TRANSLATING BIOLOGY INTO MEDICINE

Xenograft models for development of new drugs targeting Fibroblast Growth Factor Receptor (FGFR)

Lan Zhang[1], Juan Zhang[1], Ying Jin[1], Guizhong Liu[1], Meng Qiao[1], Qian Shi[1] Cancer Pharmacology, CrownBio Inc., China

3124

This poster is powered by layar INTERACTIVE PRINT

Abstract

The fibroblast growth factor receptors (FGFR) are tyrosine kinases that are present in many types of endothelial and tumor cells. There are 4 different fibroblast growth factor receptors (FGFR) in the cell: FGFR1, FGFR2, FGFR3, and FGFR4. The activation of FGFR pathway plays an important role in tumor angiogenesis, tumor growth, survival, as well as migration. Mutation or amplification/overexpresion of FGFR has been found in several types of tumors, including breast cancer, bladder cancer, gastric cancer, prostate cancer, colon cancer, multiple myeloma, and non–small cell lung carcinoma. Therefore, targeting FGFRs represents an attractive strategy for development of new cancer therapeutics. However, the lack of suitable models hinders the progress of research on FGFR inhibitors. Here we successfully validate subcutaneous models in several cancer types, in which FGFR is mutant or amplified/overexpressed, such as breast cancer MDA-MB-134 (FGFR1 amplification), gastric cancer SNU-16 (FGFR2 amplification), endometrial cancer AN3CA (FGFR2 N549K/K310R mutation), bladder cancer RT-4 (FGFR3 overexpression) and RT-112 (FGFR3 overexpression), lung cancer NCI-H1581 (FGFR1 amplification), as well as human derived gastric cancer GA114 (FGFR2 amplification). In conclusion, the validated cell line derived (CDX) and patient derived xenograft models (PDX) in several cancer types provide valuable platforms for the development of new compounds targeting FGFRs..

Materials and Methods

Animals: NOD/SCID and BALB/c nude mice purchased from Beijing HFK Bio-Technology Co. Ltd. (HFK, Beijing, China) and Shanghai Laboratory Animal Center (SLAC, Shanghai, China).

Tumor Inoculation: For subcutaneous human xenograft models, each mouse was inoculated at the right flank with tumor cells for tumor development.

Group and Treatment: The treatments for the therapeutic study were started when mean tumor size reached 150-200 mm^3 in subcutaneous model.

Endpoints: Tumor volume was calculated as the formula: $V (mm^3) = (a \times b^2)/2$, where *a* and *b* were the long and short diameters of the tumor, respectively.

Statistical Analysis: For comparison between two groups, an independent sample t-test were used. For comparison among three or more groups, a one-way ANOVA were performed followed by multiple comparison procedures. All data were analyzed using SPSS 18.0. $p < 0.05$ was considered to be statistically significant.

CrownBio FGFR Targeted Xenograft Model

Models	CDX Model						PrimeXeno
	MDA-MB-134	NCI-H1581	SNU-16	AN3CA	RT-4	RT-112	GA114
FGFR	FGFR1 amplification	FGFR1 amplification	FGFR2 amplification	FGFR2 N549K/K310R mutation	FGFR3 overexpression	FGFR3 overexpression	FGFR2 amplification

Cell-Derived Xenograft Model

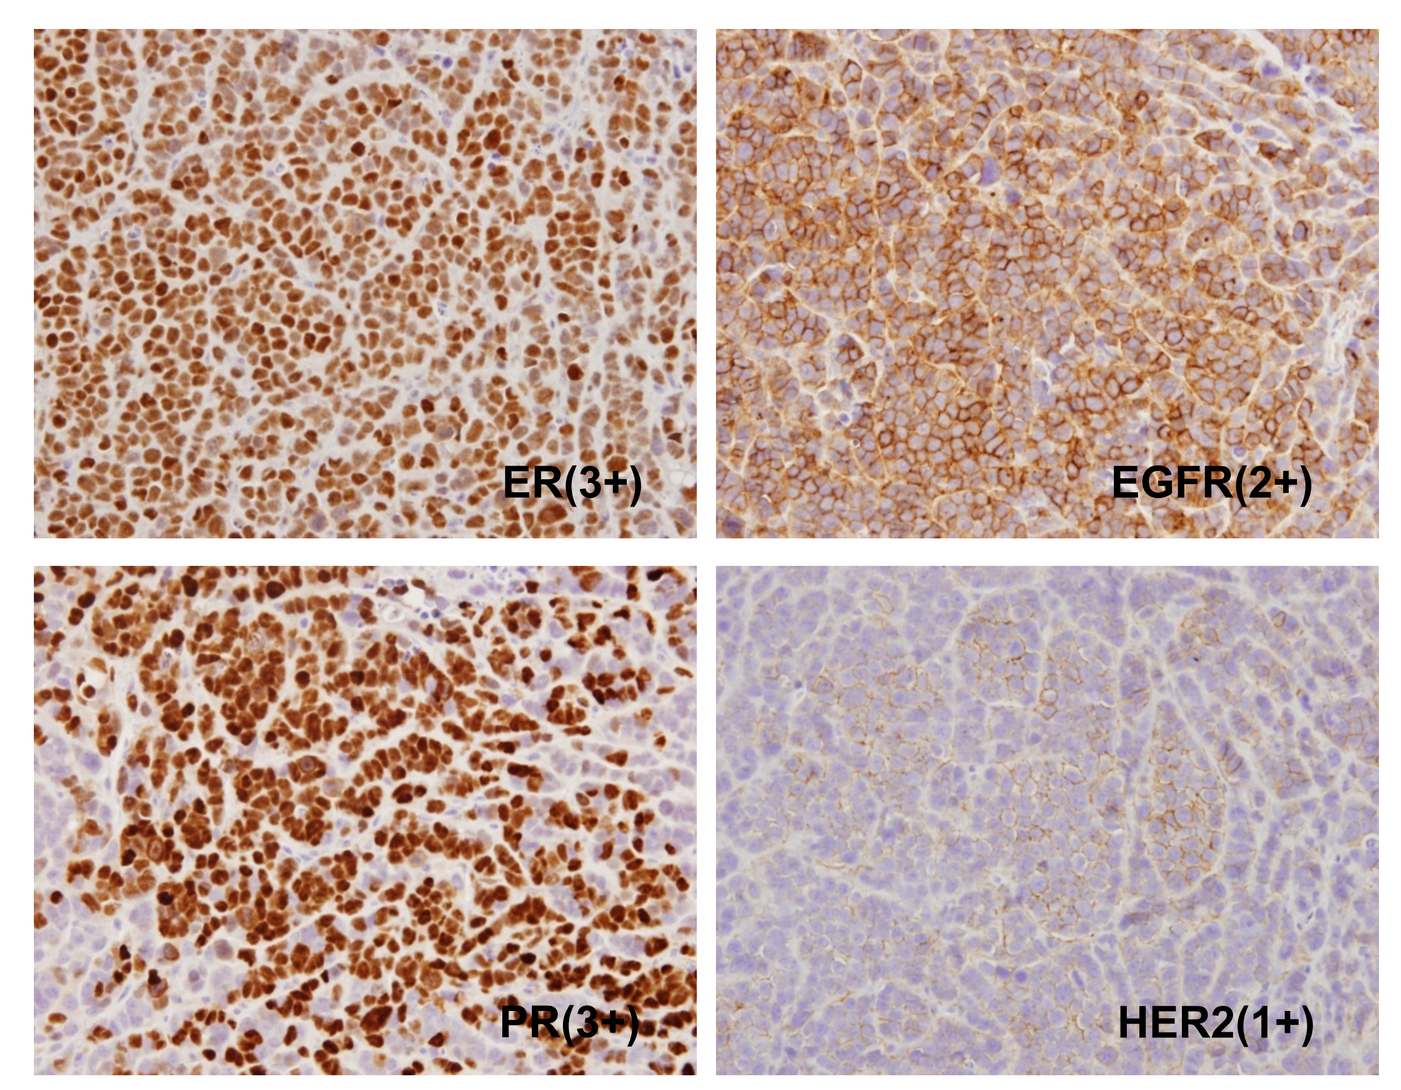
ER(3+)
EGFR(2+)
PR(3+)
HER2(1+)

