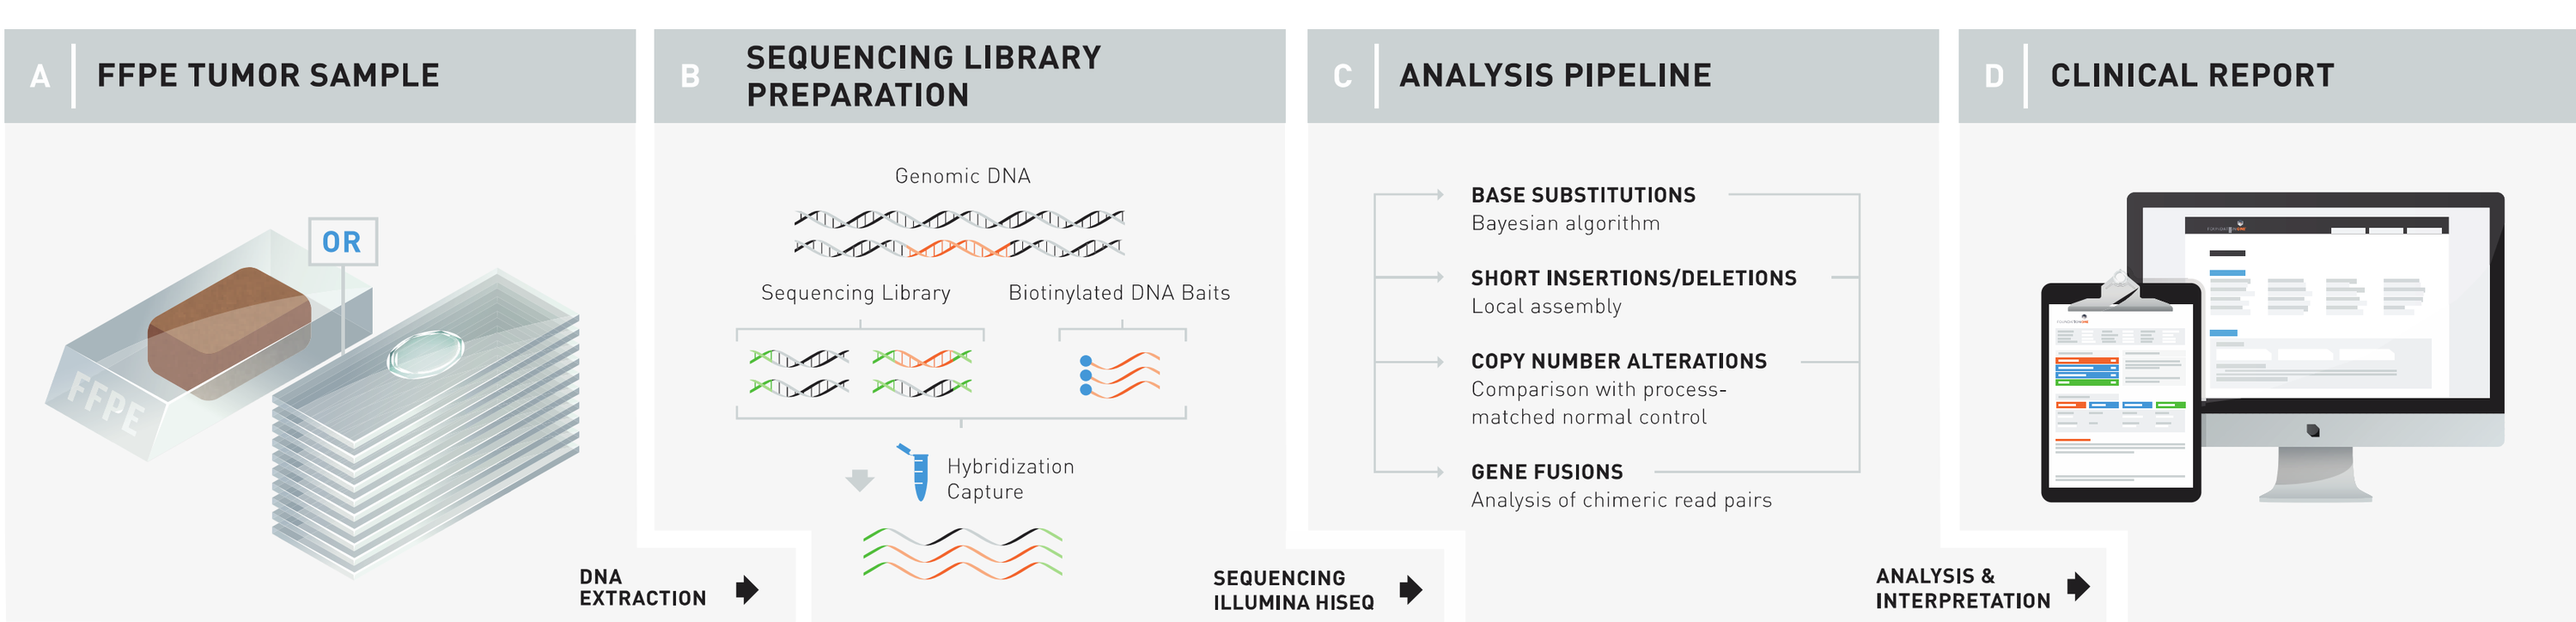

A FFPE TUMOR SAMPLE
OR
DNA EXTRACTION
B SEQUENCING LIBRARY PREPARATION
Genomic DNA
Sequencing Library
Biotinylated DNA Baits
Hybridization Capture
SEQUENCING ILLUMINA HISEQ
C ANALYSIS PIPELINE
BASE SUBSTITUTIONS
Bayesian algorithm
SHORT INSERTIONS/DELETIONS
Local assembly
COPY NUMBER ALTERATIONS
Comparison with process-matched normal control
GENE FUSIONS
Analysis of chimeric read pairs
ANALYSIS & INTERPRETATION
D CLINICAL REPORT

- ≥50 ng DNA extracted from 40 µm of FFPE sections
- Sequencing performed for up to 315 cancer-related genes and introns from 28 genes commonly rearranged in cancer
- Hybrid capture-based sequencing using adaptor ligation-based libraries
- Mean coverage depth >600X
- Base substitutions, insertions and deletions (short variants; SV), rearrangements, and copy number changes were assessed
- Tumor mutational burden (TMB) calculated from 0.83-1.14 Mb sequenced DNA

RESULTS

5	ER+/HER2-	HER2 Amp	TNBC
Cases	1,259	1,969	643
Age (range in years)	55 (23-89+)	55 (20-89+)	53 (20-85)
GA/tumor	6.1	7.3	5.9
MTOR GA	*PIK3CA* (36%) *PTEN* (6%) *NF1* (3%)	*PIK3CA* (38%) *PTEN* (5%) *NF1* (7%)	*PIK3CA* (19%) *PTEN* (15%) *NF1* (8%)
CDH1 GA	7%	4%	3%
ESR1 GA	16%	6%	0.5%
BRCA1/2 GA	3%/6%	2%/3%	7%/3%
HER2 amp	0%	100%	0%
HER2 SV	3%	7%	0%
Other Kinase targets	*FGFR1 (18%)* *FGFR2 (2%)* *EGFR (2%)* *KIT (1%)* *MET (0.4%)* *BRAF (2%)*	*FGFR1 (11%)* *FGFR2 (1%)* *EGFR (3%)* *KIT (2%)* *MET (1%)* *BRAF (1%)*	*FGFR1 (8%)* *FGFR2 (4%)* *EGFR (4%)* *KIT (2%)* *MET (1%)* *BRAF (4%)*
AR amp	1%	1%	1%
MSI High	0.2%	0.1%	0.4%
CD274 (PD-L1) amp	1%	1%	3%
TMB > 10 mut/Mb	8%	12%	9%
TMB > 20 mut/Mb	3%	2%	2%
PD-L1 IHC low/high	10%/0%	10%/0%	18%/1%
PBRM1 GA	1%	1%	1%
STK11 GA	1%	1%	2%
MDM2 amp	6%	5%	3%

