Development of patient-derived xenograft models for small cell lung cancer as a preclinical platform for drug development

Chelsea Mullins, Jill Ricono, Praveen Nair, Cyrus Mirsaidi and Thomas Broudy
Crown Bioscience San Diego Inc. & Molecular Response LLC. 11011 Torreyana Road, San Diego, CA 92121

Poster : 4209

In life for life

Introduction

Small cell lung cancer comprises 15-20% of all lung cancer cases, and is more invasive and has a higher rate of proliferation than non-small cell lung cancer, leading to a higher mortality rate. Most cases are responsive to chemotherapy, however there is a high rate of recurrence and those in advanced stage of the disease often have a refractory response. As such, the need for new treatment modalities is critical.

In previous work, we have reported using the Molecular Response tumor bank, comprising >144,000 viable tumor specimens, to establish more than 350 PDX models of various cancer types.

In this work, we describe the development of 8 new SCLC PDX models established from a collection of >300 small cell lung cancer specimens, many of which come from prior-treated and metastatic patients. We report a comprehensive characterization of these models, including: histopathology, immunohistochemistry, and mutation analysis. Additionally, we have evaluated functional response of these models with in vivo pharmacology studies. Current studies are underway to derive correlations between in vivo drug response and mutational status of these models. Our data strongly suggests the potential to use these unique PDX models to aid in efforts in drug development efforts in oncology.

Living Tumor Bank & Patient Derived Xenograft Models

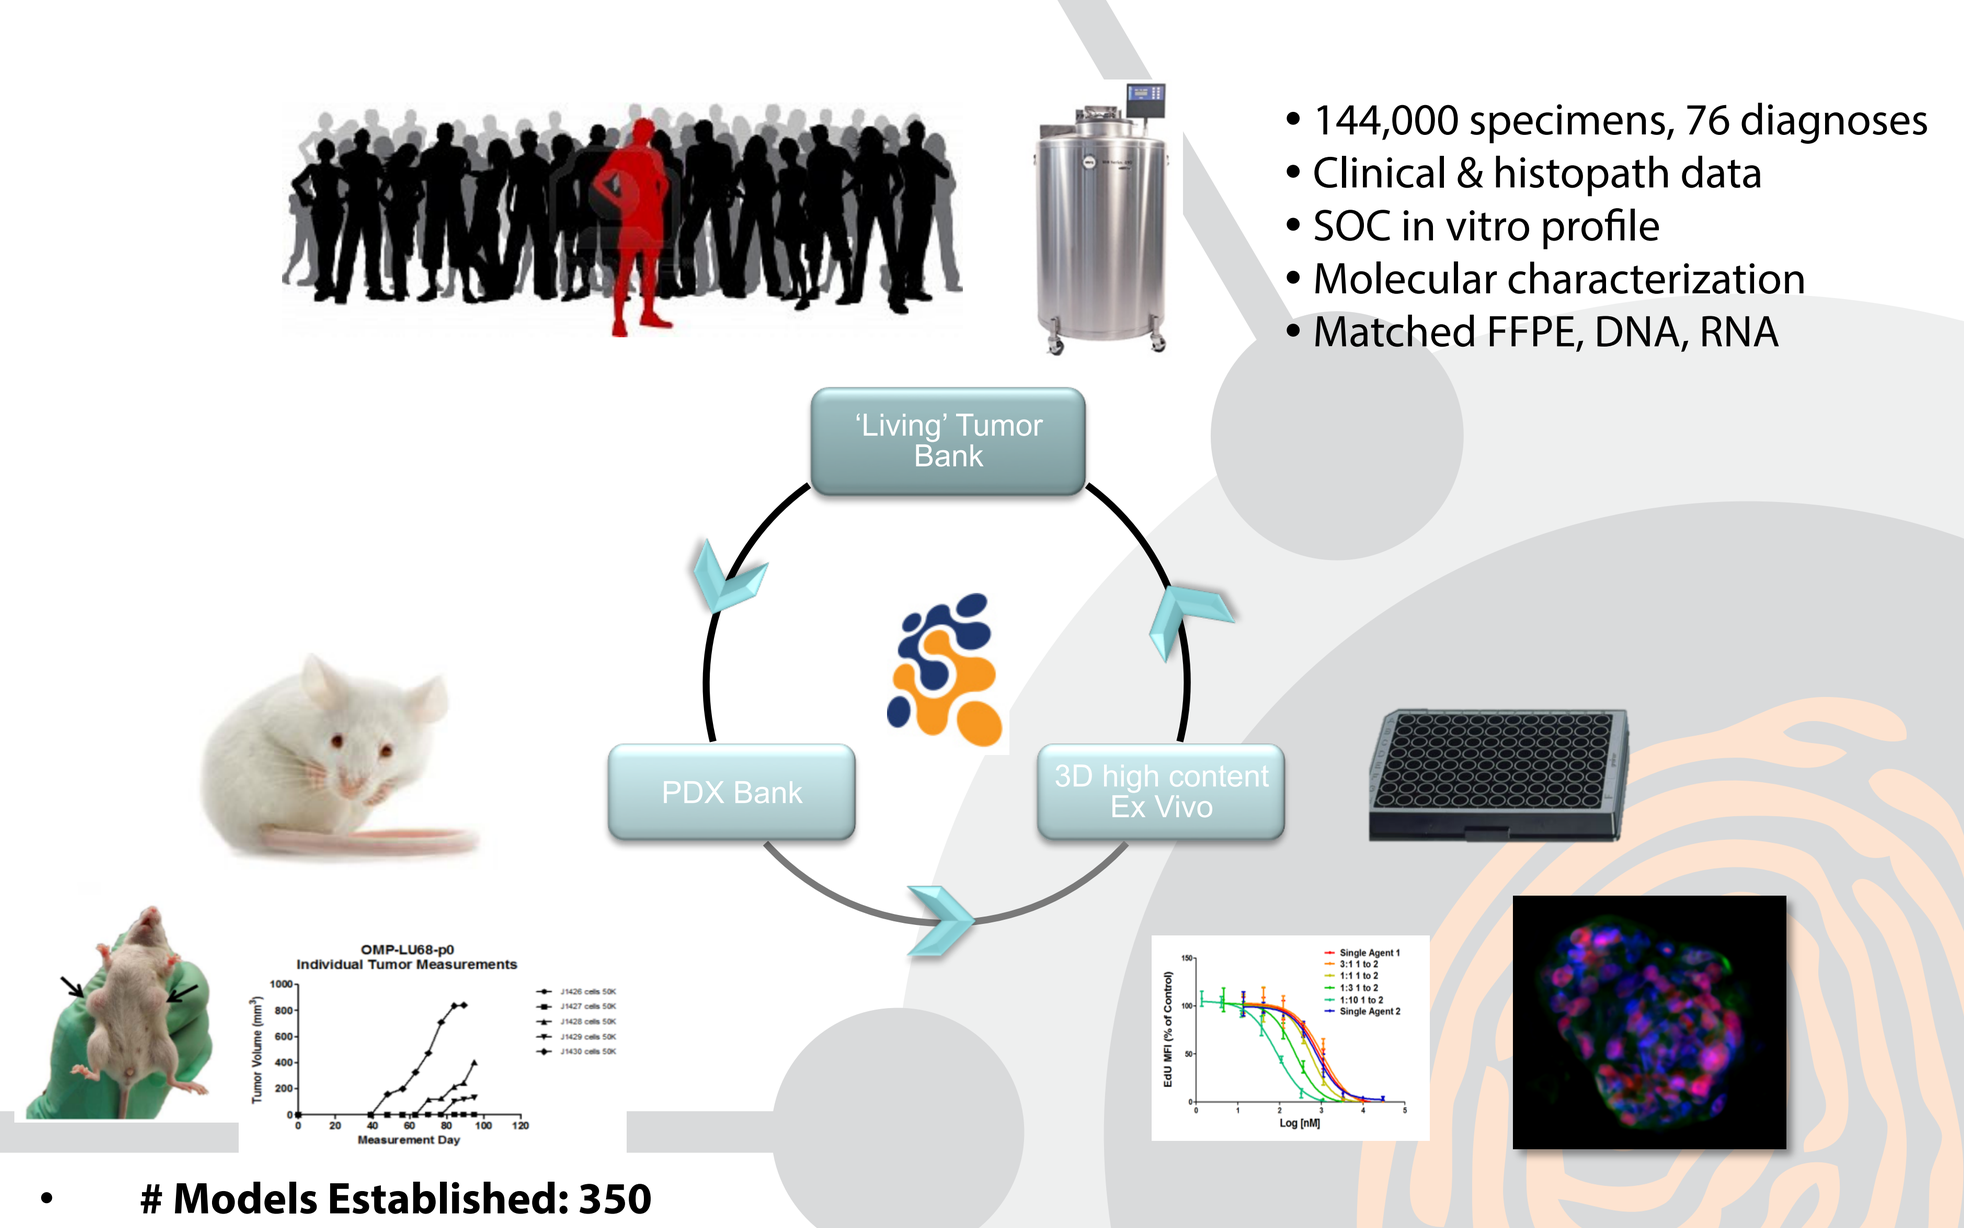
• 144,000 specimens, 76 diagnoses
• Clinical & histopath data
• SOC in vitro profile
• Molecular characterization
• Matched FFPE, DNA, RNA
'Living' Tumor Bank
PDX Bank
3D high content Ex Vivo
• # Models Established: 350
• # Models in Progess: 541