Figure 1. ER, PR, EGFR and Her2 expression in MDA-MB-134 human breast cancer xenograft model

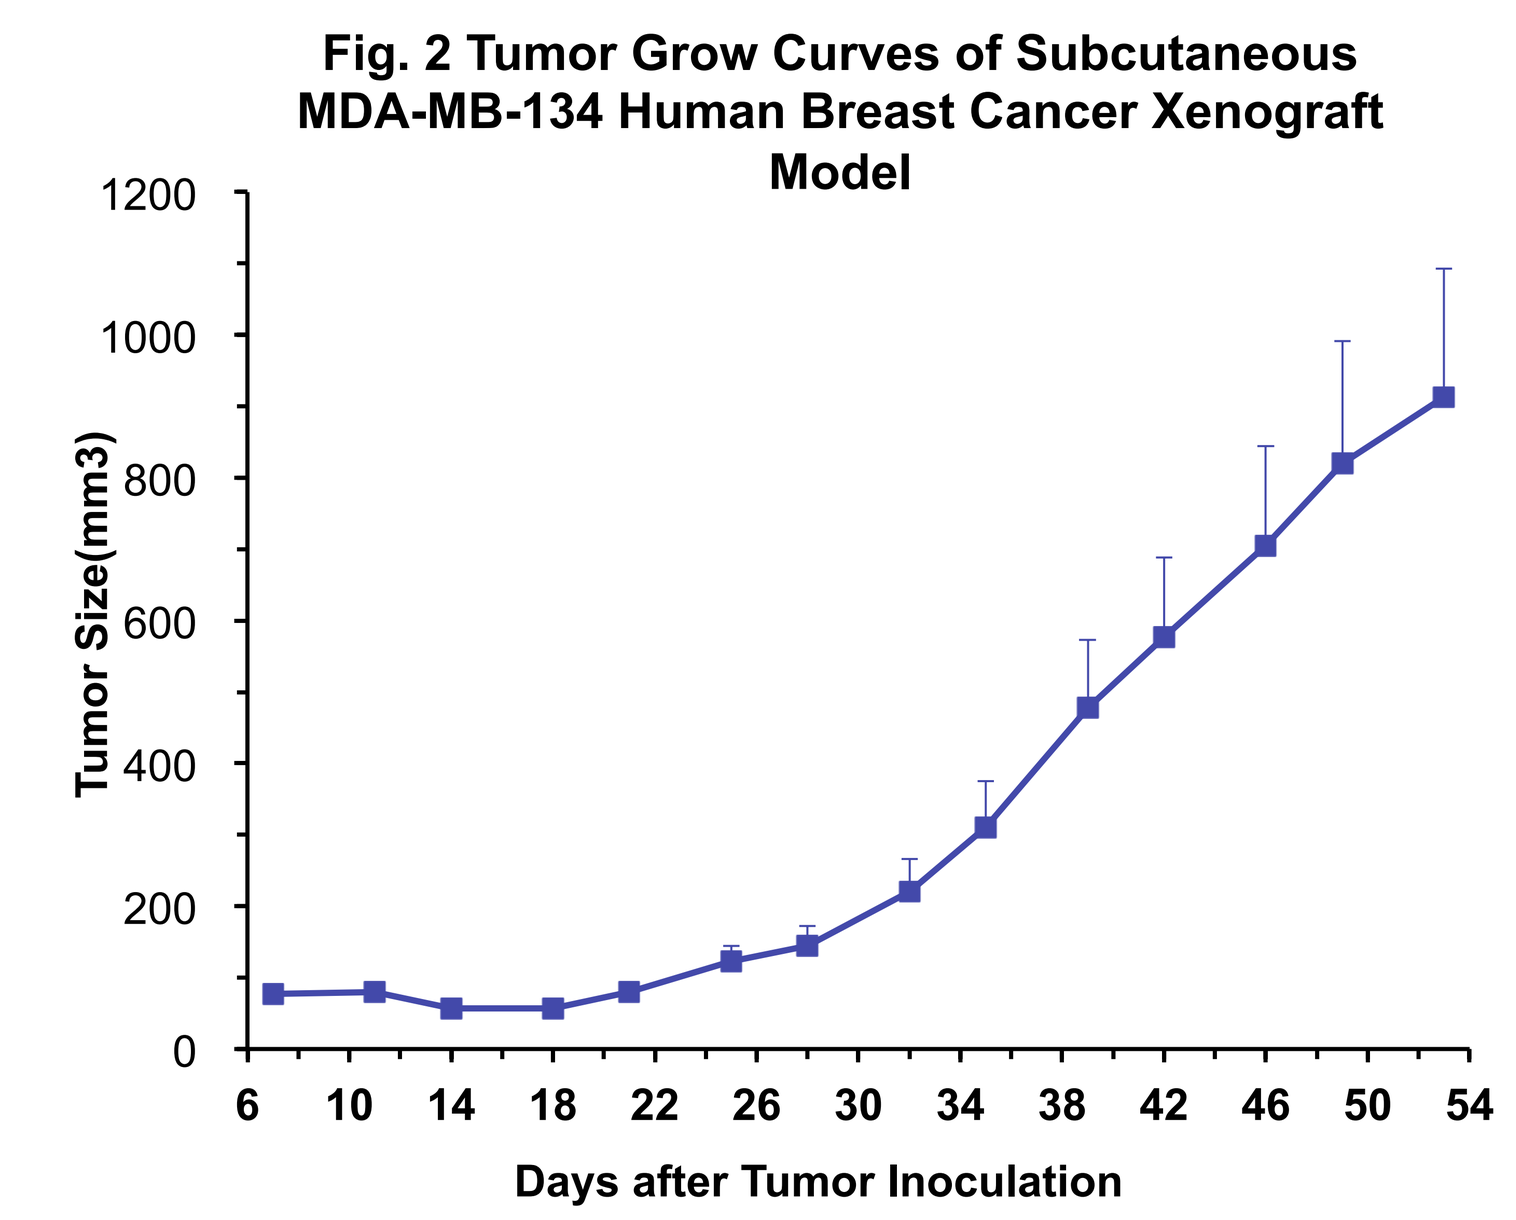
Fig. 2 Tumor Grow Curves of Subcutaneous MDA-MB-134 Human Breast Cancer Xenograft Model
Tumor Size(mm3)
Days after Tumor Inoculation