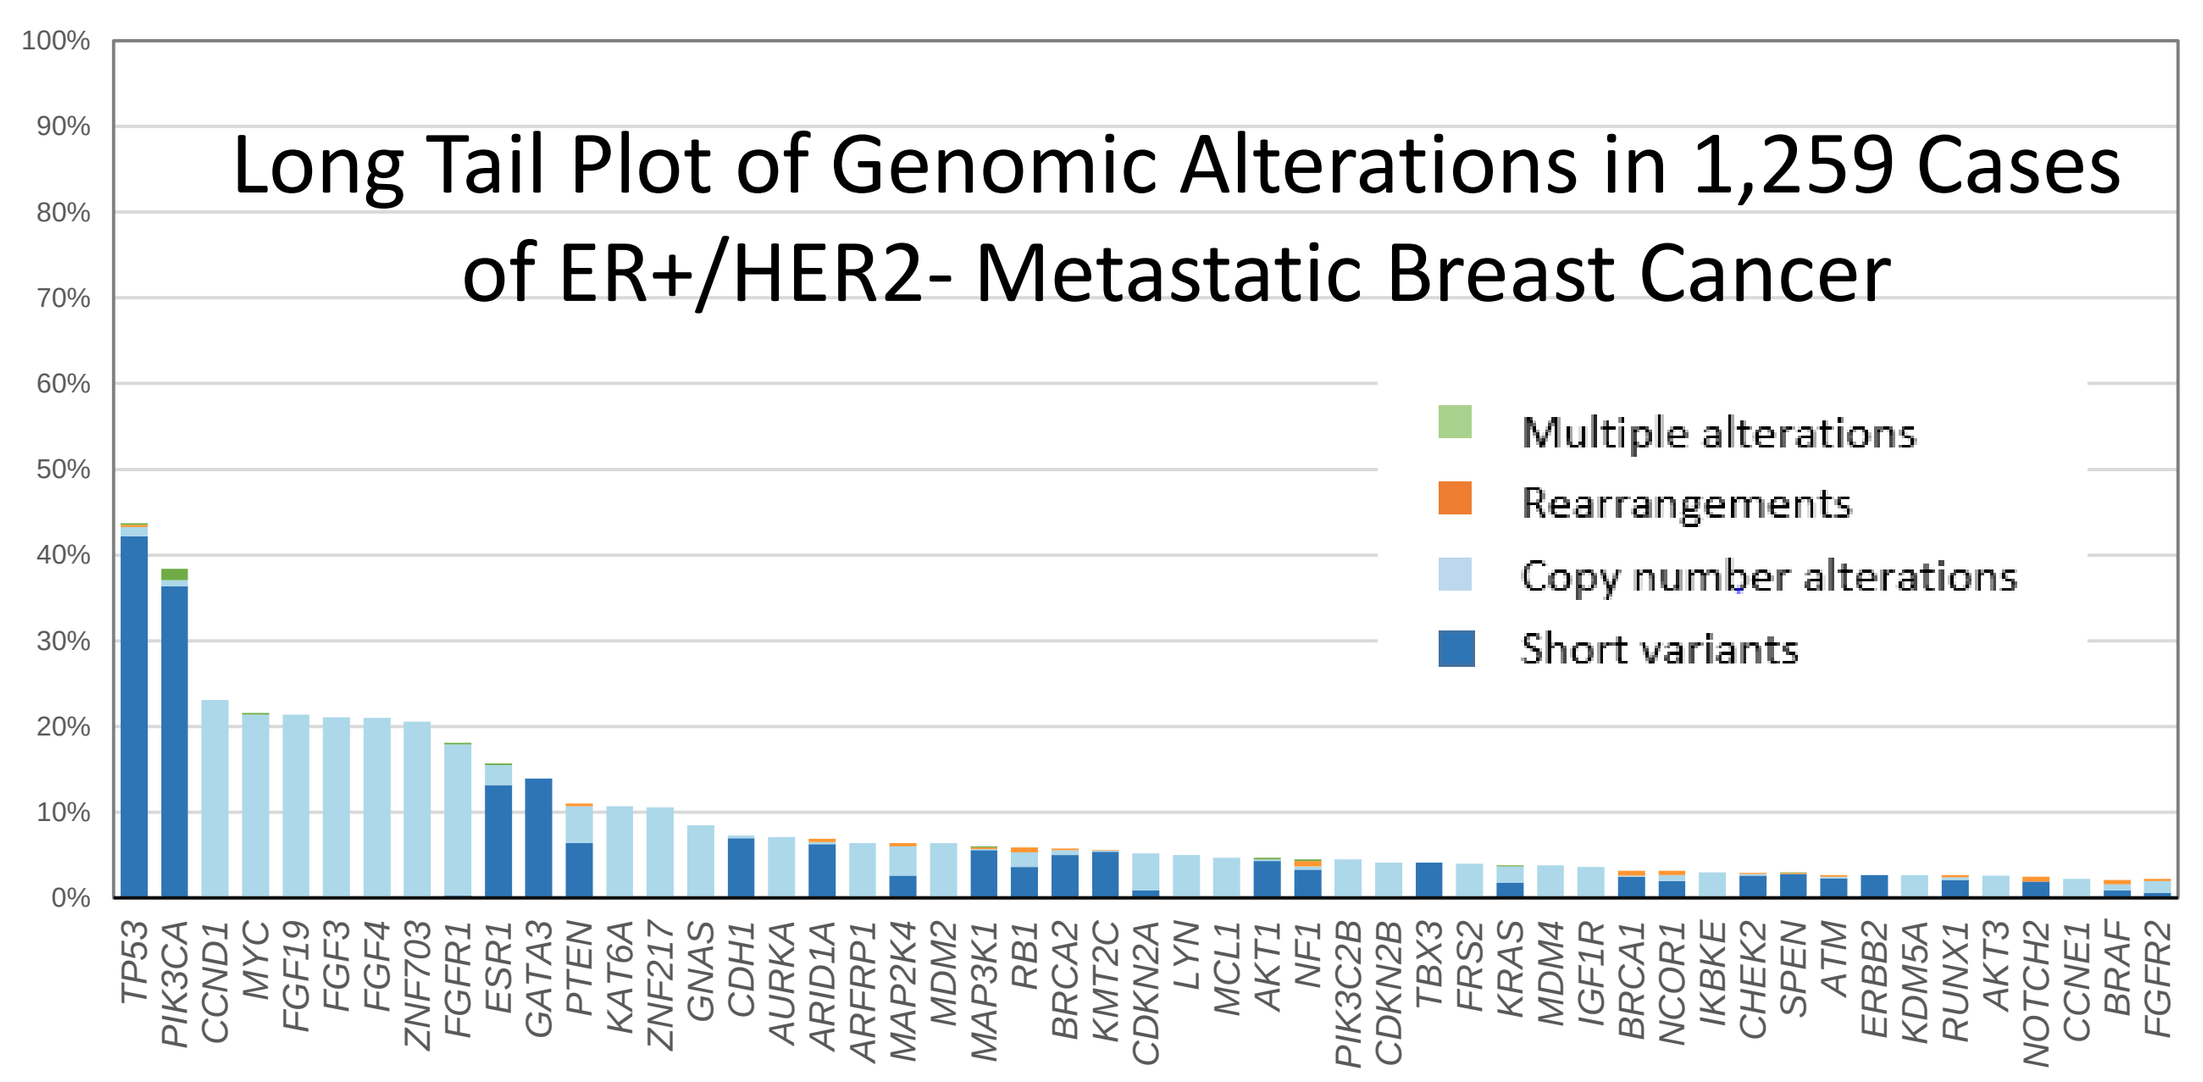

Long Tail Plot of Genomic Alterations in 1,259 Cases of ER+/HER2- Metastatic Breast Cancer
Multiple alterations
Rearrangements
Copy number alterations
Short variants

