Midazolam Measurement and Modelling using Matrix Samplers (The 4M's Study)

Bikalpa Neupane[1, 2], Hussain Mulla [2], Neil Spooner[3], Paul Abu-Rabie[4], James Rudge[5], Hitesh Pandya[1,2]

[1] University of Leicester, [2] University Hospitals of Leicester NHS Trust, [3] Spooner Bioanalytical Solutions, [4] Glaxo SmithKline PLC, [5] Neoteryx Limited

Introduction

Midazolam, a short-acting sedative, hypnotic and anxiolytic benzodiazepine, is administered intravenously (IV) to critically unwell children and peri-operatively to children undergoing elective surgical procedures. Intravenous bolus doses of midazolam may cause cardio-respiratory depression. In contrast, incomplete sedation (tolerance) and withdrawal symptoms post discontinuation of drug are commonly observed in patients infused IV drug continuously for prolonged periods. Clearly, use of IV midazolam has potential to adversely affect clinical outcomes.

As with all drugs, midazolam efficacy and toxicity are dependent on multiple factors. Recently, critical illness / systemic inflammatory responses have been linked to altered midazolam pharmacokinetics[1,2] (PK). This implies that severely ill children treated with IV midazolam are at greatest risk of drug toxicity. Developing midazolam dosing schedules based on patient physiology requires comparative analysis of PK parameters in critically-ill children and in relatively well children. Ideally, the analysis should be performed using PK parameters determined from patients recruited into the same study.

However, It is difficult to perform PK studies in children because sampling relatively large volumes of blood frequently are significant barriers to recruiting children into research trials. While families find sampling micro-volumes of blood collected on GUTHRIE cards acceptable [3], from a bioanalytical perspective, GUTHRIE card based drug and metabolite analysis often provides only semi-quantitative data because of haematocrit effects[4]. VAMS™ a new 'direct-from-capillary-prick', blood collection platform offers a potential solution to quantitative bioanalysis.

Study Hypotheses

We hypothesise that

1. Critical illness alters midazolam PK
2. Blood samples collected on VAMS™ tips reliably and accurately quantify midazolam and 1-OH midazolam (active metabolite) concentrations

Study Objectives

- To compare blood midazolam and 1-OH midazolam concentrations determined from 'wet blood samples' and 'VAMS' tips
- To compare midazolam PK in critically ill (=PICU group) and clinically well children undergoing elective surgery (=ES group) administered IV drug
- To determine how well a computer-based physiological model of midazolam PK (PB-PK) predicts actual blood midazolam concentrations.

Methods

The study received Ethical (Derby NRES: No 140935), R&D approval and first recruited a patient in April 2015. Children in the ES group were recruited into the study on the day of surgery. Critically ill children were recruited at least 24 hours following admission to PICU.

Methods (Cont'd)

Blood (venous, arterial or capillary) for study purposes was obtained (i)) at predetermined sampling times using a dedicated cannula (ES group only) (ii) from any extra blood left in EDTA tubes sent for routine clinical laboratory tests (scavenged samples) and (iii) extra blood collected when sampling was performed for routine clinical tests (opportunistic sampling). Samples were collected at 'random' time points following administration of IV midazolam. From any one child samples were obtained at a maximum of 5 different time points with a maximum total volume (Wet and Dry) of 1 ml. Blood midazolam and 1-OH Midazolam concentrations are analysed by HPLC / MS at GSK Laboratories, Ware, UK. The upper and lower limits of quantification for both metabolite assays are 5 ng/ml and 5000 ng/ml. Midazolam and 1-OH midazolam concentration data were analysed using descriptive statistics. Pearson correlation (R^2) was calculated using EXCEL software.