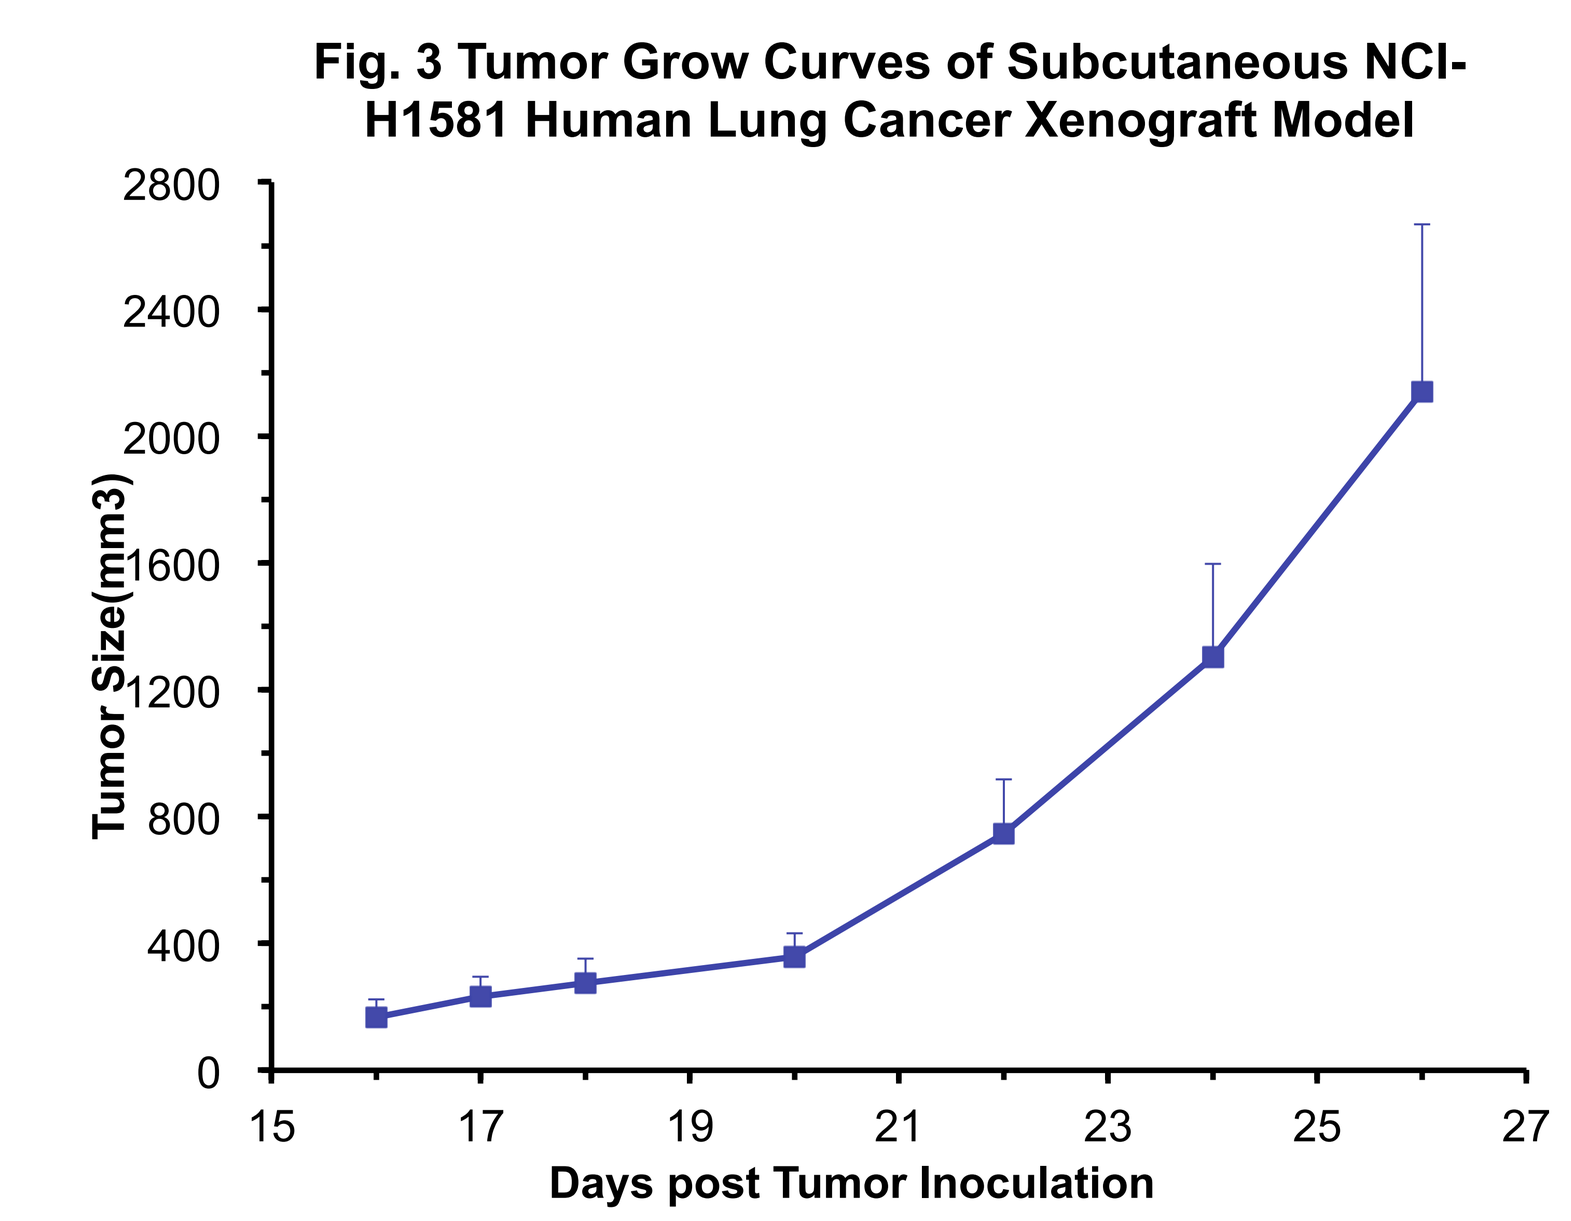
Fig. 3 Tumor Grow Curves of Subcutaneous NCI-H1581 Human Lung Cancer Xenograft Model
Tumor Size(mm3)
Days post Tumor Inoculation

