In vivo response and molecular characterization of a Caucasian NSCLC squamous cell carcinoma PDX sensitive to FGFR inhibitors

Andrew McKenzie[1], Nektaria Papadopoulou[1], Yinfei Yin[1], Simon Jiang[1], Jane Wrigley[1], Jie Cai[2], Martin Page[1], Henry Li[2] and Rajendra Kumari[1]

[1]Crown Bioscience UK Ltd, Loughborough, UK; [2]Crown Bioscience, Santa Clara, USA

In life for life

Abstract: 769

In life for life

Background

- The fibroblast growth factor receptor (FGFR) family are reported to be involved in key cellular processes such as proliferation, differentiation, migration & survival with the deregulation of signalling through genetic modification or amplification being observed in cancer.
- Non-small cell lung cancer (NSCLC) accounts for 85% of lung cancer cases, carries a poor prognosis and remains an area of high unmet need. Despite recent advances in treating NSCLC adenocarcinomas with targeted agents such as Erlotinib (Tarceva©) few advances have been made in the treatment of squamous cell carcinoma (SCC), which accounts for 25% of all NSCLC.
- FGFR1 has been reported to be amplified in squamous cell NSCLC as well as other indications (e.g. breast). Although the incidence of FGFR mutations in SCC is modest, amplification of FGFR is well documented and there are several first generation FGFR inhibitors (FGFRi) currently in clinical trial e.g. AZD4547.
- It is therefore essential to develop relevant *in vivo* models for the development and characterisation of new FGFR inhibitors and/ or combination strategies which may prolong benefit and delay the emergence of resistance.
- Patient-derived xenograft (PDX) models offer a more clinically relevant heterogeneous microenvironment for evaluating targeted agents and potential identification of biomarkers as well as establishment of Avatar trials.

Methods

- ***In vivo* xenograft studies**: Caucasian NSCLC PDX models, part of our HuPrime® platform (Table 1), are maintained subcutaneously *in vivo* in nude mice (HsdOla:MF1-*Foxn1*^{nu}) admixed with a human stromal cell component (bone marrow-derived human mesenchymal stem cells, ScienCell). Mice were dosed with AZD4547 (p.o. q.d.) and/or chemotherapy (CTx; i.p. q2w) alone, or in combination (parallel or serial dosing regimens). Tumour measurements and body weights were taken 3 times weekly and dosing initiated when the tumours reached a mean volume of ~200mm^3 unless otherwise stated.
- **Molecular Characterisation**: Tumour material was characterised for key mutations (EGFR, kras, p53) by PCR. FGFR1-3 genomic amplification by quantitative PCR. FGFR1-2 genomic amplification by FISH analysis. RNA sequencing was also carried out on some models.

Table 1: Summary of characterisation of Caucasian NSCLC PDX models

HuBase ID	LION number	Sub-type	Known Mutations	Amplifications	FGFR inhibitor	EGFR inhibitor	CTx response
LU6408	42	SCC		FGFR1	Stable disease lost with passgae	Progressive disease	Partial response
LU6426	116	SCC	p53	FGFR1 & FGFR2	Partial response	ND	Partial response
LU6429	137	SCC	p53	FGFR1 & FGFR2	Stable disease	ND	Complete response
LU6431	182	SCC		FGFR2 slightly amplified	Heterogeneic response	ND	ND
LU6438	221	SCC		FGFR2 slightly amplified	Heterogeneic response	ND	ND

In vivo characterisation of LU6429

LU6429 is a novel Caucasian NSCLC (SCC subtype from male ex-smoker) PDX model which is maintained in serial passage subcutaneously (s.c.) *in vivo* admixed with a human stromal cell component. Significant reduction in tumour growth resulting in stabilisation of tumour growth is seen following treatment with 25mg/kg AZD4547 p.o. q.d. The response was reproducible over different passages (p8 and p12 shown below).

LU6429 *in vivo* response to AZD4547 at passage 8 (left) and passage 12 (right)