VAMS ™ TIPS

- 10 µl blood per tip
- Maximum 3 tips/ sample point
- Storage/ transportation: Ambient temperature

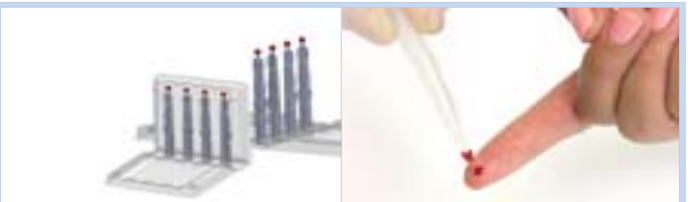

Results

50 (27 Male, 23 Female) critically-ill children and 21 (15 Male, 6 Female) children undergoing elective surgery have been recruited to the study (Figure 1). So far, 251 'paired' wet and dried blood samples (N = 179 PICU group, 72 = ES group) have been collected from these children. No blood samples either wet or dry, have been discarded or rejected for bio analysis due to collection problems. The correlation coefficients between concentration of [midazolam] and [1-OH midazolam] obtained via wet and VAMS dried blood were respectively 0.97 and 0.97 (Figure 4) with a bias of 3.5 %.

Figure 1. Age, Sex and Illness severity of children recruited into the study; *N= 71*

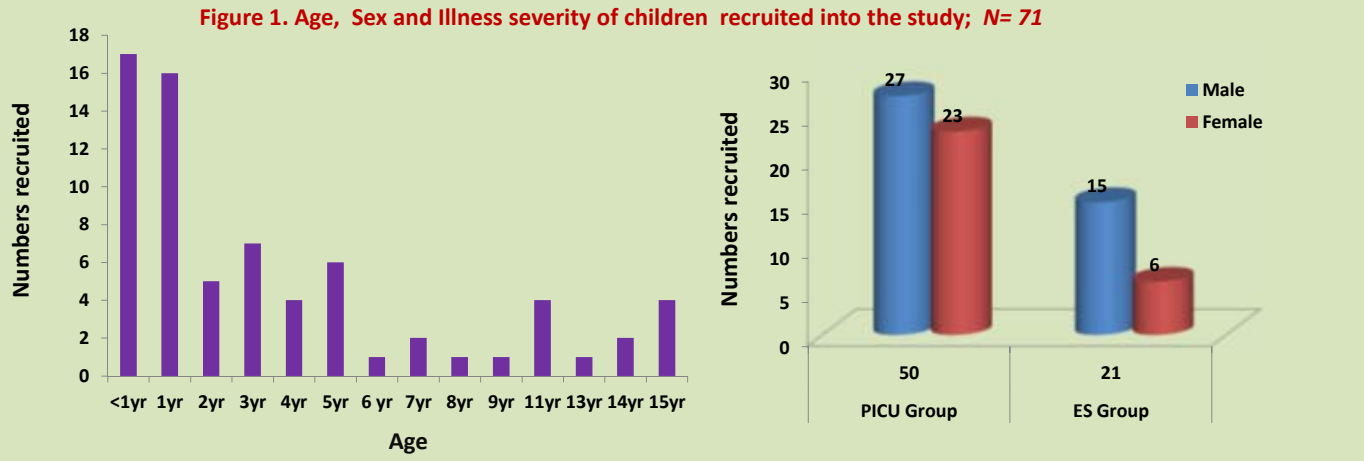
Numbers recruited
<1yr 1yr 2yr 3yr 4yr 5yr 6 yr 7yr 8yr 9yr 11yr 13yr 14yr 15yr
Age
Male
Female
27
23
15
6
50
PICU Group
21
ES Group