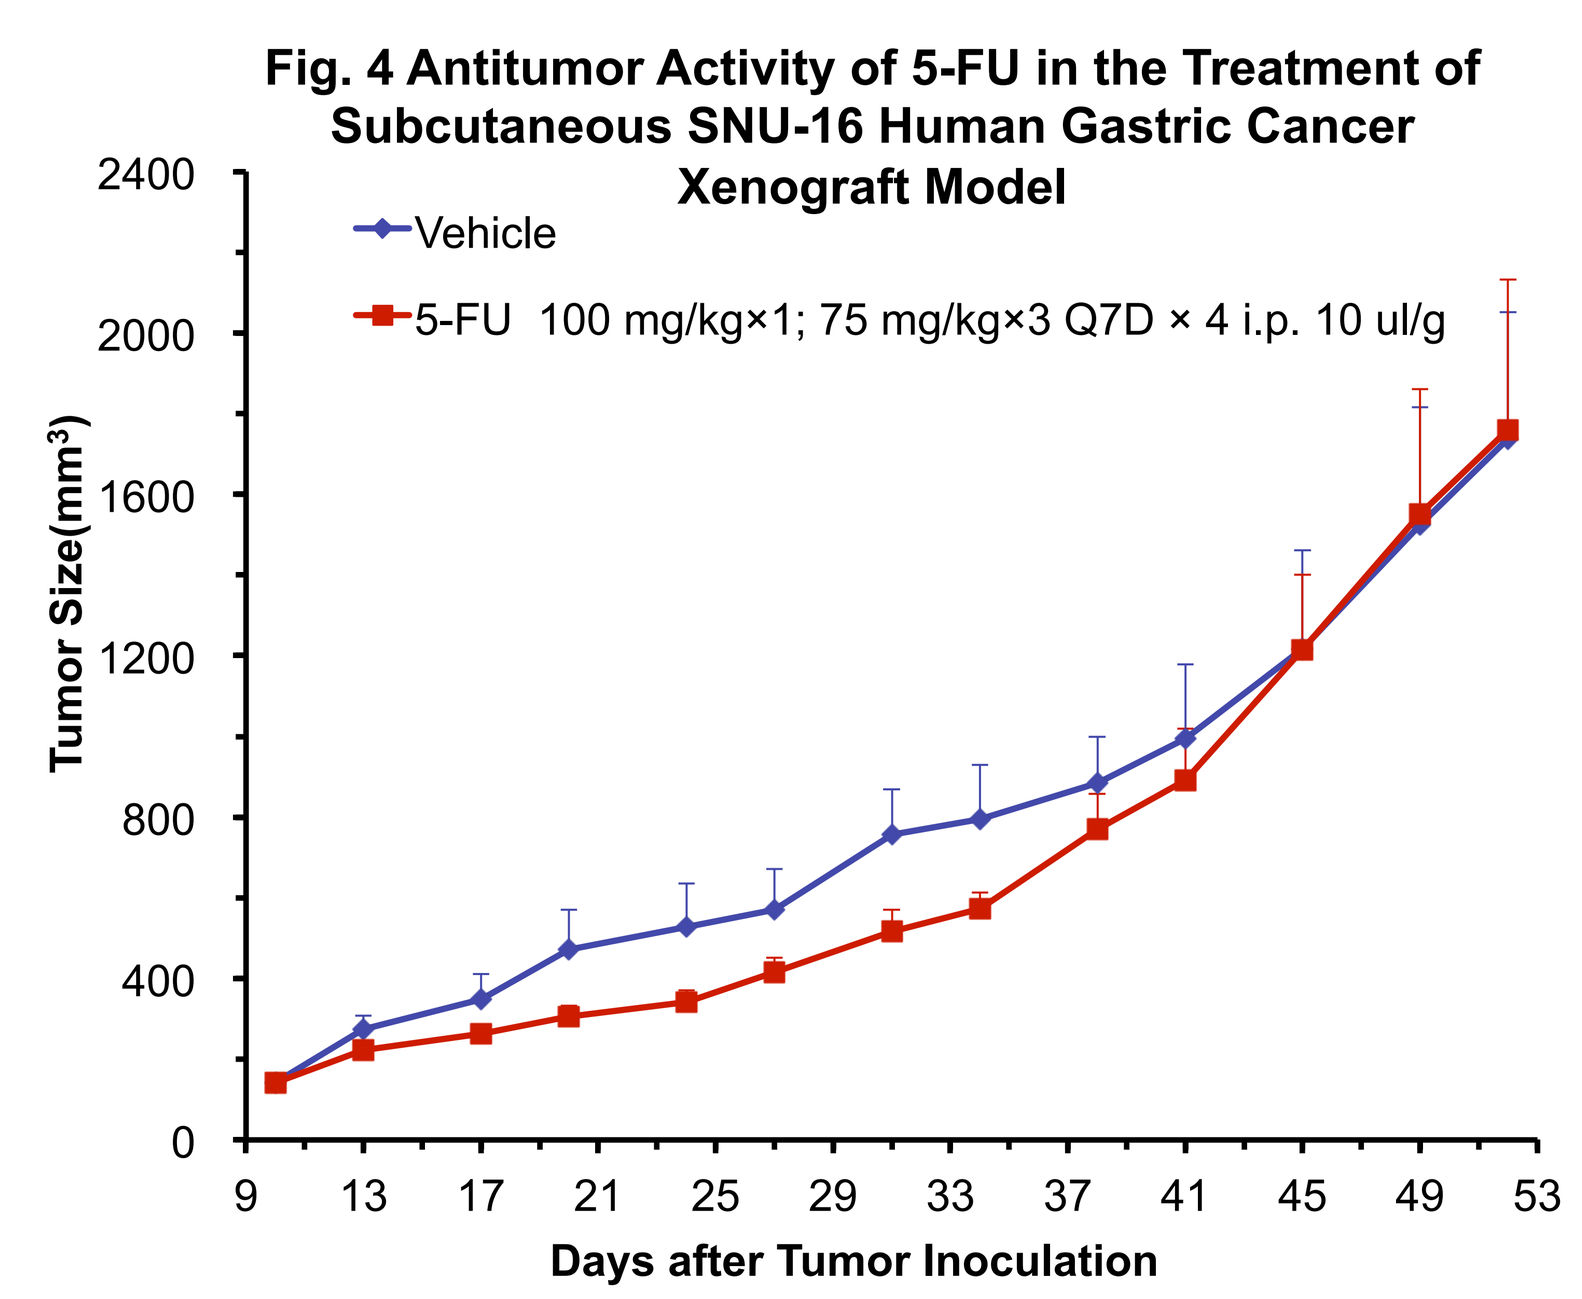
Fig. 4 Antitumor Activity of 5-FU in the Treatment of Subcutaneous SNU-16 Human Gastric Cancer Xenograft Model
Vehicle
5-FU 100 mg/kg×1; 75 mg/kg×3 Q7D × 4 i.p. 10 ul/g
Tumor Size(mm³)
Days after Tumor Inoculation

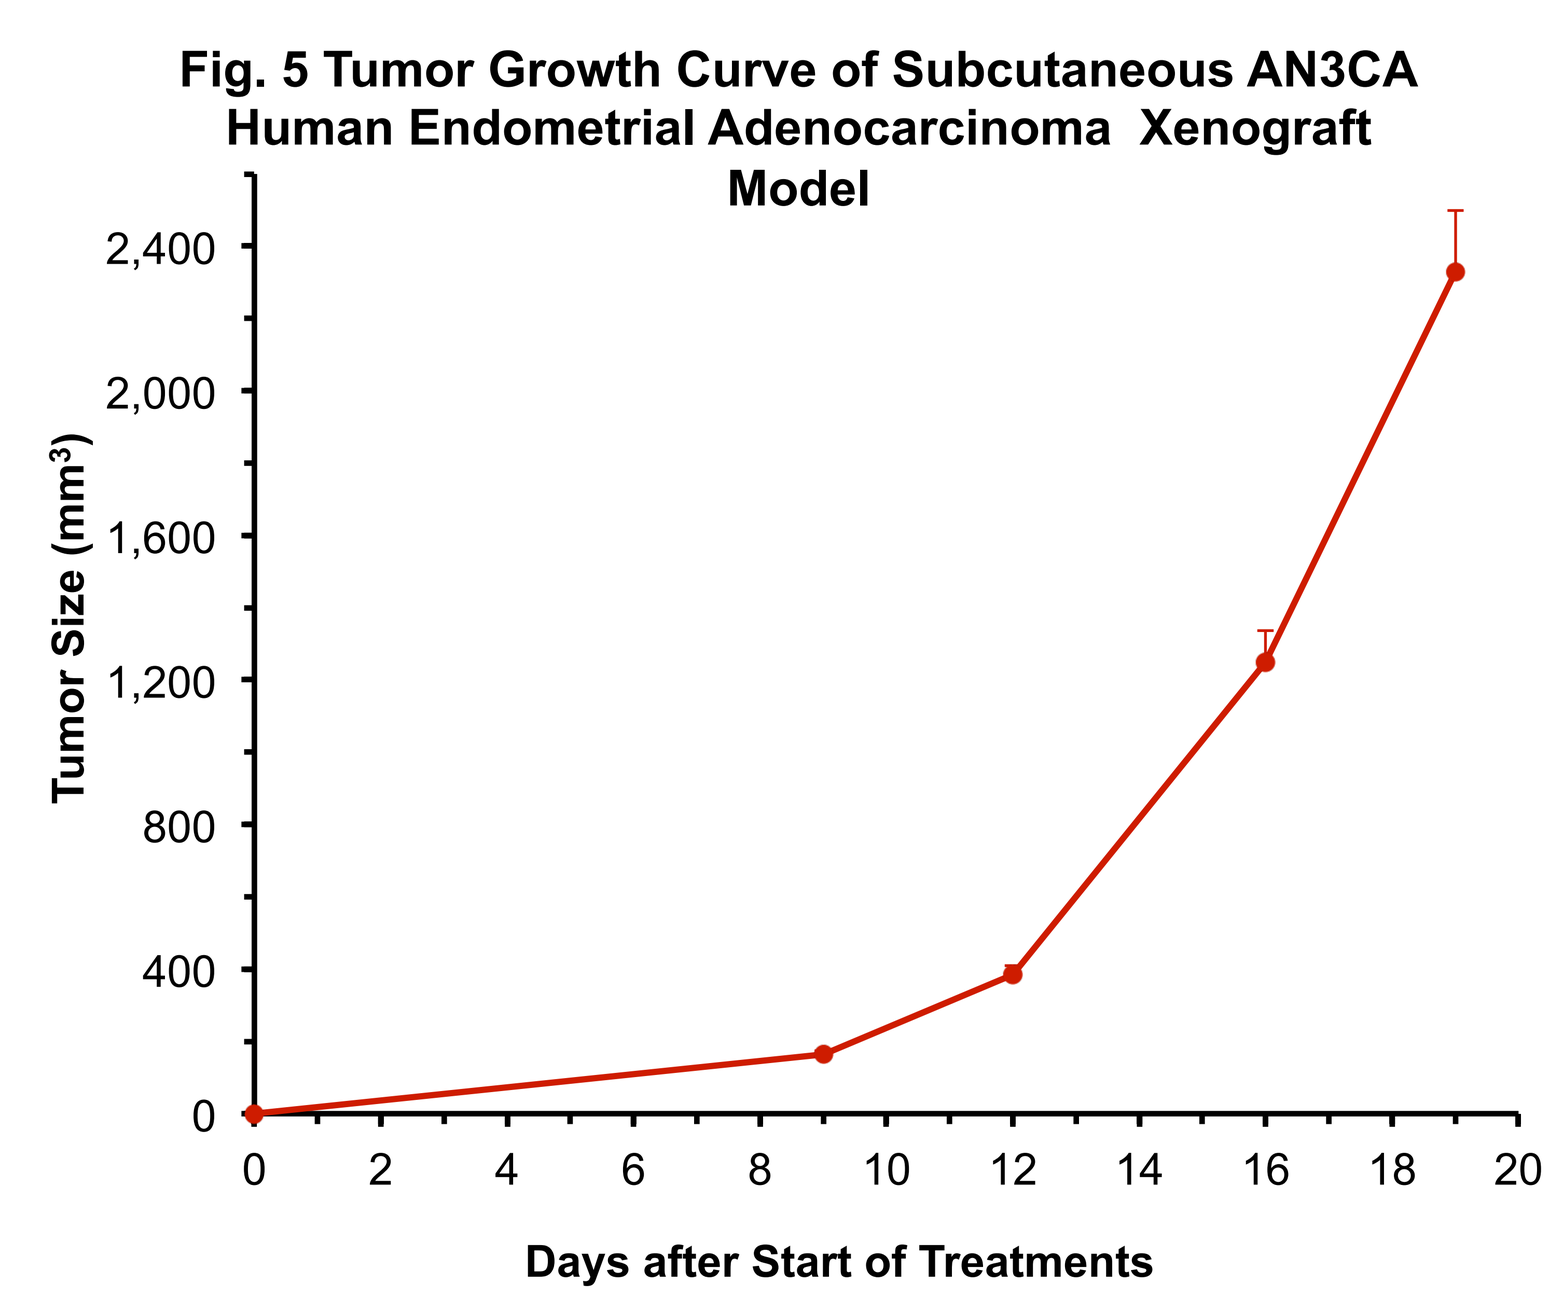
Fig. 5 Tumor Growth Curve of Subcutaneous AN3CA Human Endometrial Adenocarcinoma Xenograft Model
Tumor Size (mm³)
Days after Start of Treatments