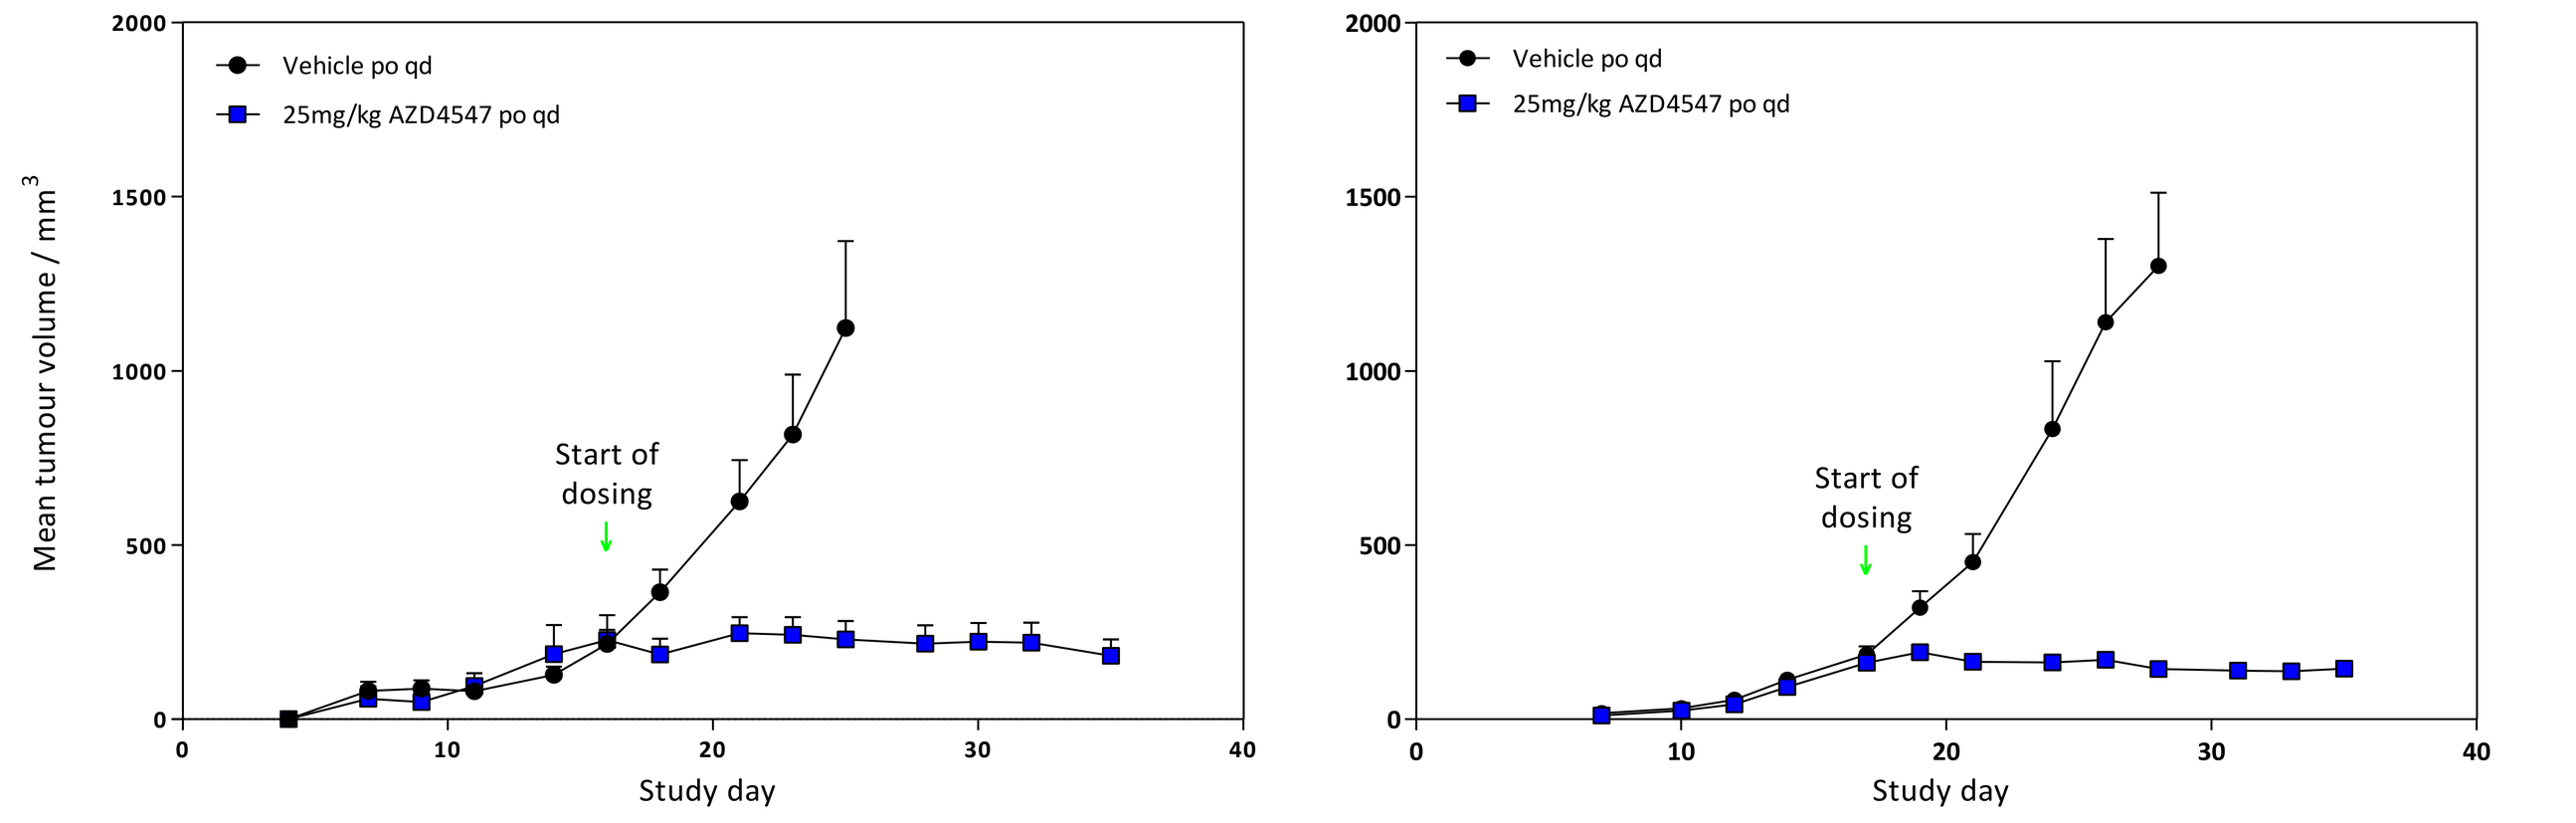

Vehicle po qd
25mg/kg AZD4547 po qd
Start of dosing
Mean tumour volume / mm3
Study day

LU6429 tumours with a volume of >1000mm^3 also responded to treatment with AZD4547 resulting in partial regression of growth (a). Resuscitation, growth and subsequent response to 25mg/kg AZD4547 p.o. q.d. following cryopreservation of LU6429 passage 7 (P7) PDX material at -196ºC is shown in the figure below (b).