Figure 2. Whole blood Midazolam concentrations in (a) Wet and (b) Dry samples

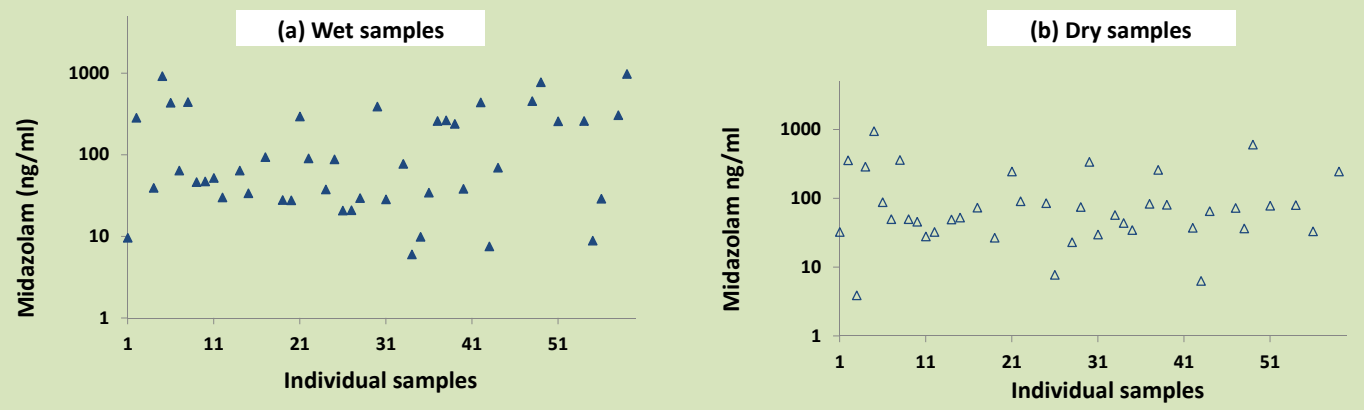
(a) Wet samples
Midazolam (ng/ml)
Individual samples
(b) Dry samples
Midazolam ng/ml
Individual samples

	PICU Group	ES Group
Mean Midazolam Concentration (ng/ml)	322	28
Range Midazolam Concentration (ng/ml)	5 - 1987	5 - 161

N = 56 patients
N = 199 time points

Figure 3. Whole blood 1-OH Midazolam concentrations in (a) Wet and (b) Dry samples

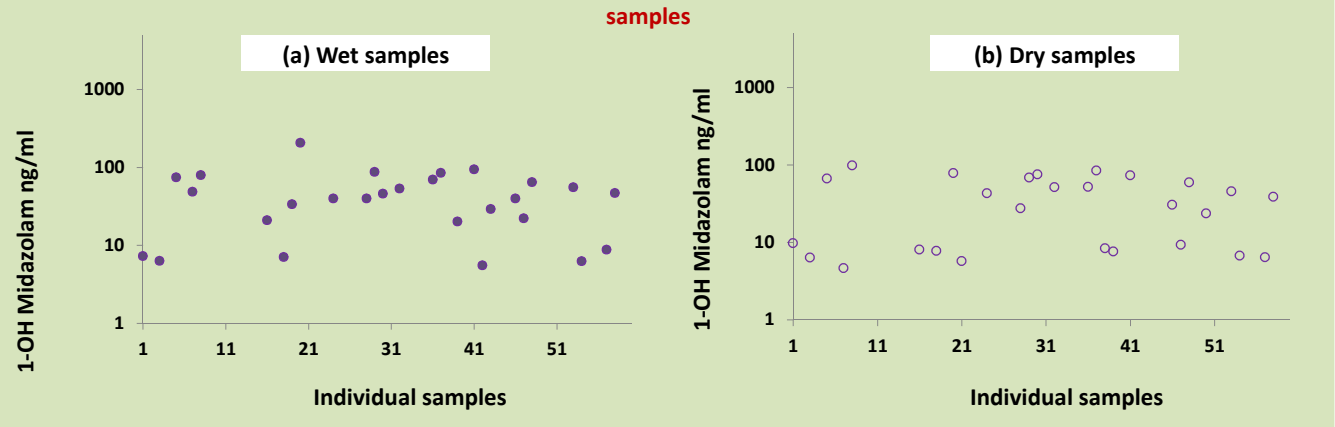
(a) Wet samples
1-OH Midazolam ng/ml
Individual samples
(b) Dry samples
1-OH Midazolam ng/ml
Individual samples

	PICU Group	ES Group
Mean 1-OH Midazolam Concentration (ng/ml)	42	6.6
Range 1-OH Midazolam Concentration (ng/ml)	5 - 211	5 - 8

N = 56 patients
N = 199 time points

Figure 4. Relationship between (a) Midazolam and (b) 1-OH Midazolam concentrations determined from Wet blood samples and VAMS tips