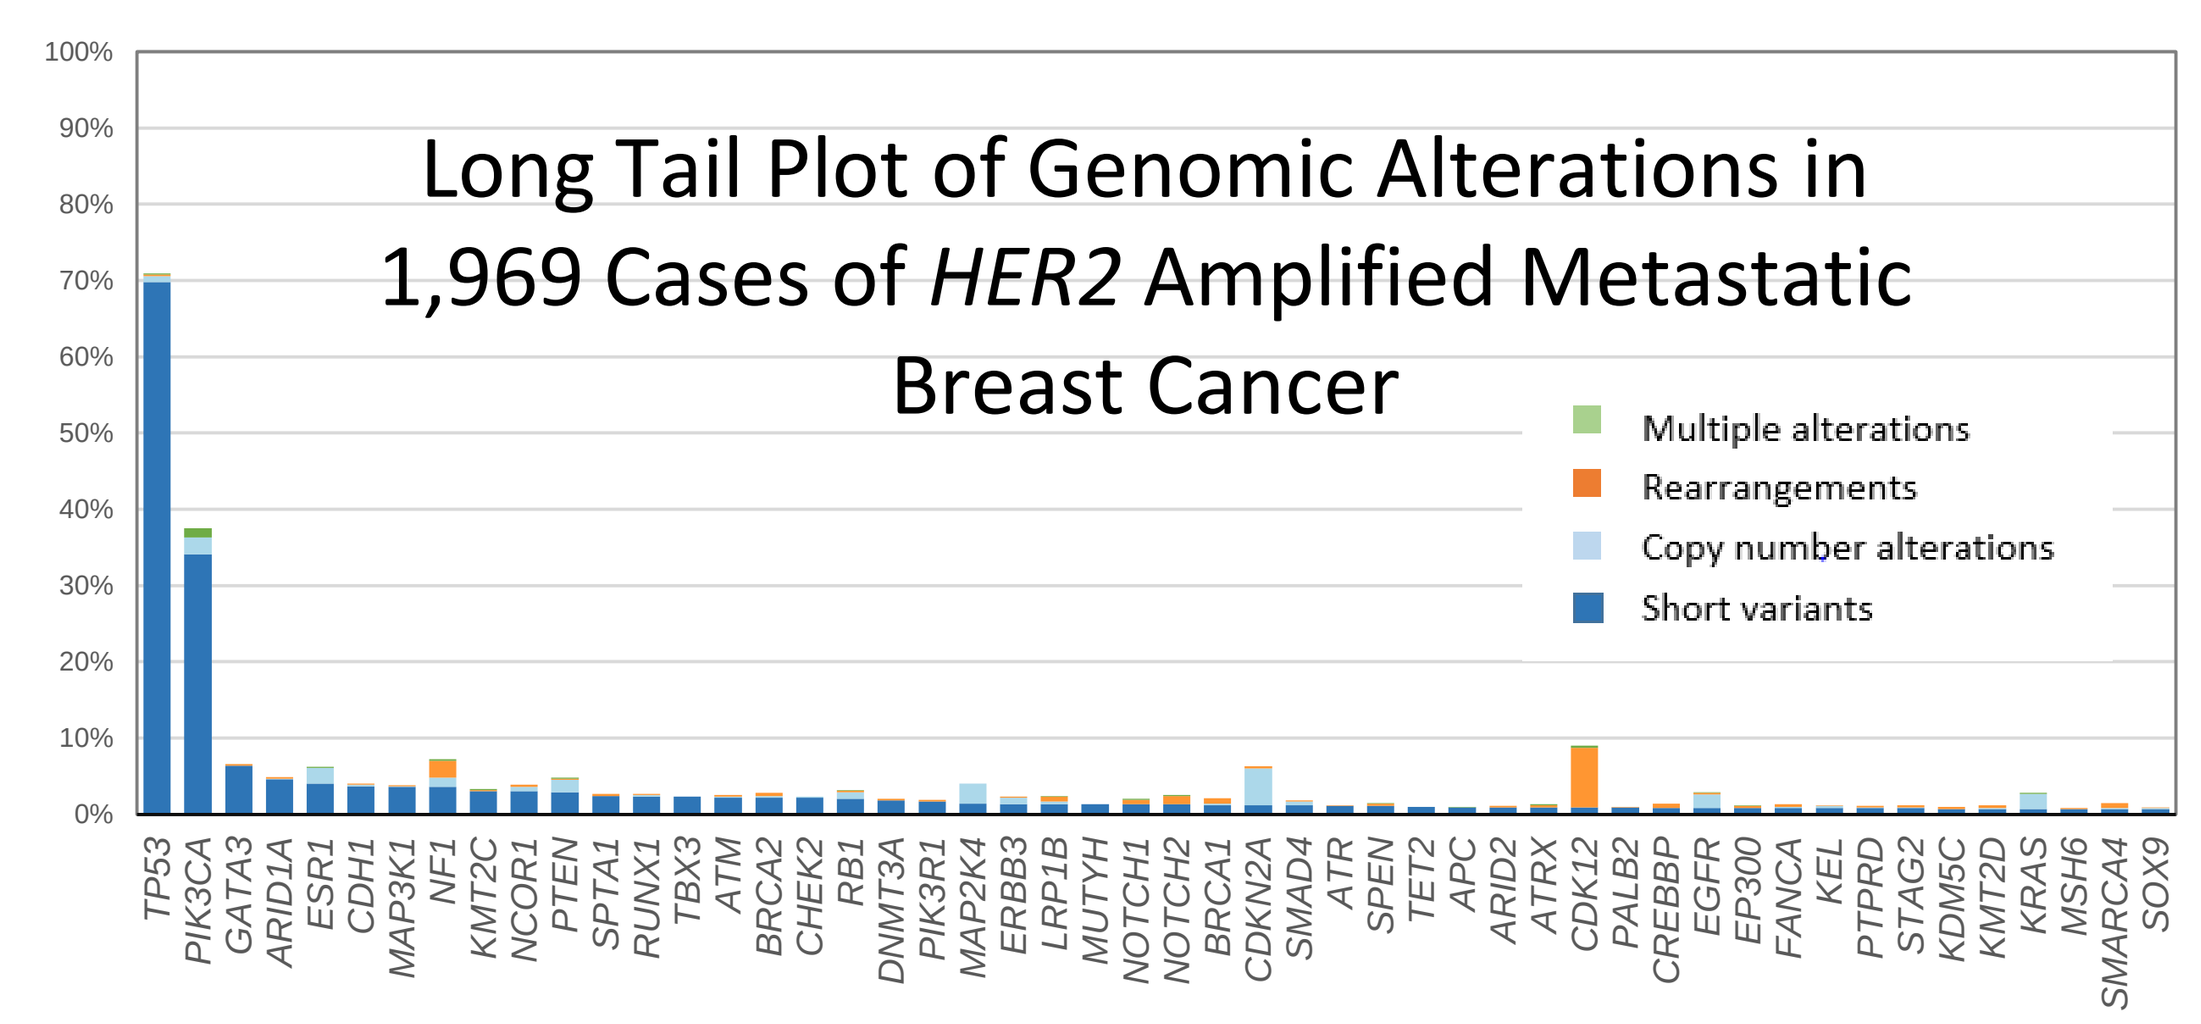

Long Tail Plot of Genomic Alterations in 1,969 Cases of HER2 Amplified Metastatic Breast Cancer
Multiple alterations
Rearrangements
Copy number alterations
Short variants

