Poster : #90

High Efficacy of a Novel and Specific c-MET TKI in a Cohort of PDXs from Selected NSCLC and HCC Patients

Dawei Chen[1], Mengmeng Yang[1], Boyu Zhong[1], Deyi Zhang[1], Zhenjian Du[1], Juan Zhang[1], Guizhong Liu[1], Qian Shi[1], Jie Cai[1], Davy Xuesong Ouyang[1], Jiping Zha[1], Joe Shih[1], Jean Pierre Wery[1], Henry Q.X. Li[1, 2*]

[1]Crown Bioscience Inc., 3375 Scott Blvd., Suite 108, Santa Clara, CA 95054, USA; [2]State Key Laboratory of Natural and Biomimetic Drugs, Peking University, 38 Xueyuan Road, Beijing 100191, China

Introduction

Turning on c-Met signaling, through gene amplification, activating mutations or interaction with its only known high affinity ligand - hepatocyte growth factor (HGF), is associated with invasive progression of many types of cancers, such as papillary renal, gastric, hepatocellular (HCC) and non-small cell lung carcinoma (NSCLC). The use of antibodies to block HGF/c-MET binding, or small molecule tyrosine kinase inhibitors (TKI) against c-MET has shown to be a promising therapeutic approach for certain tumor types. Several c-MET inhibitors have been developed and have been extensively tested in the clinic. However, they have met only limited success, either because of associated side effects or for lack of an appropriate clinical strategy. Therefore, improving the pharmacological properties of c-MET TKI and testing them in the right patient population become critical to the success of any new c-MET TKI.

Results

Enzyme-Inhibitory Activity
(TR-FRET assay)

Compound ID	IC50 (nM)
M101	31.6
M102	50.7

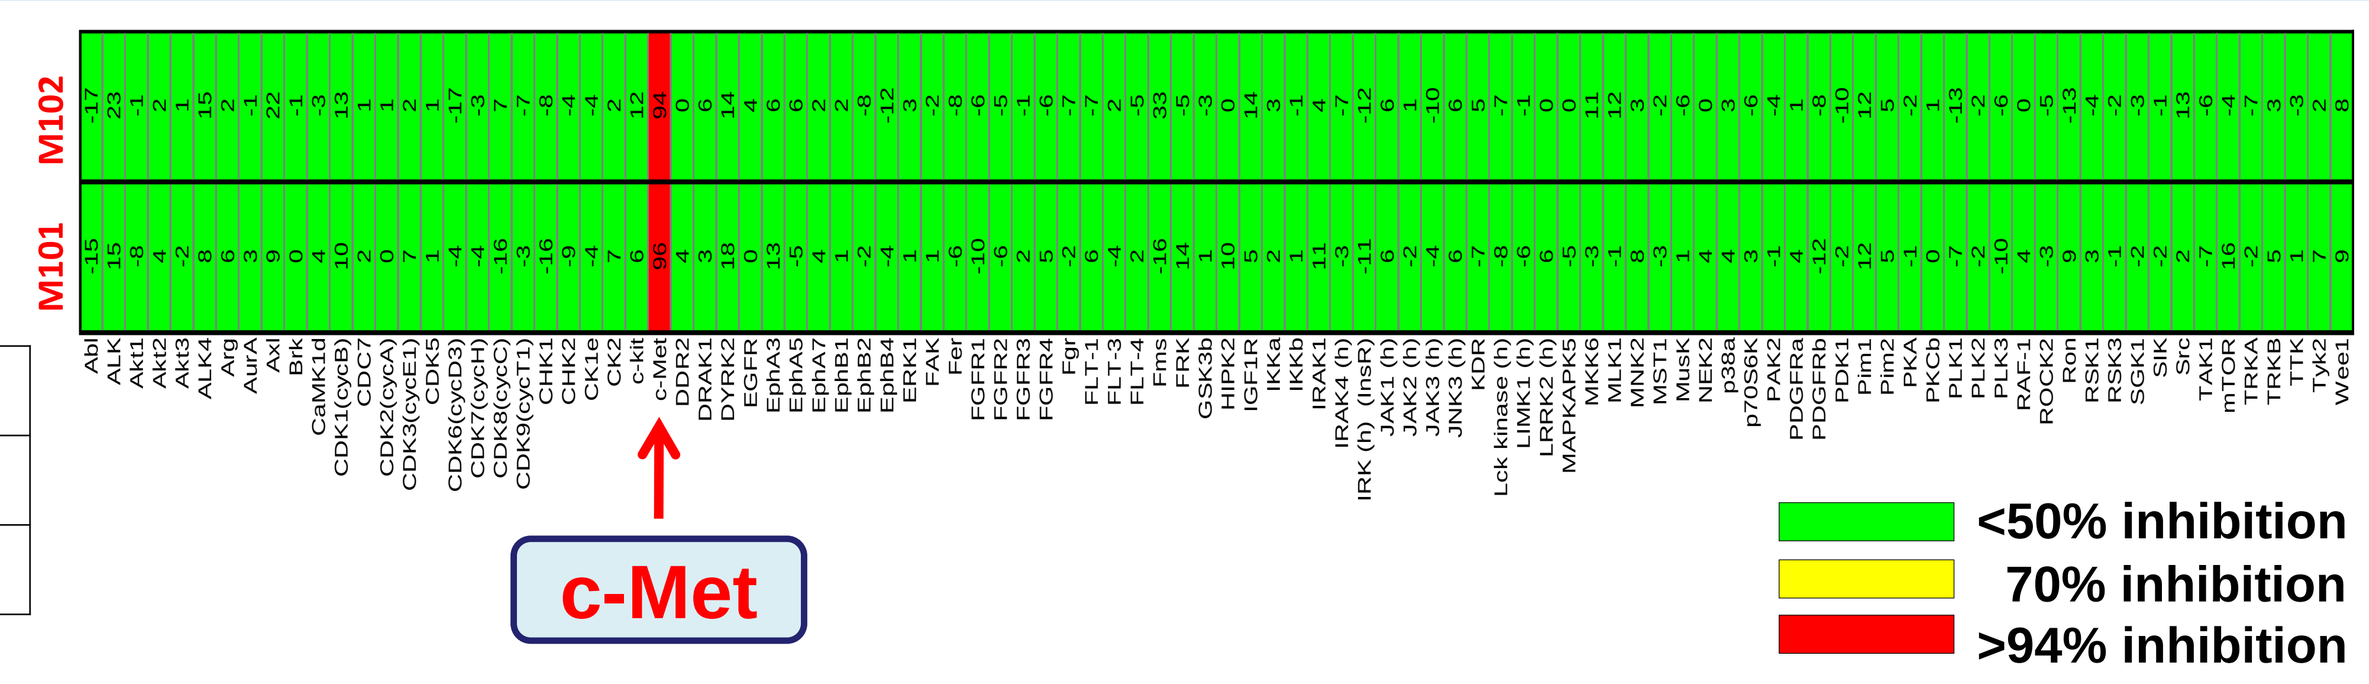
M102
M101
c-Met
<50% inhibition
70% inhibition
>94% inhibition

Figure 1. Enzymatic assay of M101 & M102. c-MET specific activity.

