Modeling anti-leukemic therapy by patient derived AML xenografts with distinct phenotypes/geneotypes

J Liu[1*], **X An[1#*]**, N Wang[2], D Wang[2], L Huang[2], R Wu[1], J Cai[1], C Li[1], L Zhao L[1], X Song[1], S Yu[1], L Zhang[1], J-P Wery[1] and **H Li[1,3]**

[1]Crown Bioscience, Inc., 3375 Scott Blvd, suite 108, Santa Clara, CA 95054, USA; [2]Cancer Biology Research Center, Tongji Hospital, Tongji Medical College, Huazhong University of Science and Technology, Wuhan, Hubei 430030, China; [3]State Key Laboratory of Natural and Biomimetic Drugs, Peking University. [#]: presenter

CROWNBIO TRANSLATING BIOLOGY INTO MEDICINE

This poster is powered by layar INTERACTIVE PRINT

3123

ABSTRACT

We have recently successfully engrafted leukemic cells, isolated from bone marrows of 3 AML patients with subtypes of M5 (AM7577), M2 (AM8096) and M6 (AM8070), into immunocompromised NOD/SCID mice. The transplanted mice developed AML leukemia with leukemic cells found in bone marrow as in human diseases and also can be found in peripheral organs (*e.g*. spleen, blood, *etc*) for some of them (AM7577), seemingly reflective of disease subtypes. Growth of the leukemia cause typical leukemia symptoms and eventual mortality as seem in human. The leukemic cells can be serially passed in animals (re-engraftment) with 100% take-rate and with stability in disease development and phenotype/ genotype-maintenance, thus creating useful experimental systems modeling AML leukemogenesis and pharmacology, both in vivo and ex vivo. In particular, specific oncogenic mutation drivers found in these models, e.g. IDH2-R142Q and FLT3-LTD, could be particularly useful in evaluating investigational drugs targeting these disease pathways.

Experimental Schemes

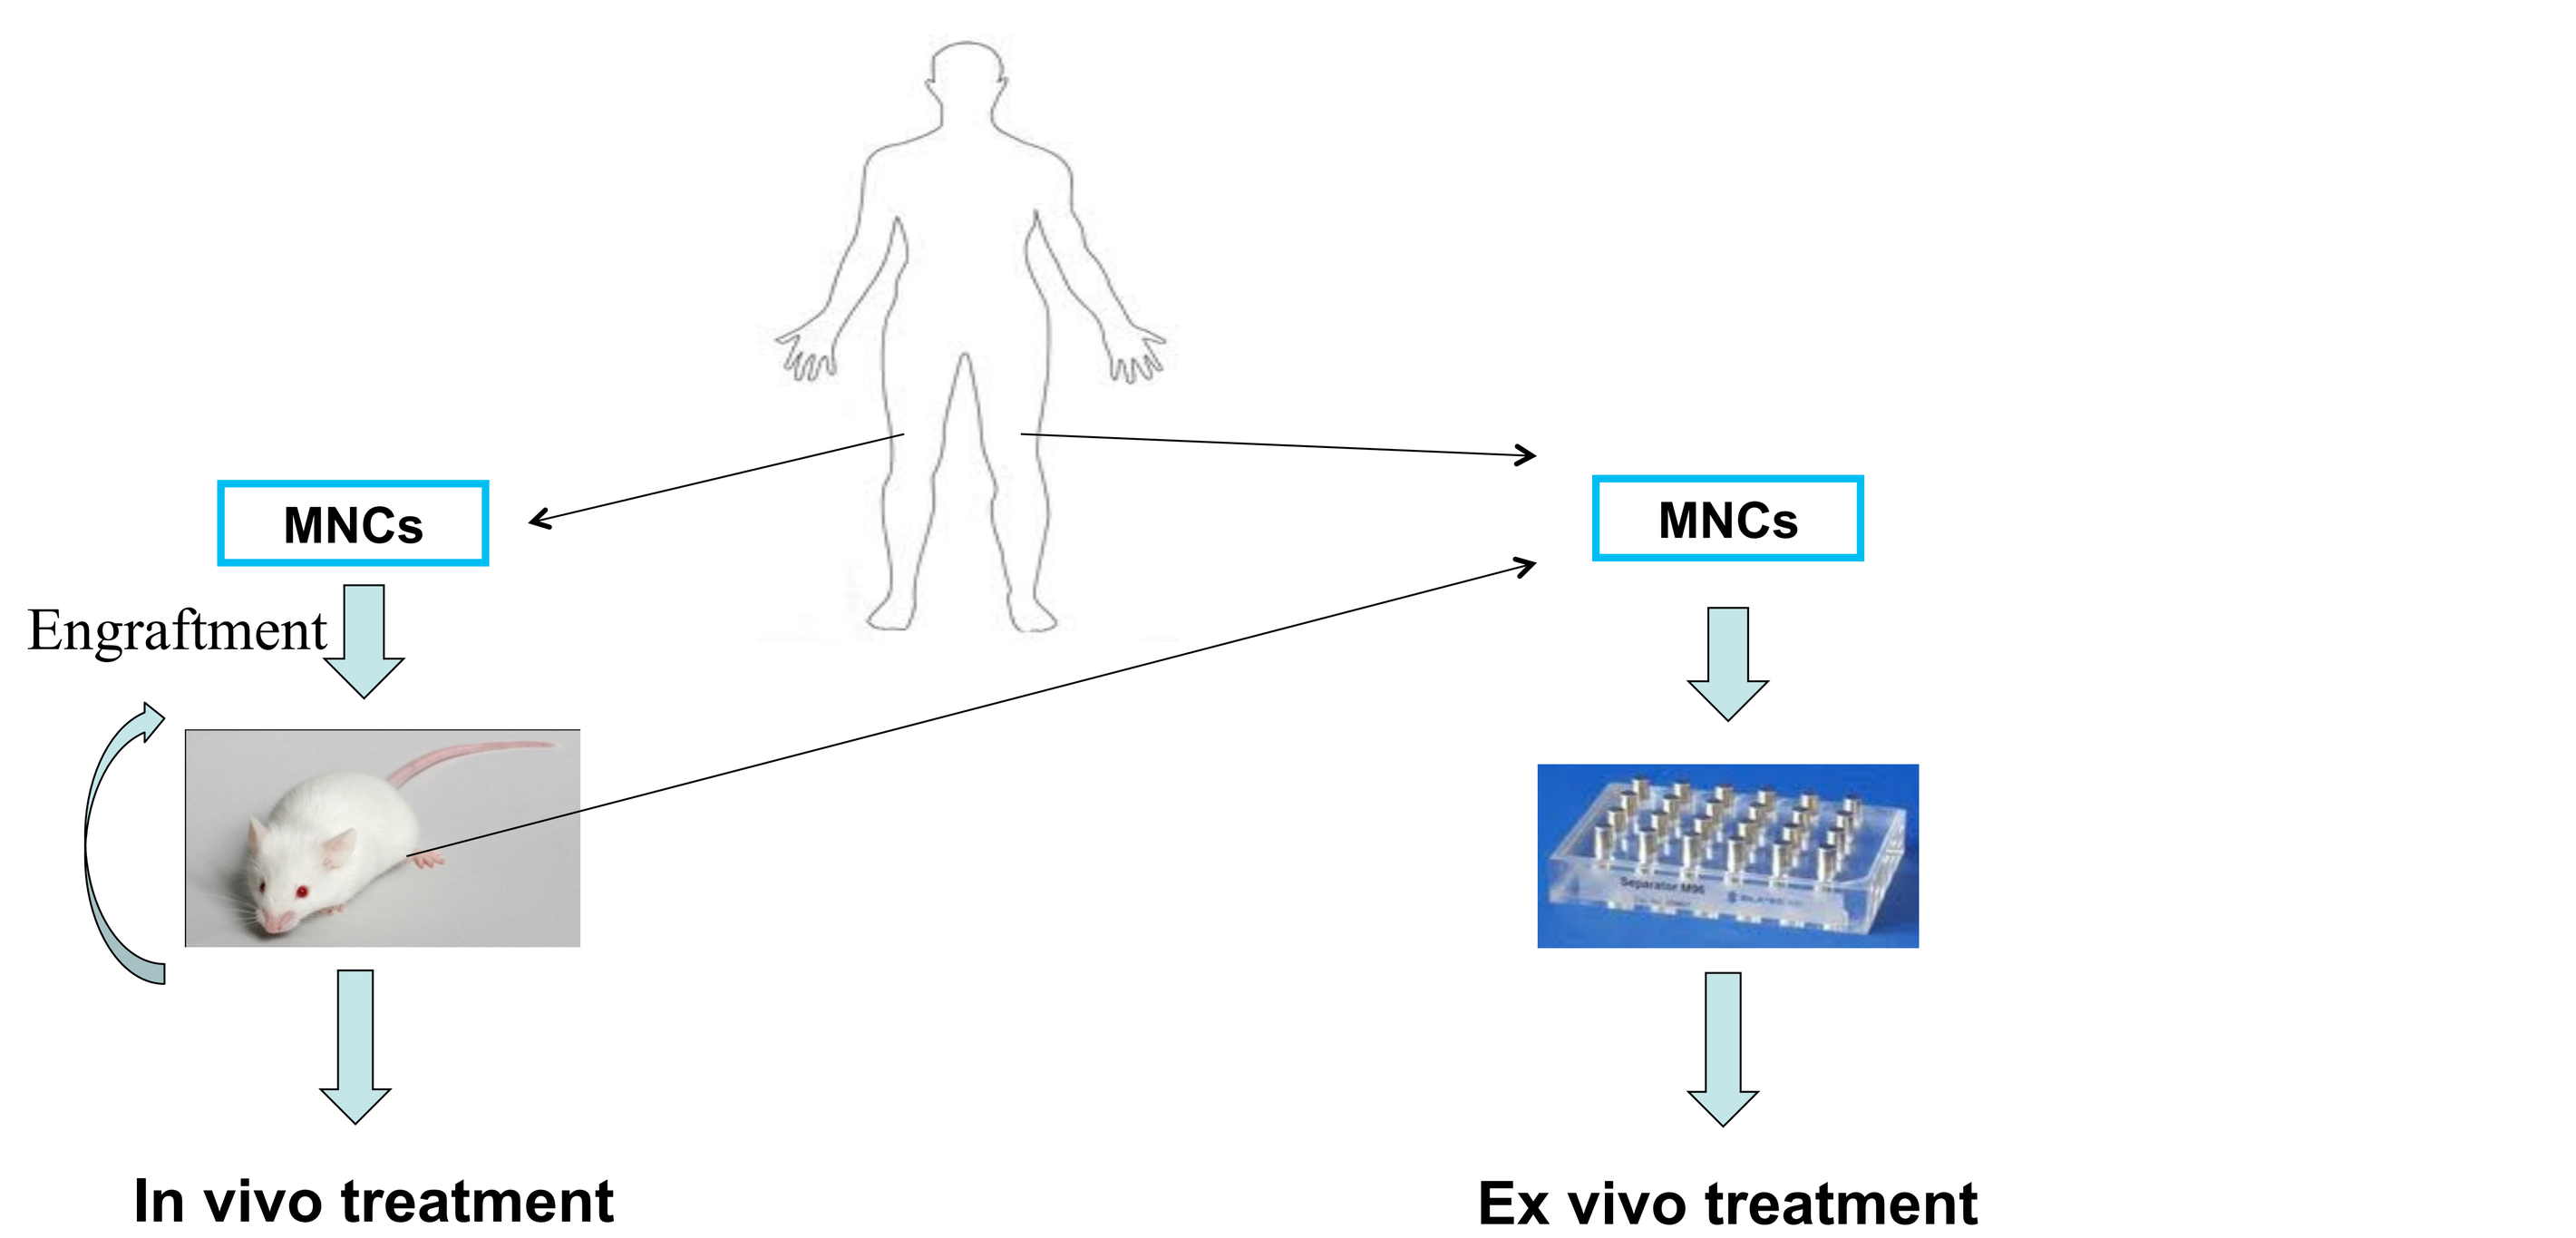
MNCs
Engraftment
MNCs
In vivo treatment
Ex vivo treatment