PDX Bank: Molecular Response PDX models have been combined with the Crown Bioscience HuPrime PDX collection now comprising more than 1,600 PDX models across 70 cancer indications. KIT mutant SCLC tumors are an example patient type found in the collection.

PDX Model Development

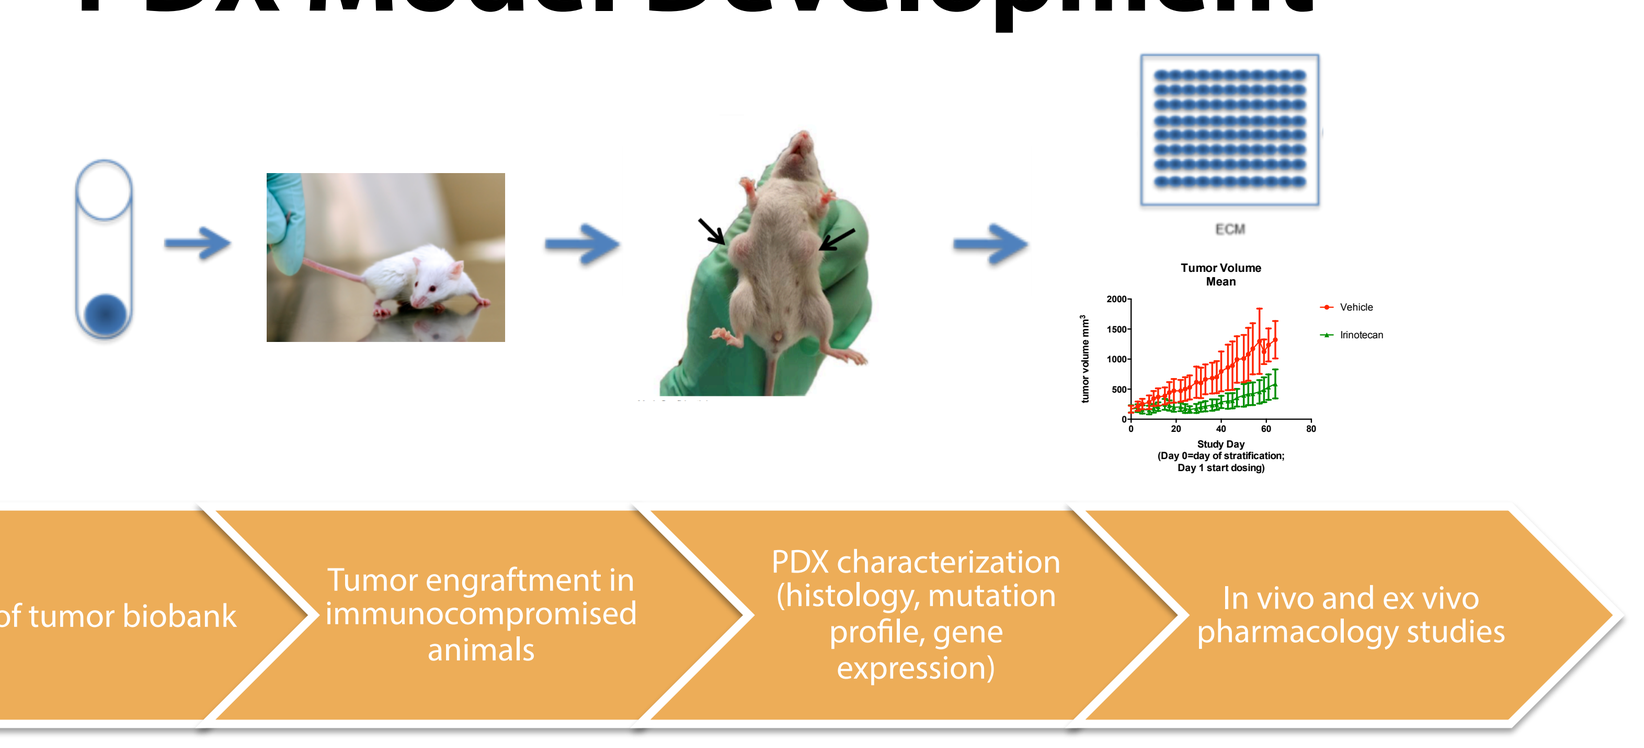
Prescreening of tumor biobank
Tumor engraftment in immunocompromised animals
PDX characterization (histology, mutation profile, gene expression)
In vivo and ex vivo pharmacology studies

- Improved preclinical models are required to advance our understanding of the molecular aberrations that underpin cancer
- Patient-derived primary tumor cell models grown in mice have proven critical for developing and deploying targeted therapies to improve patients' lives.