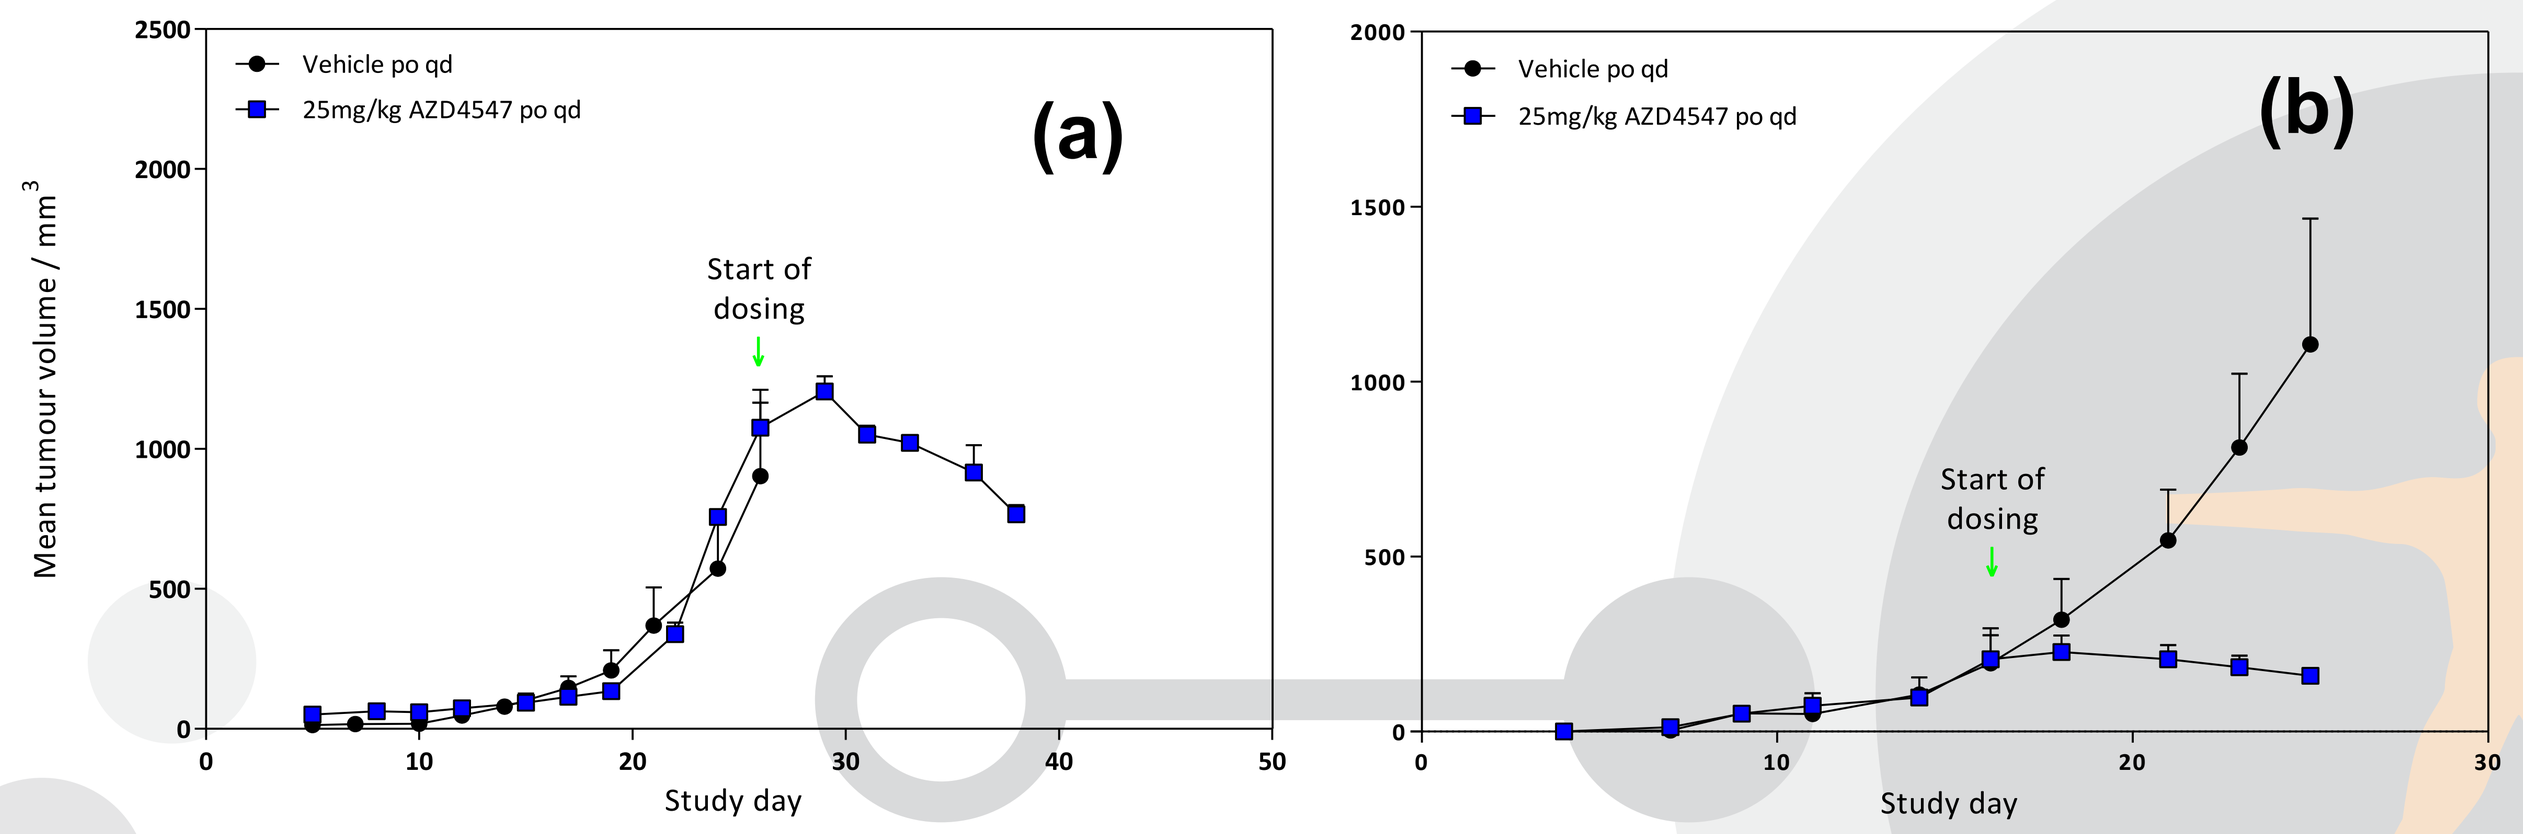

Vehicle po qd
25mg/kg AZD4547 po qd
(a)
(b)
Start of dosing
Mean tumour volume / mm3
Study day

Regression with standard of care CTx

LU6429 is responsive to treatment with paclitaxel/carboplatin doublet chemotherapy (CTx) cycled at 5/50mg/kg i.p. (↑) every 2 weeks which is also reproducible over different passages (a; Left hand side of graph below).

LU6429 is responsive following partial regression with paclitaxel/ carboplatin doublet chemotherapy treatment (5/50mg/kg i.p. q2w, ↑) LU6429 is still responsive to treatment to 25mg/kg AZD4547 p.o. q.d. (↑) (b; Right hand side of graph below) This setting is highly relevant to the likely clinical presentation in which an FGFRi will be tested.