Pharmacology endpoints
- •Survival
- •Clinical signs,
- •Leukemic burden (blood, LN, SLN, BM, etc.
- •PD-biomarkers
- •PK-PD
- •Ex vivo readout
- •Re-engraftment

Pharmacology endpoints
- •Survival/apoptosis/cell cycle
- •2D, 3D
- •PD-biomarkers
- •IC50
- •Re-engraftment

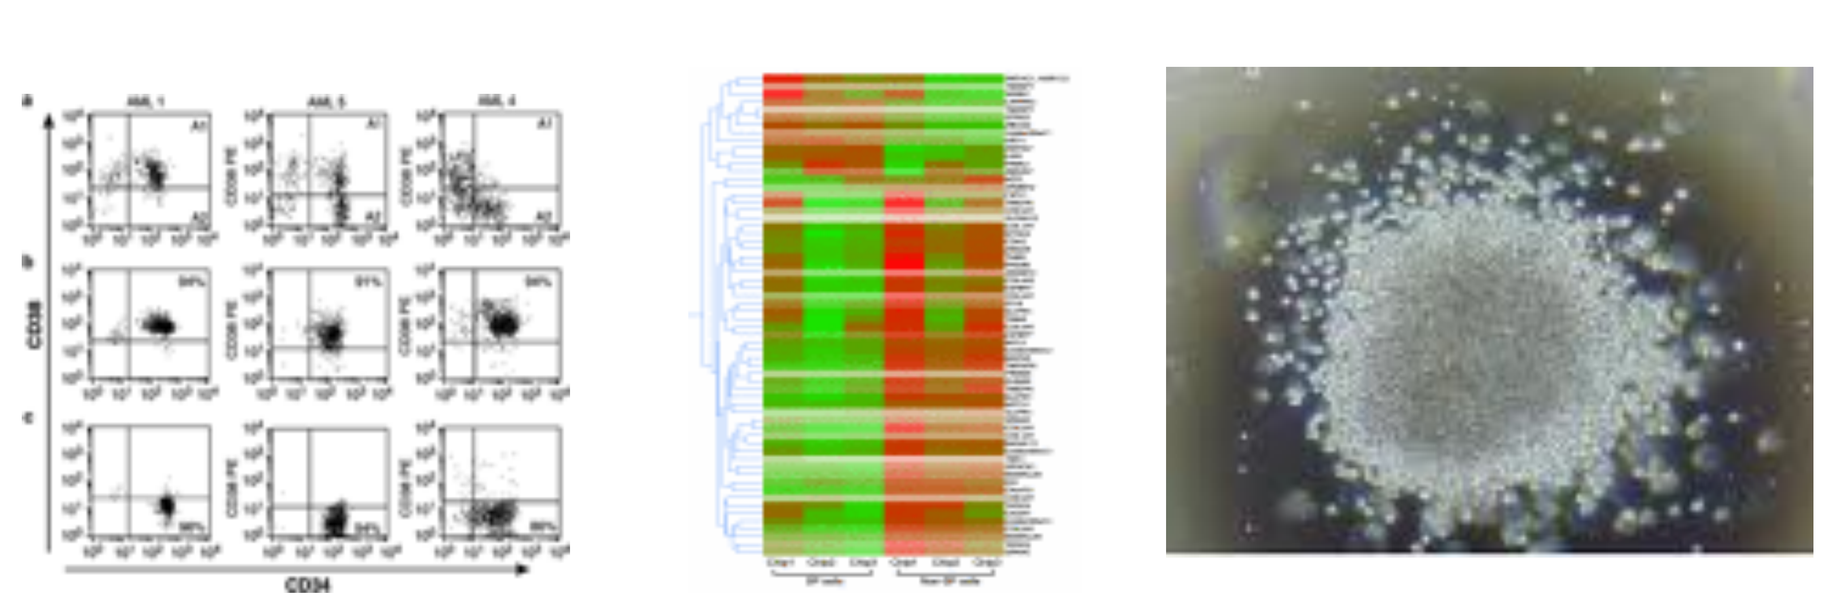

AML models information

Table 1. Patient Information

Model ID	Patient ID	Subtype	Phenotyping	Fusion/mutation
AM7577	7577	M5	Express: CD13,33, HLA-DR, CD117, 38, 71. partly express: MPO, CD15, 19, 7. no express: CD34, 10, 20, 79a, 3, 5, 11b, 14, 56, GlyA	IDH2(R140Q); *FLT3*-ITD; CEBPA insertion (SNP)
AM8096	8096	M2	Express: CD13,CD33,HLA-DR, CD117, CD38, CD71.	(-)
AM8070	8070	M6	Express: MPO,CD13,15,33,117,7,34,38, HLA-DR	(-)

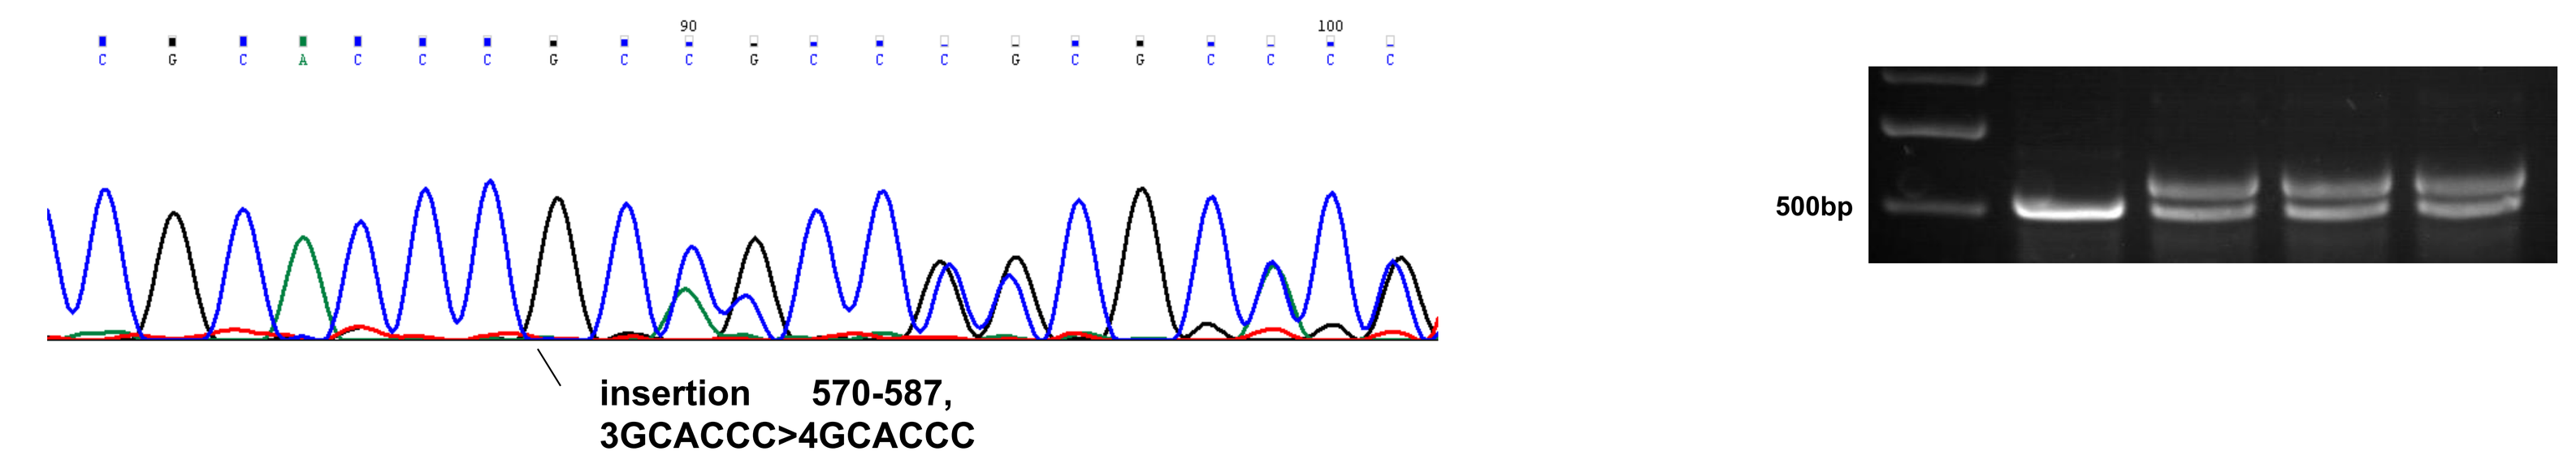
insertion 570-587, 3GCACCC>4GCACCC
500bp

Figure 1: Genotype analysis of AM7577. Left: IDH2-R140Q; Right: FLT3-LTD.