Patient Information

ID	Sex	Age	Cancer	Collection Site	Prior Treatment
LU90	M	69.2	SCL	L Lower Lobe Mass	No
LU89	F	61.6	SCL	Ovary	Yes
LU86	M	57.8	SCL	R Neck Node	Yes
LU87	F	69.8	SCL	L Lung Nodule	No
LU67	F	60.3	SCL	R Neck Mass	No
LU68	F	48.9	SCL	Omentum	No
LU65	M	16.8	SCL	Left pleura	Yes
LU66	F	69.6	SCL	Right axillary lymph node	Yes

SCL Patients Information: A summary of relevant patient data for the SCL models developed. Prior treatment history, metastatic vs. primary information available.

Mouse Model Builds

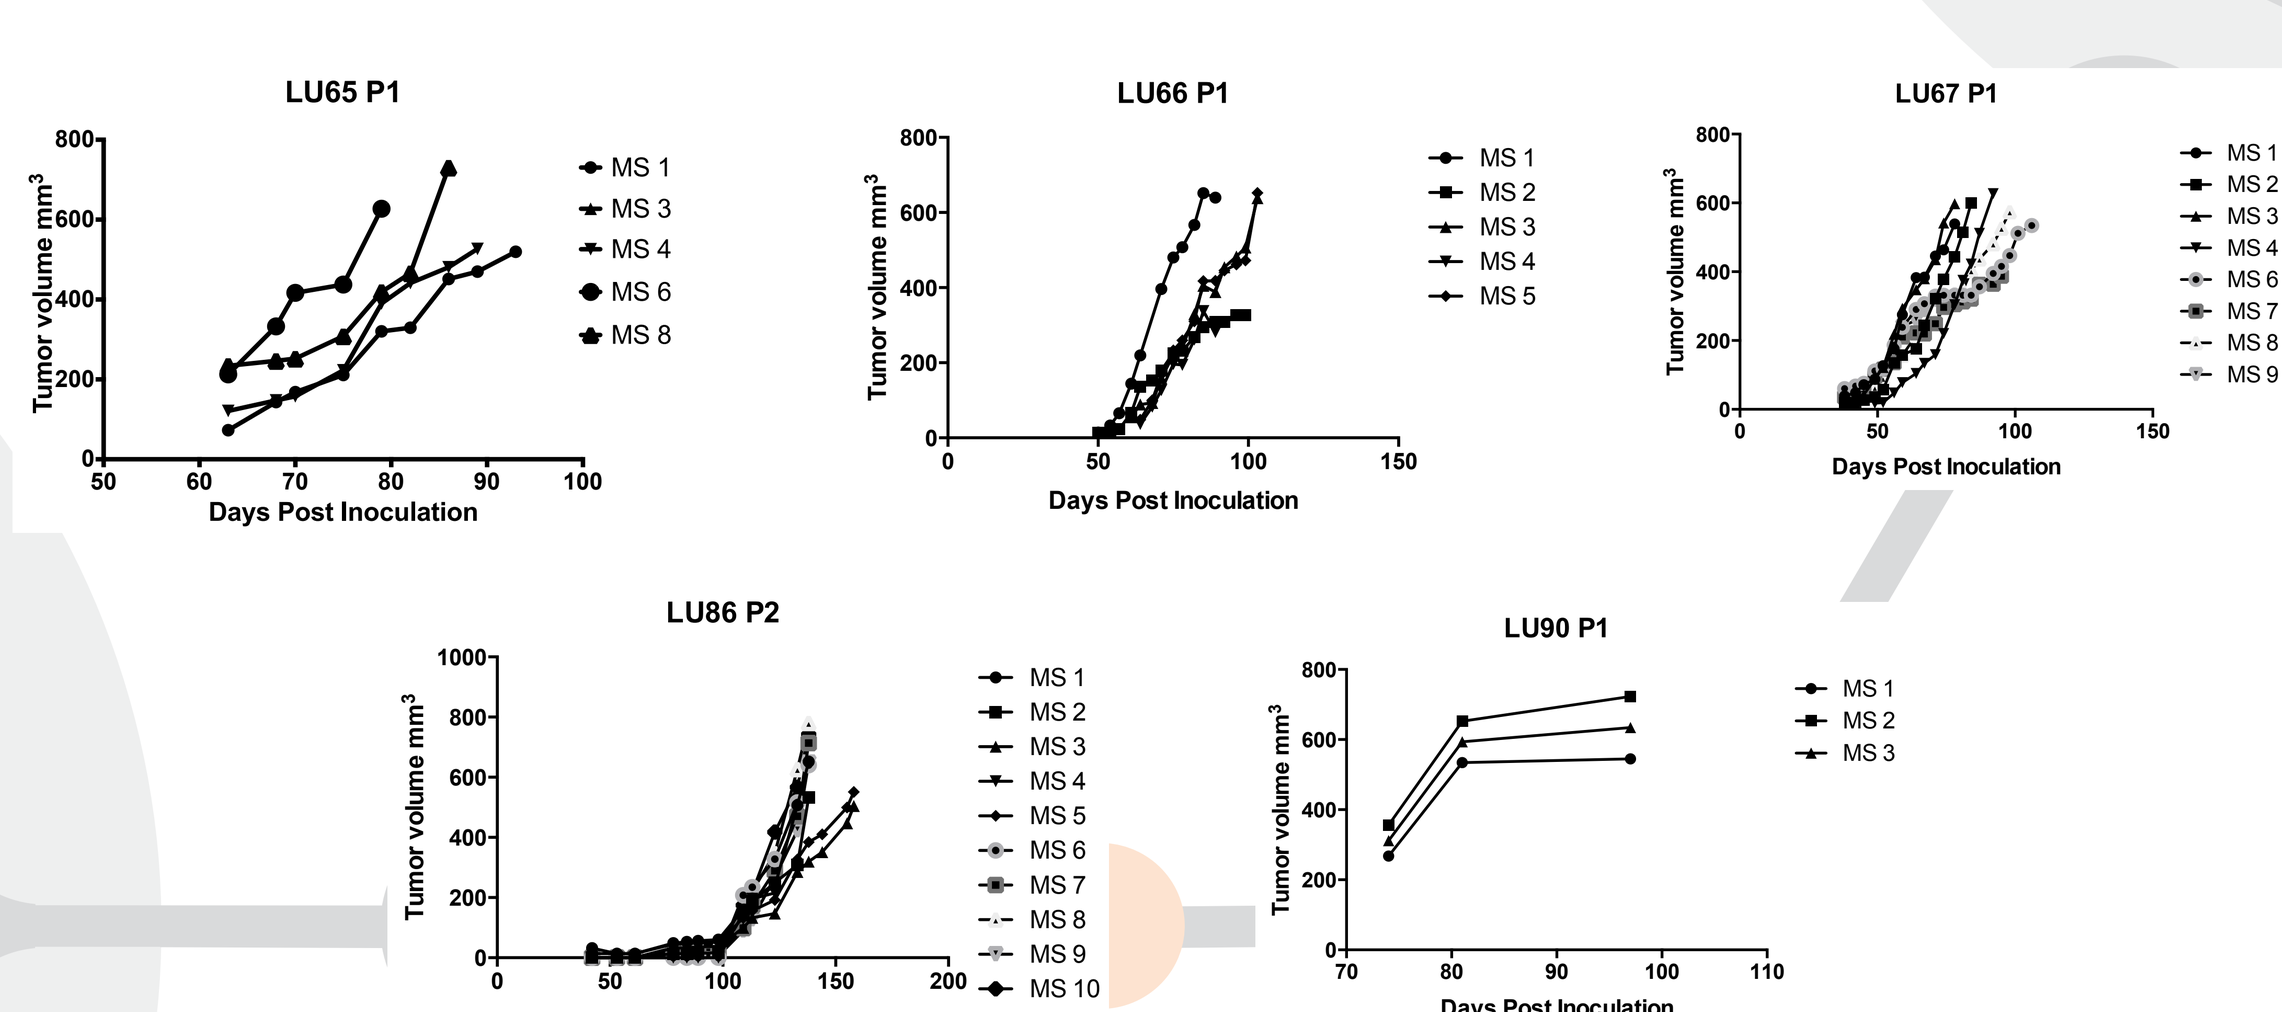
LU65 P1
LU66 P1
LU67 P1
LU86 P2
LU90 P1
Tumor volume mm³
Days Post Inoculation