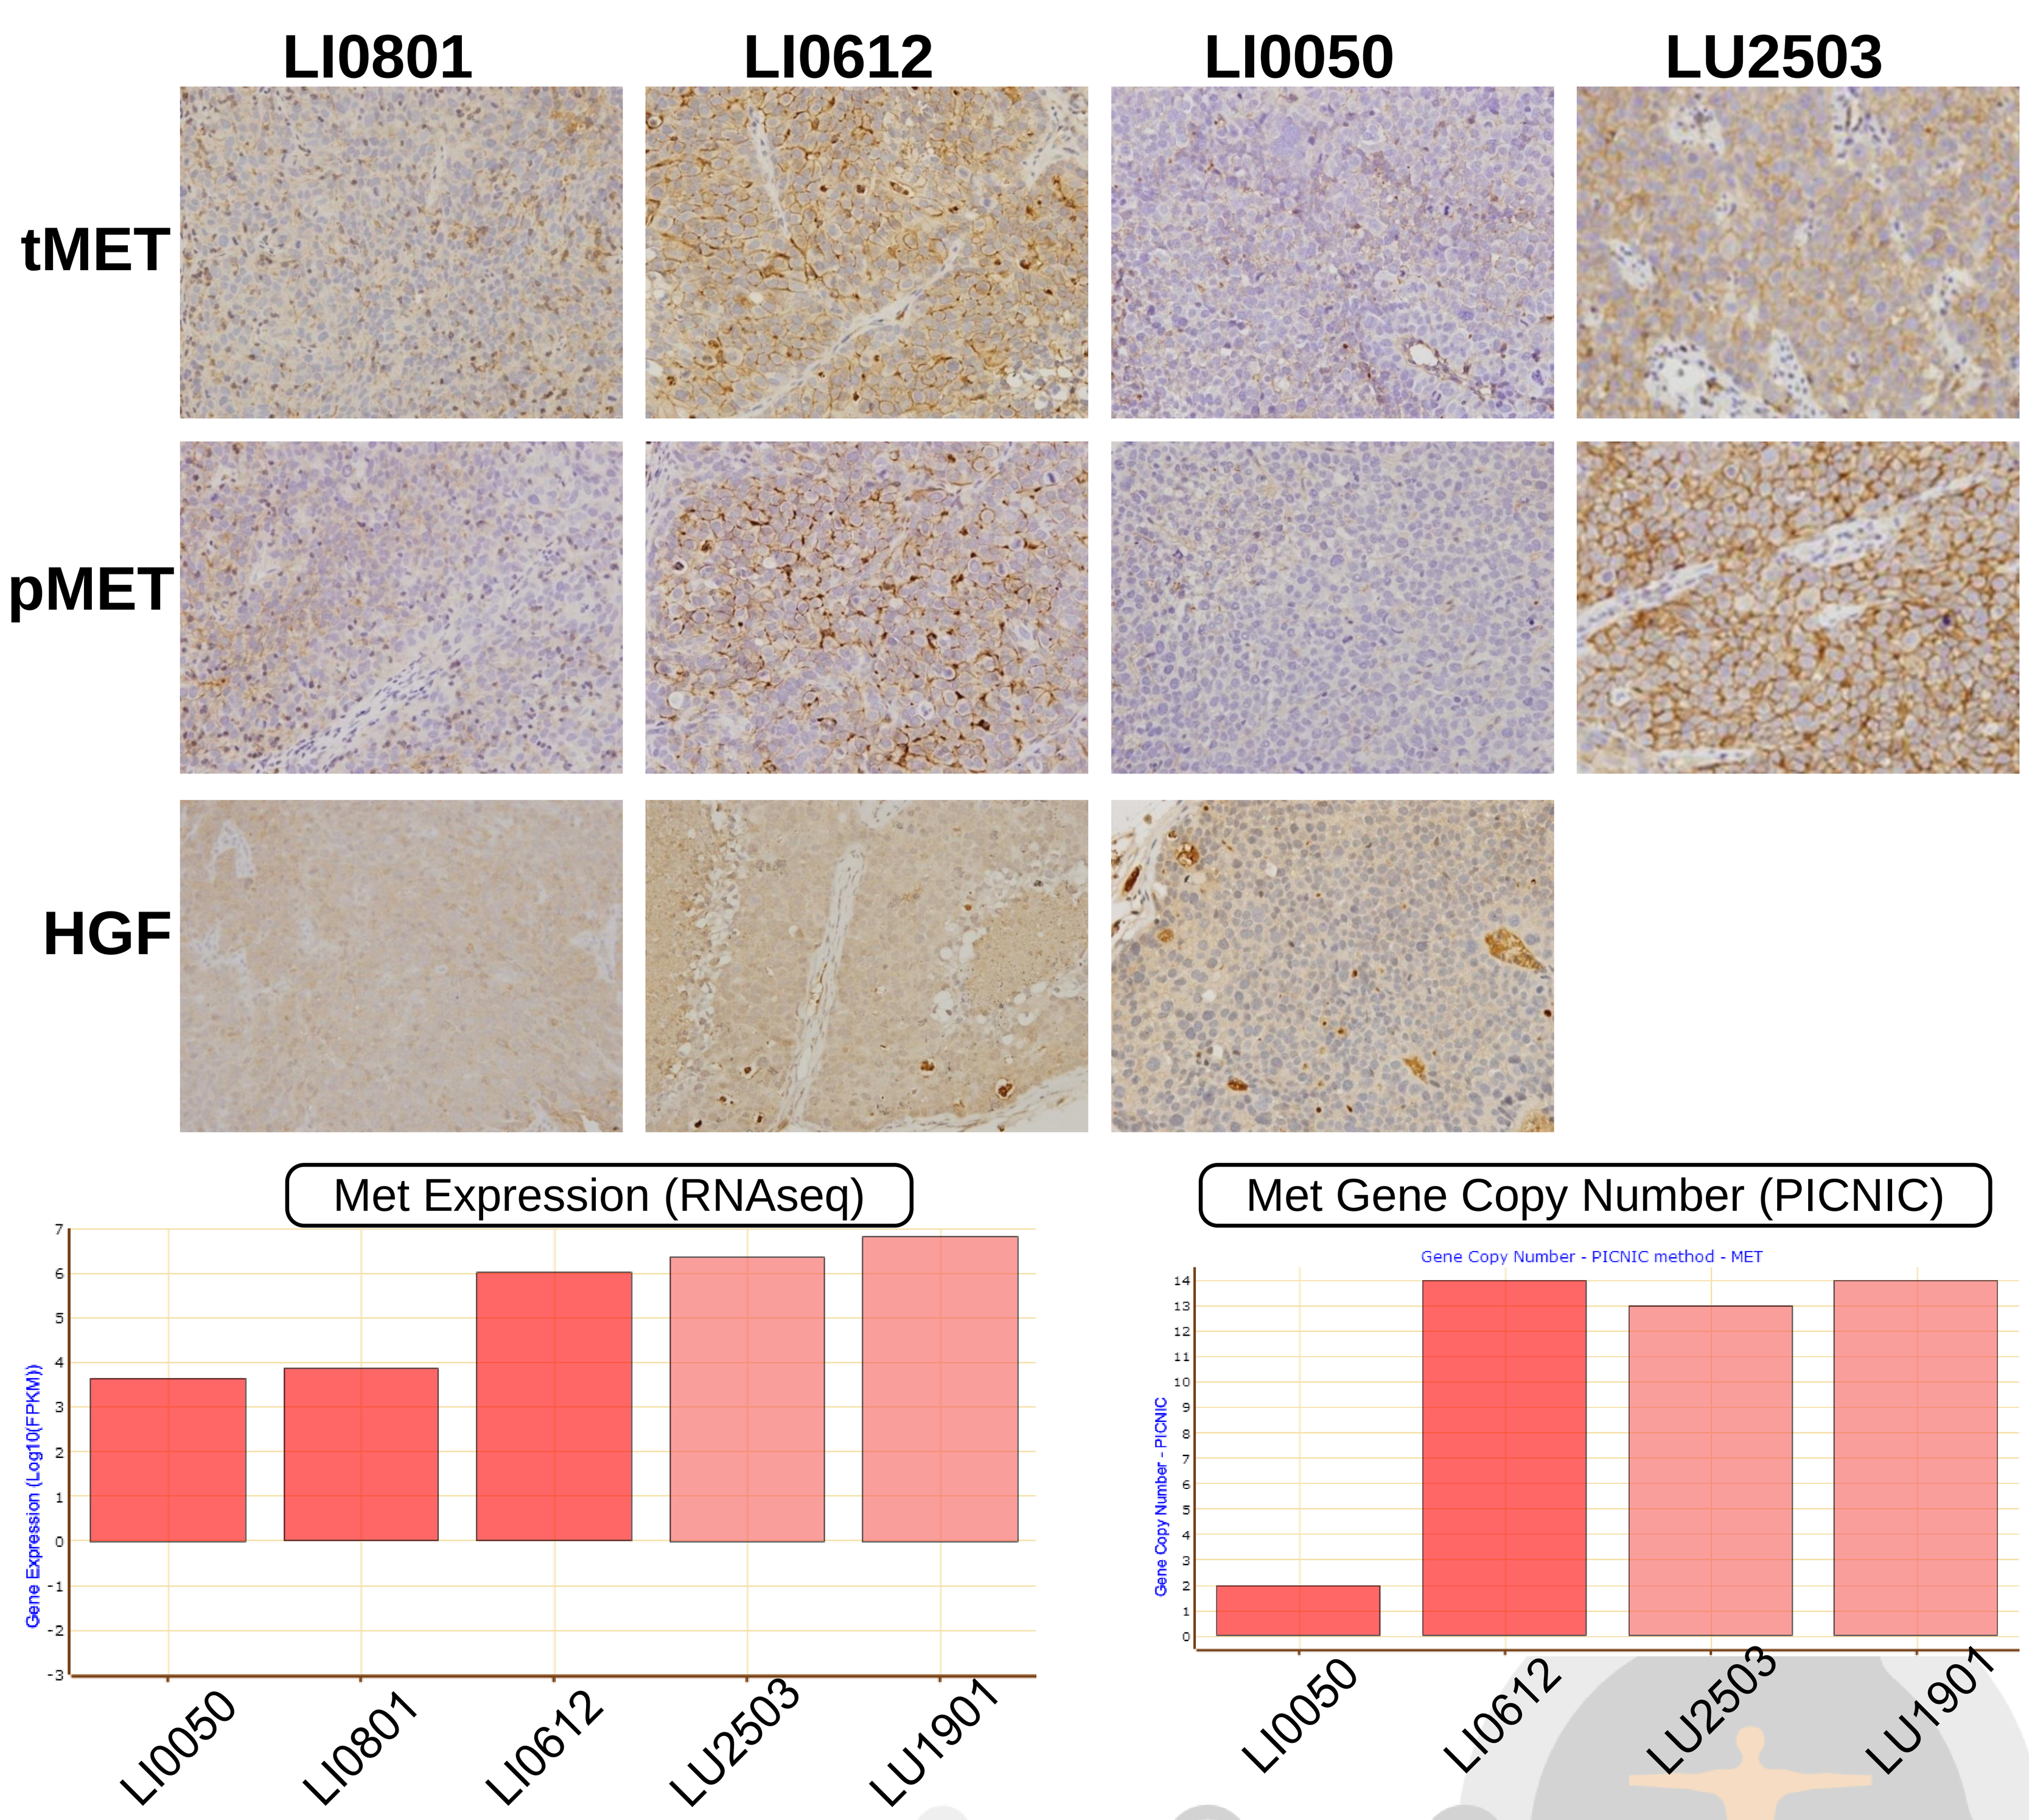
LI0801
LI0612
LI0050
LU2503
tMET
pMET
HGF
Met Expression (RNAseq)
Gene Expression (Log10(FPKM))
LI0050
LI0801
LI0612
LU2503
LU1901
Met Gene Copy Number (PICNIC)
Gene Copy Number - PICNIC method - MET
Gene Copy Number - PICNIC
LI0050
LI0612
LU2503
LU1901

