Patient-derived xenograft (PDX) models for triple negative breast cancer (TNBC): a pre-clinical platform for drug discovery

Jayant Thatte, Miguel Meza, Jill Ricono, Cyrus Mirsaidi and Thomas Broudy
Crown Bioscience San Diego Inc. & Molecular Response LLC. 11011 Torreyana Road, San Diego, CA 92121

Poster : 3226

In life for life

Abstract

In the United States, TNBC comprises 15-20% of breast cancers and are characterized by the lack of expression of estrogen receptor, progesterone receptor and receptor tyrosine protein kinase ERBB2 or Her-2/neu oncogene amplification. As a result, this type of breast cancer is difficult to treat as most of the chemotherapies target these 3 receptors. TNBC is highly aggressive and is associated with high morbidity, mortality and shorter disease-free survival. Given its heterogeneous clinical presentation, histology and response to therapy, prognosis and management of TNBC is complicated. In this study, we report development and characterization of the TNBC PDX model. We used patient derived TNBC tissues to generate 5 new TNBC models in NOD-SCID mice. Patient tumors were pre-screened for their ER, PR and Her-2/Neu expression by IHC, prior to inoculation in mice. Efficacy of two standard of care drugs, Cisplatin and Vinorelbine demonstrate the utility of these models in drug discovery for treatment of TNBC.