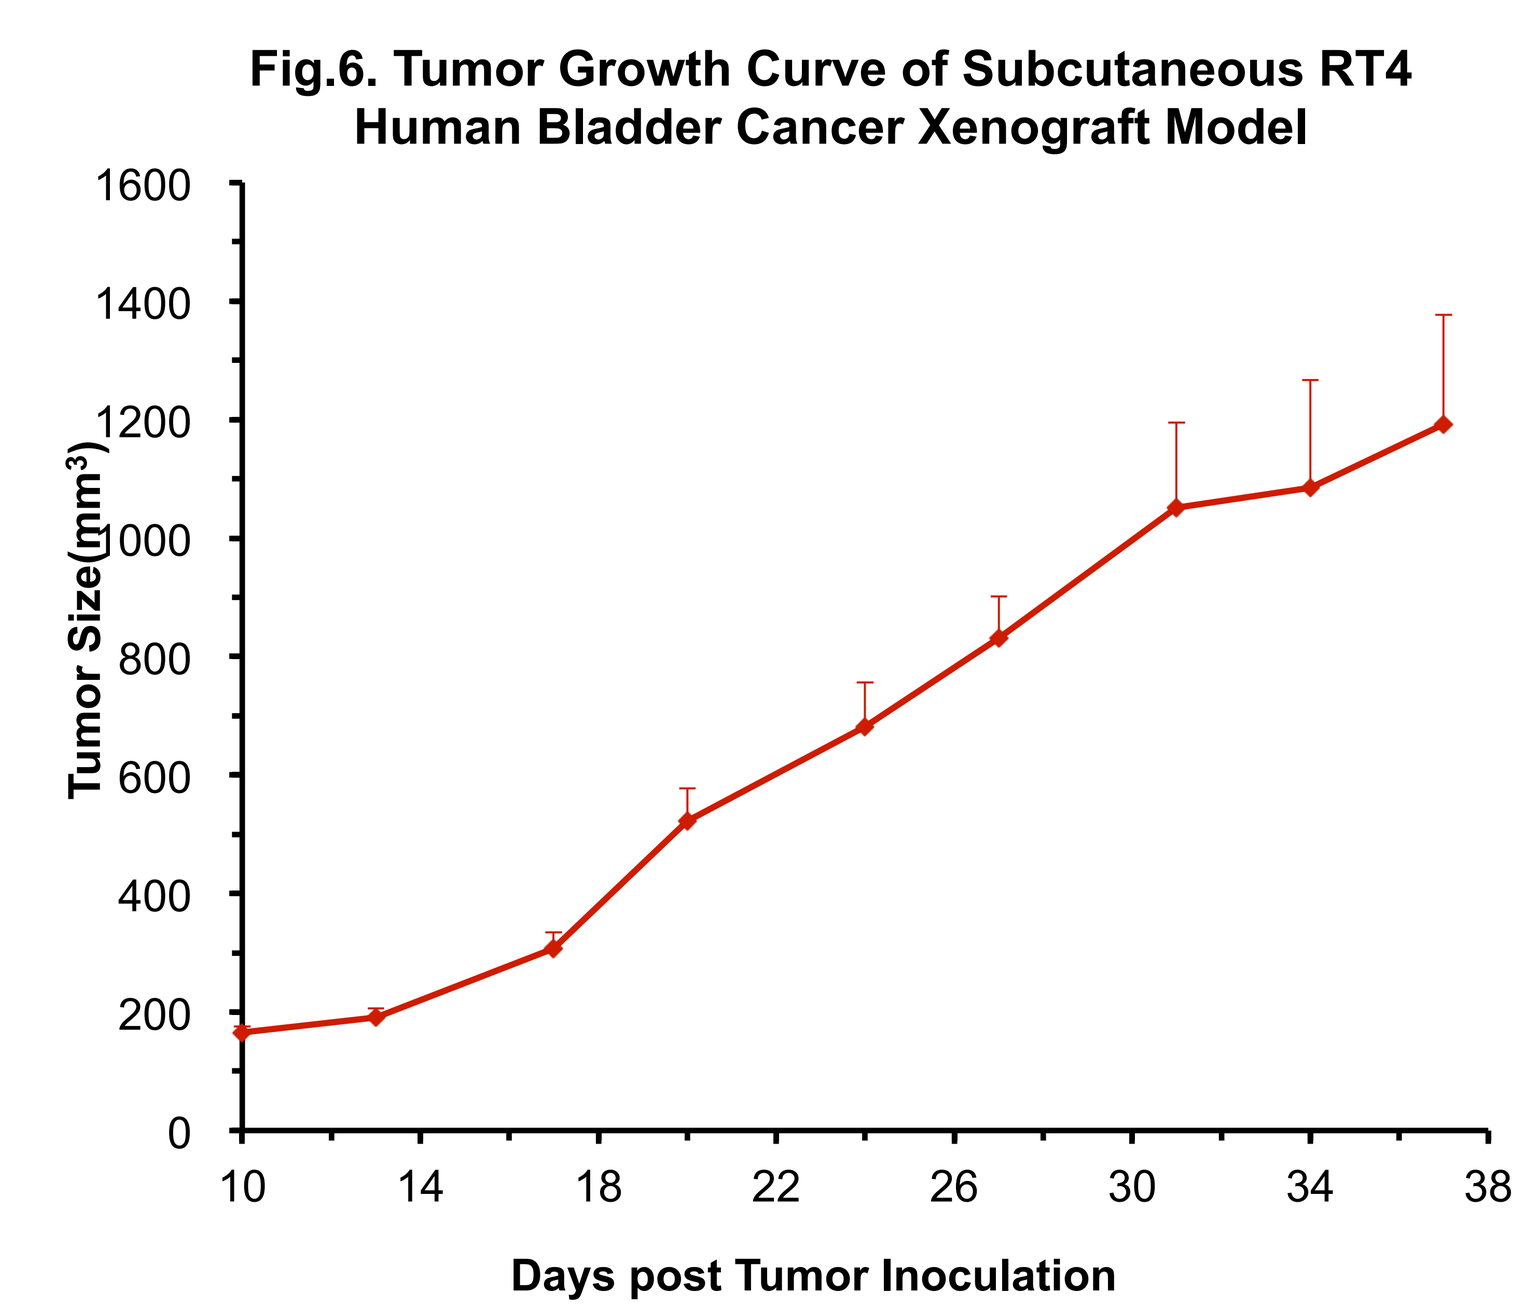
Fig.6. Tumor Growth Curve of Subcutaneous RT4 Human Bladder Cancer Xenograft Model
Tumor Size(mm³)
Days post Tumor Inoculation