2A

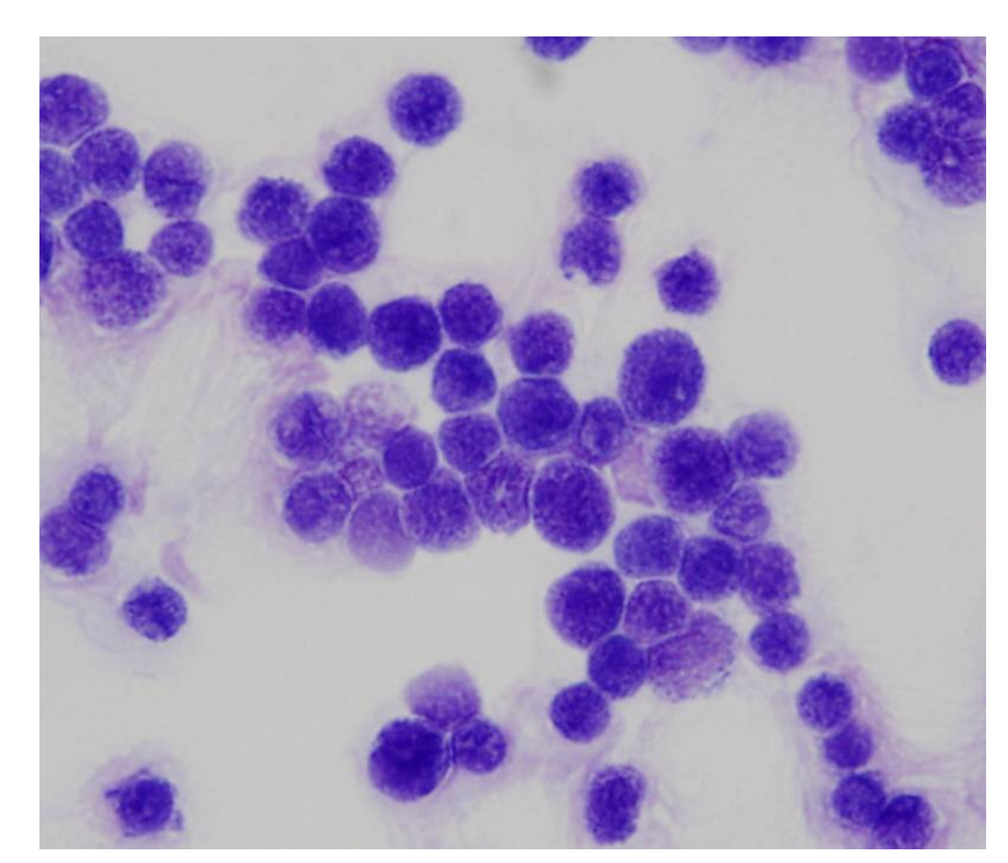

2B

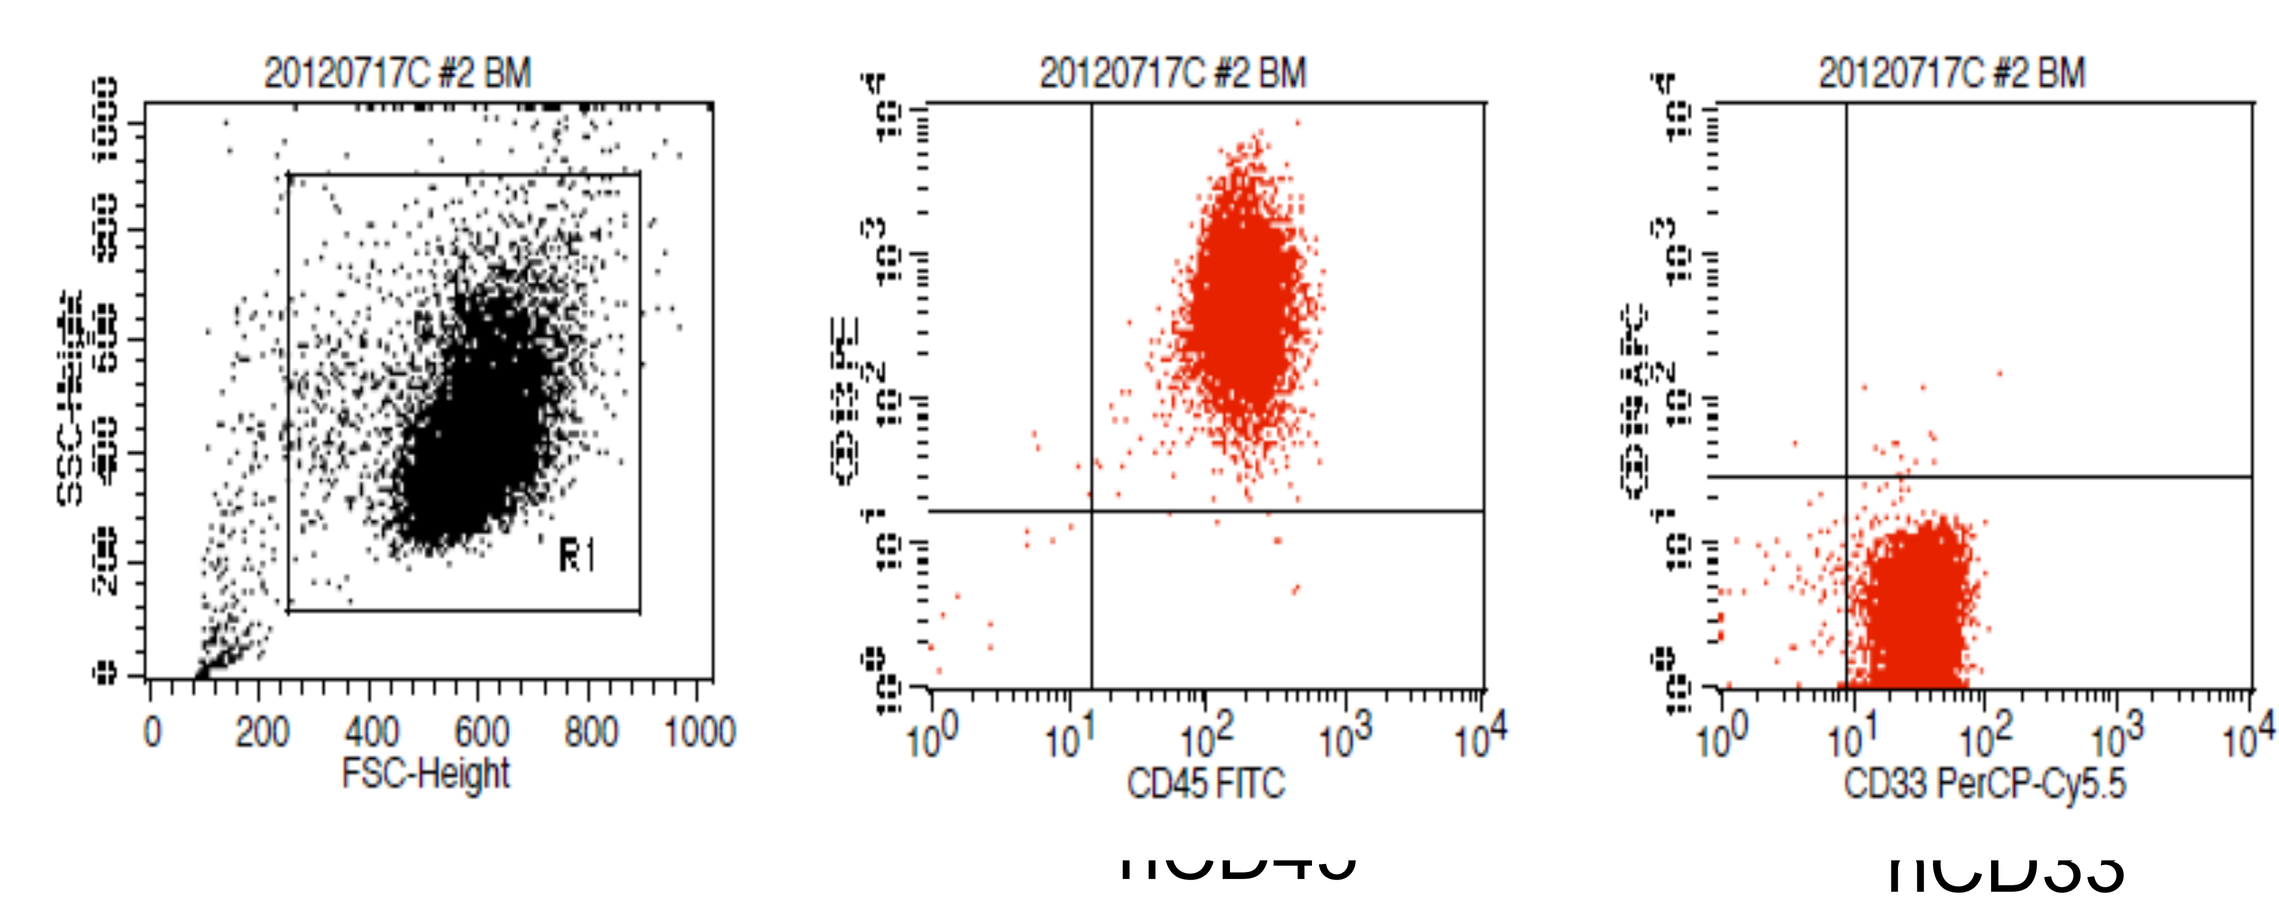
20120717C #2 BM
SSC-Height
FSC-Height
R1
CD45 FITC
CD33 PerCP-Cy5.5

Figure 2: Histopathology and phenotypic analysis of AM8070. A. Giemsa staining of BM smear; B: Flow cytometry analysis.