PDX Models

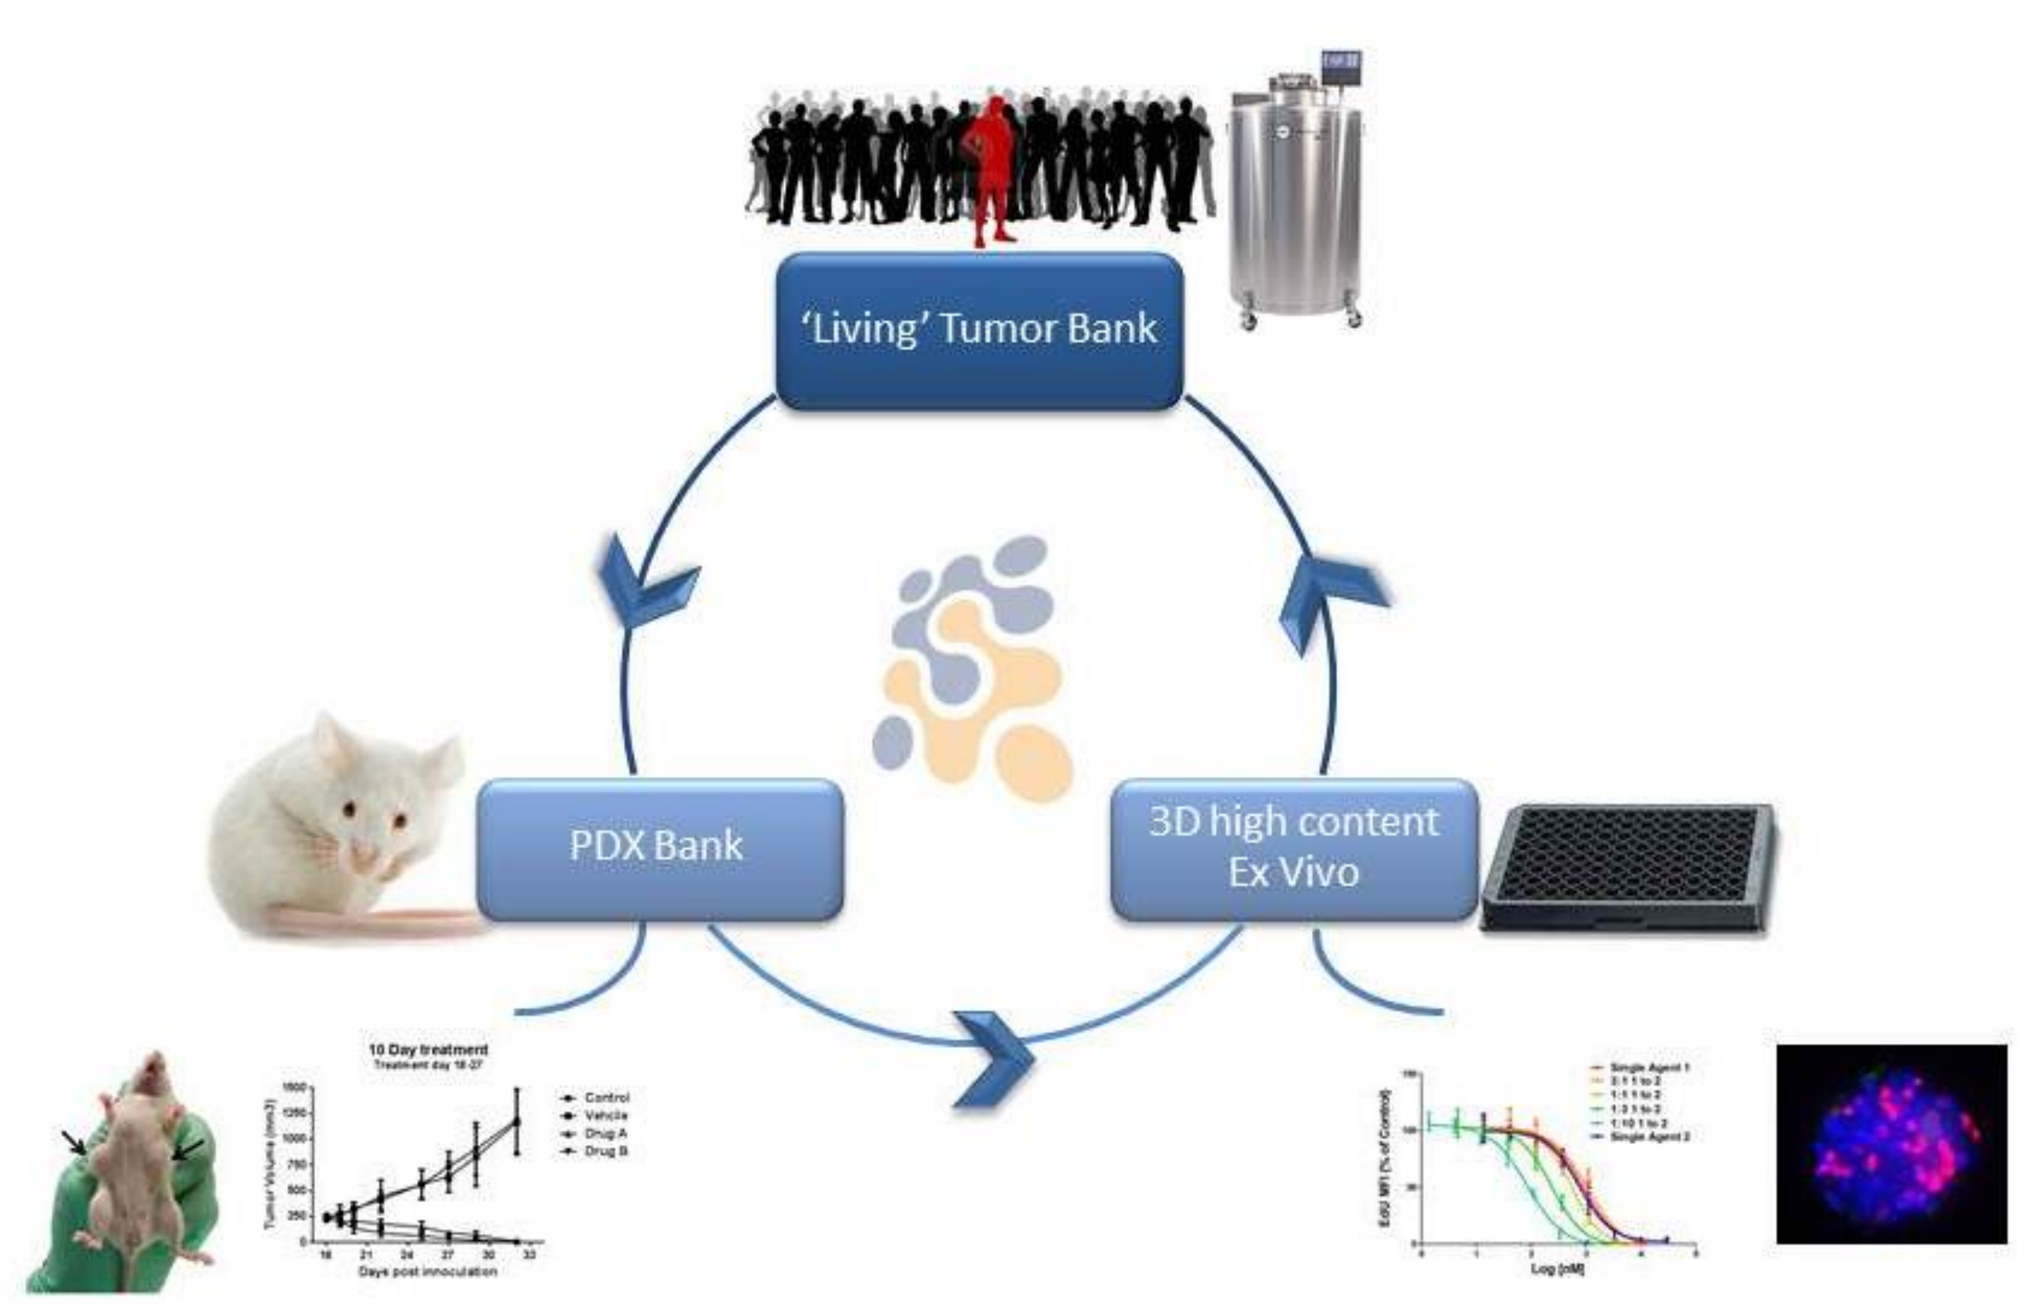
'Living' Tumor Bank
PDX Bank
3D high content
Ex Vivo