***In vivo* response to serial CTx and FGFRi dosing**

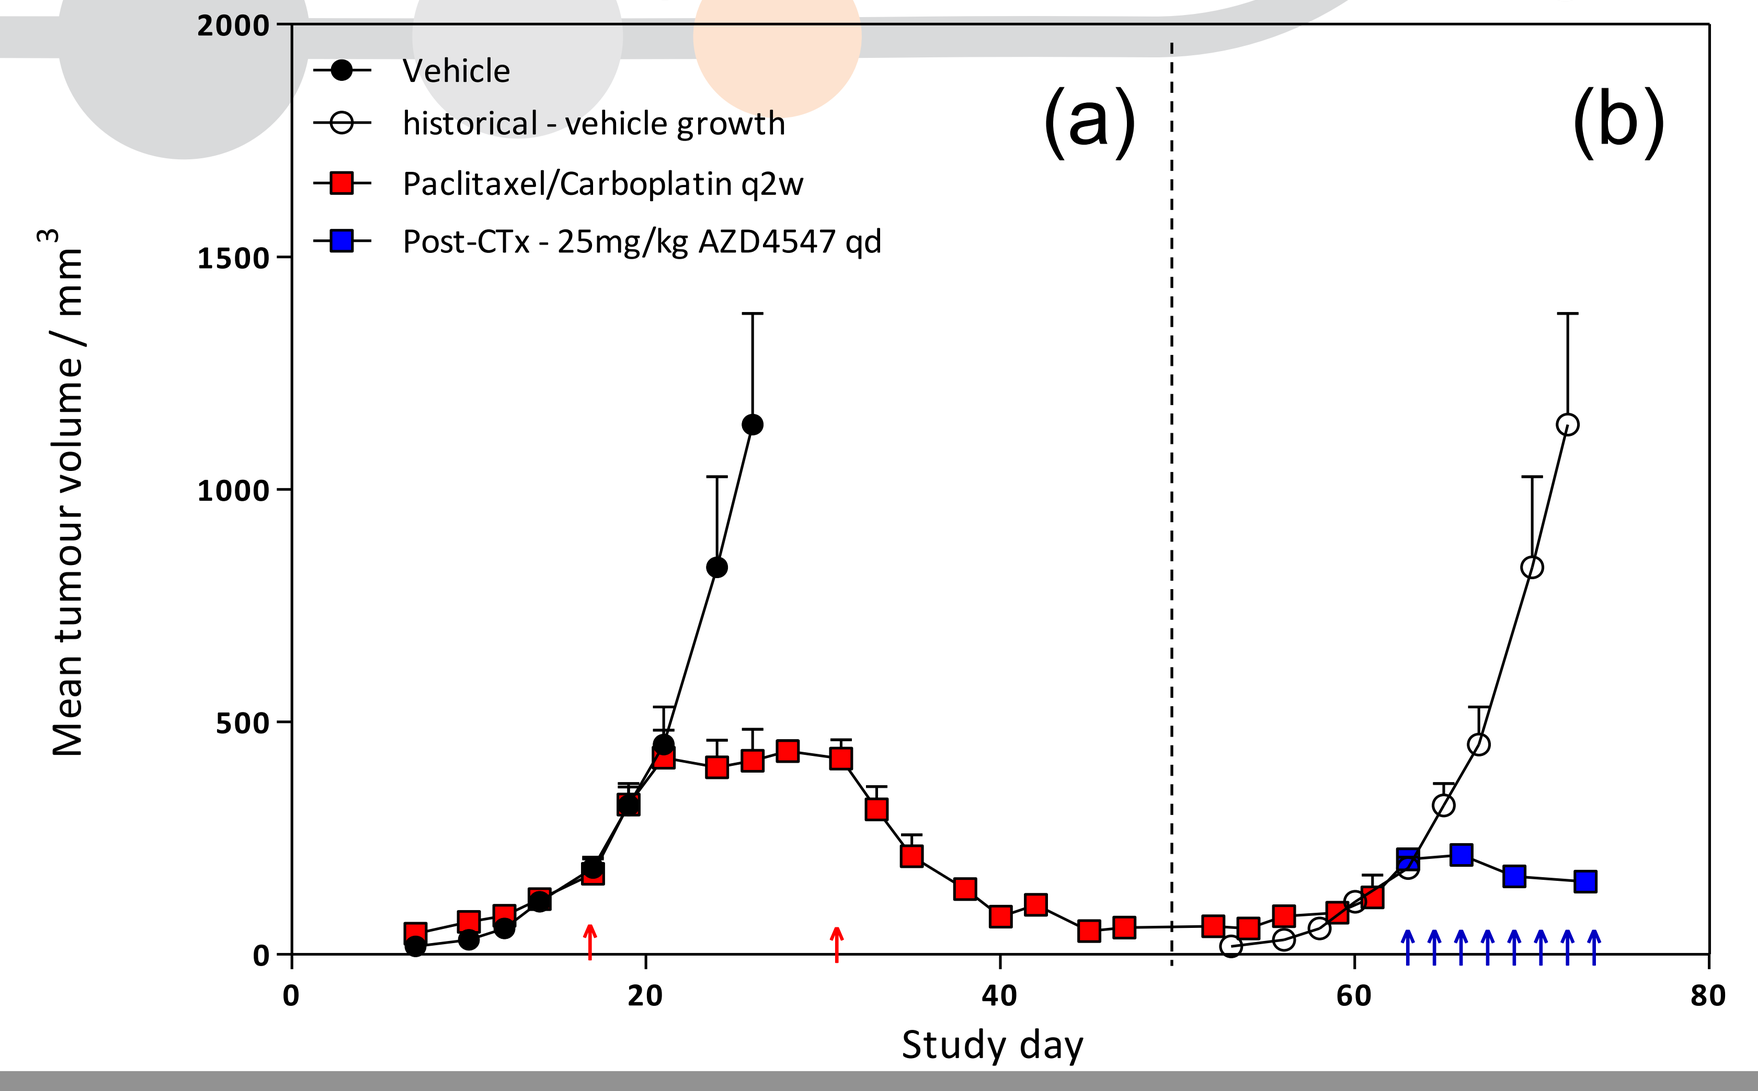

Vehicle
historical - vehicle growth
Paclitaxel/Carboplatin q2w
Post-CTx - 25mg/kg AZD4547 qd
(a)
(b)
Mean tumour volume / mm3
Study day

FGFRi and CTx combination therapy

Parallel combination dosing with 25mg/kg AZD4547 (↑) p.o. q.d. and paclitaxel/carboplatin doublet therapy treatment (5/50mg/kg i.p. q2w, ↑) is shown below. Combination dosing appears to offer no significant advantage, in reduction of tumour volume, over single agent therapy. Tumour regrowth was significantly delayed for the combination group versus the AZD4547 group alone but not compared to the CTx treatment.