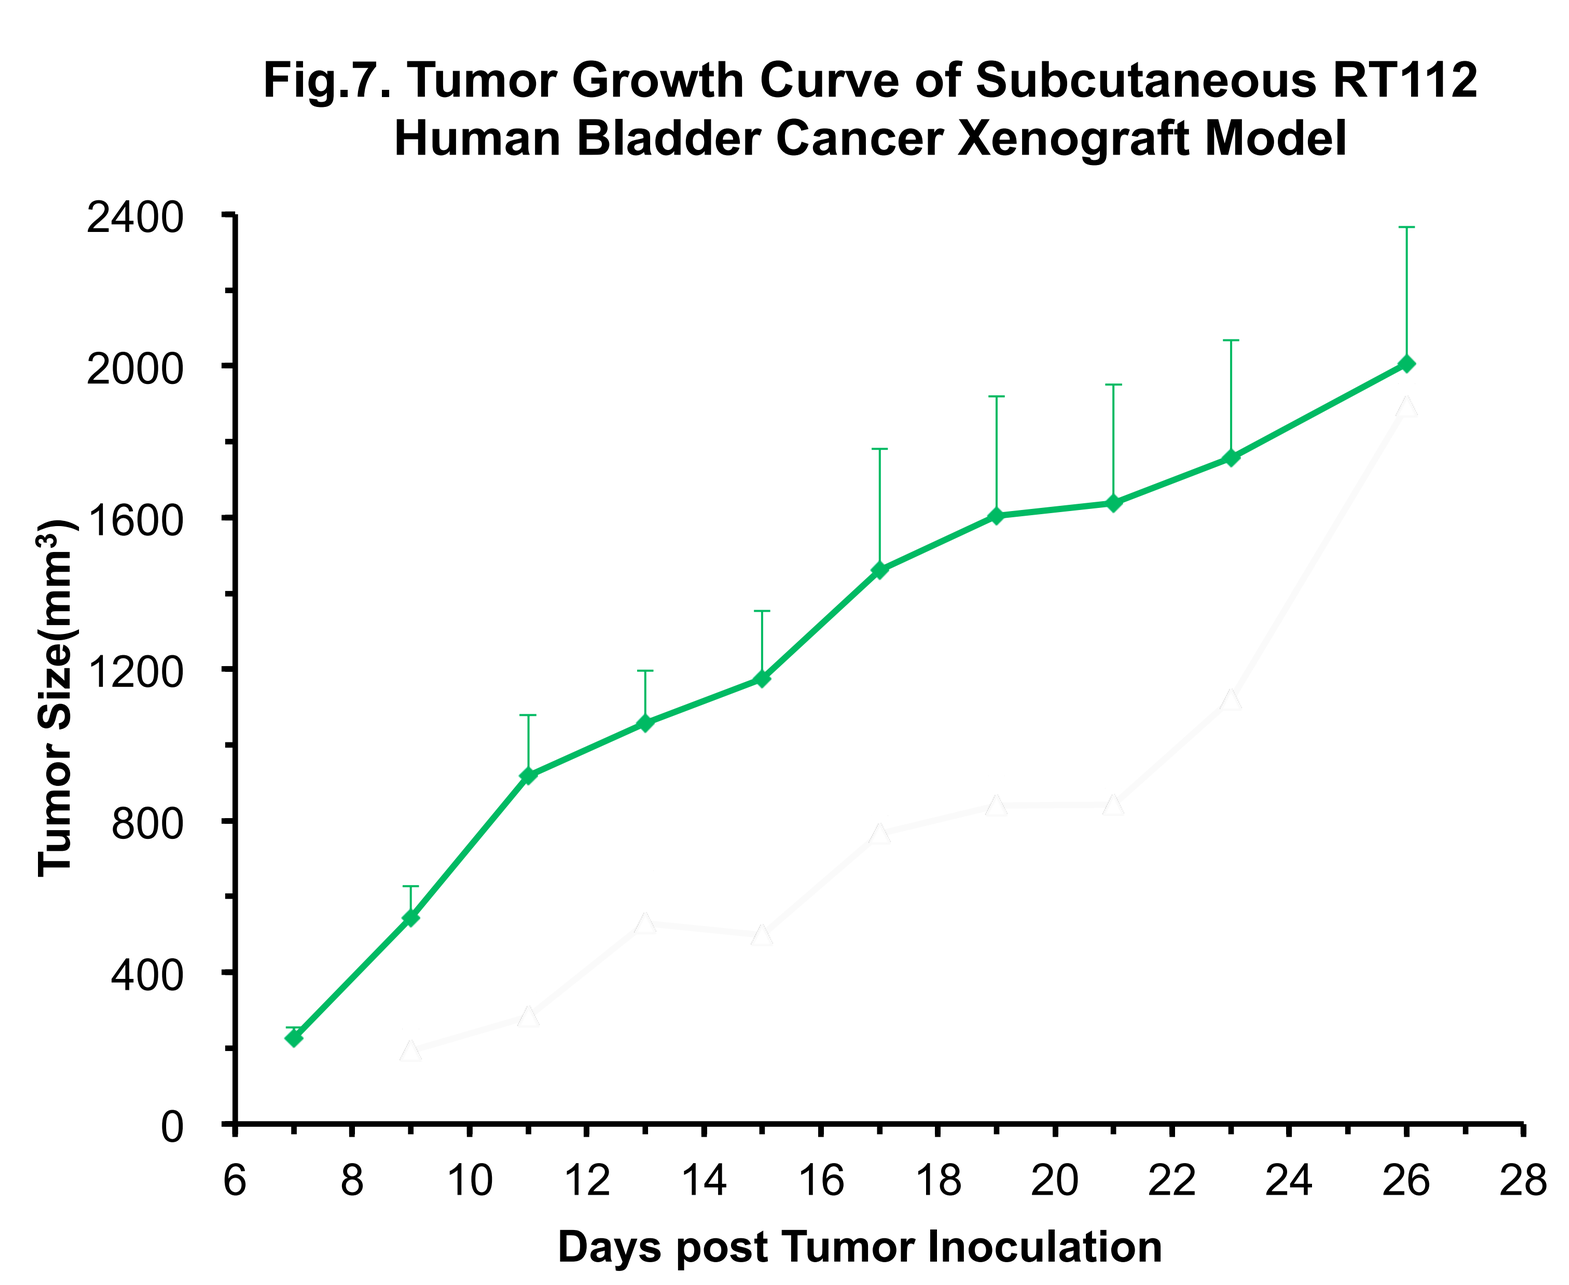
Fig.7. Tumor Growth Curve of Subcutaneous RT112 Human Bladder Cancer Xenograft Model
Tumor Size(mm³)
Days post Tumor Inoculation