Figure 2. MET Gene and Protein Expression in PDX Models

IC50 (nM)	LU1901	LU2503	EBC-1	MKN45	LI612	H1993	KP4	LU858b*
M101	5.8	6.9	12.2	14.3	17	18.6	176	>1000

* LU858b cells express mutant EGFR

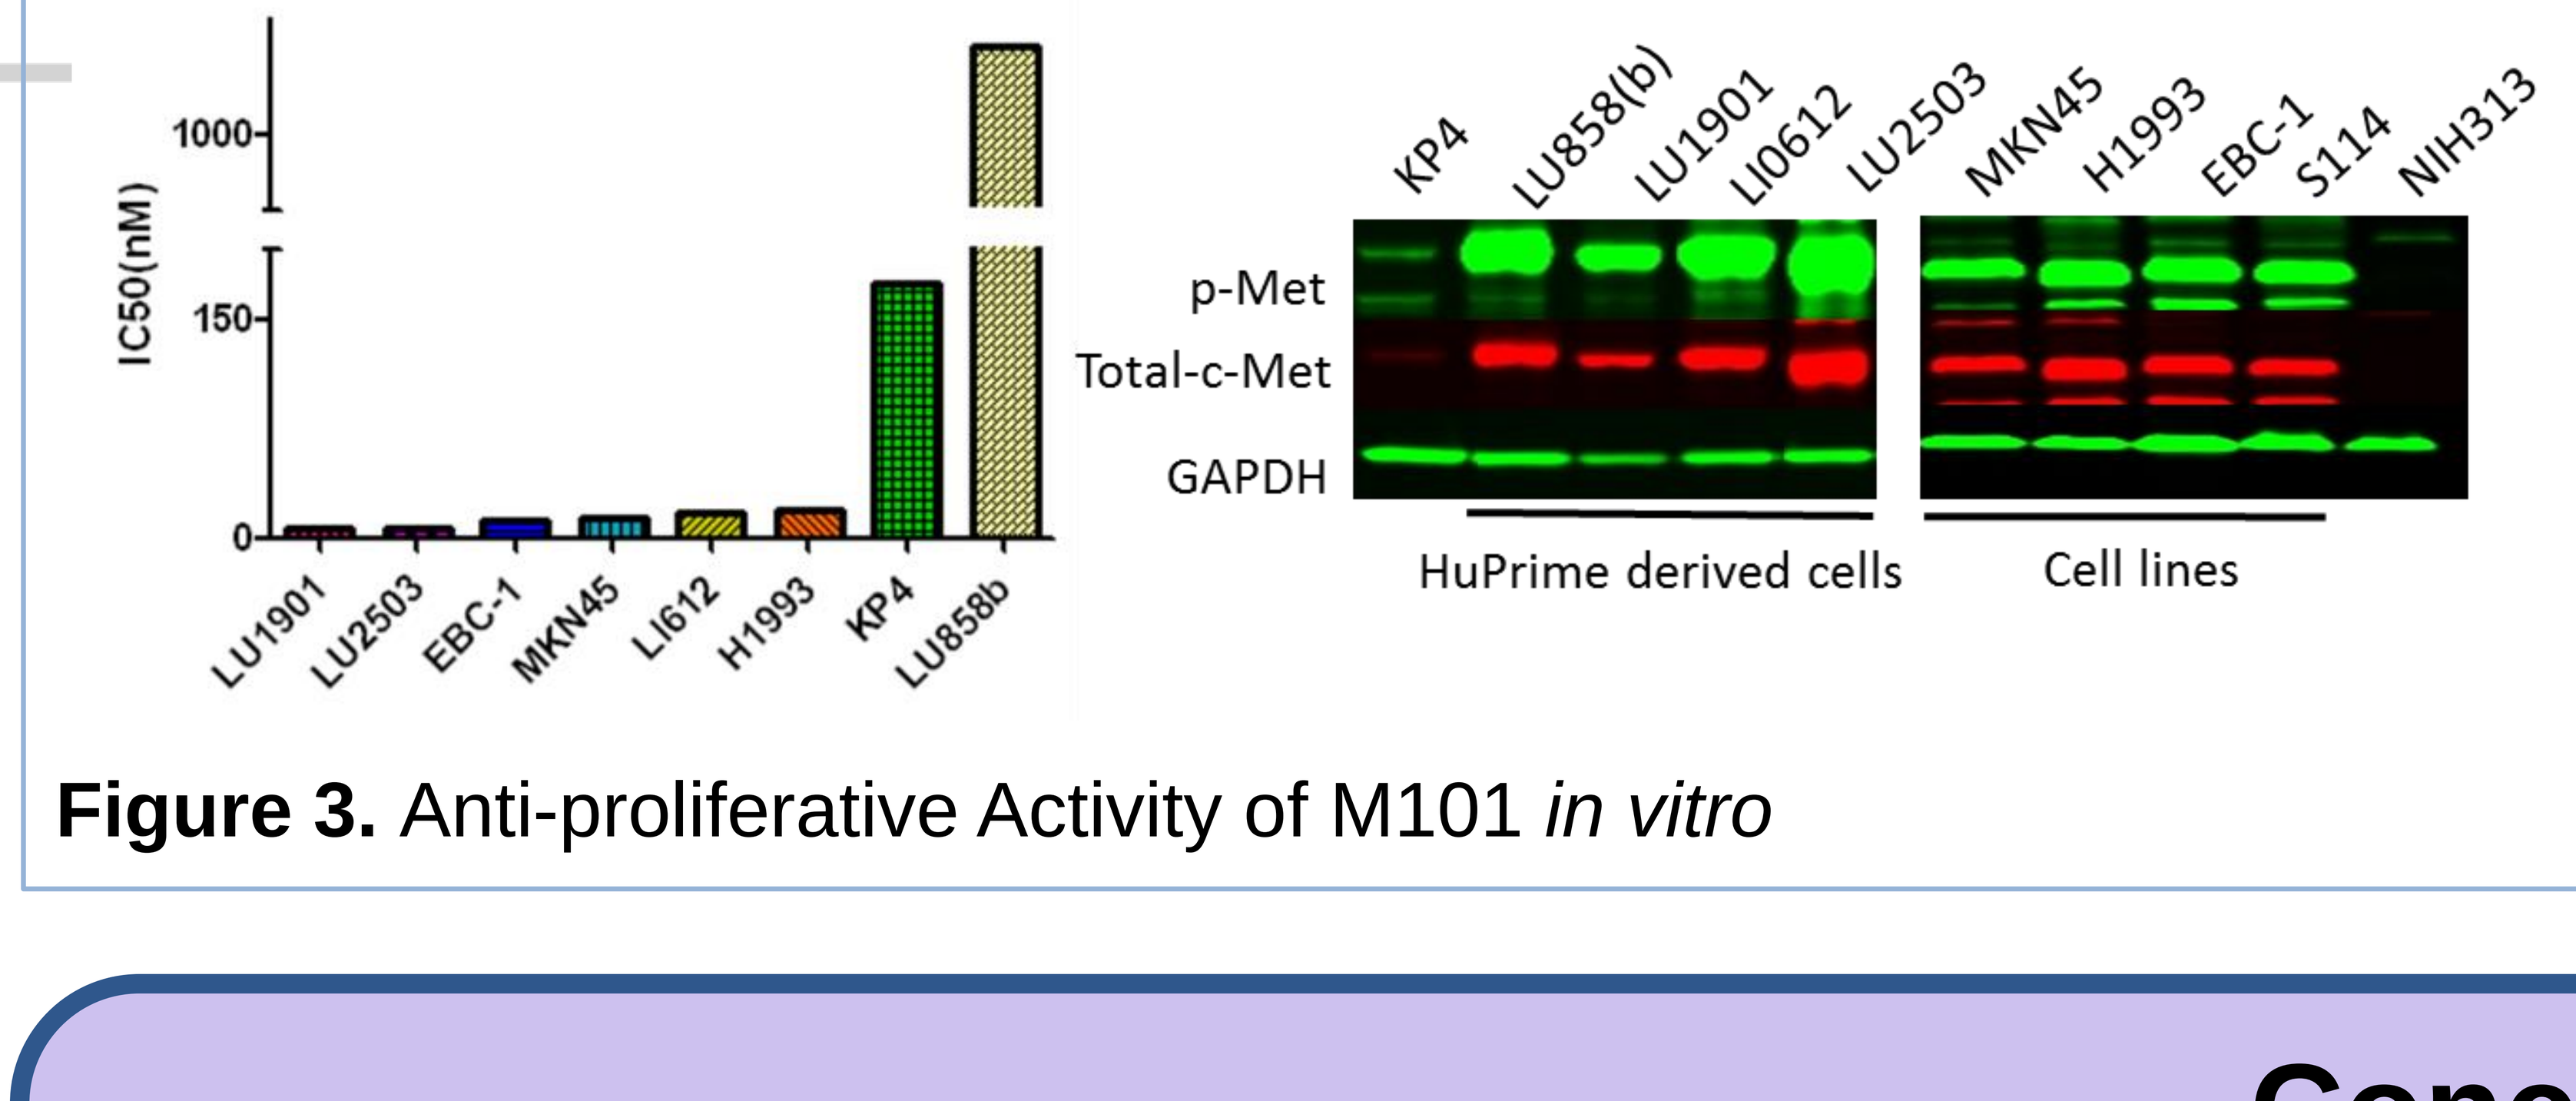
IC50(nM)
1000
150
0
LU1901
LU2503
EBC-1
MKN45
LI612
H1993
KP4
LU858b
KP4
LU858(b)
LU1901
LI0612
LU2503
MKN45
H1993
EBC-1
S114
NIH313
p-Met
Total-c-Met
GAPDH
HuPrime derived cells
Cell lines