***In vivo* response to combination dosing of AZD4547 + CTx**

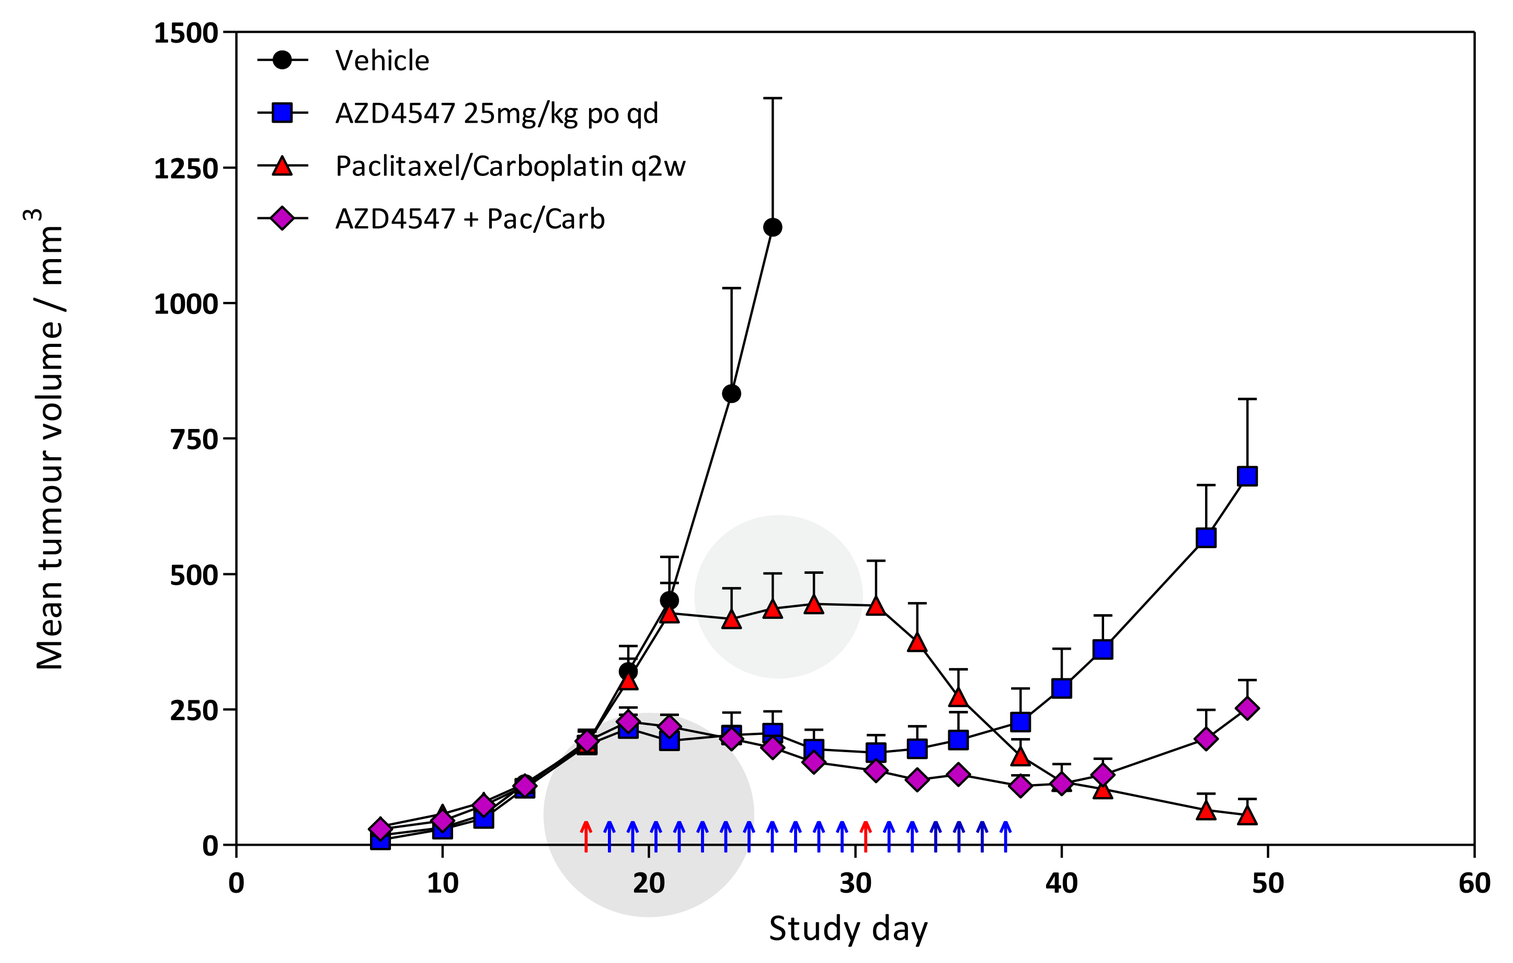

Vehicle
AZD4547 25mg/kg po qd
Paclitaxel/Carboplatin q2w
AZD4547 + Pac/Carb
Mean tumour volume / mm3
Study day

Refinement of the dosing regimens could improve the combination efficacy. Models such as LU6429 are therefore valuable in predicting the different outcomes that may arise in the clinic in response to targeted agents.

Molecular Characterisation of LU6429

Sequencing analysis with Ion TorrentTM sequencing (Ion Ampliseq™ Cancer Hotspot Panel v2) has identified the following mutations:

- p53 homozygous mutations (R117P, R156P, R24P)
- PI3KCA (M1040I) heterozygous mutation
- KDR (Q472H) heterozygous mutation

LU6429 is wild-type for FGFR1, FGFR2, KRAS, EGFR, KRAS & LKB1.

The expression of FGFR1, FGFR2 and FGFR3 (figures a-c respectively) in LU6429 has been confirmed by Taqman copy number analysis 2-3 fold relevant to housekeeping gene Rnase P and FISH analysis (d).

FGFR1, FGFR2 and FGFR3 copy number expression *in vivo* LU6429 samples pre and post cryopreservation/resuscitation and at late passage