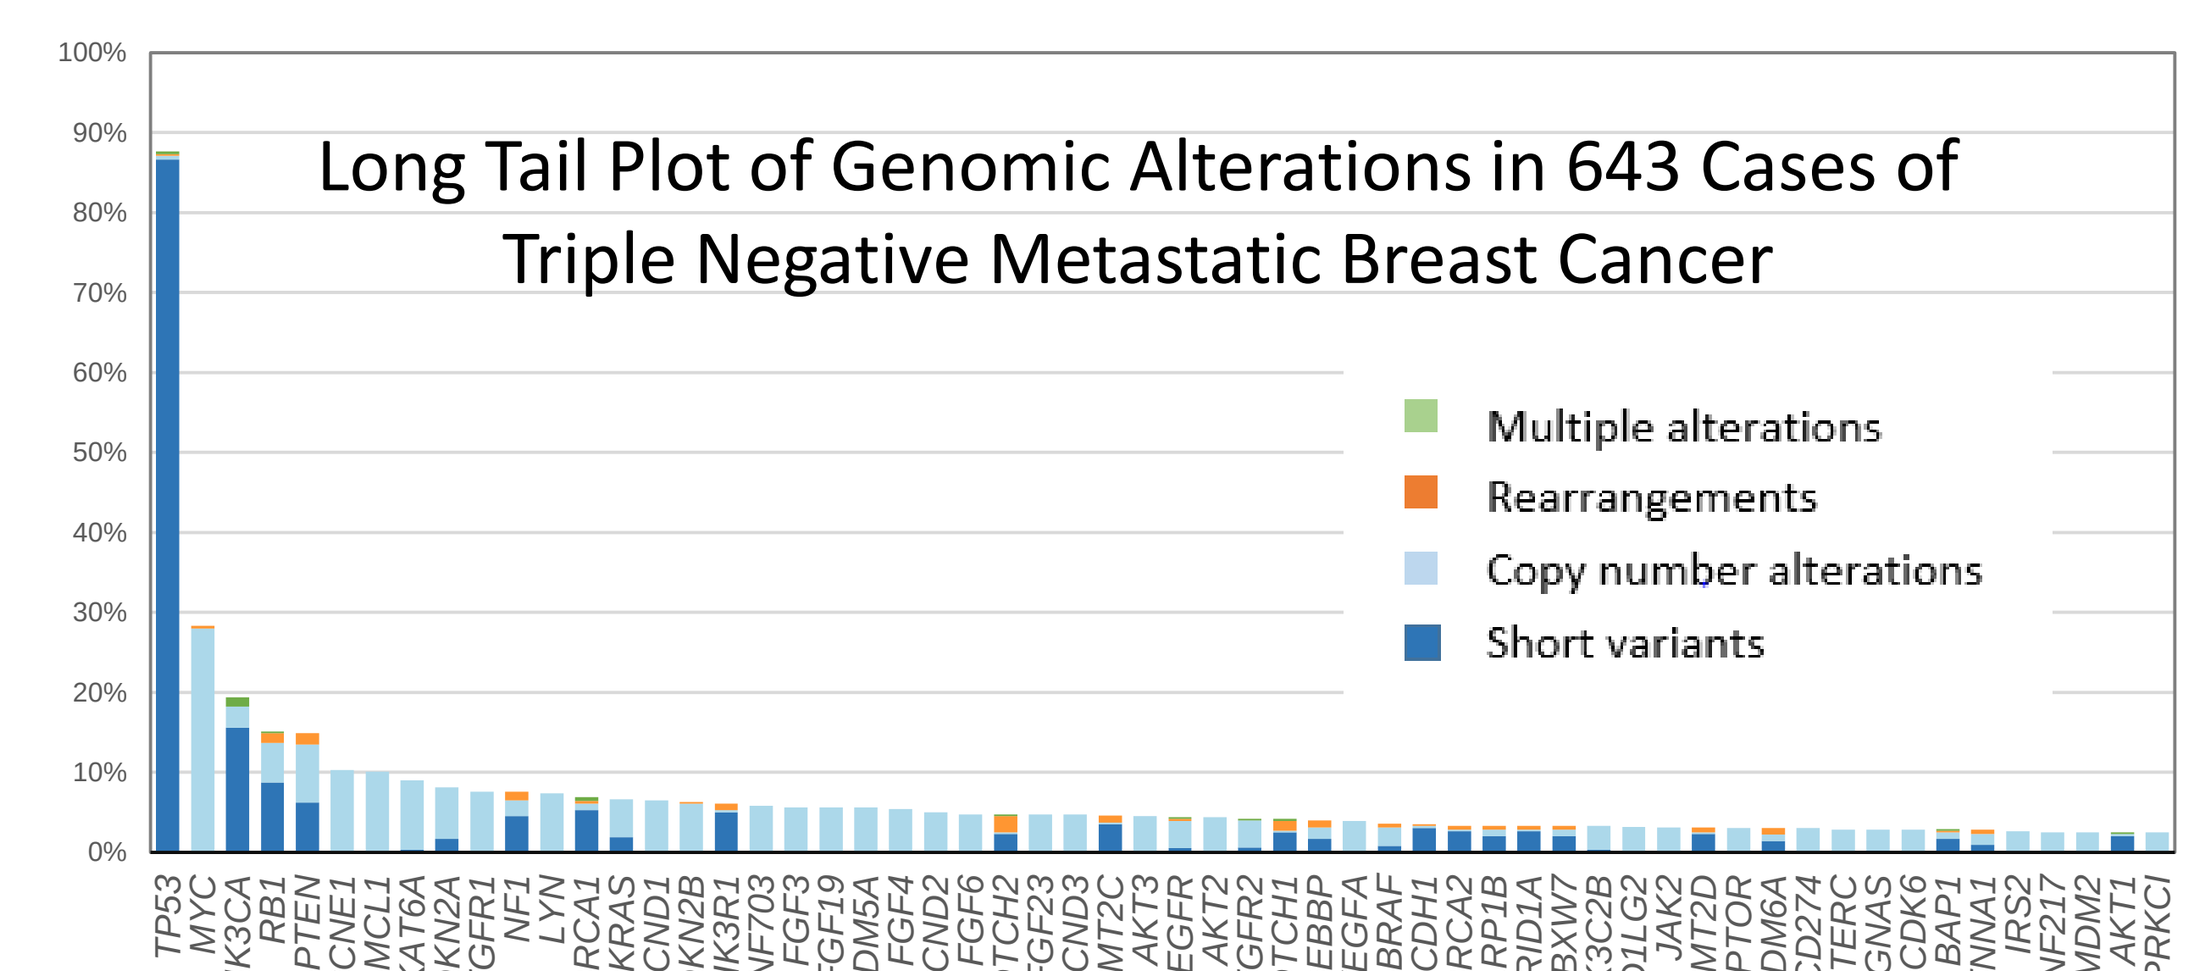

Long Tail Plot of Genomic Alterations in 643 Cases of Triple Negative Metastatic Breast Cancer
Multiple alterations
Rearrangements
Copy number alterations
Short variants

- Patient ages and GA/ tumor were similar
- MTOR pathway targets were common and lowest in TNBC
- *CDH1* and *ESR1* GA most frequent in ER+/HER2- cases
- *ERBB2* short variant (SV) mut were most frequent in ER+/HER2- and *HER2* amp
- Other kinase targets were uncommon except for *FGFR1*
- *BRCA1/2* GA least frequent in *HER2* amp

Markers of potential IO benefit:

-*CD274* (*PD-L1*) amp (1-3%)
-*BRAF* GA (1-4%)
-TMB of ≥10 mut/Mb (8-12%)
-TMB of ≥20 mut/Mb (2-3%)
-MSI-High (0.1-0.4%)
-*PBRM1* GA (1%)
-low (10%-18%) or high (0-1%) PD-L1 staining

Potential markers of potential IO resistance:

-inactivating GA in *STK11* (1-2%)
-*MDM2* amplification (3-6%)

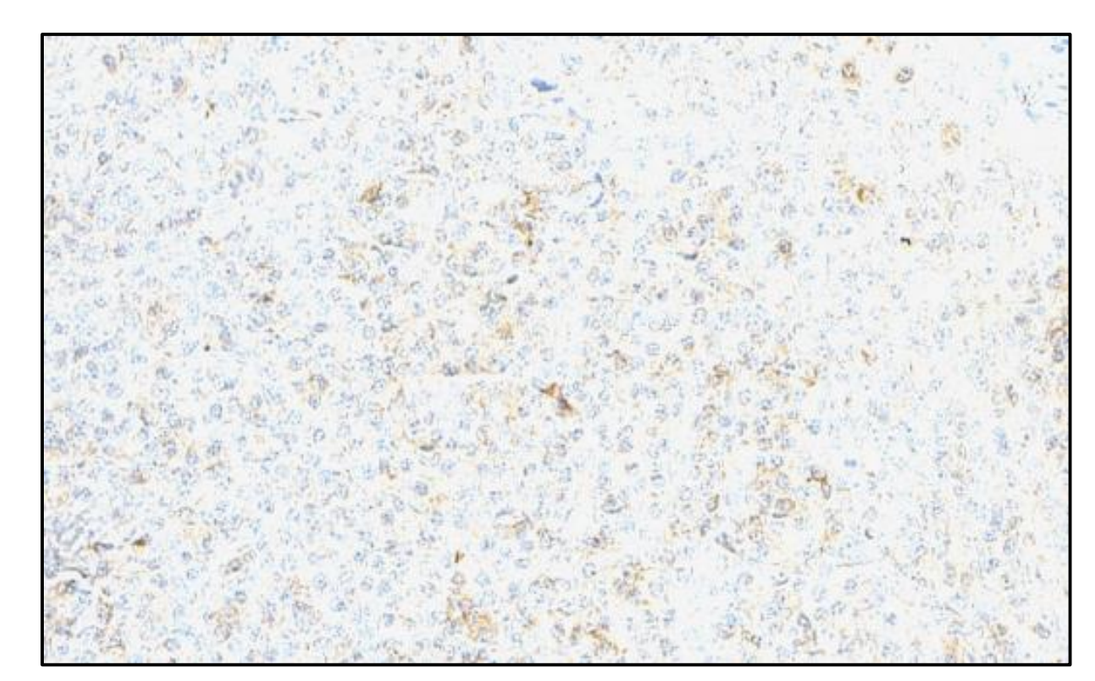

PD-L1 Expression of TILs in TNBC

Using the FDA-approved Ventana SP142 antibody, immunostaining for PD-L1 in tumor infiltrating lymphoid cells was highest in the triple negative breast cancer group. This result is similar to previously reported studies. In one published report, 20% of TNBC express PD-L1 and PTEN is deficient in almost half of the PD-L1-expressing tumors. The Phase III Impassion130 study led to the approval of the combination of chemotherapy (specifically Abraxane®, nab-paclitaxel) and immunotherapy (Tecentriq®, atezolizumab) which significantly reduced the risk of disease progression among patients with metastatic triple negative breast cancer.