Figure 3. Anti-proliferative Activity of M101 *in vitro*

Abstract

A novel and highly specific c-MET TKI (M101) was discovered and characterized for inhibition of kinase activity and cell growth *in vitro*. We found that M101 strongly inhibits the c-MET kinase (IC_{50} of ~35 nM) with high specificity and little activity against other kinases (Fig. 1). M101 suppresses cell growth driven by activated c-MET at EC_{50} of ~8 nM. In order to investigate its antitumor activity in patient-relevant models, we specifically selected a cohort of patient-derived xenografts (PDXs) from HCC and NSCLC tumors with c-MET signaling activation[1,2] (the LI0801 HCC model has high level of paracrine HGF from human stromal cells, while the LI0050 model over-expresses autocrine HGF in tumor cells; finally c-Met is amplified in models LI0612, LU2503, and LU1901), along with two cell line-derived xenografts (CDXs), i.e., the c-Met amplified MKN45 (gastric) and autocrine KP4 (pancreatic). M101 inhibits tumor growth in all these models with daily oral administration 2.3-21 mg/kg, accompanied by deactivation of p-c-MET and the downstream markers pERK and p-Akt. Complete tumor regression was observed in models LI0612, LU2503, and LU1901 after 21 days of treatment, while partial response was observed in models LI0801 and LI0050 where c-MET is not the sole oncogenic driver. M101 efficacy was superior to Crizotinib (50 mg/kg, q.d.) in all cases.
Our data suggest that this novel c-MET TKI is a promising new candidate targeting c-MET that might be particularly efficacious in patients where c-MET is the main oncogenic driver.

Reference

1. *Chen, D. Autocrine c-met/HGF HCC Patient Derived Xenograft (PDX) Models for Evaluating c-Met Inhibitors. AACR Annual Meeting 2012* ***Poster*** *(2012).*
2. *Bladt, F., Friese-Hamim, M., Ihling, C., Wilm, C. & Blaukat, A. The c-Met Inhibitor MSC2156119J Effectively Inhibits Tumor Growth in Liver Cancer Models. Cancers* ***6****, 1736-1752 (2014).*