Establishment of PDX Models: Mice were inoculated from frozen patient tumor samples (P0) and monitored for engraftment. Cells from the tumors were reinoculated into animals to confirm the growth as a PDX model (≥P1) and banked down for future use.

Histology

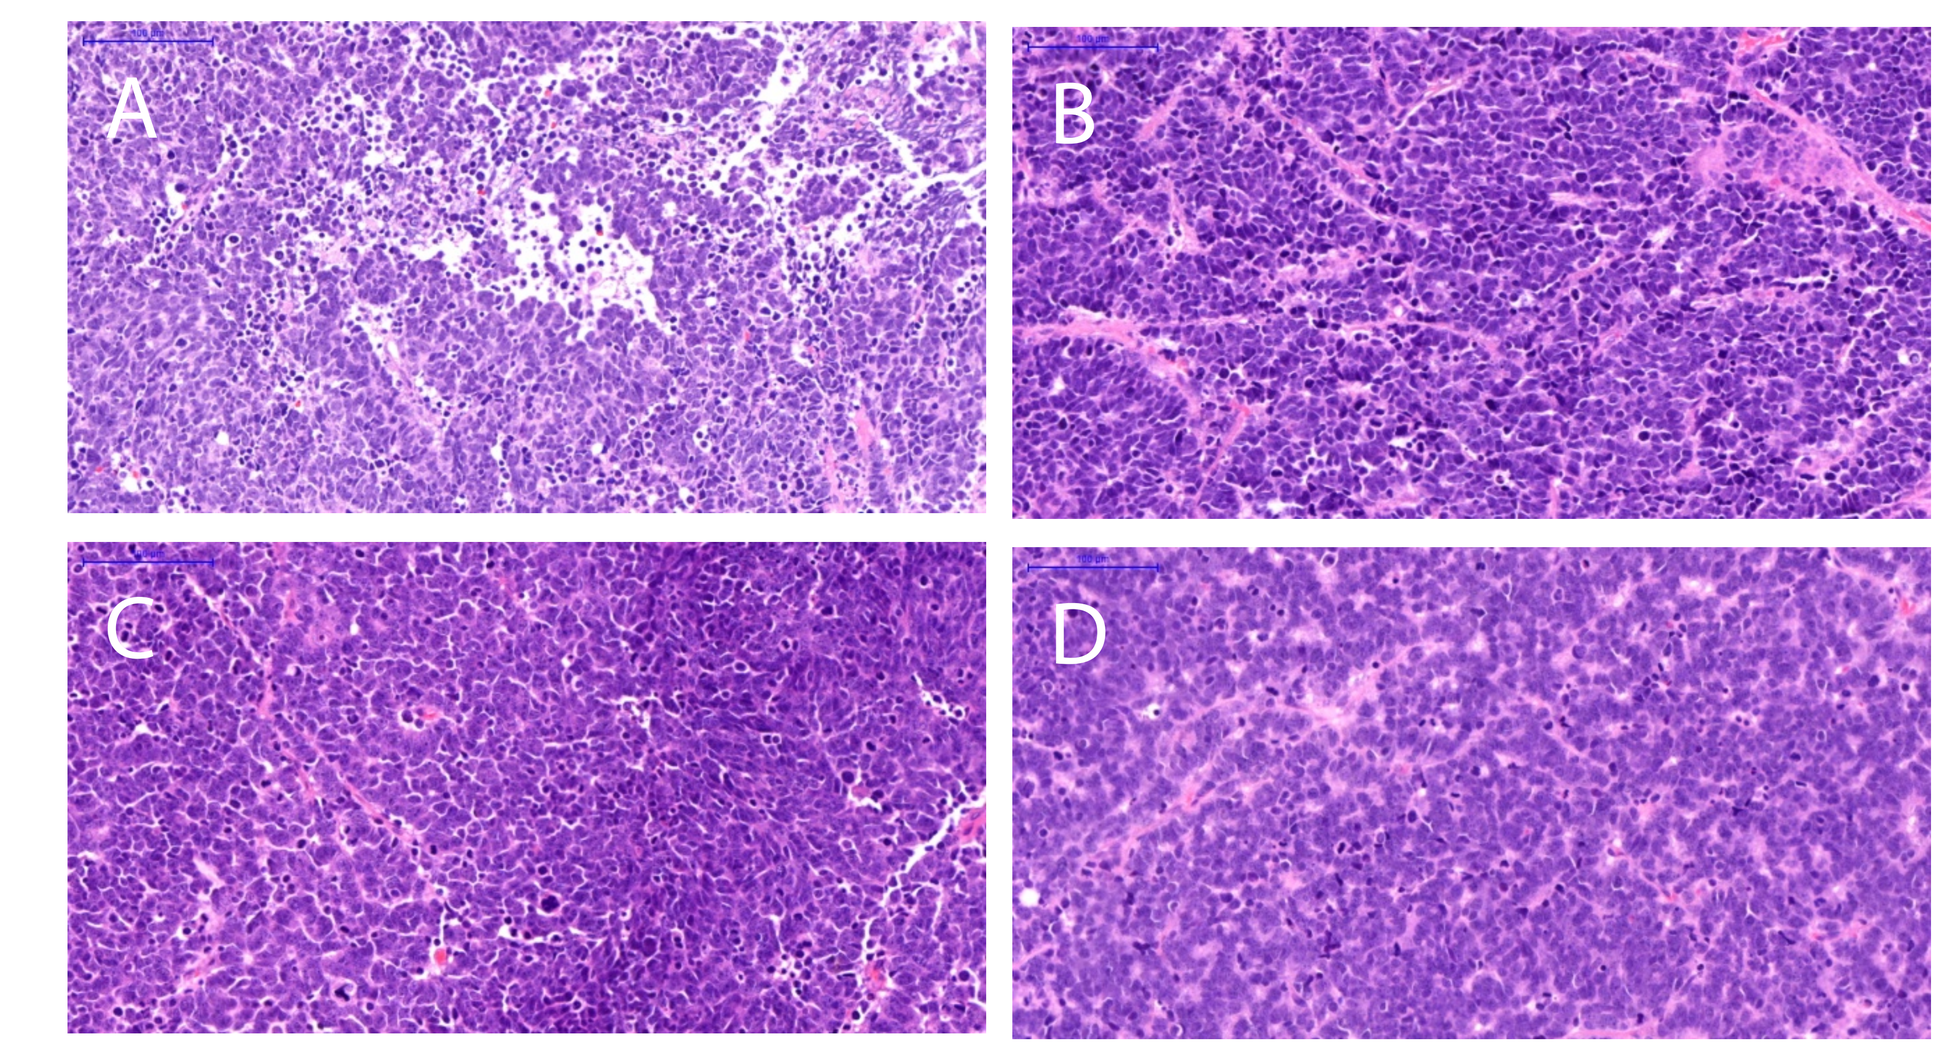
A
B
C
D

H&E Slides: (A) Original Patient Slide (B) P0 PDX Tumor (C) P1 PDX Tumor (D) P2 PDX Tumor. Each patient features unique SCL histologies which are maintained throughout PDX model development from patient through P2.