CASES

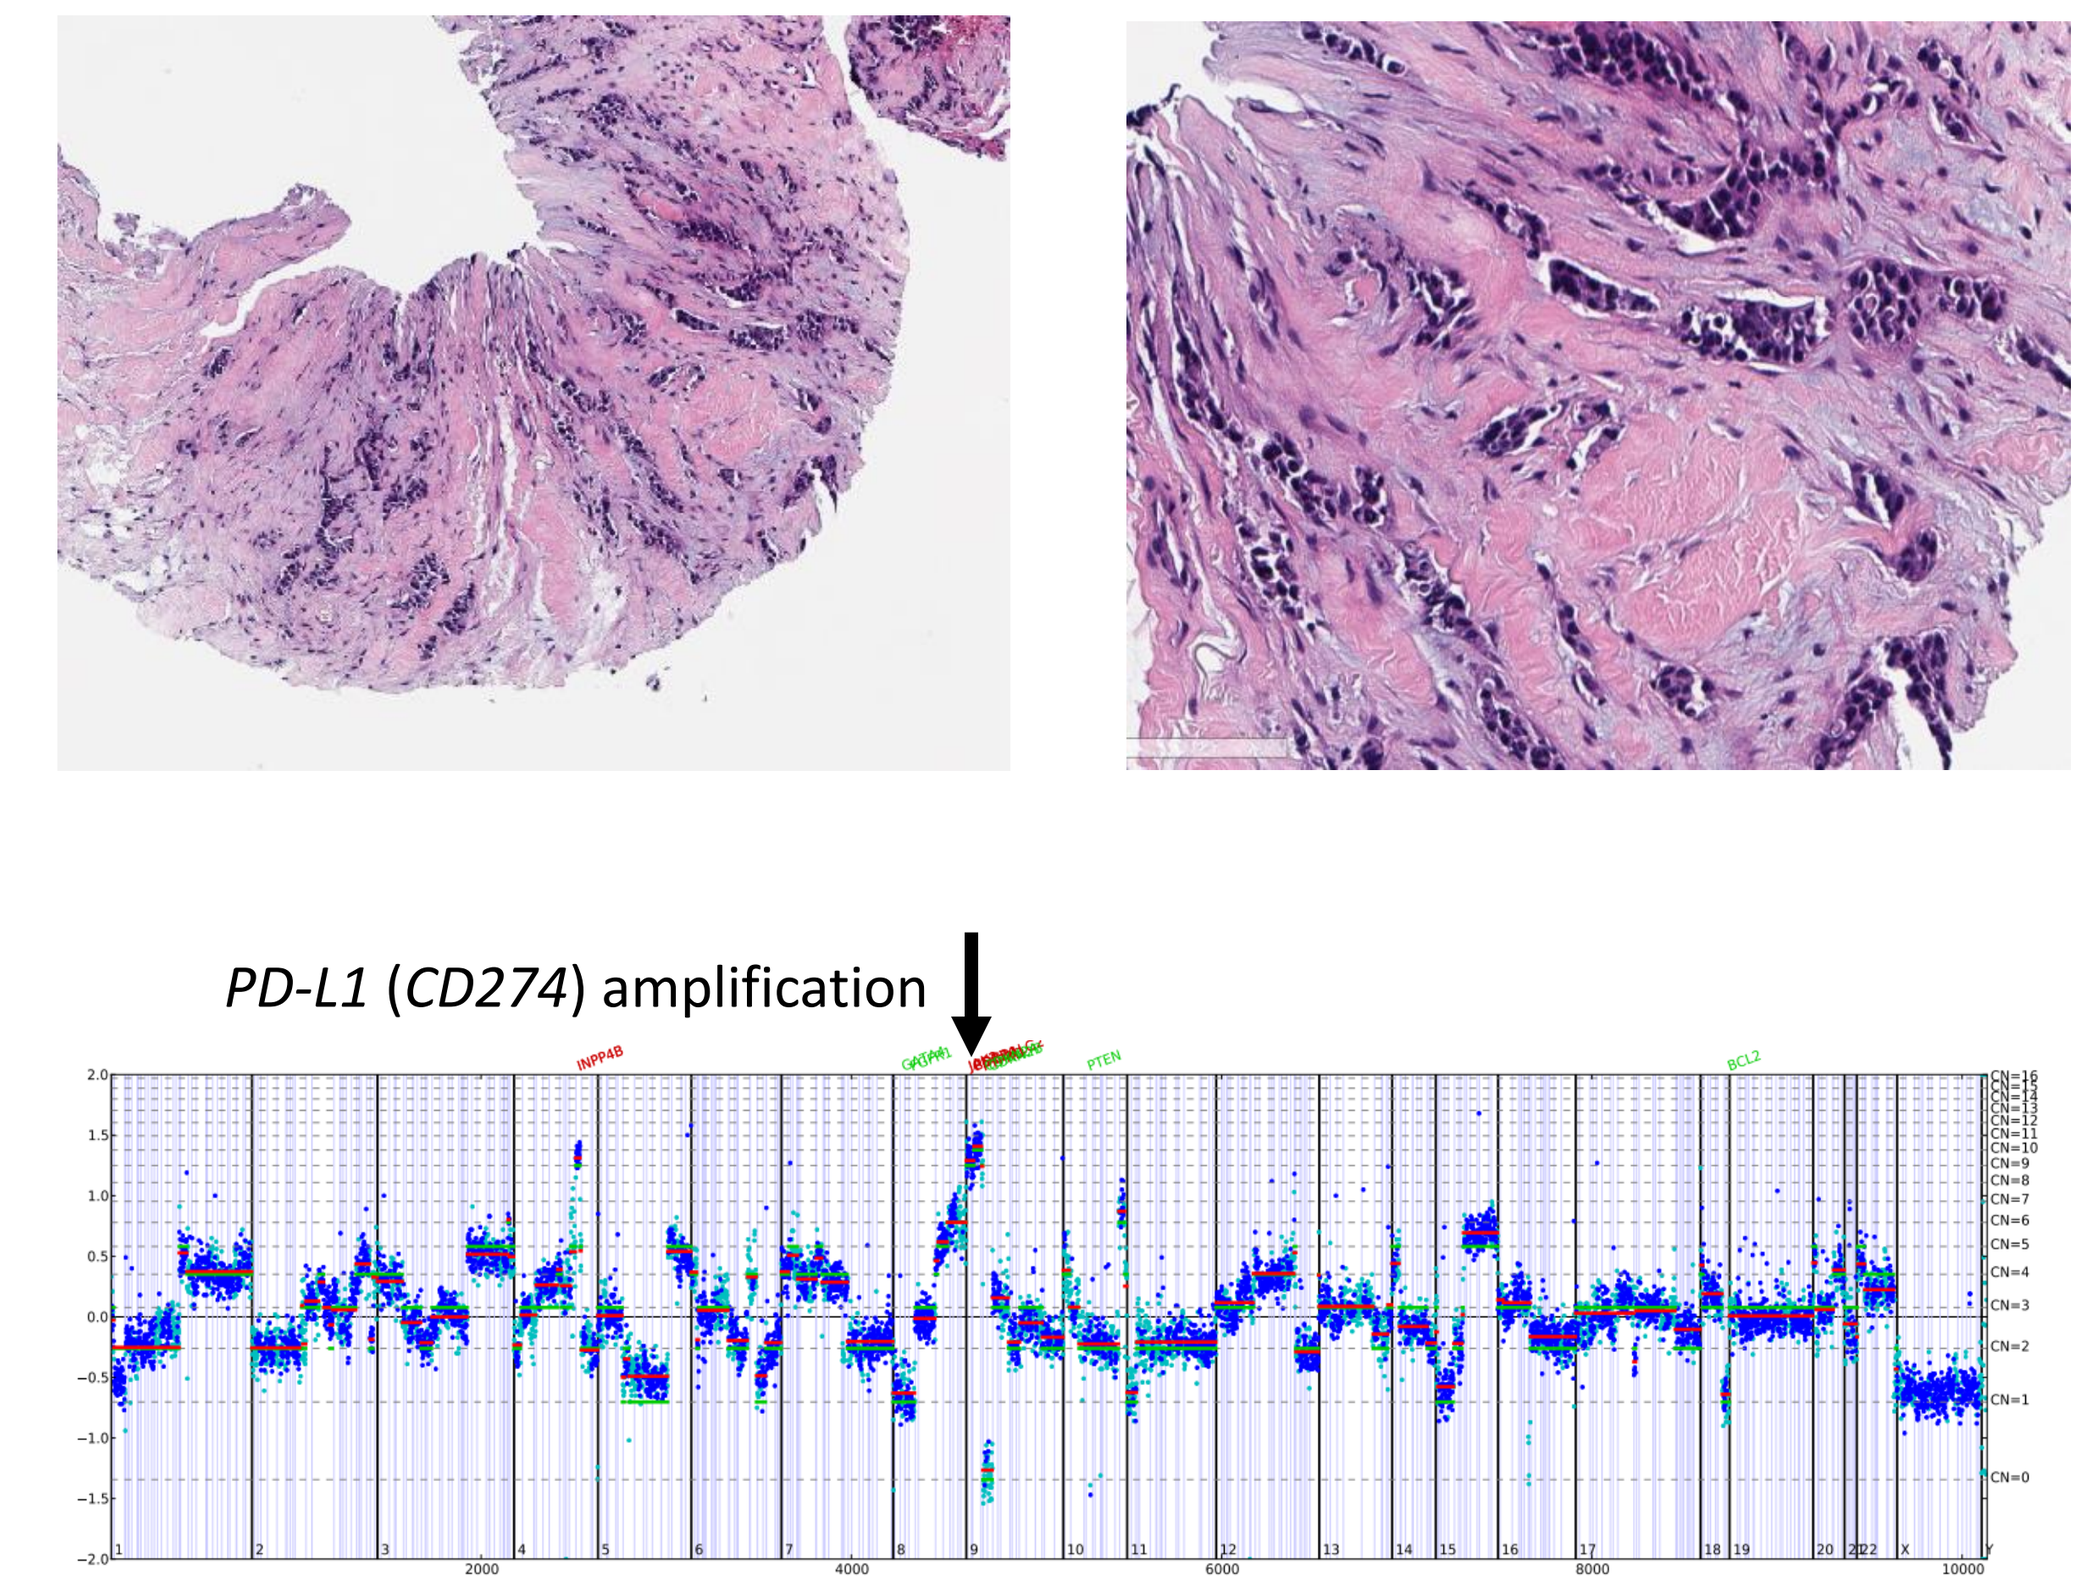

PD-L1 (CD274) amplification

Needle biopsy of a sternal wall mass in a 72 year old TNBC patient with stage IV disease. Comprehensive genomic profiling revealed amplification of the PD-L1 gene (CD274) and *PTEN* loss exon 7, *PDCD1LG2 (PD-L2)* amplification, *CDKN2A/B* loss, *CREBBP* Q1104fs*23, *JAK2* amplification and *TP53* K132E mutations. Amplification of *CD274* has been reported in 1% of breast cancers In the TCGA dataset. Amplification of chromosomal fragment 9p24, which includes *CD274*, has been reported in breast cancer. PD-L1 overexpression in breast cancer has been reported in 19-23% of cases and significantly associated with adverse clinic-pathological features and overall survival. On the basis of strong clinical evidence, *CD274* amplification may predict sensitivity to antibodies targeting PD-L1 or PD-1. In one recent study, a multivariate analysis suggested that PD-L1 amplification was highly predictive of immune checkpoint inhibitor efficacy in a wide variety of tumor types.