3A

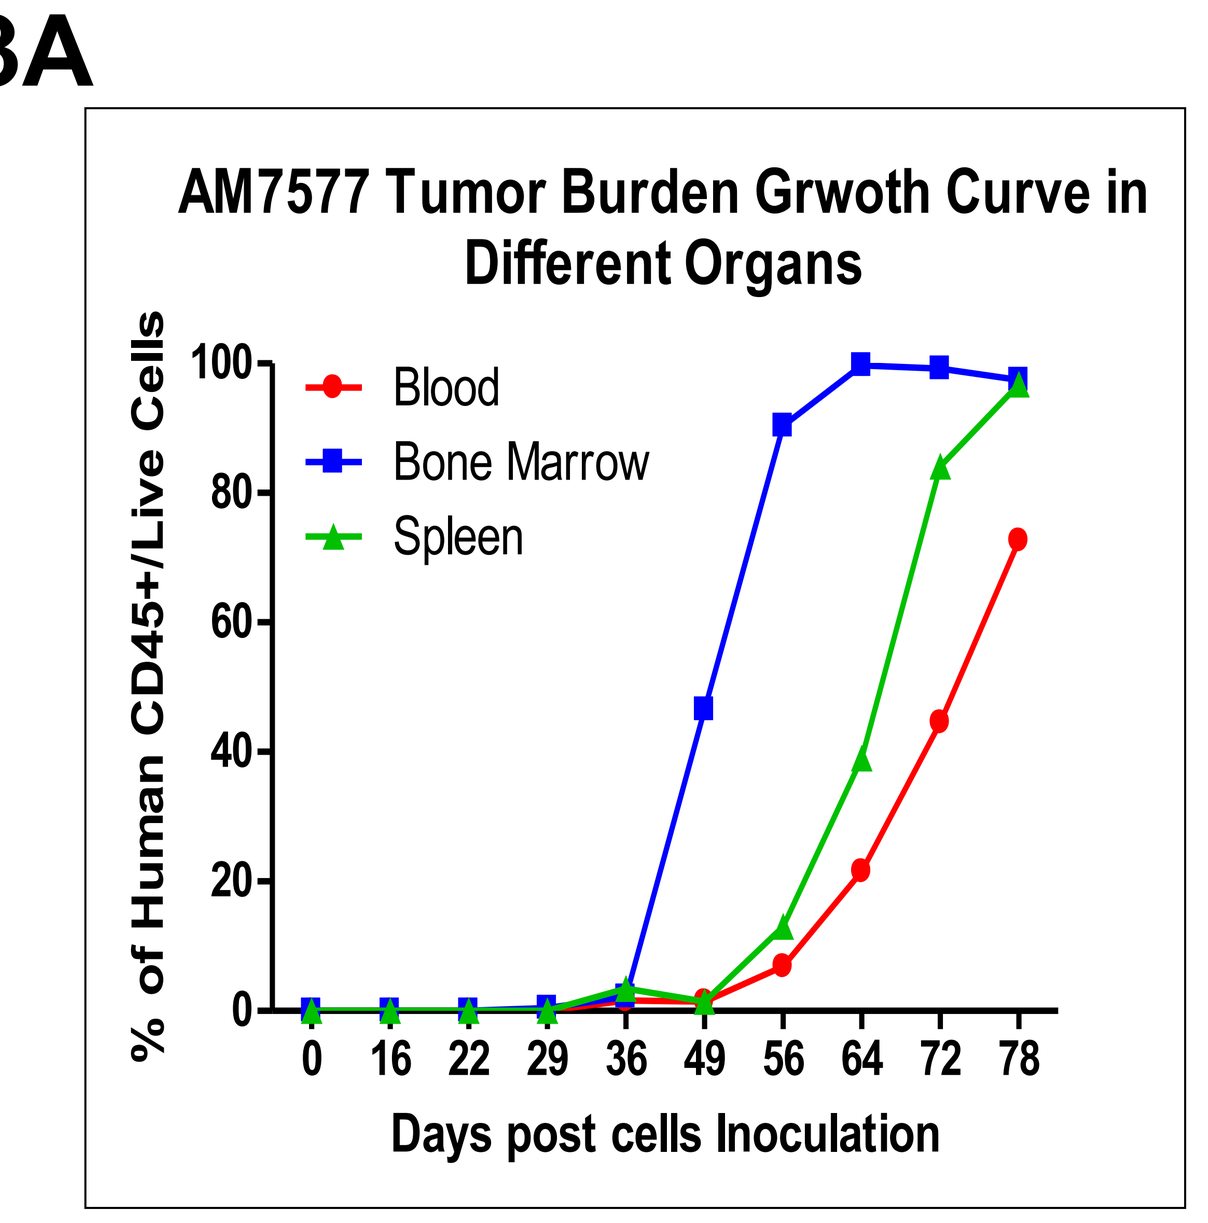
AM7577 Tumor Burden Grwoth Curve in Different Organs
Blood
Bone Marrow
Spleen
% of Human CD45+/Live Cells
Days post cells Inoculation