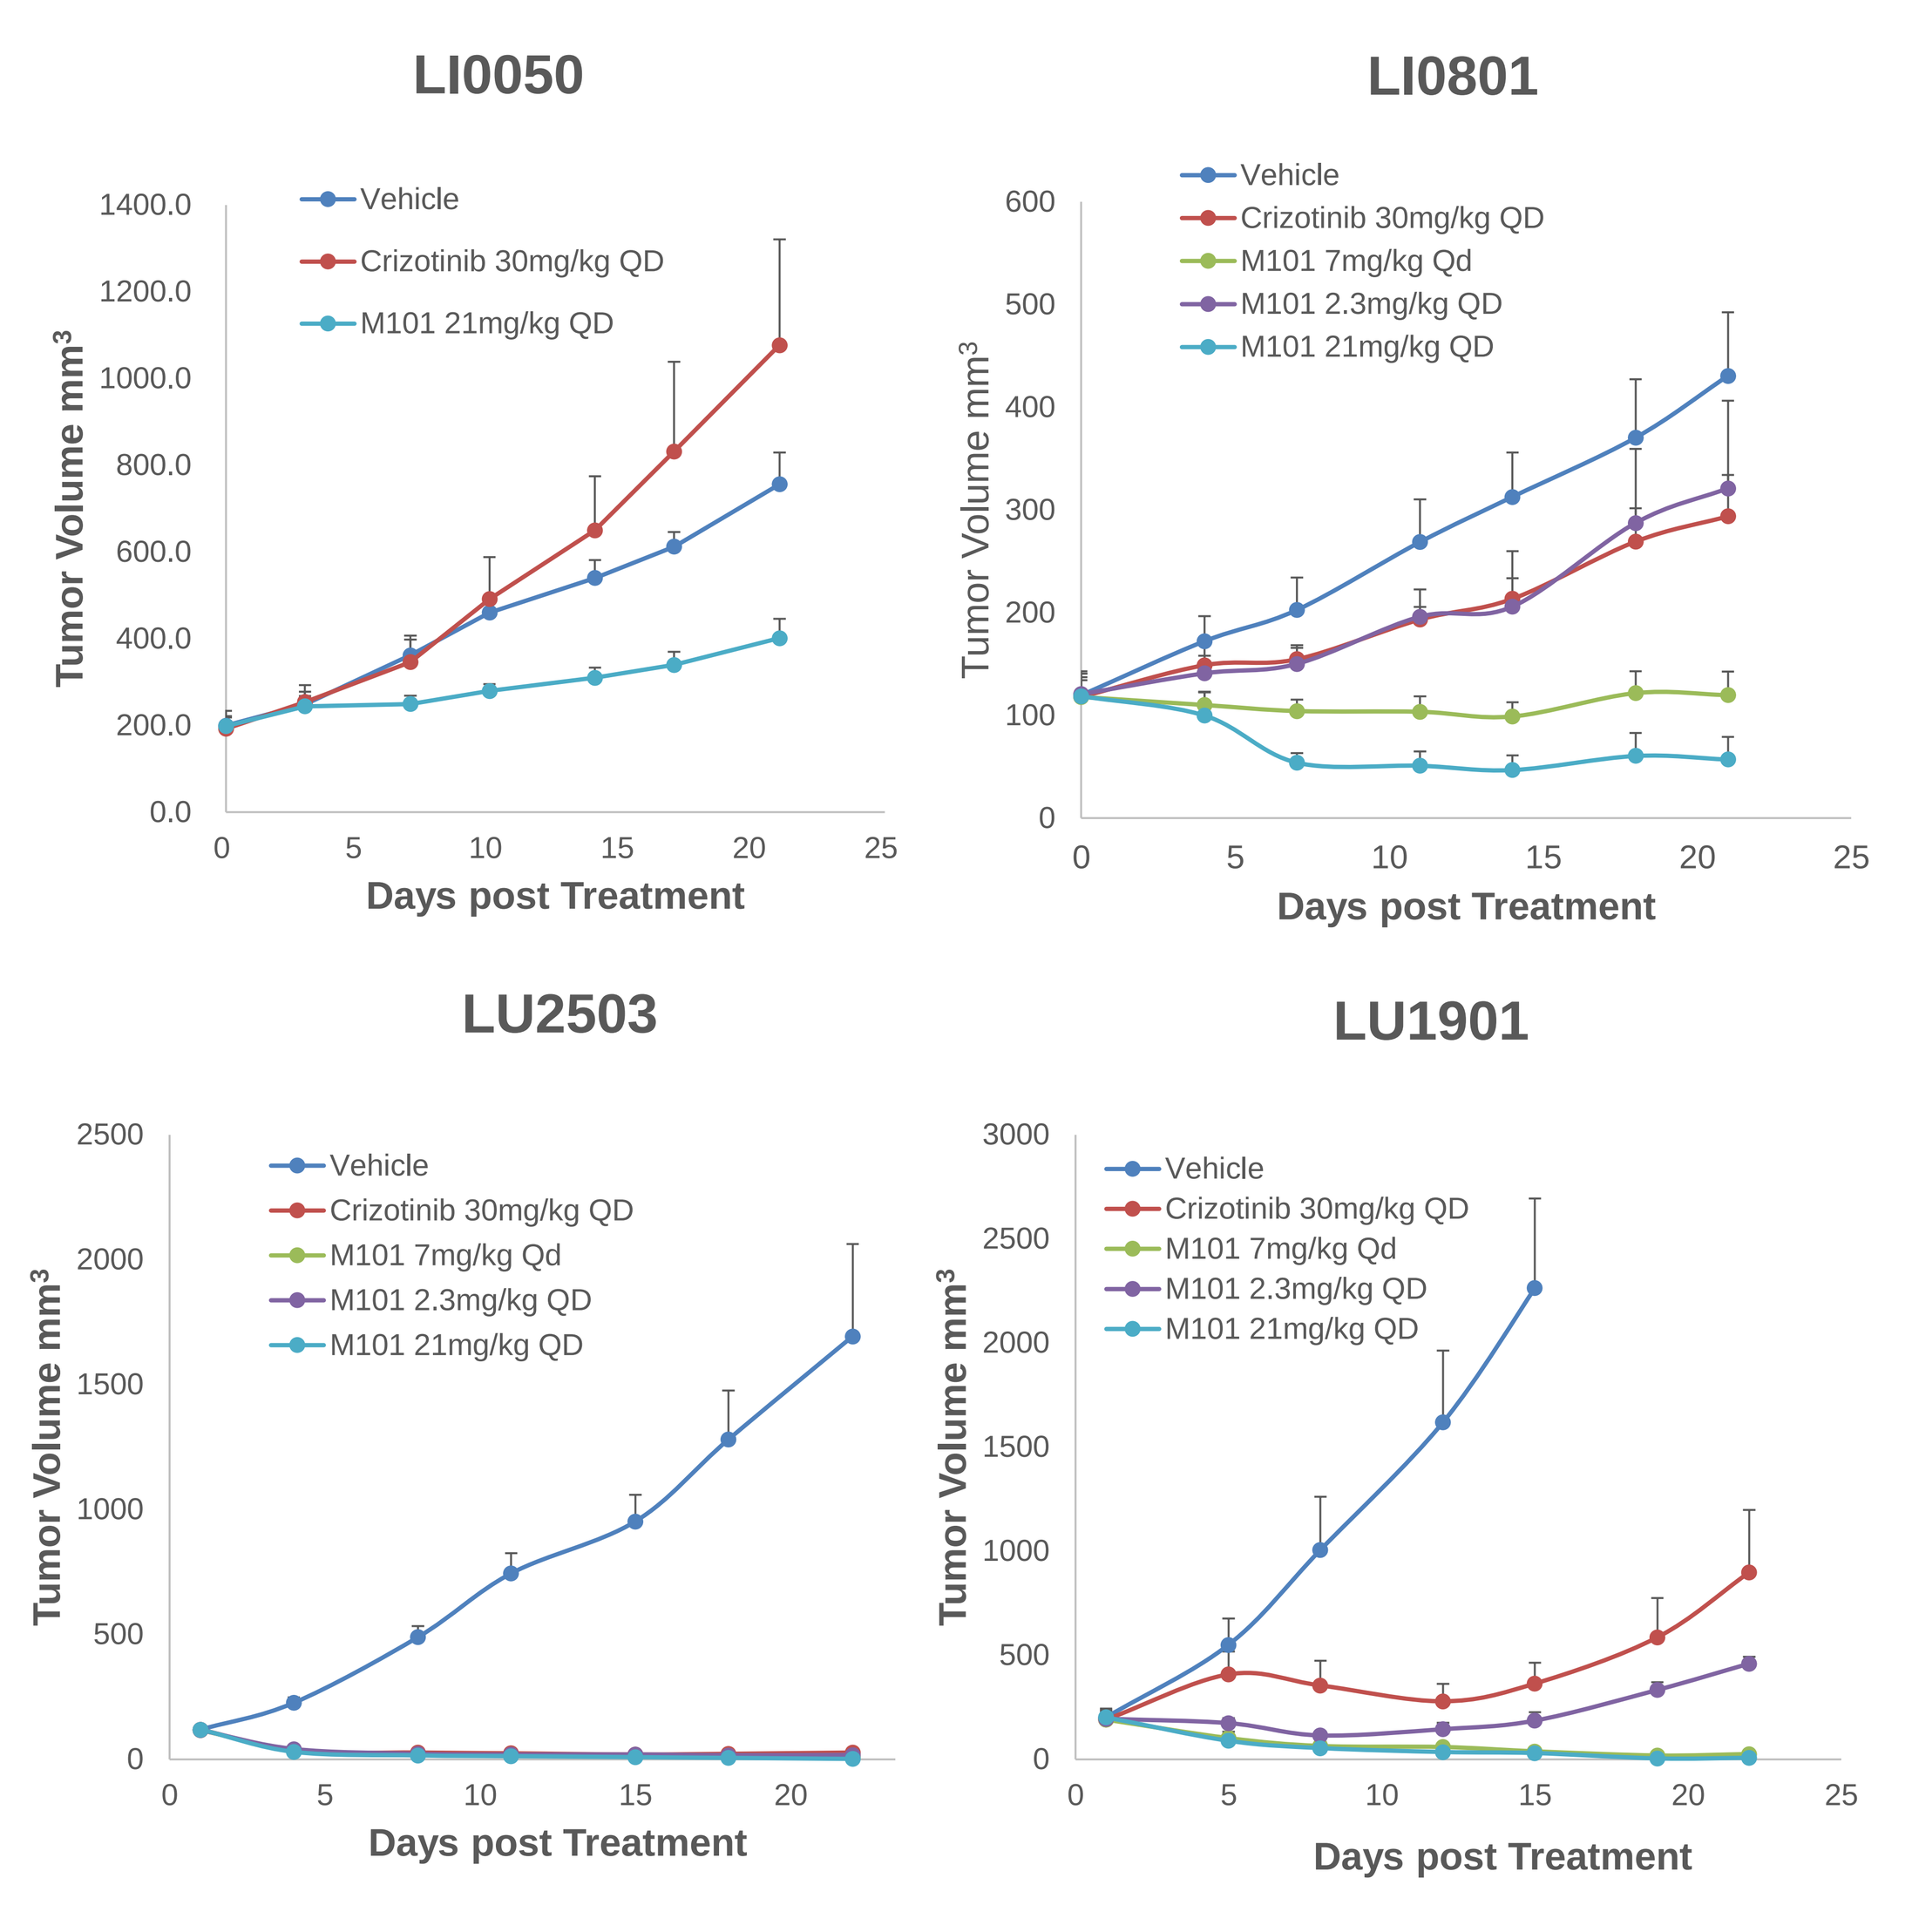
LI0050
LI0801
LI0612
LU2503
LU1901
Vehicle
Crizotinib 30mg/kg QD
M101 7mg/kg Qd
M101 2.3mg/kg QD
M101 21mg/kg QD
Tumor Volume mm³
Days post Treatment