Figure 2. Tumor grow curves of subcutaneous MDA-MB-134 human breast cancer xenograft model.
Figure 3. Tumor grow curves of subcutaneous NCI-H1581 Human Lung cancer xenograft model.
Figure 4. Antitumor activity of 5-FU in the treatment of subcutaneous SNU-16 Human Gastric cancer xenograft model.
Figure 5. Tumor grow curves of subcutaneous AN3CA Human Endometrial Adenocarcinoma xenograft model.
Figure 6. Tumor growth curve of subcutaneous RT4 Human bladder cancer xenograft model.
Figure 7. Tumor growth curve of subcutaneous RT112 Human bladder cancer xenograft model
Figure 8. Tumor grow curves of subcutaneous GA114 human gastric cancer xenograft model

PrimeXeno

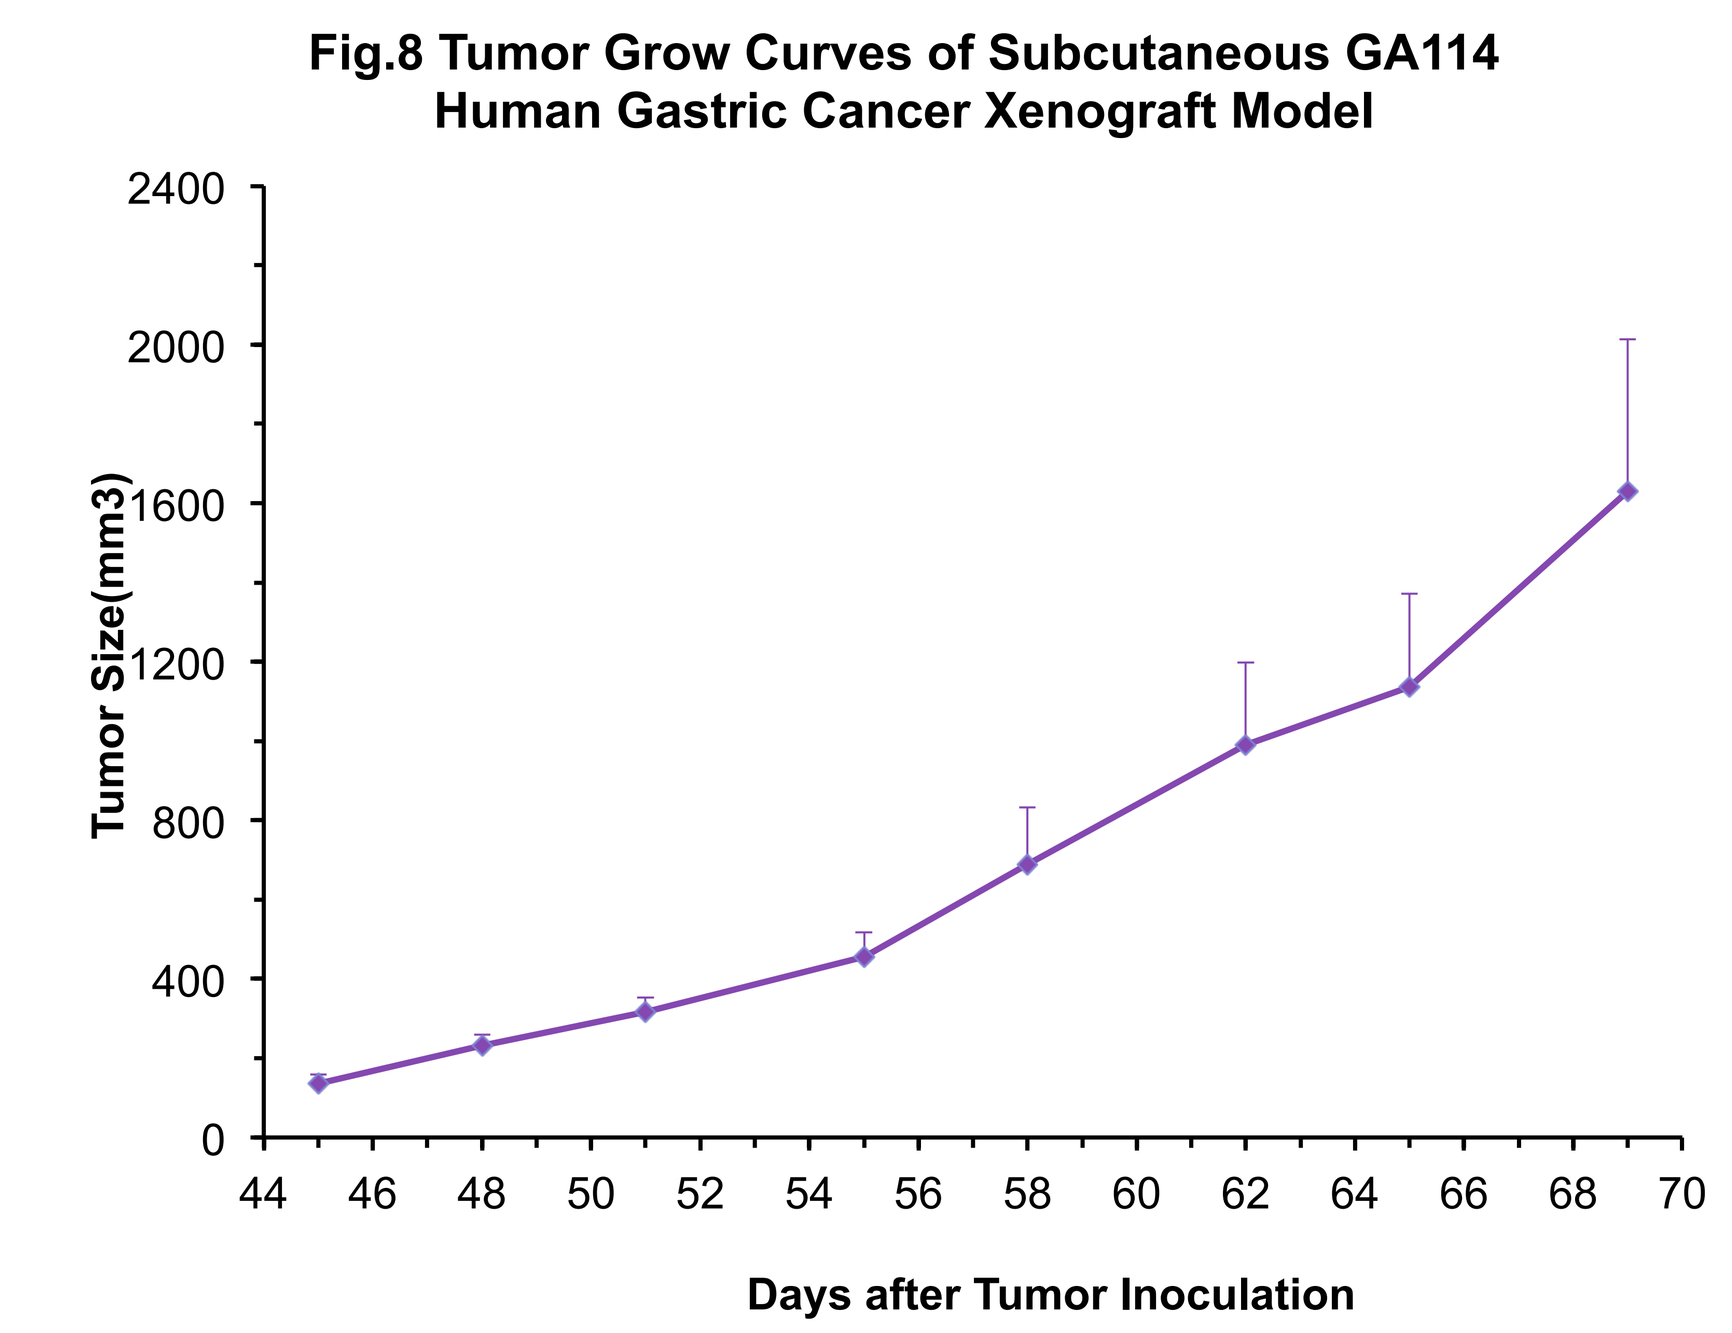
Fig.8 Tumor Grow Curves of Subcutaneous GA114 Human Gastric Cancer Xenograft Model
Tumor Size(mm3)
Days after Tumor Inoculation