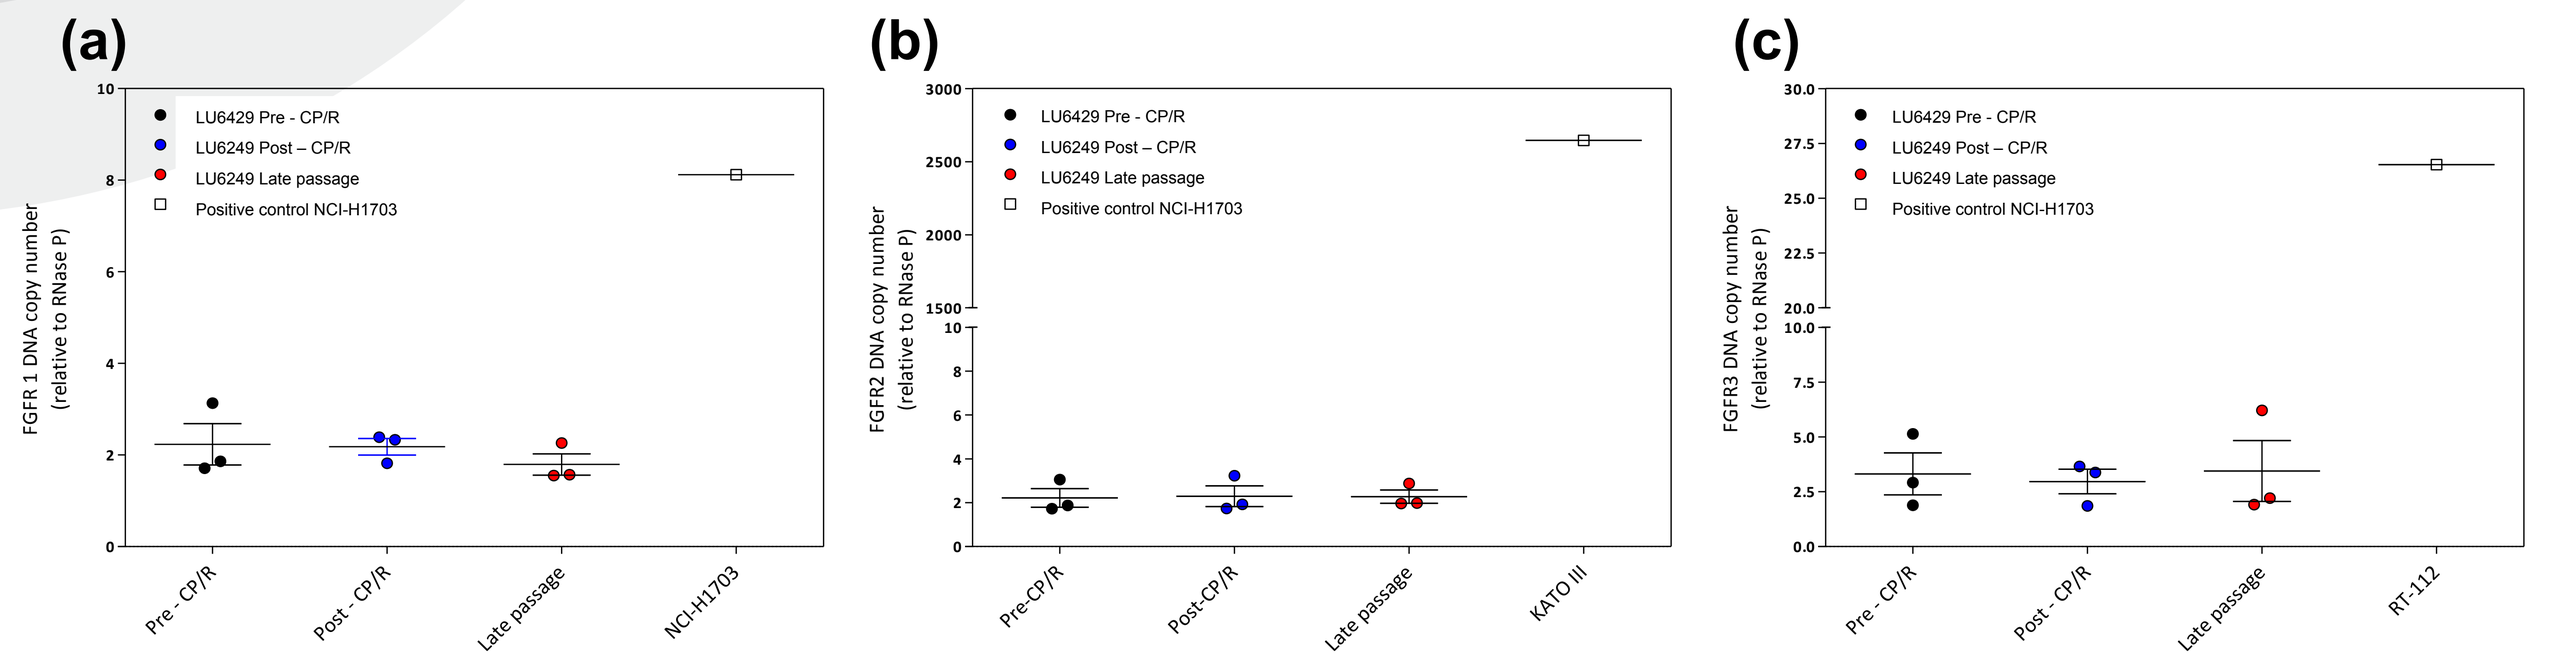

(a)
(b)
(c)
LU6429 Pre - CP/R
LU6249 Post – CP/R
LU6249 Late passage
Positive control NCI-H1703

(d) FISH copy number analysis shows amplification of FGFR1.

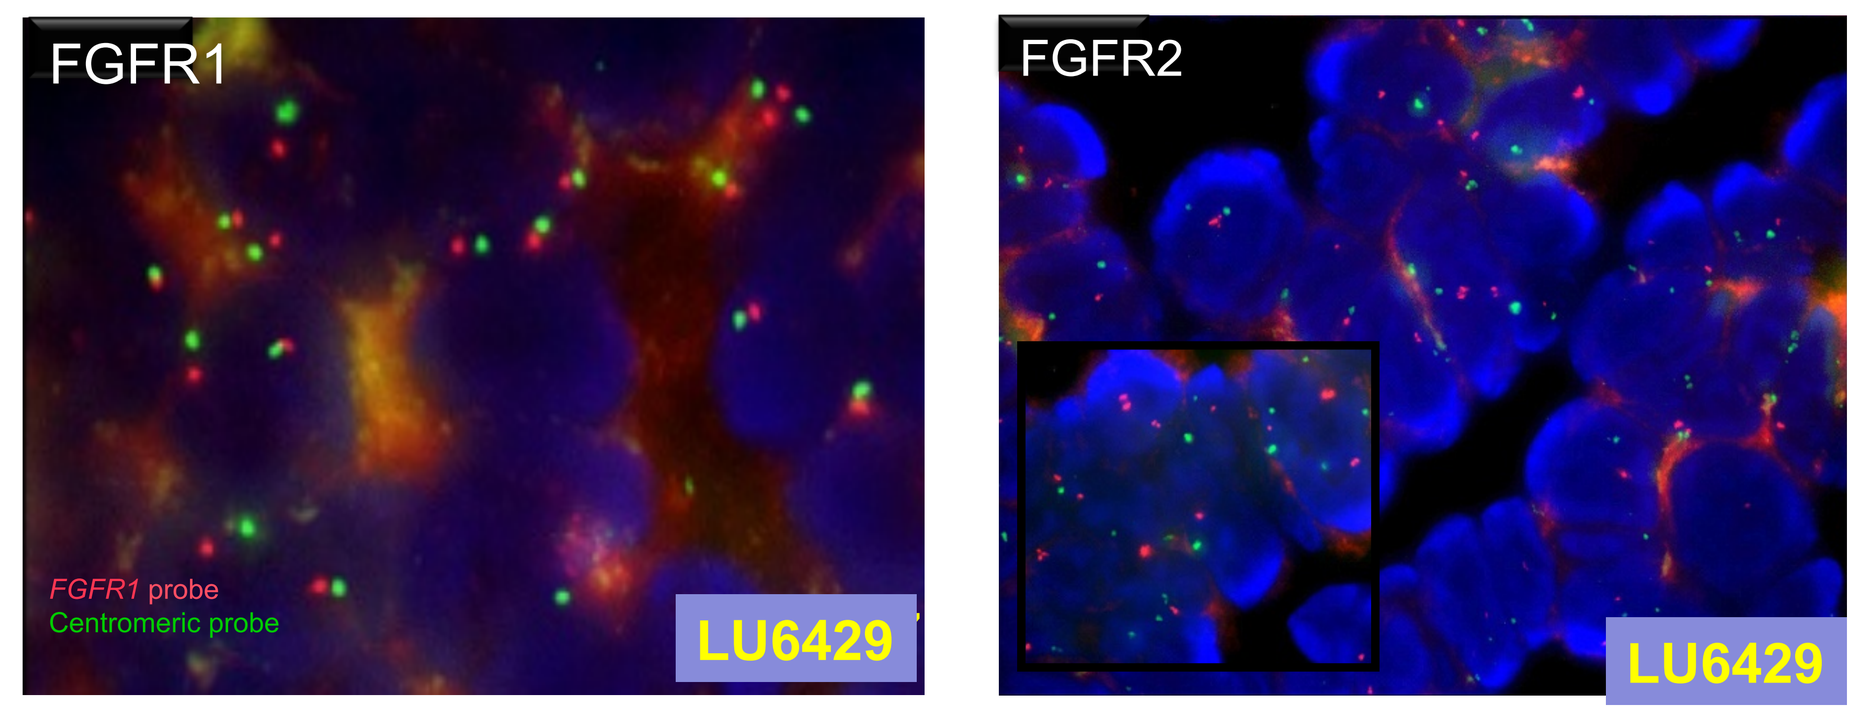

FGFR1
FGFR1 probe
Centromeric probe
LU6429
FGFR2
LU6429

*Contact information: Dr. Rajendra Kumari, rajendra.kumari@crownbio.com

FGFRi response in CTx resistant variant

To generate resistance to CTx mice were dosed with paclitaxel/ carboplatin doublet chemotherapy (CTx; 5/50mg/kg i.p q2w or qw) for up to 7 cycles, followed by outgrowth and re-passage of refractory tissue + hMSC into donor mice; this process was repeated several times before the emergence of resistance. Chemo-resistant LU6429 tumours were challenged with 25mg/kg AZD4547 p.o. q.d. following CTx treatment.