Figure 4. M101 Efficacy in PDX Models

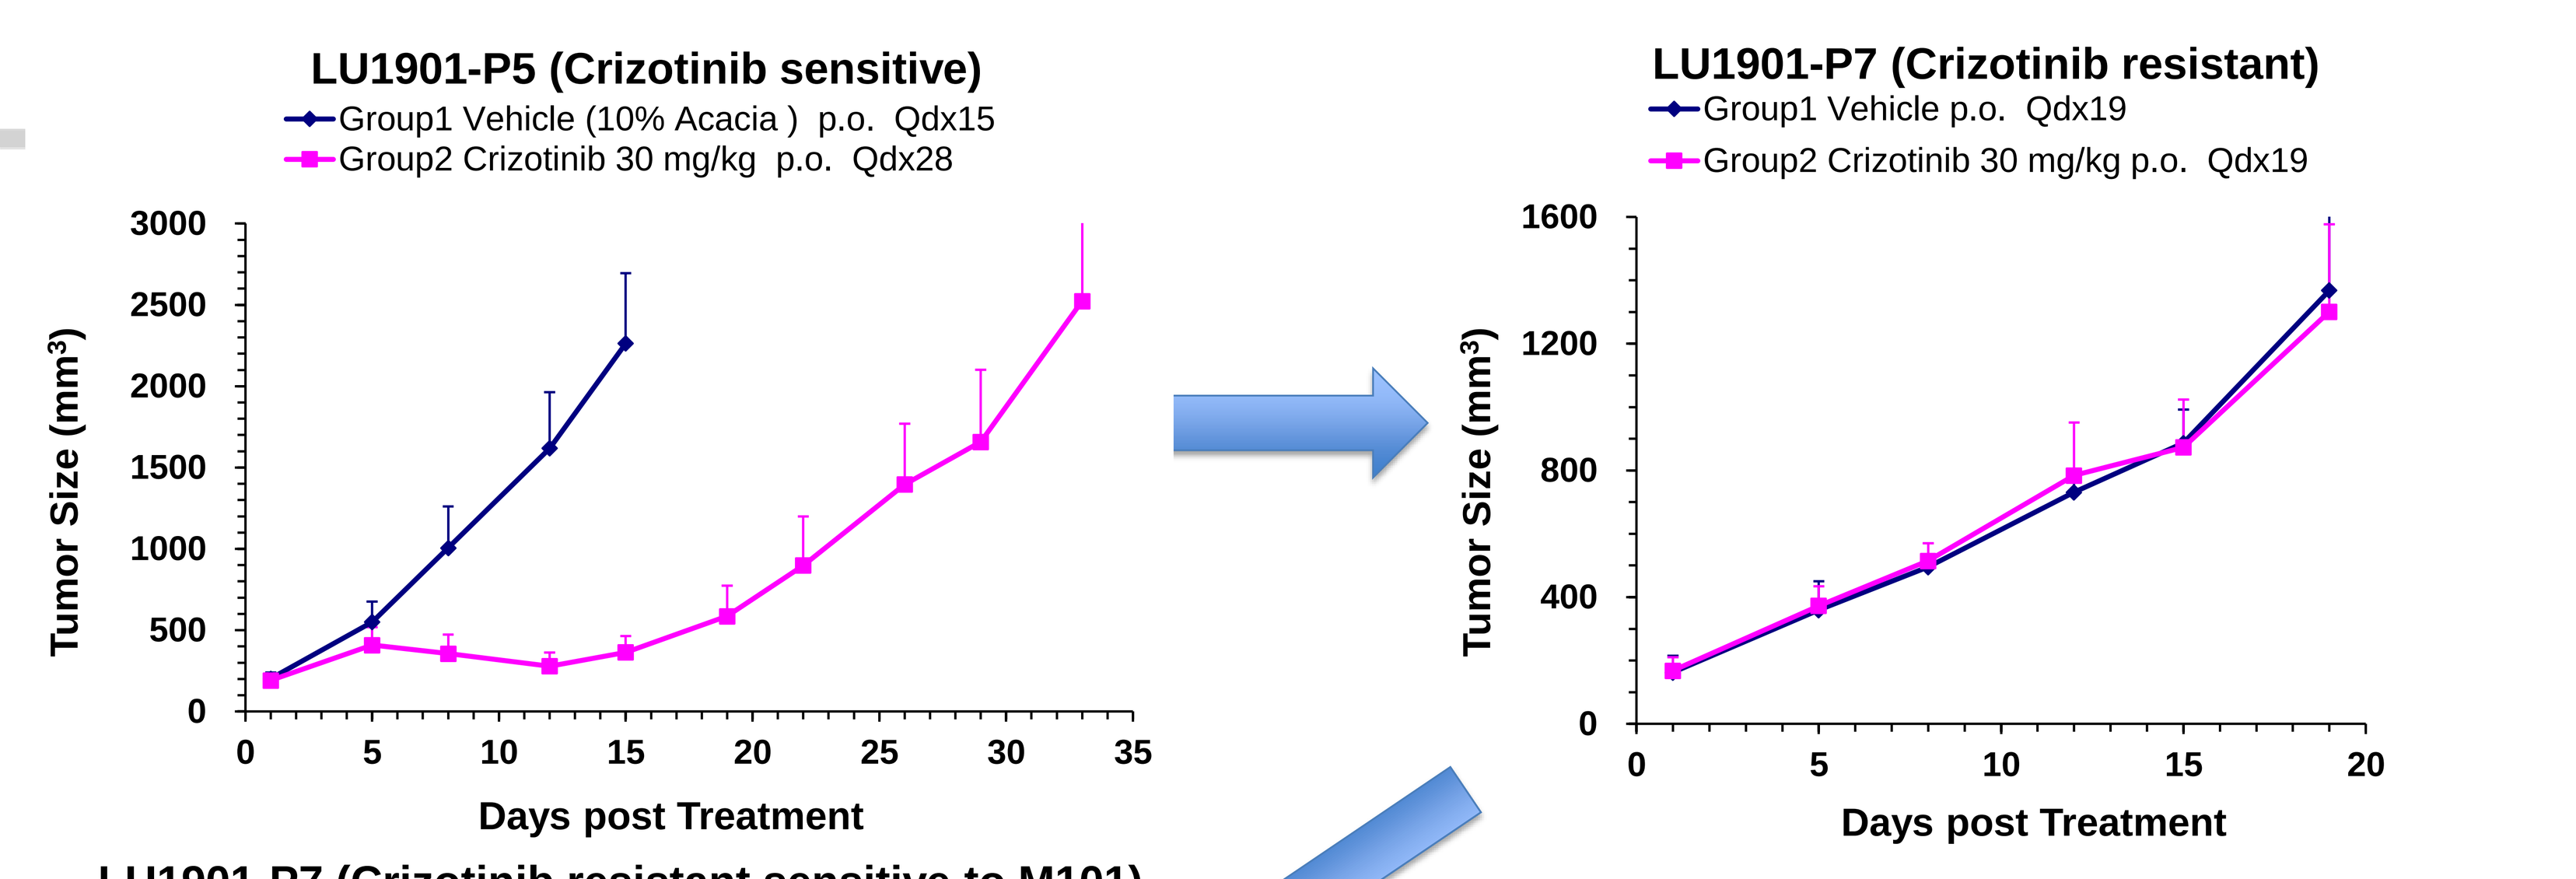
LU1901-P5 (Crizotinib sensitive)
Group1 Vehicle (10% Acacia) p.o. Qdx15
Group2 Crizotinib 30 mg/kg p.o. Qdx28
Tumor Size (mm³)
Days post Treatment
LU1901-P7 (Crizotinib resistant)
Group1 Vehicle p.o. Qdx19
Group2 Crizotinib 30 mg/kg p.o. Qdx19
Tumor Size (mm³)
Days post Treatment