3B

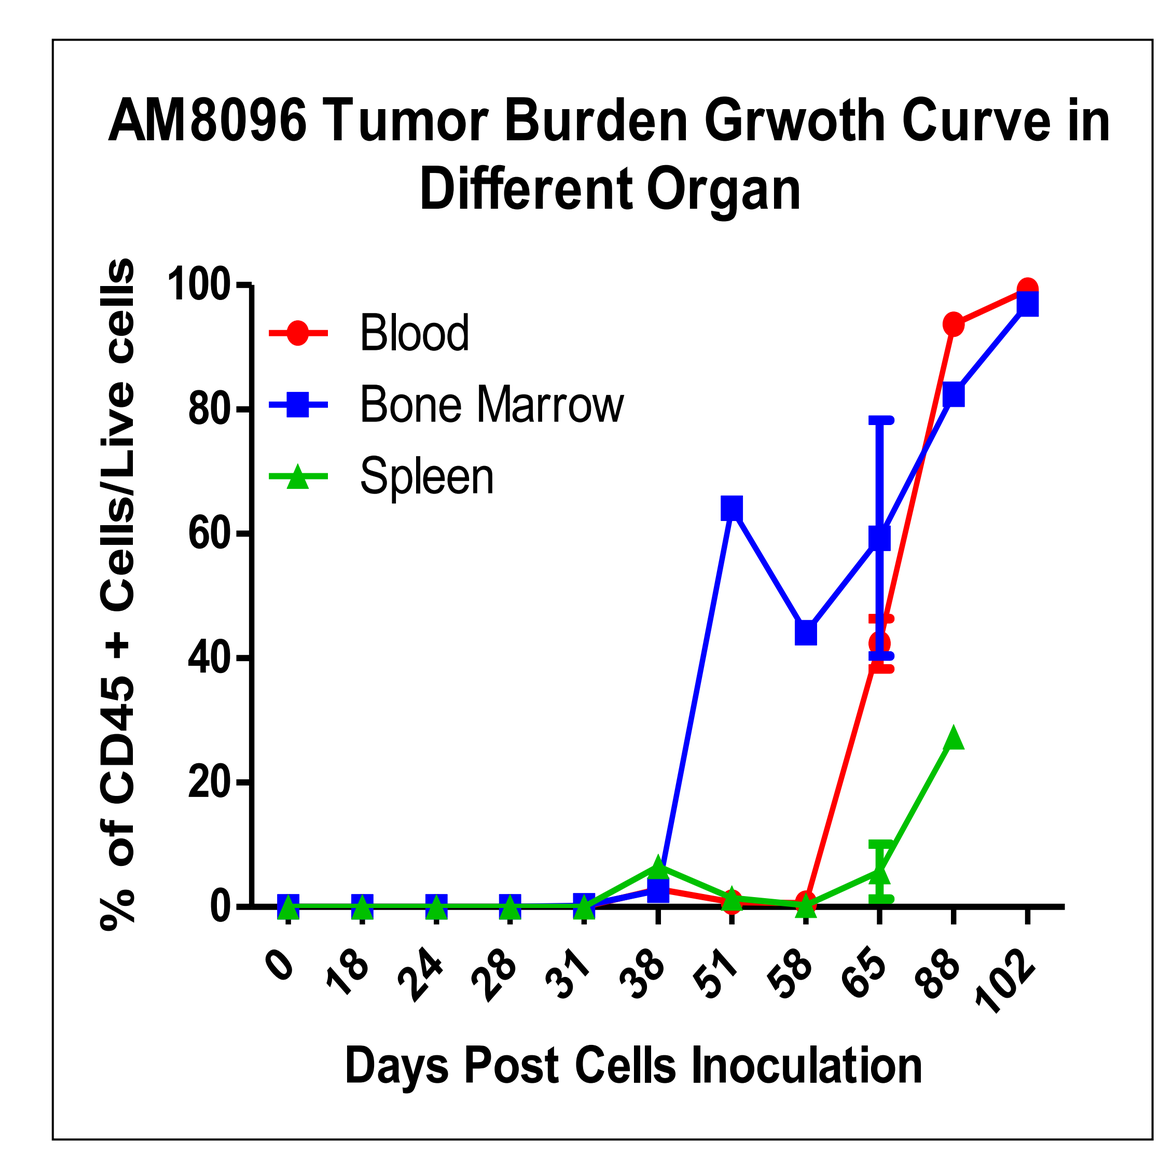
AM8096 Tumor Burden Grwoth Curve in Different Organ
Blood
Bone Marrow
Spleen
% of CD45 + Cells/Live cells
Days Post Cells Inoculation

Figure 3: Tumor burden growth curve in different organs. A:AM7577; B: AM8096.