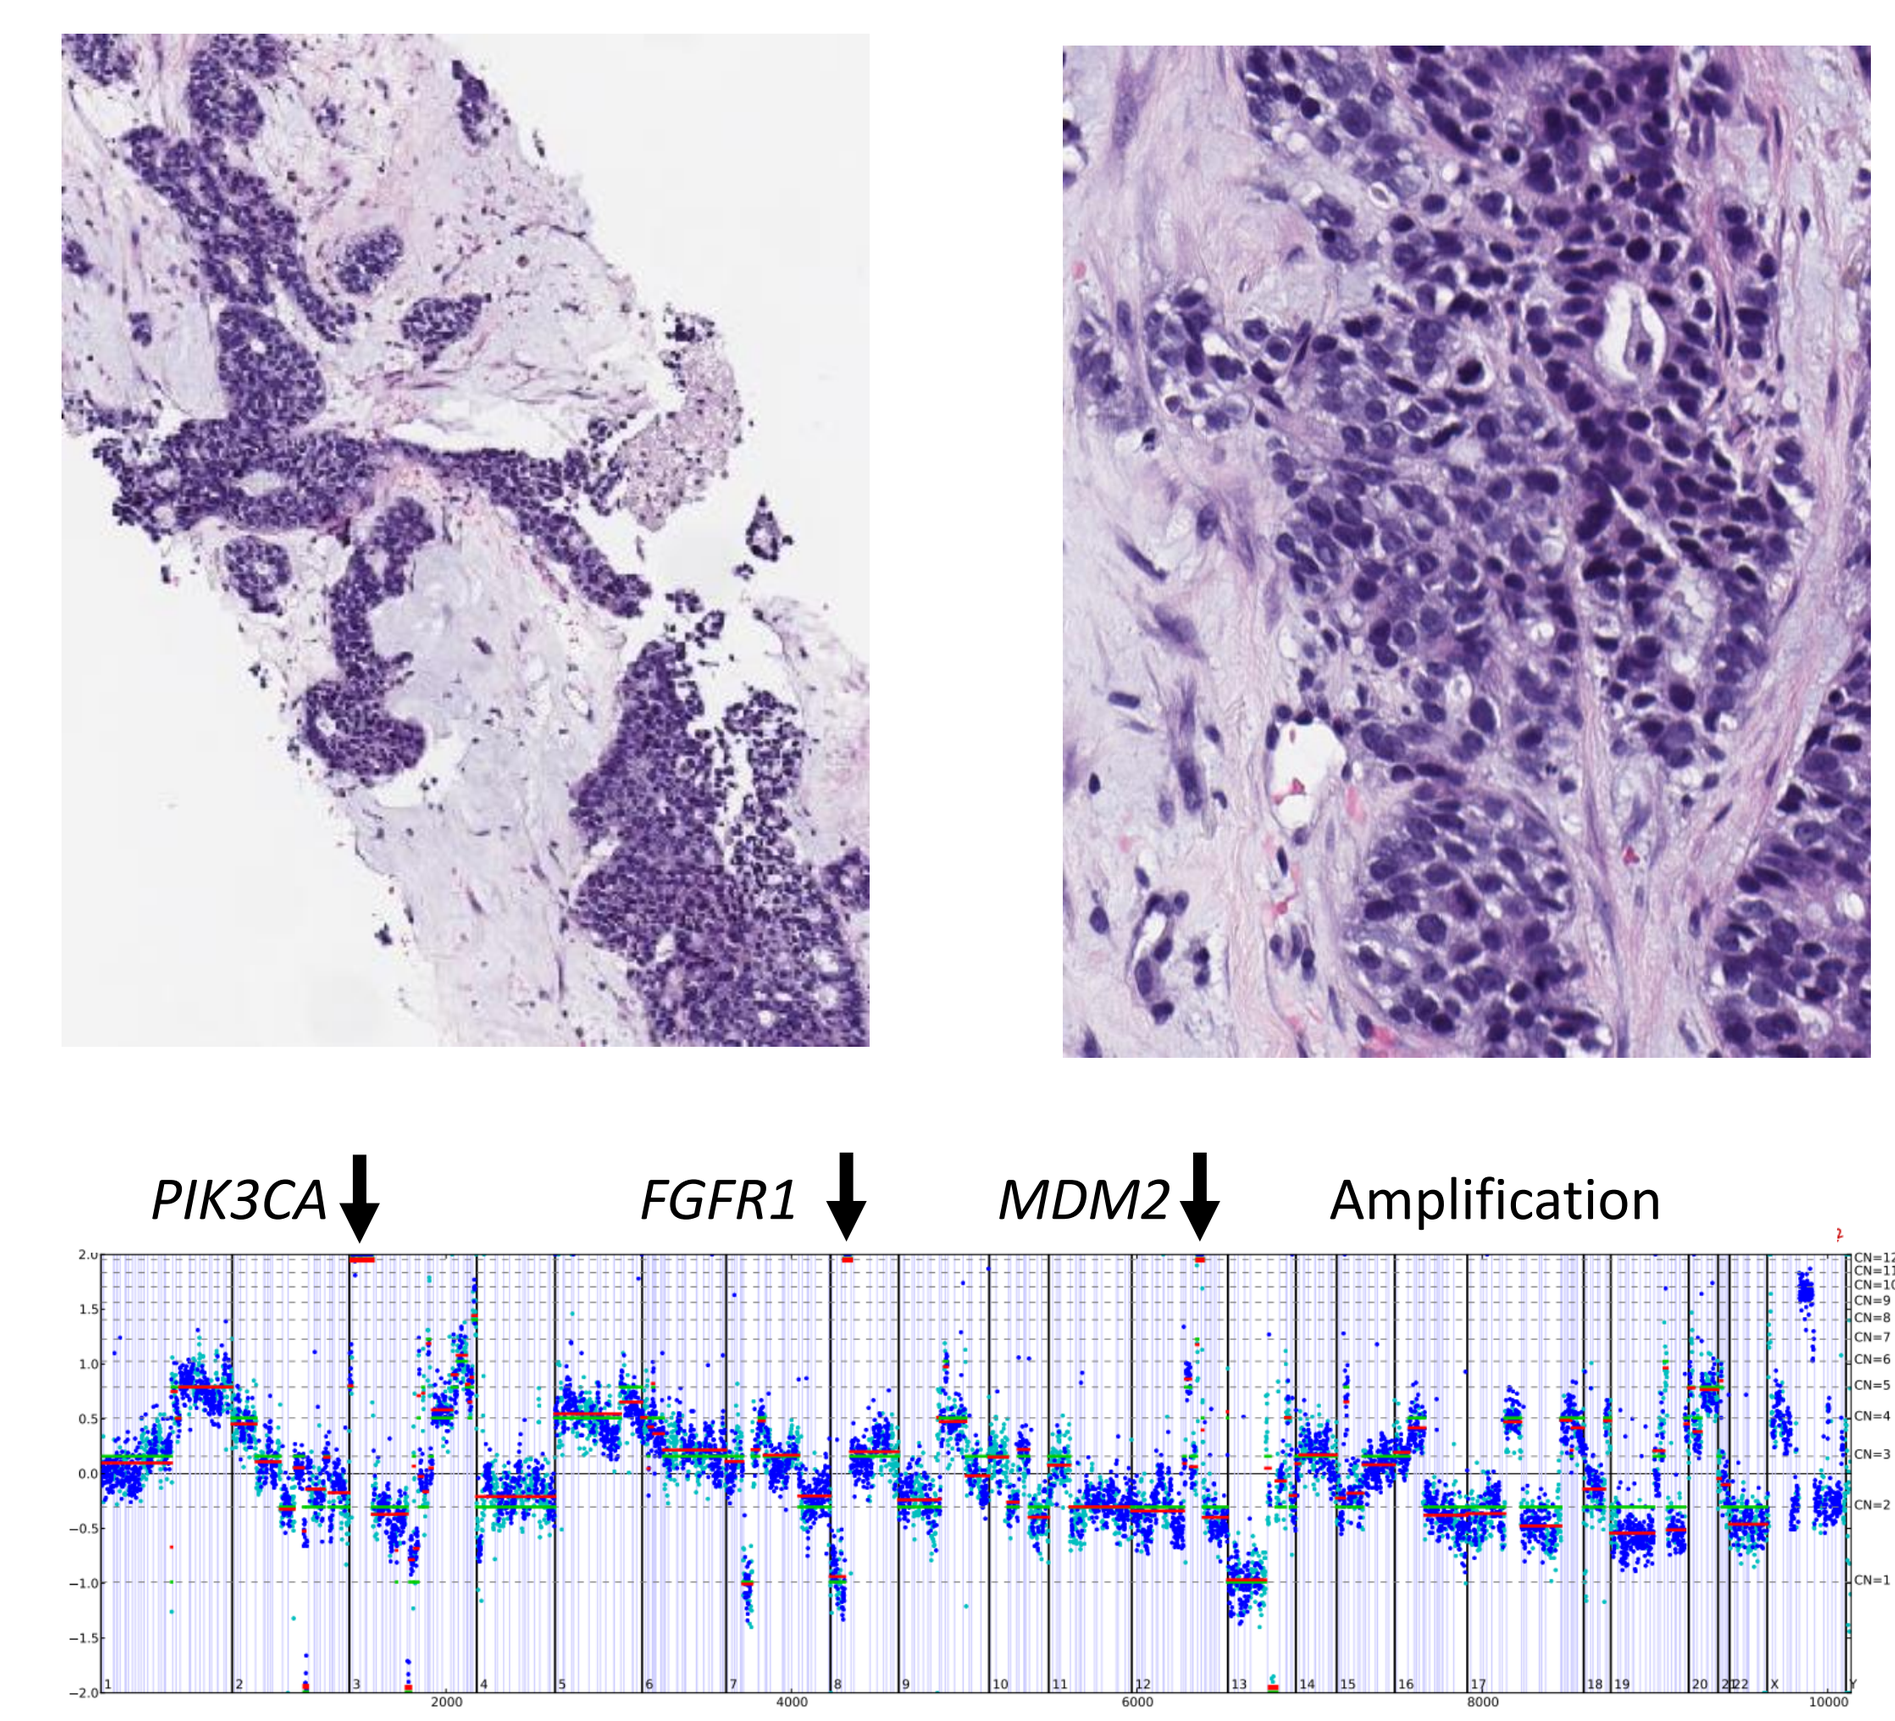

PIK3CA
FGFR1
MDM2
Amplification

Liver biopsy showing metastatic high grade TNBC in a 57 year old patient. Comprehensive genomic profiling of this tumor revealed *MDM2* amplification as well as amplifications of *FGFR1*, *PIK3CA* and *FRS2.* Short variant mutations were also present in *LRP1B*, *MAP2K4*, *MUTYH*, *SPTA1* and *TP53*. *RB1* loss was also found. *MDM2* amplification has been linked to hyper-progression of a wide variety of tumor types when treated with immune checkpoint inhibitors. *FGFR1* is a recognized target in TNBC. Although initial studies using established kinase inhibitors have not been impressive, newer anti-FGFR drugs are showing renewed promised in mid-stage clinical trials. The recent success of using the new anti-PIK3CA inhibitor alpelisib in ER+ breast cancer has created significant interest in using this agent in TNBC cases with *PIK3CA* genomic alterations demonstrated on tumor sequencing.

CONCLUSIONS

- In addition to guiding targeted therapy selection, CGP shows potential to identify GA linked to response and resistance to IO in mBC

- The demonstrations of clinical benefit of immunotherapy in mBC supports the need for the development of biomarkers used to guide the use of ICPI drugs for these patients