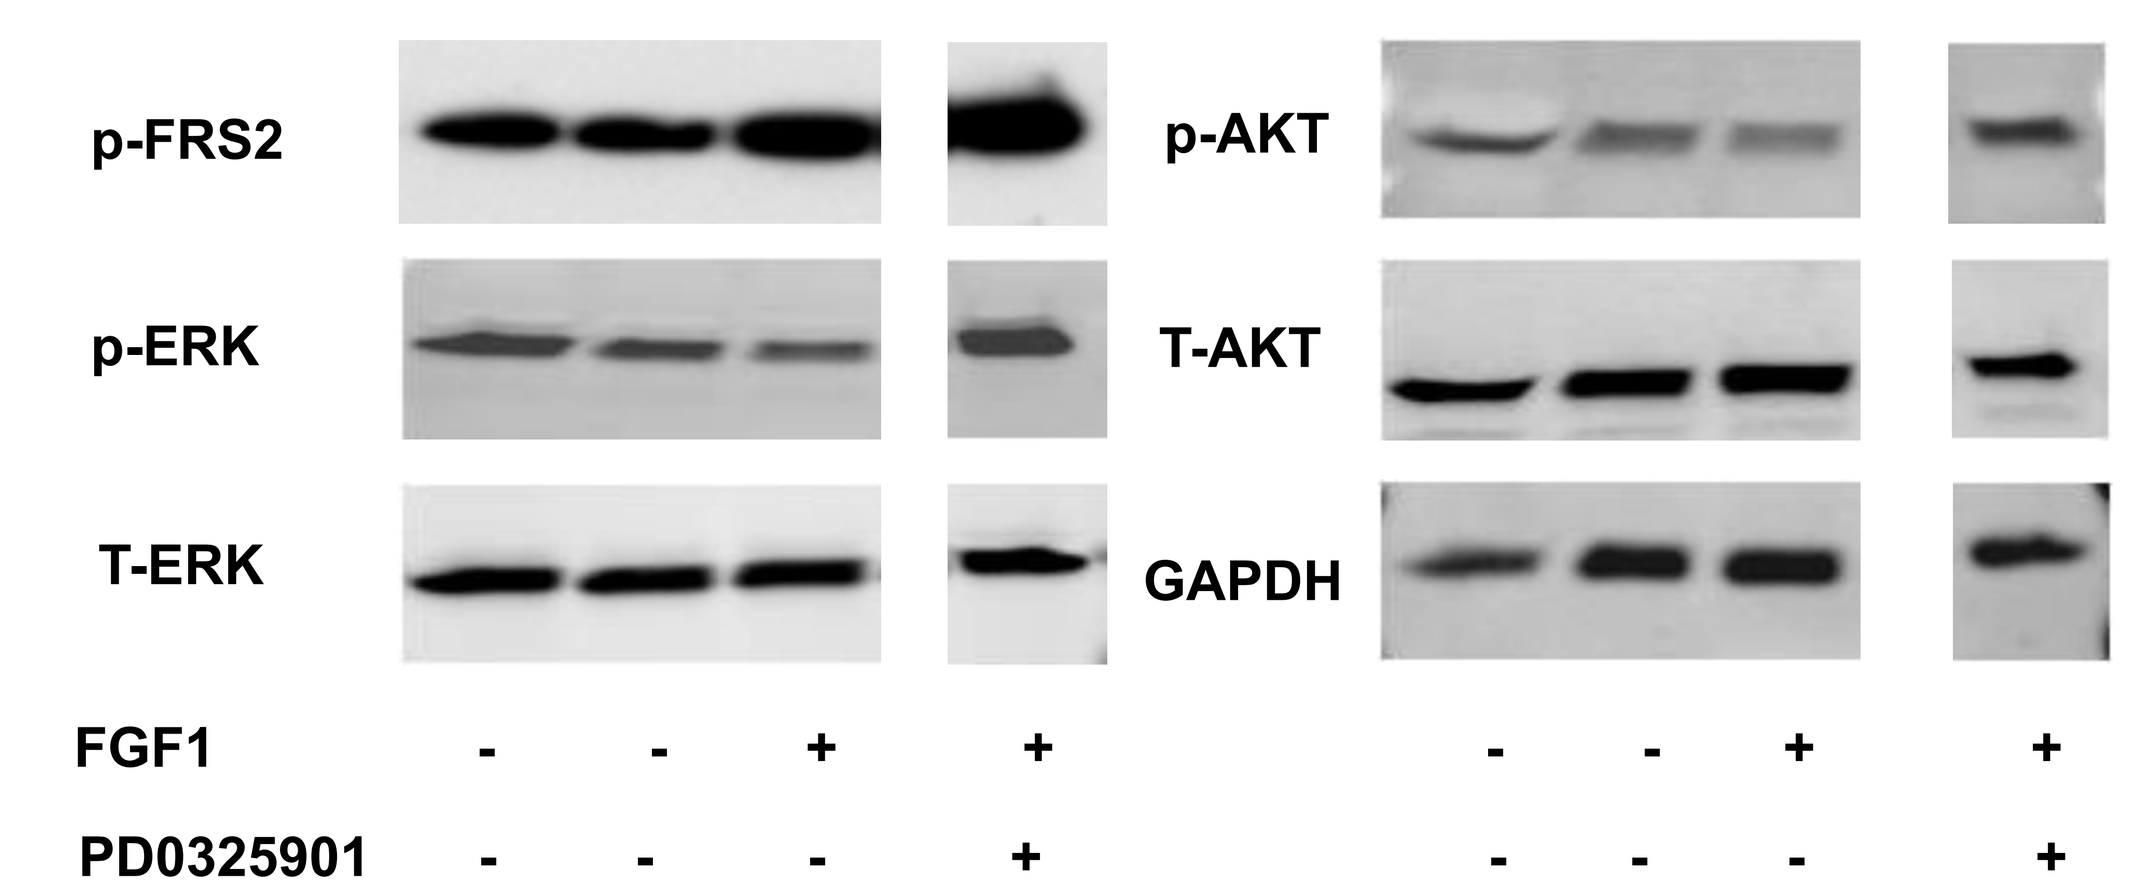
p-FRS2
p-ERK
T-ERK
p-AKT
T-AKT
GAPDH
FGF1 - - + + - - + +
PD0325901 - - - + - - - +

Figure 9. p-FRS2 expression and itsdownstream p-ERK, P-AKT expression after FGF1 stimulation in GA114 gastric cancer xenograft model.

Conclusion

In conclusion, we have established several FGFR targeted CDX and PrimeXeno models, including FGFR1 and FGFR2 amplification and FGFR2 mutation.

References

1. Yi-Mi Wu, Fengyun Su, Shanker Kalyana-Sundaram, et al.. Identification of Targetable FGFR Gene Fusions in Diverse Cancers. Cancer Discovery 2013;3:636-647.
2. Joseph M. Gozgit, Matthew J. Wong, Lauren Moran, et al. Ponatinib (AP24534), a Multitargeted Pan-FGFR Inhibitor with Activity in Multiple FGFR-Amplified or Mutated Cancer Models. Mol Cancer Ther 2012;11:690-699.
3. Genshi Zhao, Wei-ying Li, Daohong Chen, et al.. A Novel, Selective Inhibitor of Fibroblast Growth Factor Receptors That Shows a Potent Broad Spectrum of Antitumor Activity in Several Tumor Xenograft Models. Mol Cancer Ther 2011;10:2200-2210.

Contact information:
Dr. Juan Zhang, zhangjuan@crownbio.com; Dr. Qian Shi, shiqian@crownbio.com

Download the free Layar App
Scan this page
Discover interactive content