Ion AmpliSeq Cancer Hotspot Panel v2

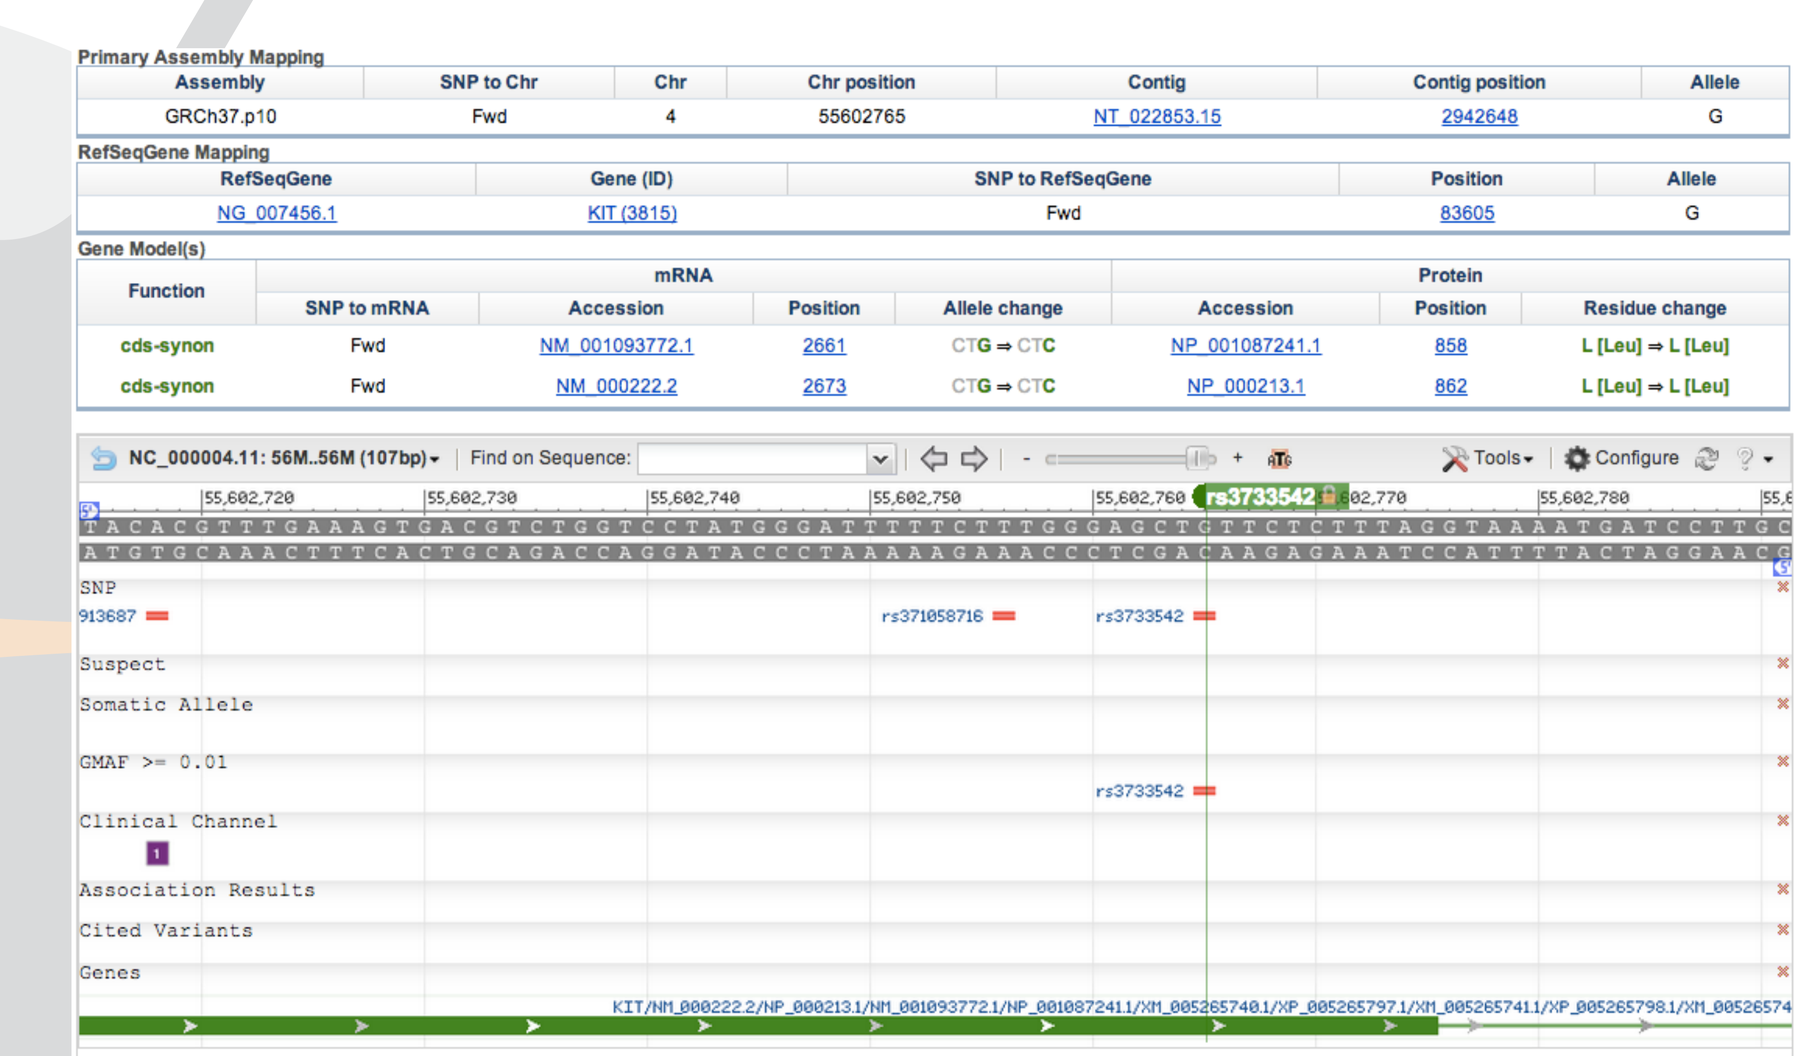

AmpliSeq Results:
All SCL samples were run on a panel targeting 50 oncogenes and tumor suppressor genes. LU65 showed the presence of the KIT gene and was used for further pharmacological testing.