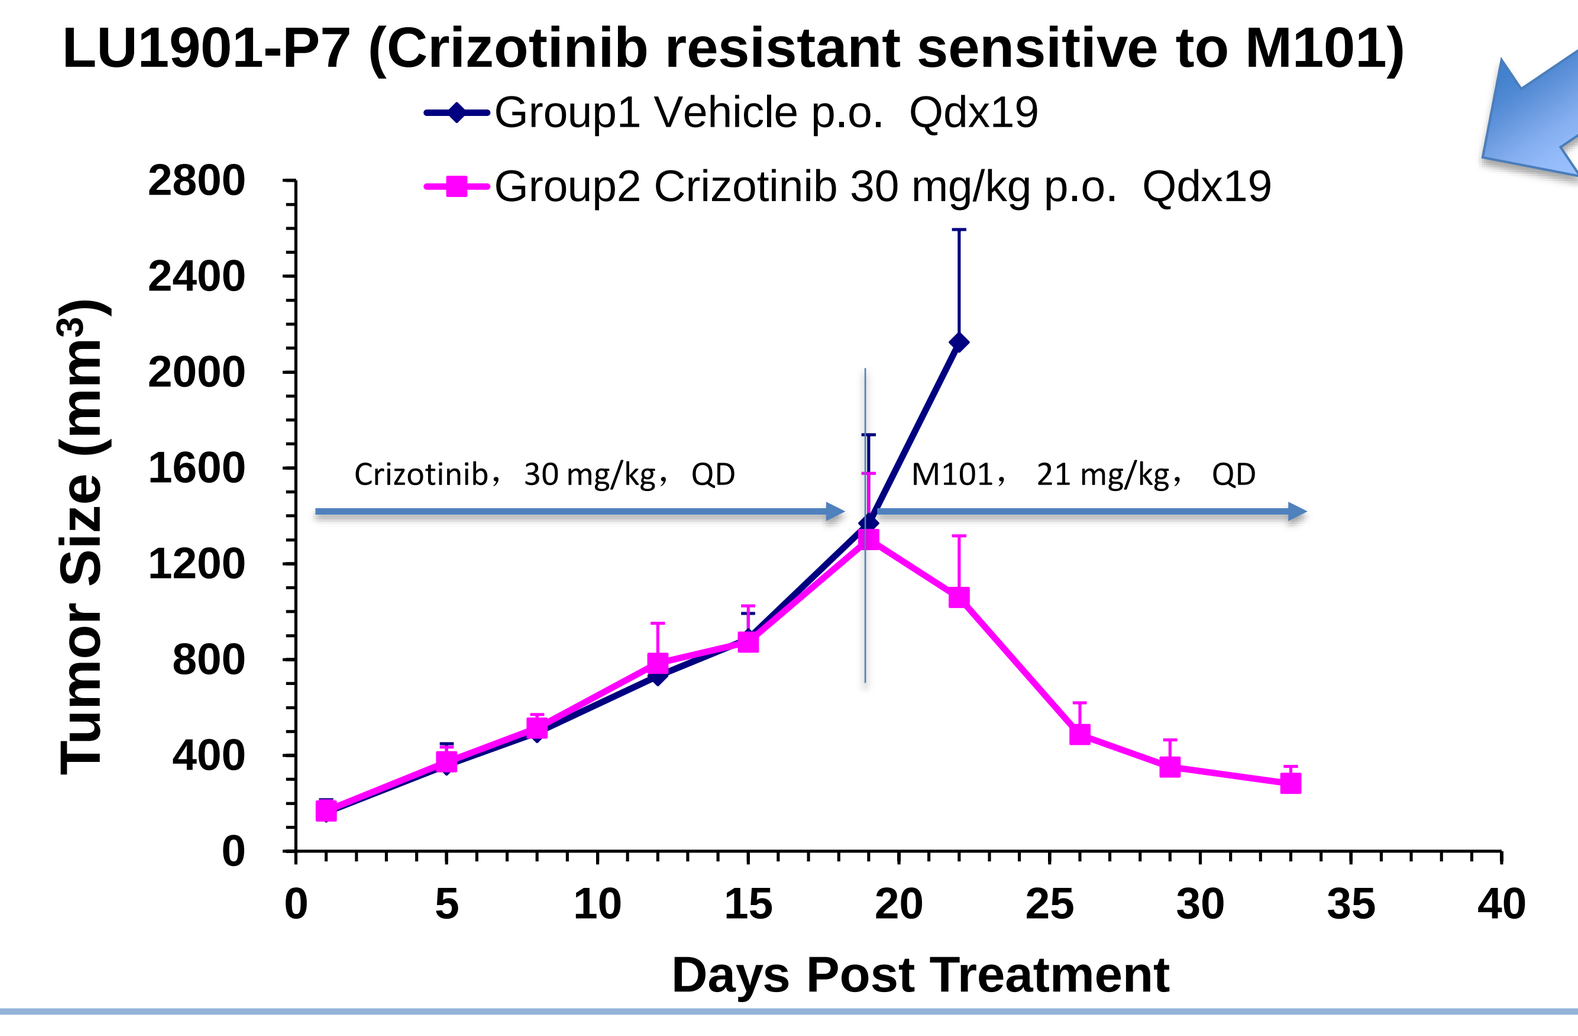
LU1901-P7 (Crizotinib resistant sensitive to M101)
Group1 Vehicle p.o. Qdx19
Group2 Crizotinib 30 mg/kg p.o. Qdx19
Crizotinib，30 mg/kg，QD
M101， 21 mg/kg， QD
Tumor Size (mm³)
Days Post Treatment

Figure 5. Exposure of LU1901 to crizotinib caused rapid development of resistance, while crizotinib resistant tumors are still sensitive to M101

Conclusions

1. M101 is a highly specific c-MET TKI
2. Models with activated MET signaling *via* gene amplification, over expression, and auto/paracrine binding of HGF can be classified as responders
3. M101 shows superior efficacy *versus* Crizotinib in both PDX and CDX models.
4. M101 is effective on crizotinib-resistant models and might be considered a good candidate for treatment of relapsed tumors.

Contact information: HenryLi@crownbio.com