Note: Red: Peripheral Blood; Blue: Bone Marrow; Green: Spleen.

4

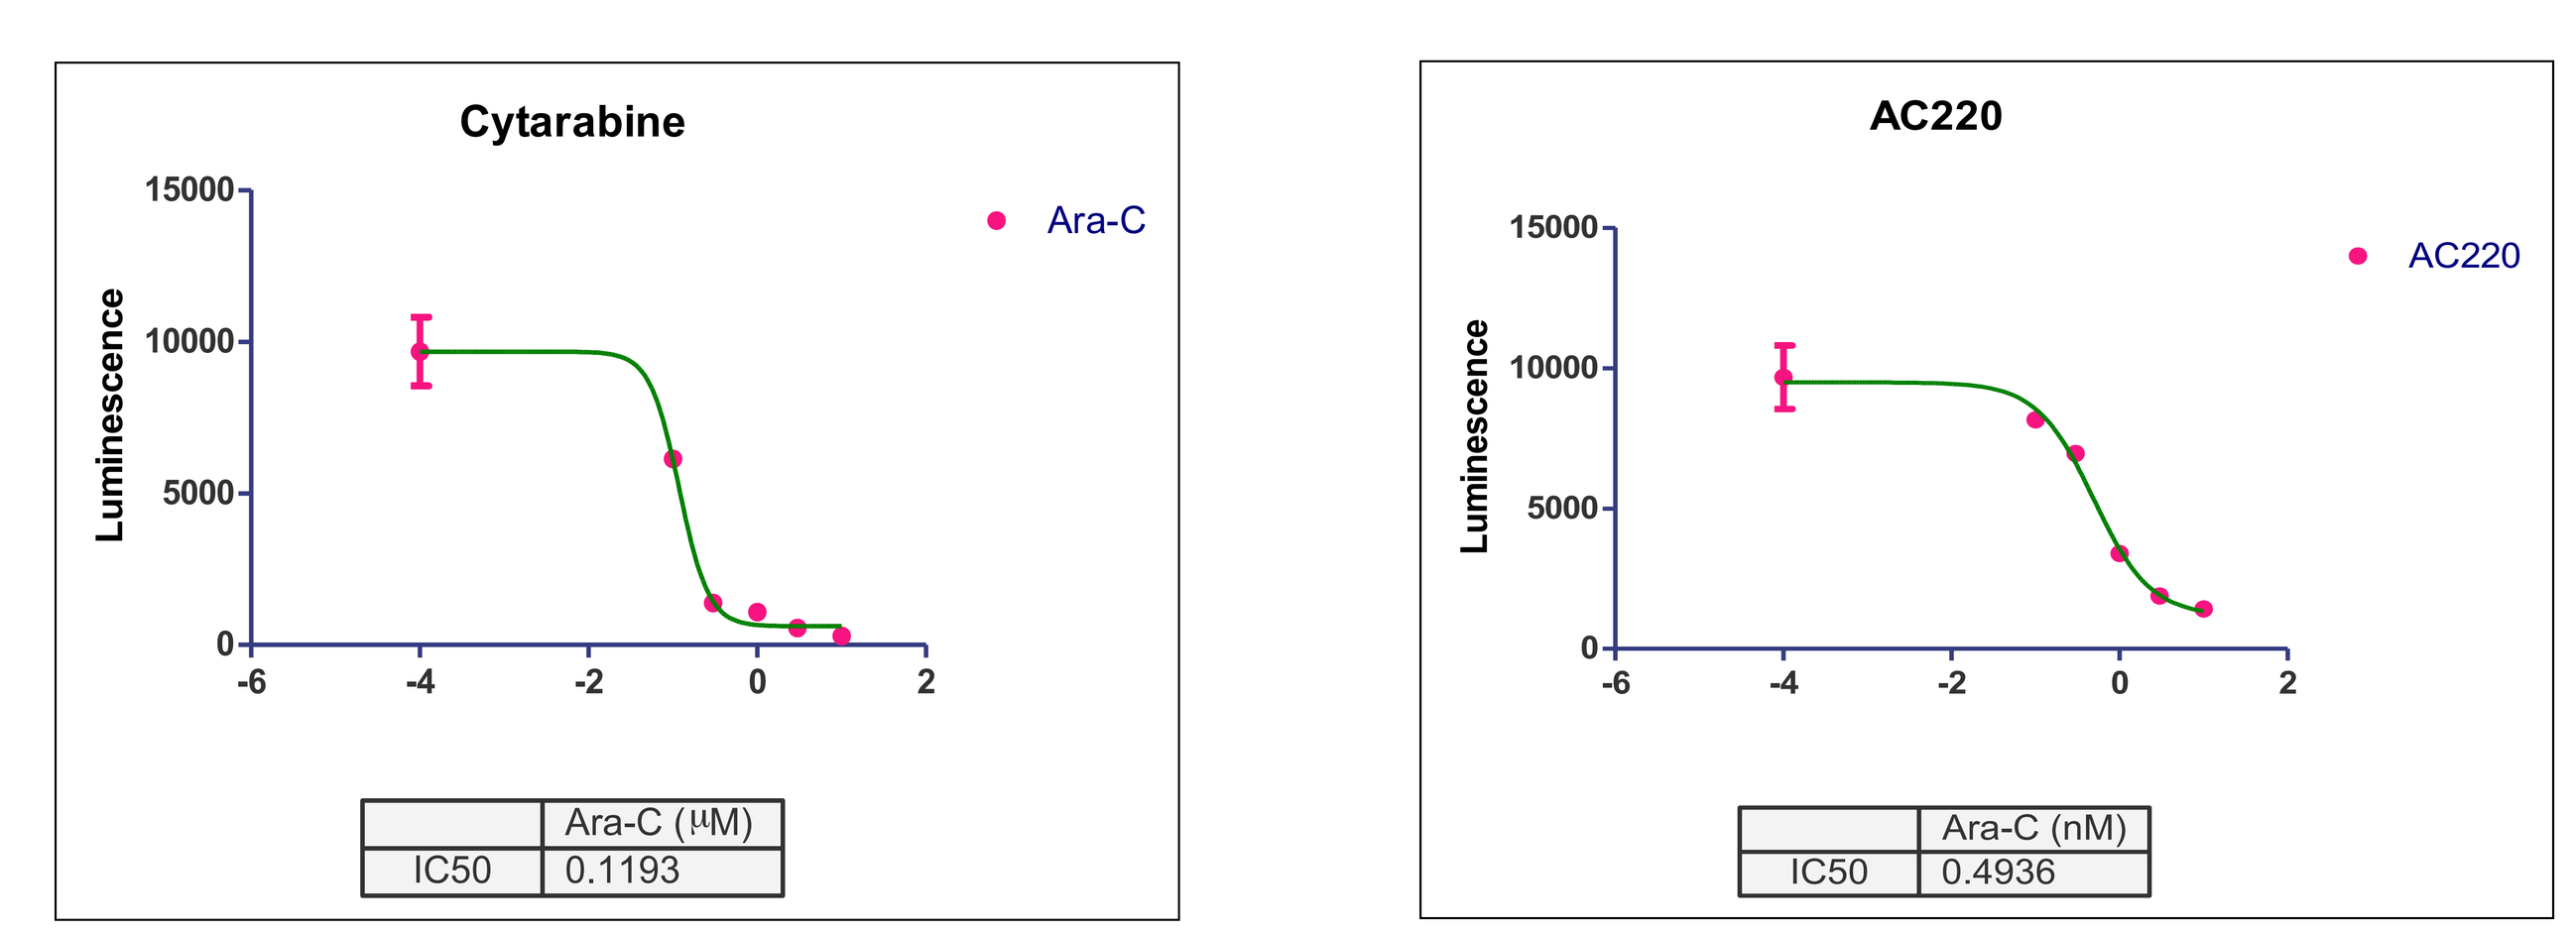
Cytarabine
Ara-C
Luminescence
Ara-C (µM)
IC50 0.1193
AC220
AC220
Luminescence
Ara-C (nM)
IC50 0.4936