in vivo Pharmacology

Study 1

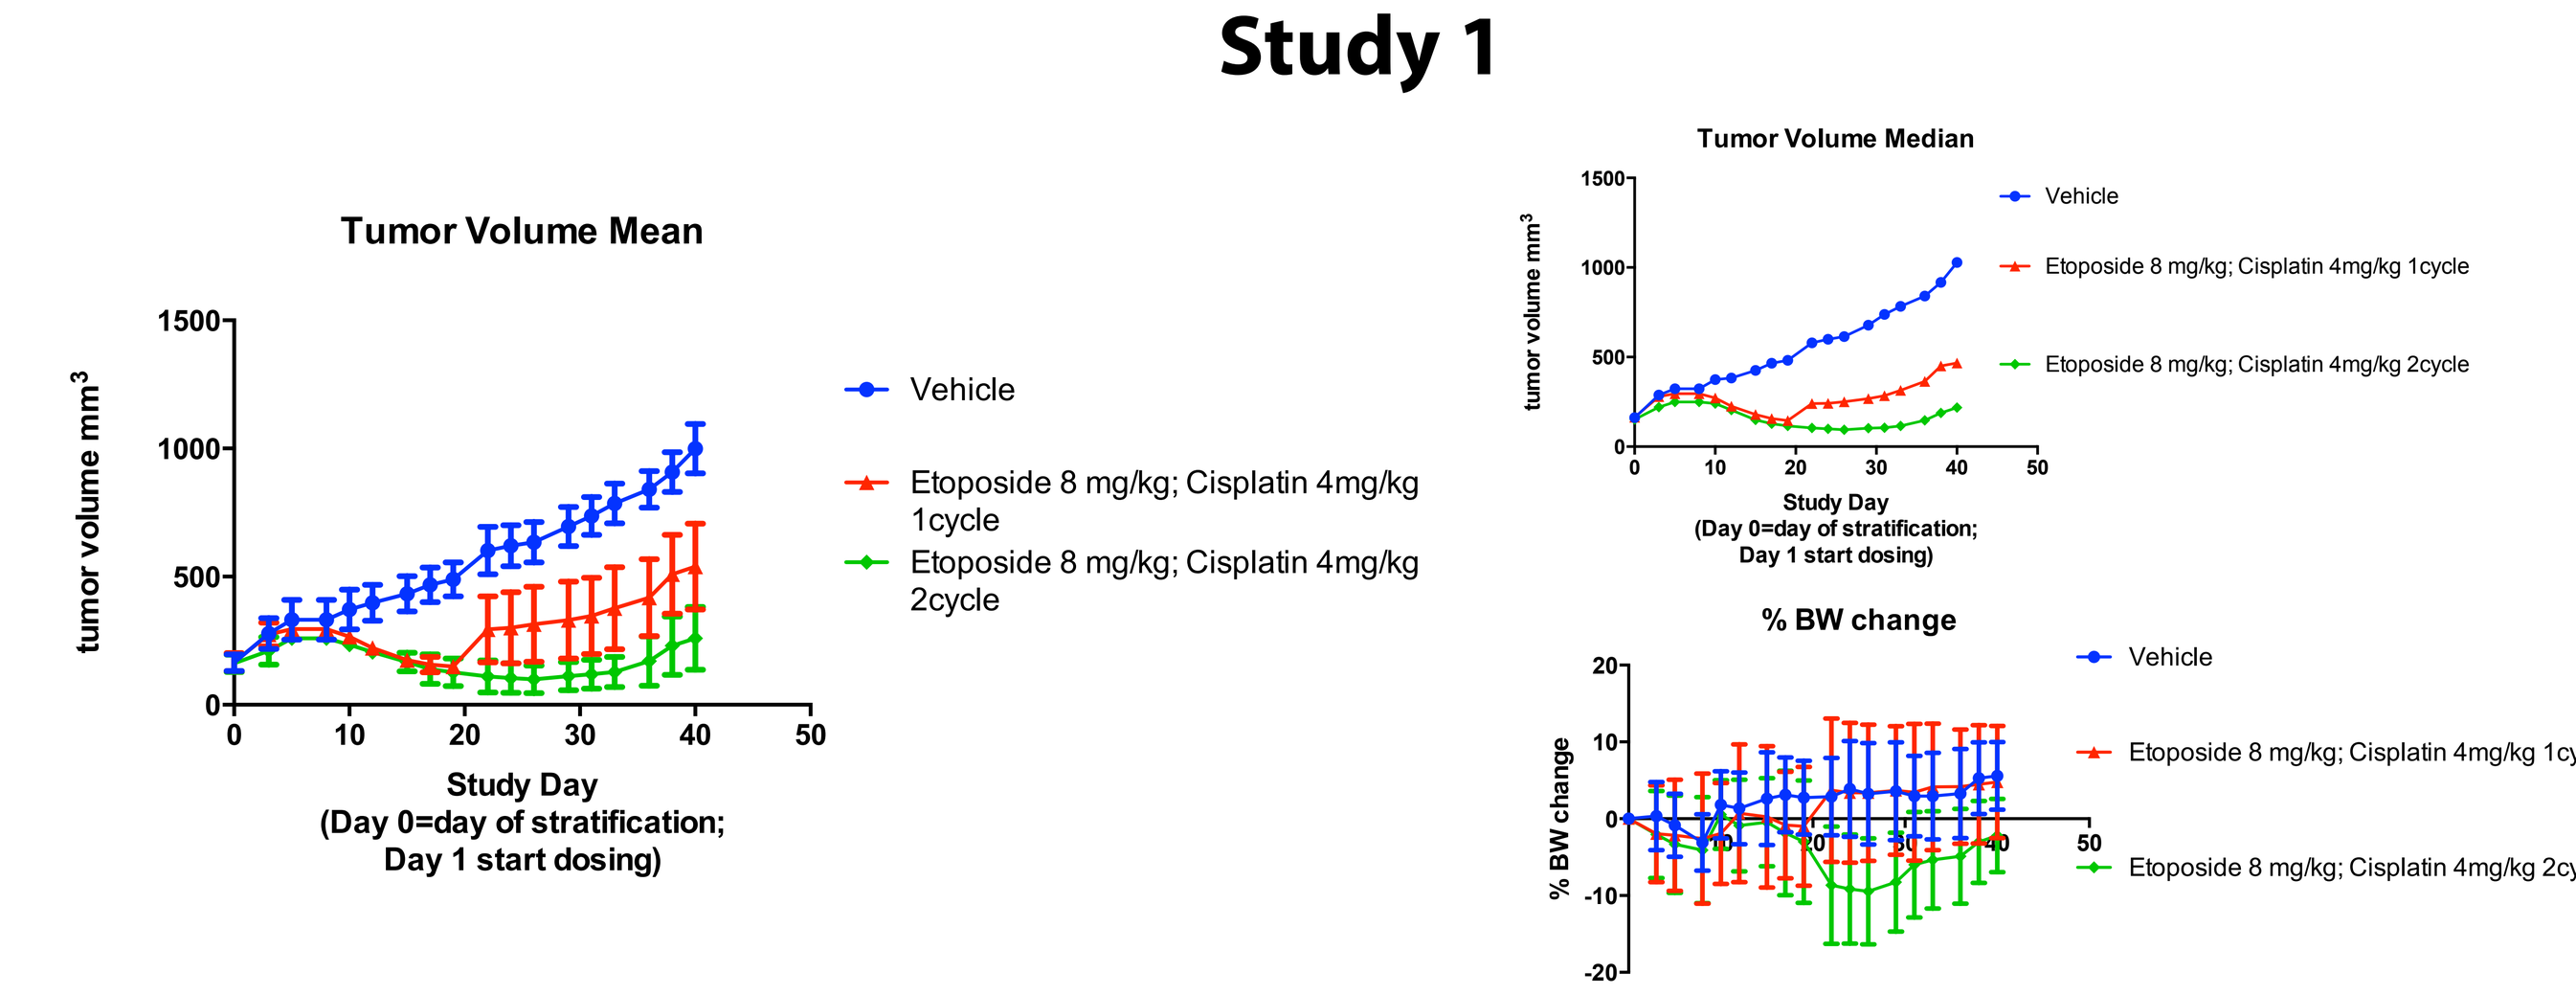
Tumor Volume Mean
Tumor Volume Median
% BW change
Vehicle
Etoposide 8 mg/kg; Cisplatin 4mg/kg 1cycle
Etoposide 8 mg/kg; Cisplatin 4mg/kg 2cycle
Study Day (Day 0=day of stratification; Day 1 start dosing)