Schema for utilization of tumor bank for oncology drug development:

The 'living' tumor bank can be screened for the patient population of interest (based on molecular characterization or other characteristics), test a hypothesis or develop new hypotheses. Cryopreserved cells were injected into immunocompromized mice to develop xenografts. Tumor cells from those xenograft models can be grown as spheroids to conduct *Ex vivo* drug treatment studies using high content imaging platform.

TNBC Patient Information

Patient ID	2002080607	2003100375	2004070737	2004120492	2009110002
Age	46.7	34.3	37.1	39.8	49.2
Sex	Female	Female	Female	Female	Female
Clinical Diagnosis	Breast Carcinoma	Recurrent Breast Carcinoma	Breast Carcinoma	Breast Carcinoma	Breast carcinoma
Collection Site	L Pleural Fluid	L Chest Wall Resection	R Breast	Omental Nodule	L breast
Prior Treatment	Treated	Treated	Treated	Treated	Treated
IHC for ER/PR/ HER2	Negative	Negative	Negative	Negative	Negative

TNBC Models: *in vitro* Drug Resistance

Patient ID	4HC	5FU	CPCB	CPLAT	DOXO	GMCB	NAVBL	TAXOL	TXTR
2002080607	E	E	--	E	L	E	L	L	E
2003100375	L	E	--	I	E	E	L	I	L
2004070737	L	E	L	L	E	L	L	L	L
2004120492	L	E	E	L	L	L	L	L	L
2009110002	L	I	L	L	L	L	L	L	L