Figure 4: The half maximal inhibitory concentration (IC50) curve for AM7577 by treating with A. Ara-C and B. AC-220. *Note: The data was done by CellTiter-Glo Luminescent Cell Viability Assay.*

In vivo pharmacology analysis of AraC treatment

5A

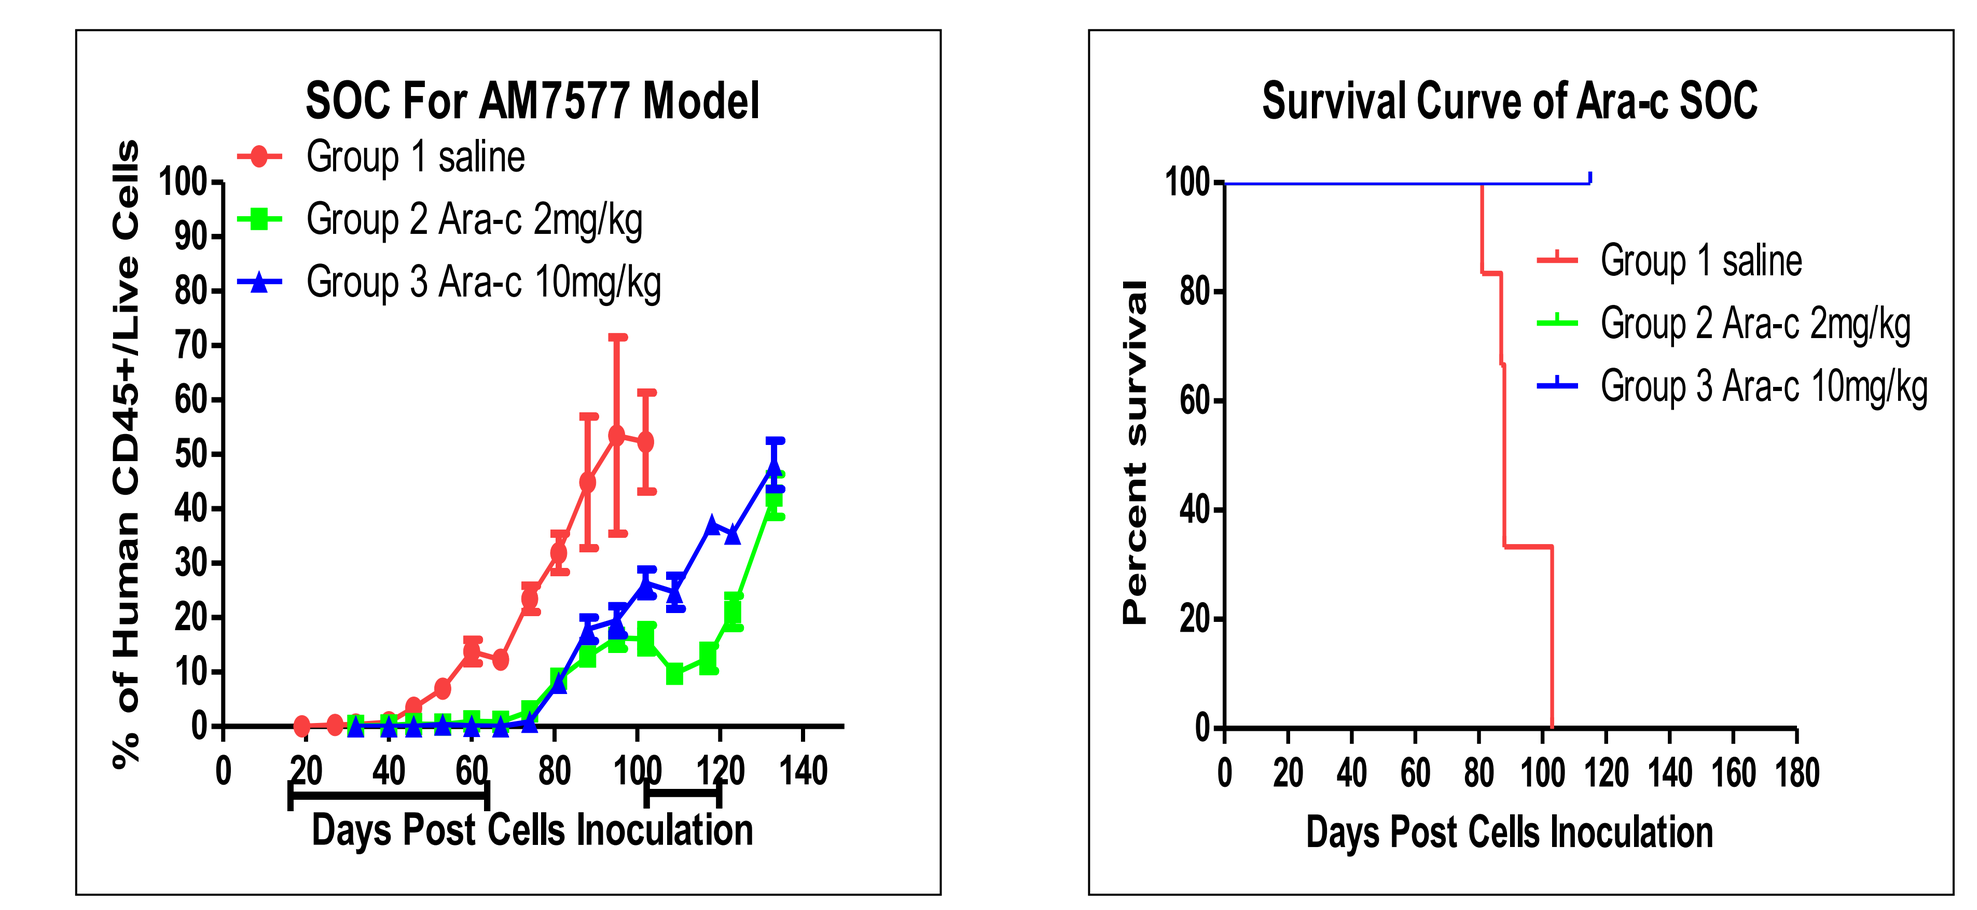
SOC For AM7577 Model
Group 1 saline
Group 2 Ara-c 2mg/kg
Group 3 Ara-c 10mg/kg
% of Human CD45+/Live Cells
Days Post Cells Inoculation
Survival Curve of Ara-c SOC
Group 1 saline
Group 2 Ara-c 2mg/kg
Group 3 Ara-c 10mg/kg
Percent survival
Days Post Cells Inoculation