in vivo Pharmacology

Study 2

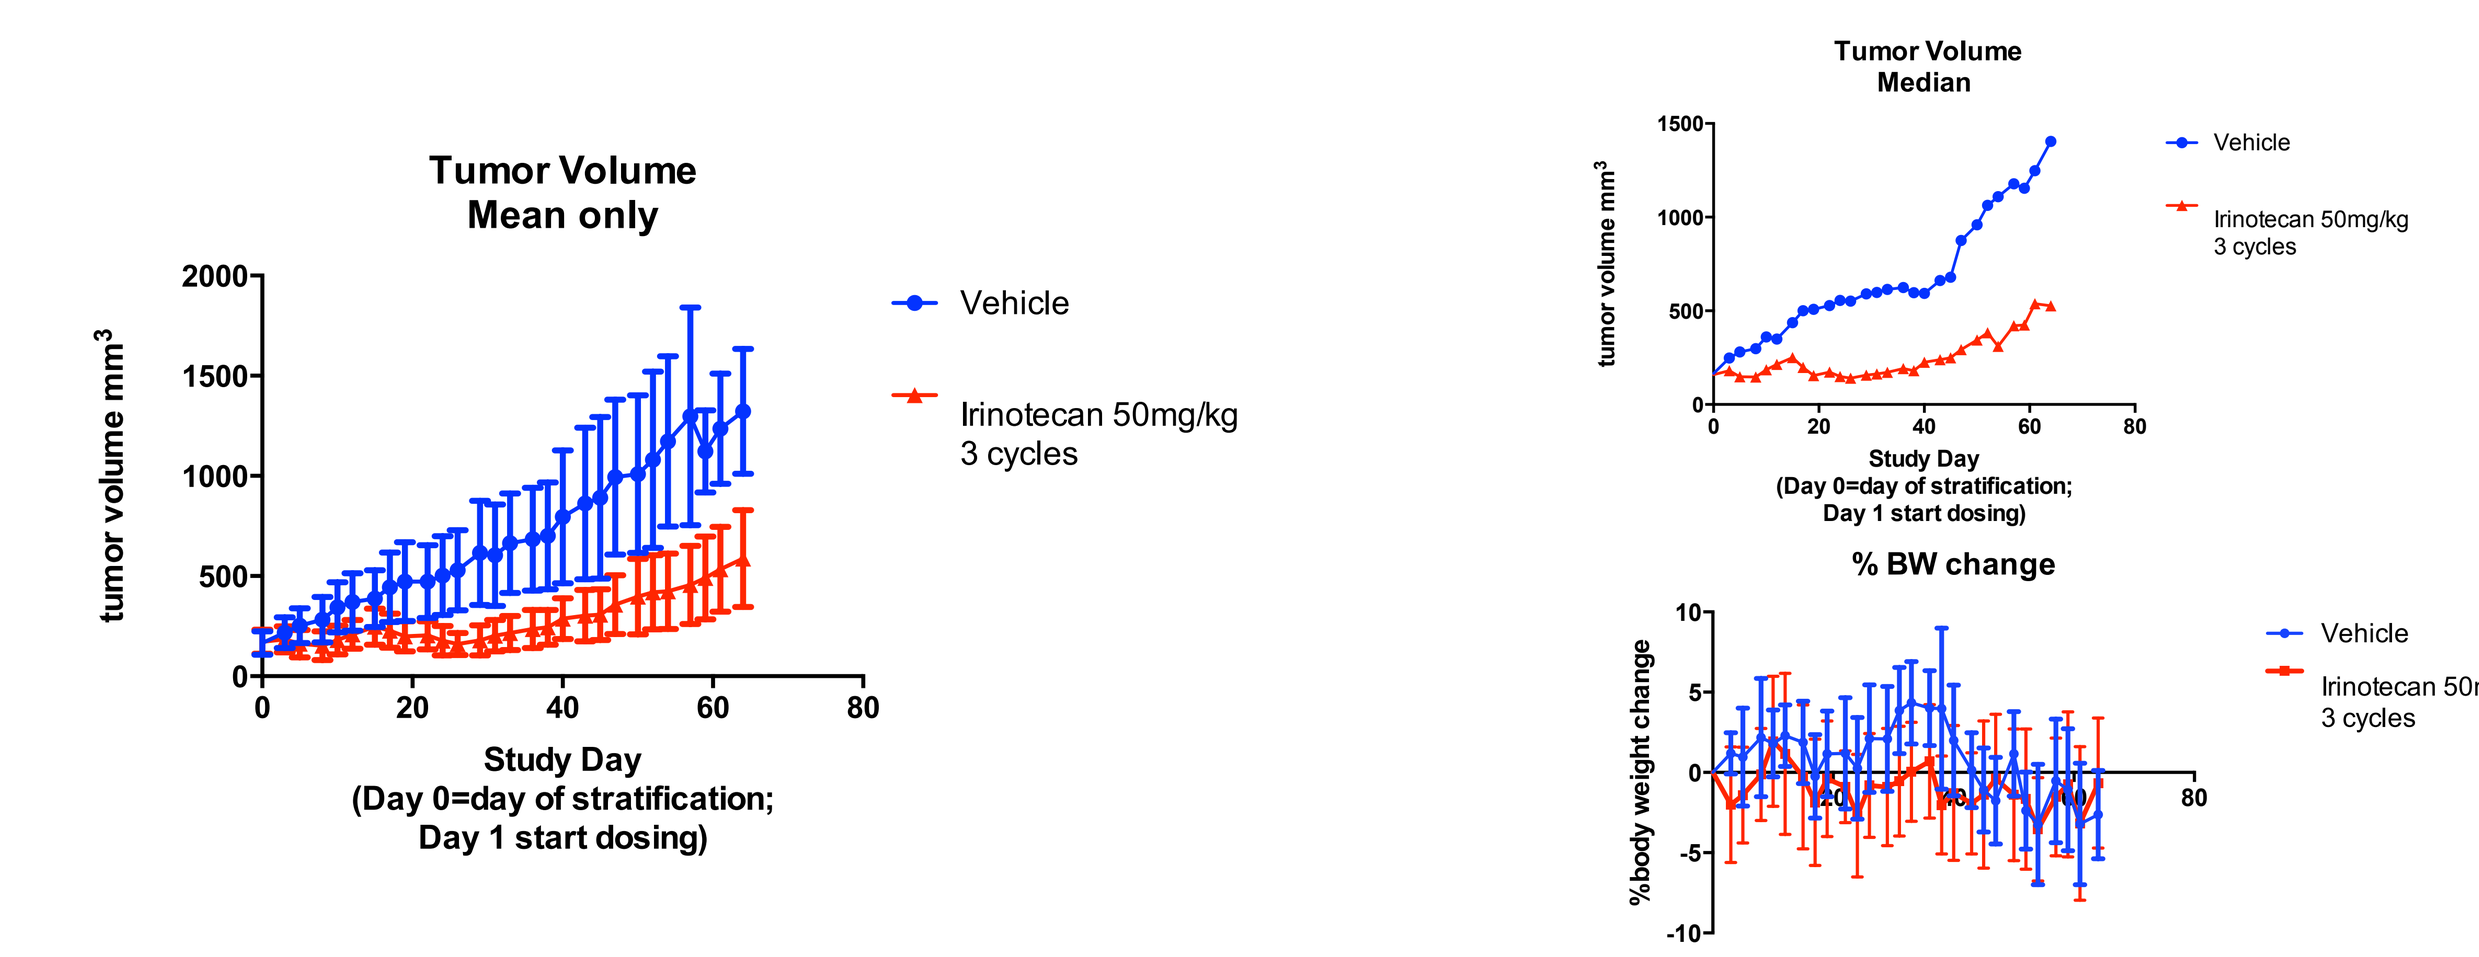
Tumor Volume Mean only
Tumor Volume Median
% BW change
Vehicle
Irinotecan 50mg/kg 3 cycles
Study Day (Day 0=day of stratification; Day 1 start dosing)

***In vivo* Pharmacology Studies:**

Study 1 : Female NOD-SCID mice were inoculated with LU65 and stratified into groups once the group average tumor volume reached between 161mm^3 – 166mm^3. Each group represents 8 mice: Vehicle group was dosed QDx5 for 4 weeks. Mice shown in red were dosed with etoposide on day 1, 2, and 3 and cisplatin on day 1. Mice shown in green were dosed with etoposide on day 1, 2, 3, 15, 16 , 17 and cisplatin on day 1 and 15 (2 cycles). Statistical analysis: Tukey's multiple comparisons test revealed significant efficacy of the test articles in inhibiting tumor growth compared to vehicle (not shown).

Study 2 : Female NOD-SCID mice were inoculated with LU65 and stratified into groups once the group average tumor volume reached between 165mm^3 –173mm^3. Each group represents 8 mice: Vehicle group was dosed QDx5 for 4 weeks, irinotecan group was dosed Q7Dx3. Statistical analysis: Tukey's multiple comparisons test revealed significant efficacy of the test article in inhibiting tumor growth compared to vehicle (not shown).

Conclusions

- Our novel small cell lung cancer models retained their unique histology over passage.
- Patient information concerning prior drug treatment as well as sequencing information looking at presence of oncogenes and tumor suppressor genes provide valuable tools towards choosing the right model for drug development.
- Our SCL models show significant drug response in vitro and in vivo.