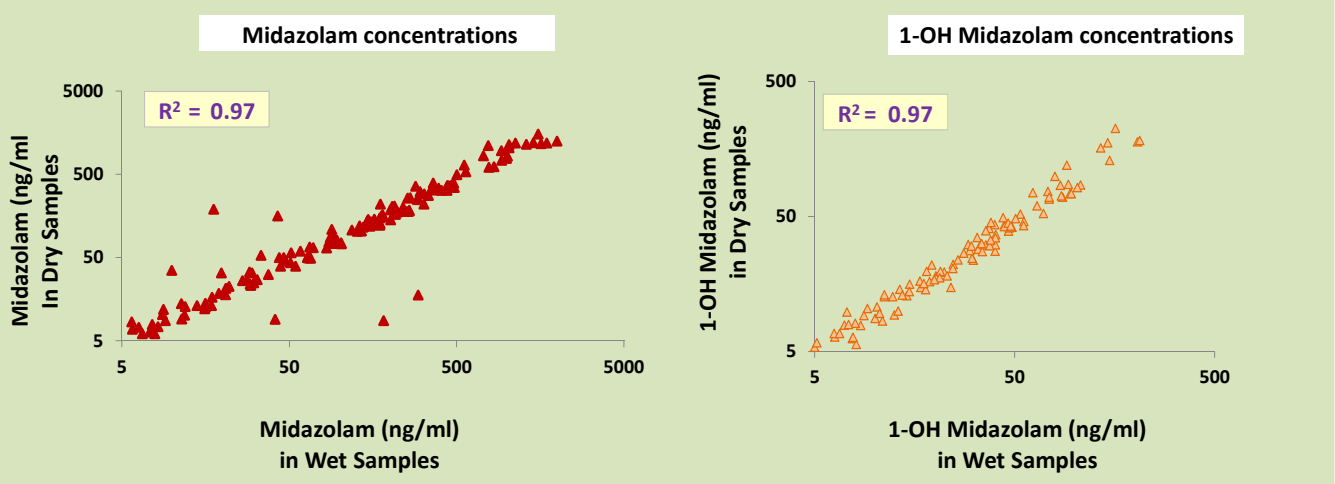
Midazolam concentrations
R² = 0.97
Midazolam (ng/ml In Dry Samples
Midazolam (ng/ml) in Wet Samples
1-OH Midazolam concentrations
R² = 0.97
1-OH Midazolam (ng/ml) in Dry Samples
1-OH Midazolam (ng/ml) in Wet Samples

Conclusions

Our preliminary data show

(i) Over 300-fold difference in blood midazolam concentrations and a 30-fold difference in blood 1-OH midazolam concentrations in children receiving IV drug

(ii) Concentrations of midazolam and 1-OH midazolam observed in wet blood samples and VAMS tips were closely correlated with no systematic bias.

Our experience of the VAMS blood collection platform is positive. There have been no specific difficulties in blood collection or in assay methods. We plan to continue recruitment until September 2016 to achieve a recruitment target of 65 patients in both ES and PICU groups. At present there is insufficient data to determine whether critical illness alters midazolam PK in critically ill children.

References

1. Ince I, de Wildt SN, Peeters MYM, et al. Critical illness is a major determinant of midazolam clearance in children aged 1 month to 17 years. Ther Drug Monit 2012; 34:381–9.
2. Vet NJ, Brussee JM, de Hoog M, et al. Inflammation and organ failure severely affect midazolam clearance in critically ill children. J Respir Crit Care Med. 2016 Jan 21.
3. Soto C, Terry HK, Pandya H, Field DJ, Kairamkonda V, Boyle EM. "Attitudes of neonatal nurses to research, and their role in the research process", Oral presentation at the European Society of Paediatric and Neonatal Intensive Care congress, November 2011
4. Patel P, Mulla H, Kairamkonda V, Spooner N, Gade S, Della Pasqua O, Field DJ, Pandya HC. (2013). Dried blood spots and sparse sampling: a practical approach to estimating pharmacokinetic parameters of caffeine in preterm infants. Br J Clin Pharmacol. 75(3):805-13

Acknowledgements

We would like to thank GSK Plc and Neoteryx Limited for supporting the study. VAMS is a trademark of Neoteryx LLC.