***In vivo* response to AZD4547 following generation of resistance to CTx**

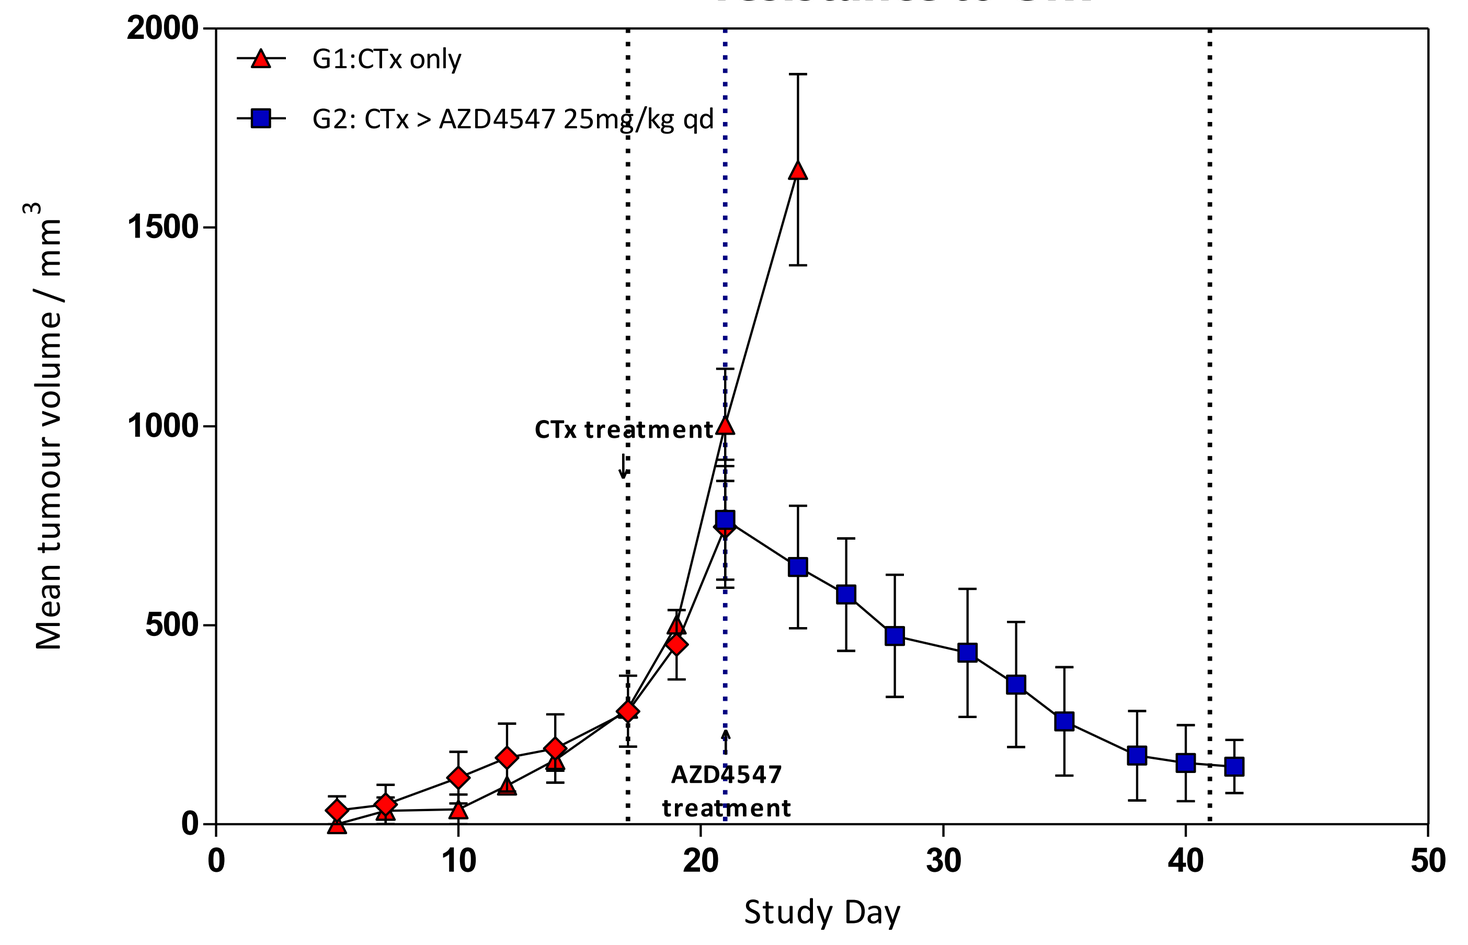

G1:CTx only
G2: CTx > AZD4547 25mg/kg qd
CTx treatment
AZD4547 treatment
Mean tumour volume / mm3
Study Day

LU6429 did not respond to CTx treatment, but did respond to AZD4547 resulting in regression of growth.
Models such as LU6249 are therefore valuable in predicting the different outcomes that may arise in the clinic in resistant or relapsed patients.

FGFRi resistance

Resistance to AZD4547 was established through continuous cycling of dosing and outgrowth until emergence of resistance appeared (figure a below). However, when grafted on into new cohorts of mice + hMSCs and dosed with AZD4547 in non-established/established regimens, AZD4547 sensitivity was retained suggesting resistance to AZD4547 was not stable in this model.

a) LU6429 outgrown under dosing pressure – AZD454 12.5mg/kg p.o. q.d.

b) Comparison of treatment of established & non-established tumours passaged from AZD4547 refractory tumours.