L : Low resistance
I : Intermediate resistance
E : Extreme resistance

TNBC Model Builds & SOC Efficacy

A

B

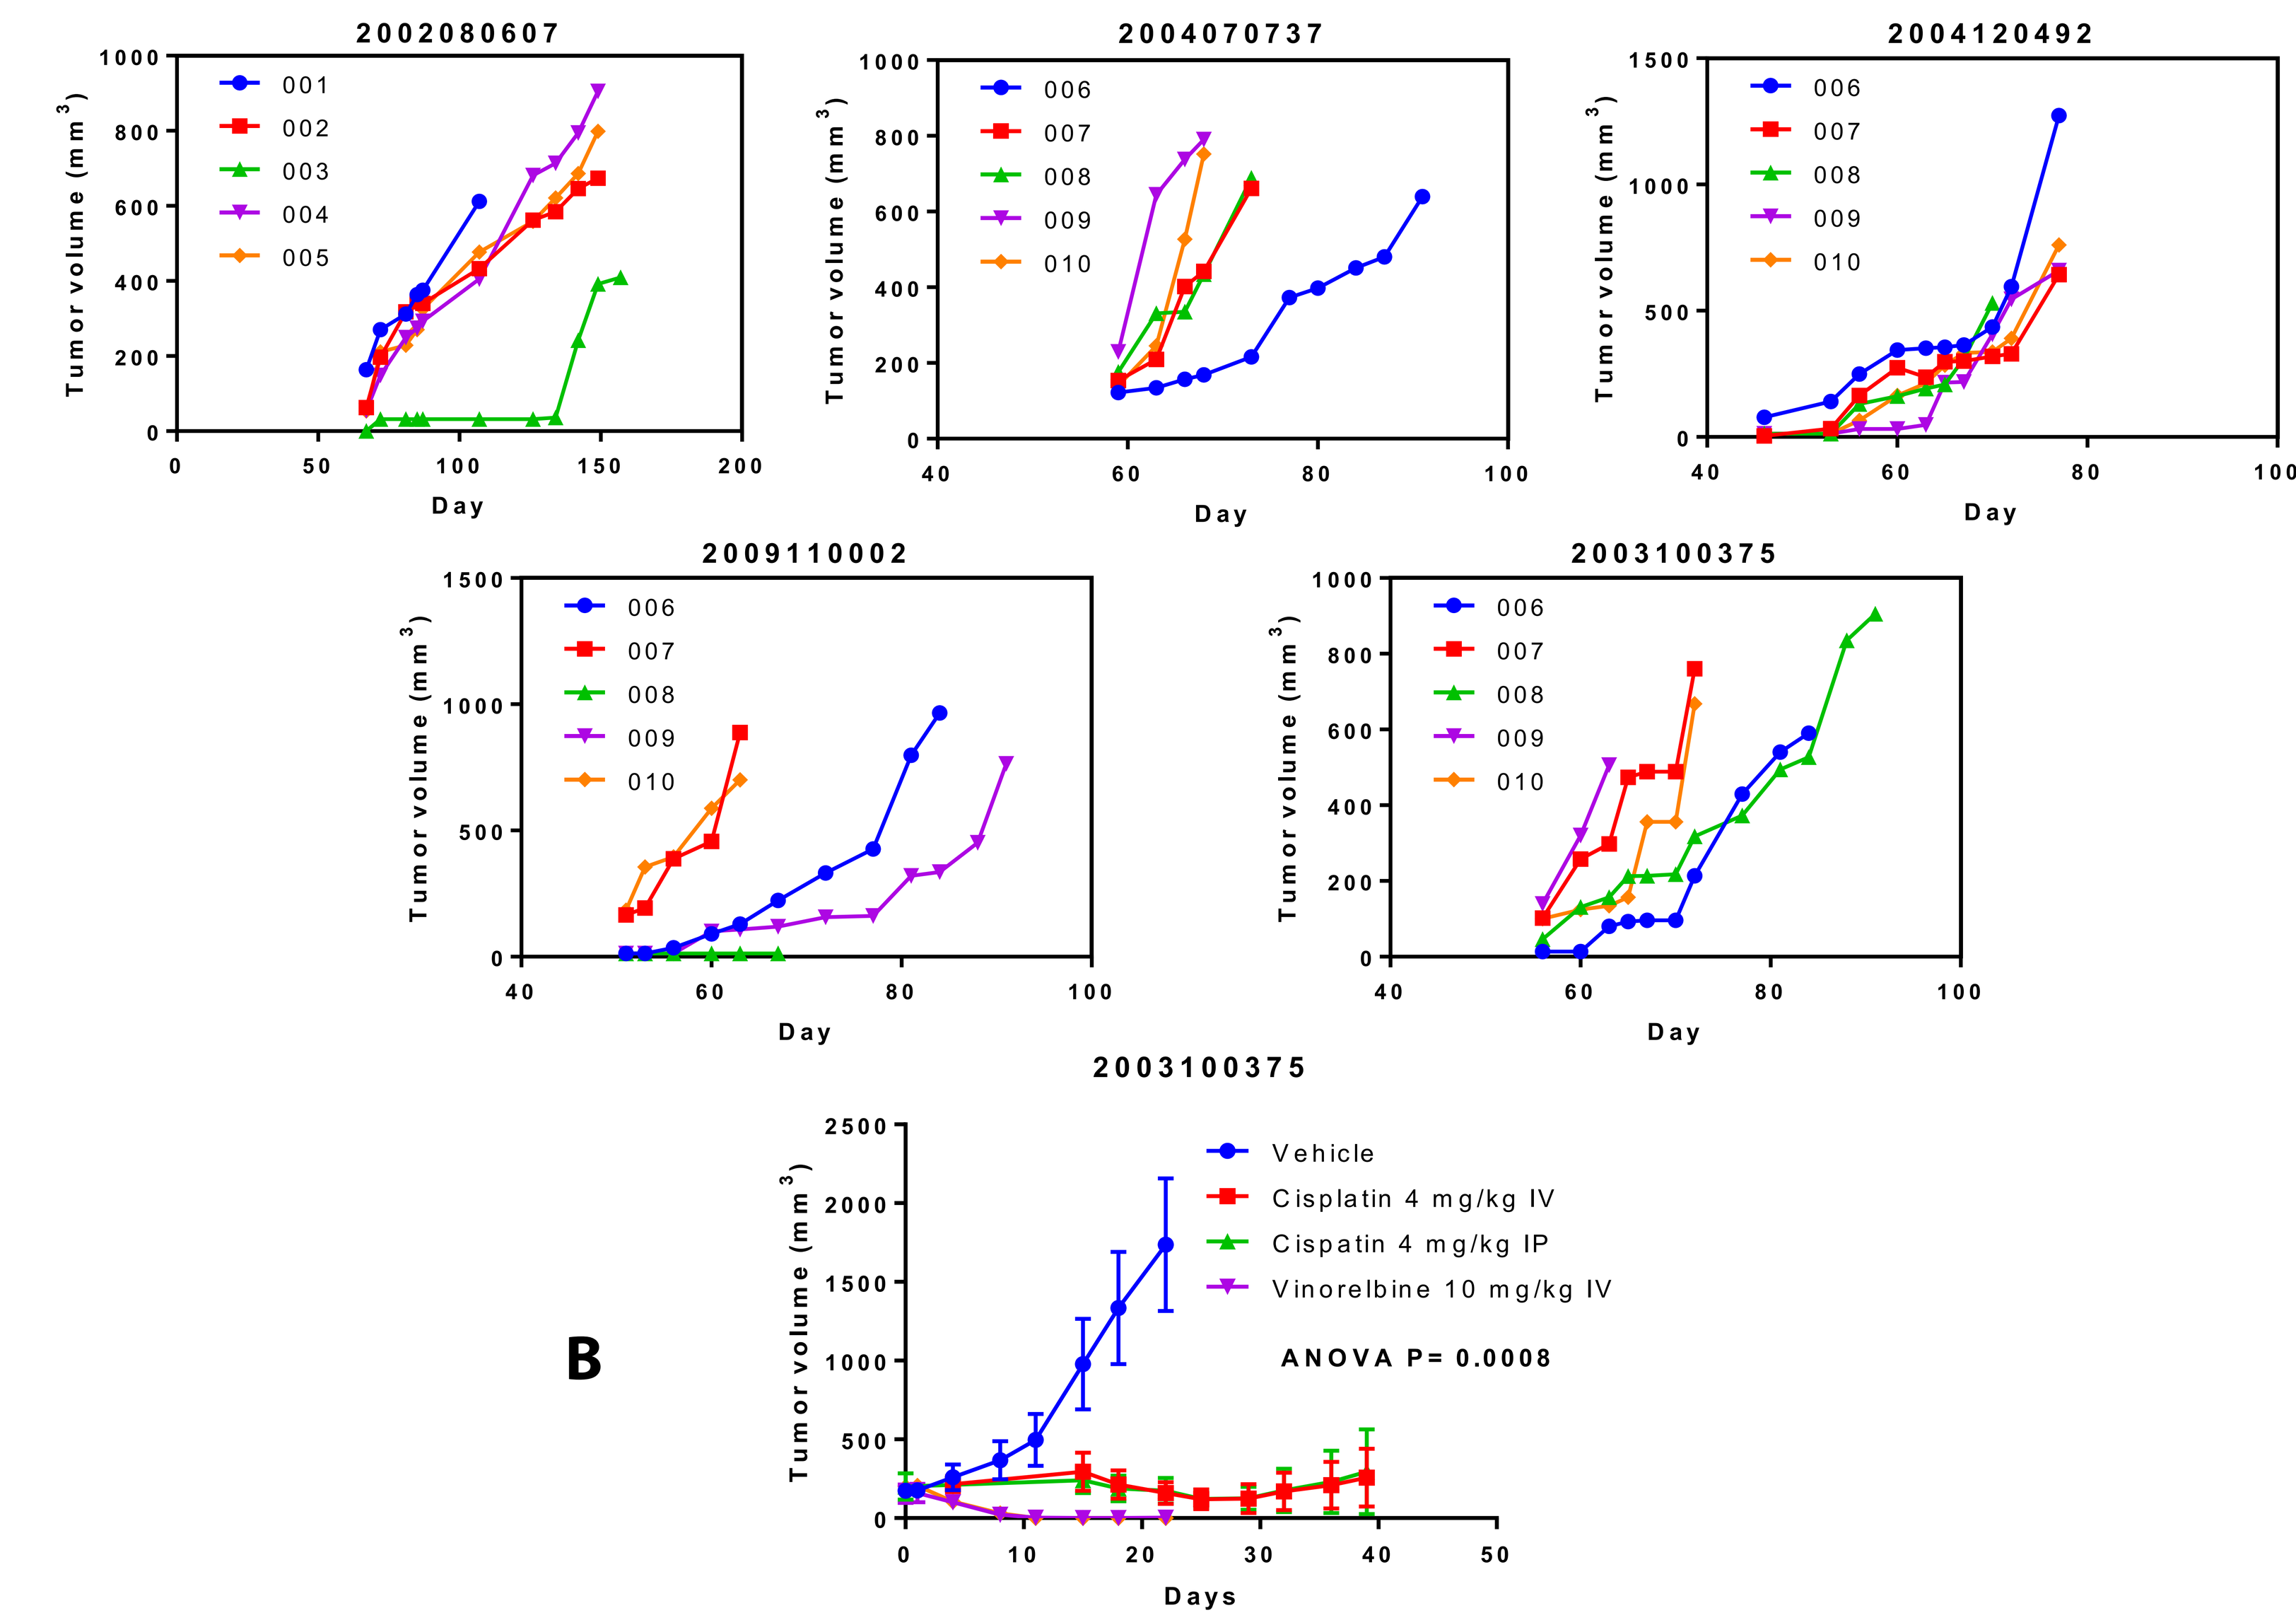
2002080607
2004070737
2004120492
2009110002
2003100375
Tumor volume (mm3)
Day
001
002
003
004
005
006
007
008
009
010
2003100375
Vehicle
Cisplatin 4 mg/kg IV
Cispatin 4 mg/kg IP
Vinorelbine 10 mg/kg IV
ANOVA P= 0.0008
Days

Triple negative breast carcinoma xenograft model

A) Mice were inoculated subcutaneously into immunocompromised mice with patient tumor samples (P0) and monitored for engraftment. Tumor growth was measured using calipers and tumors were harvested during exponential growth phase when tumors reached >500 mm^3. Tissue were fixed and stained to verify the pathology. Triple negativity was confirmed by immunohistochemistry analysis.

B) For the SOC efficacy study, animals were randomized and drug treatment was initiated on day 1 when average tumor volume reached 150-200 mm^3.

Summary & Conclusions

- Crown Bioscience's tumor bank is a reliable source of patient-derived tumor cells for drug testing
- Treatment with Cisplatin and Vinorelbine significantly suppressed tumor growth in the TNBC model
- Crown Bioscience's TNBC PDX models are powerful tools to interrogate novel drug candidates

Further characterization of these TNBC models is currently in progress