5B

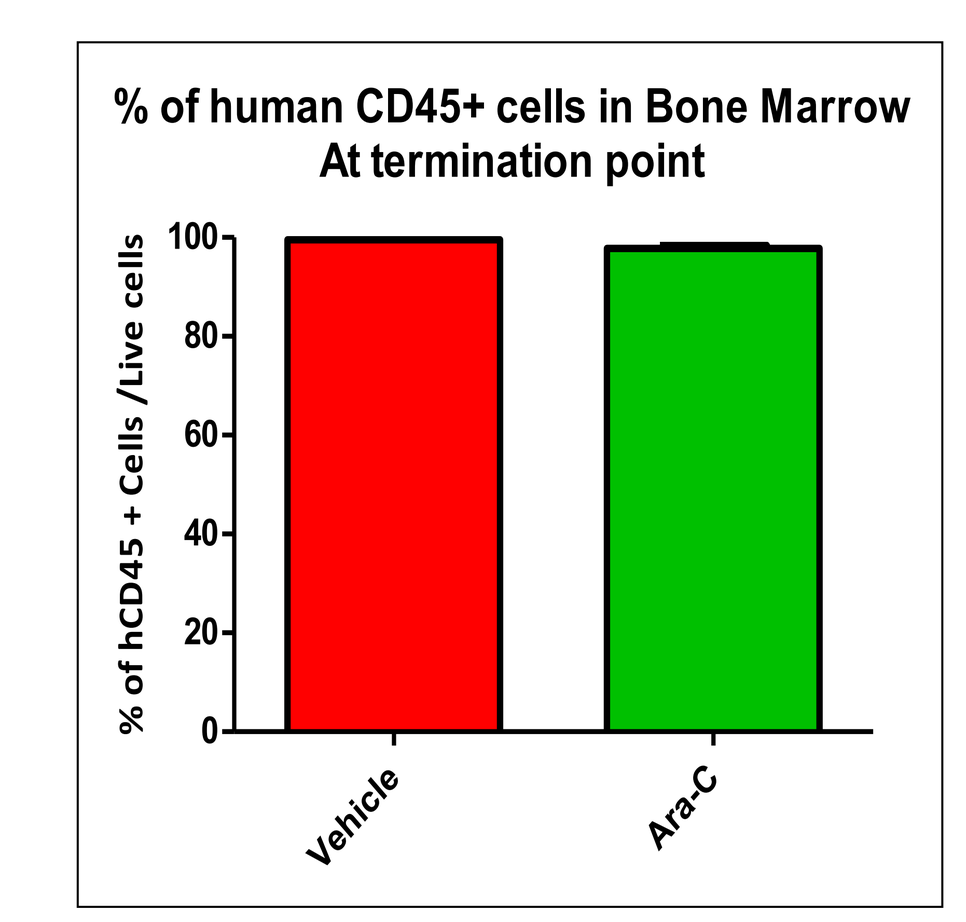
% of human CD45+ cells in Bone Marrow At termination point
% of hCD45 + Cells /Live cells
Vehicle
Ara-C

Figure 4 (A&B)): Example of Standard of Care (SOC) experiment by using Ara-C. A: Tumor burden growth curve and survival curve of AM7577 with Ara-C treatment; B: Bone marrow's tumor burden at termination point compared between vehicle group and Ara-C treated Group.

Conclusions

1. **3 PDX-AML models have been established;**
 a) With typical AML symptoms
 b) Leukemic load found primarily in BM, as well as in peripheral organs for some models, reflective of patient diseases.
 c) Serially passable in vivo with demonstrated stable genotypes and phenotypes
2. **Primary leukemia cells can be used for ex vivo testing**.
 a) 2D culture for IC50 (araC, AC220)
 b) 3D (CFC) (araC and AC220)
3. **In vivo administration of AraC yield rapid elimination of leukemic load in blood and partial reduction in spleen, but not in BM.**
 a) Relieve symptoms and extend life, but without cure
 b) Rapid relapse upon the treatment withdrawal in peripheral
4. **AML-7577 with FLT3-LTD respond completely to AC220**
 a) High dose treatment lead to cure without relapse long after treatment withdrawal
5. **In summary, these models may serve as useful experimental system for modeling leukemogenesis and pharmacology**

Contacts:

•X (Annie) An: anxiaoyu@crownbio.com

•H Li: henryli@crownbio.com

Download the free Layar App · Scan this page · Discover interactive content