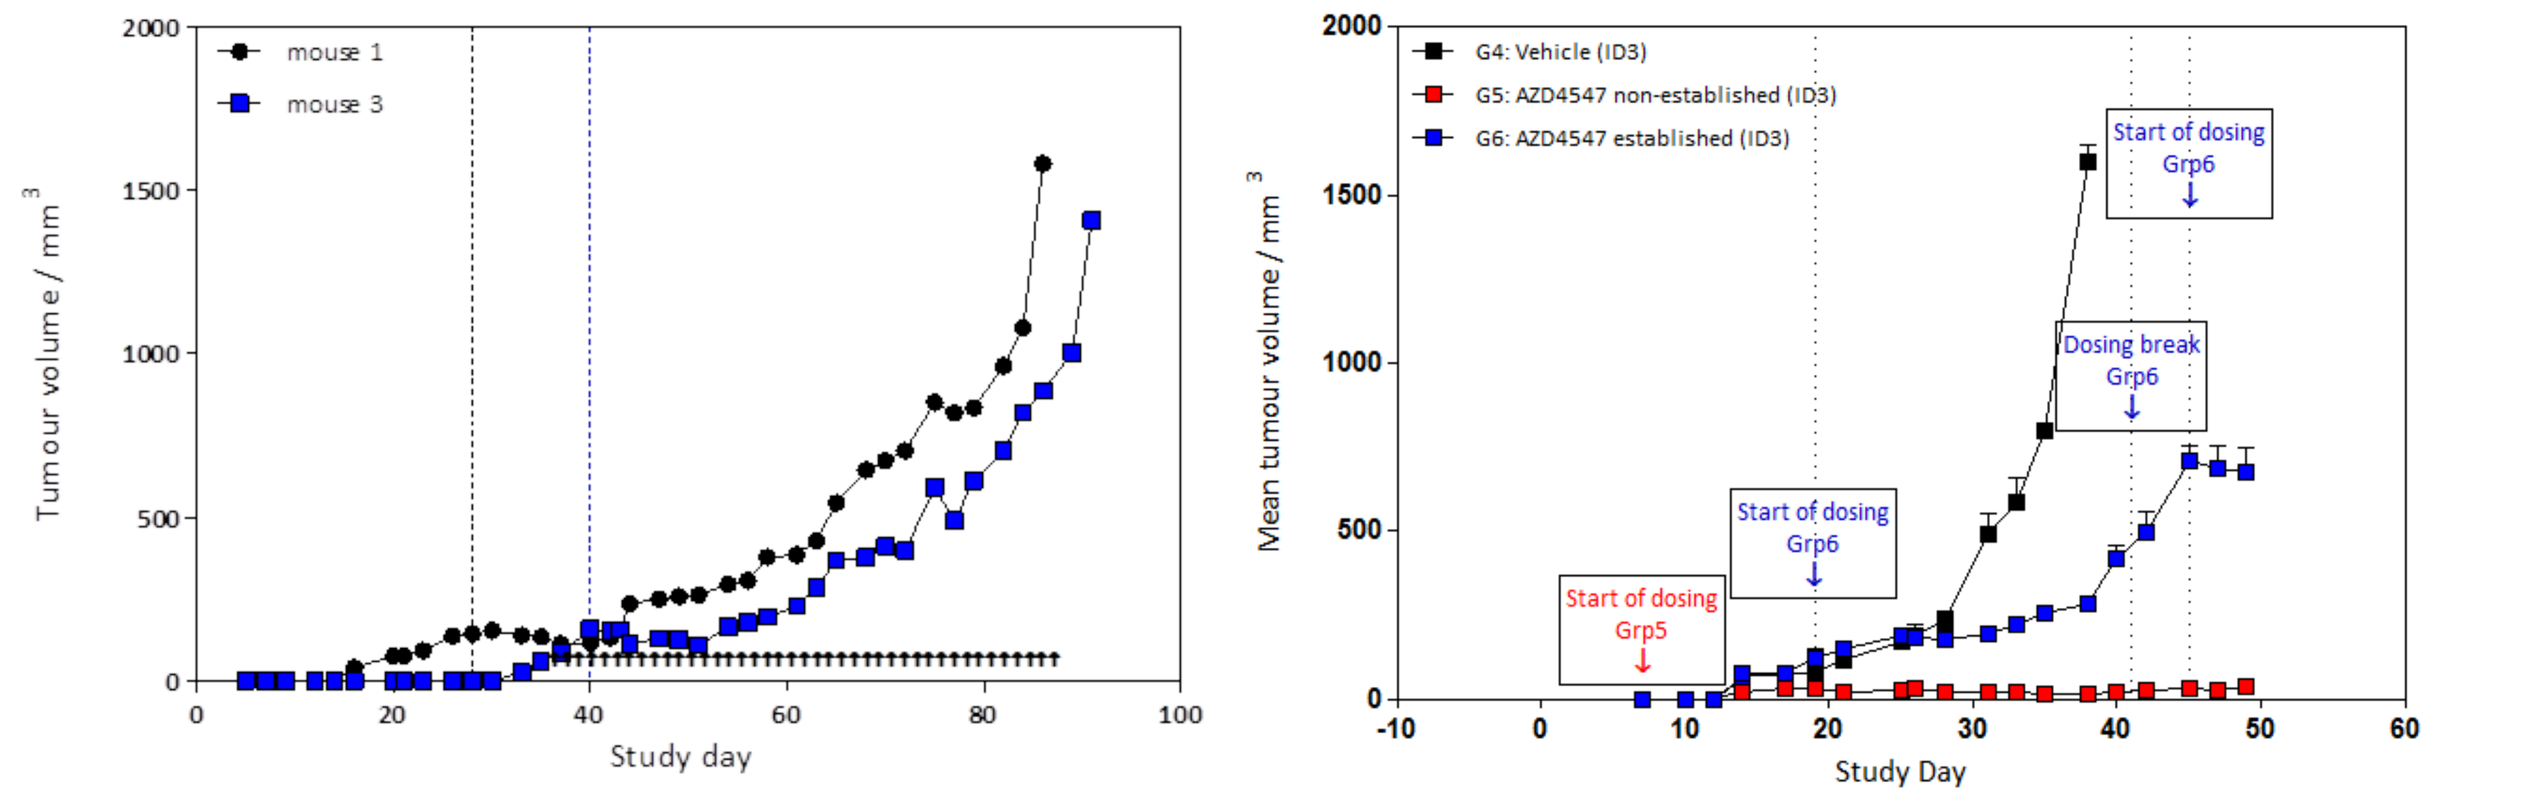

mouse 1
mouse 3
Tumour volume / mm3
Study day
G4: Vehicle (ID3)
G5: AZD4547 non-established (ID3)
G6: AZD4547 established (ID3)
Start of dosing Grp5
Start of dosing Grp6
Dosing break Grp6
Start of dosing Grp6
Mean tumour volume / mm3
Study Day

Conclusion

- Heterogeneity is represented by NSCLC PDX models in vivo and provides unique models for preclinical evaluation of both response and resistance.
- The LU6429 SCC PDX model showed reproducible FGFRi-sensitivity and moderate FGFR1-3 copy number expression.
- This model is invaluable in assessing novel agents targeting the FGFR pathway and in the development of new combination strategies which may prevent or overcome resistance
- In addition, models of acquired resistance to both CTx and FGFR inhibitors can offer substantial insight into understanding mechanisms of resistance in the clinic as well as assessing new agents and combination strategies.