A Novel RET TKI Inhibits Growth of Patient-Derived Xenografts with RET Fusion and Overcomes Ponatinib-Induced Resistance

Poster : #87

In life for life

Yanrui Song[1]*, Xiaoxi Xu[1], Mengmeng Yang[1], Deyi Zhang[1], Meiling Zheng[1], Jinying Ning[1], Jie Cai[1], Sheng Guo[1], Davy Xuesong Ouyang[1], Jean Pierre Wery[1], and Henry Q.X. Li[1,2]
[1]Crown Bioscience, Inc., 3375 Scott Blvd, suite 108, Santa Clara, CA 95054; [2]State Key Laboratory of Natural and Biomimetic Drugs, Peking University, 38 Xueyuan Road, Beijing, 100191, China

Introduction

Activation of RET, a RTK proto-oncogene, is associated with the development of several human malignancies. RET activation occurs either through germ-line mutations, leading to the inheritable expression of a constitutively active form of RET, or more frequently, *via* somatic fusion with various activating genes. This results in aberrant RET expression and constitutive dimerization with the PTC oncoprotein in thyroid follicular cells. RET gene fusion has been identified in 10-20% of sporadic papillary thyroid cancer and 1-2% of non-small cell lung cancer (NSCLC). Fusion events involve the kinesin family member 5B (KIF5B-RET), the coiled-coil domain-containing protein 6 (CCDC6-RET), the nuclear receptor coactivator 4 (NCOA4-RET), and tripartite motif containing 33 (TRIM33-RET). Therefore, targeting RET signaling with selective tyrosine kinase inhibitors (TKI) has been suggested to be an important therapeutic approach for a subset of human cancers.

Abstract

We established two CRC PDX models carrying two different types of RET fusions, i.e. CCDC6-RET (CR2518) and NCOA4-RET (CR1520), as revealed by RNAseq/Sanger sequencing. These two PDXs were found to be sensitive to ponatinib (AP24534), a TKI with high RET inhibitory activity(1). Most interestingly, ponatinib treatment caused rapid induction of drug resistance in the CR2545 PDX, hallmarked by introduction of a gate keeper mutation, V804M(2).

We set out to test a novel RET-TKI, R101 (it inhibits the wild-type enzyme at IC_{50} of 1.6 x 10-7M) for overcoming the ponatinib induced resistance. R101 demonstrated similar potency as ponatinib (EC_{50} 0.56 vs. 0.47uM) in inhibiting *in vitro* 2D proliferation of CR1520 PrimePanel cell line, while it resulted significantly less potent in HUVEC normal endothelial cells (EC_{50} 5.0 vs. 0.25uM). *In vivo* efficacy study showed strong antitumor activity of R101 in both CR2518 and CR1520. Interestingly, the ponatinib resistant CR2545 also demonstrated robust response to R101, with greatly extended overall survival of tumor bearing mice following treatment. Our data suggest that R101 could potentially be a good candidate for the treatment of tumors with RET gene fusion, and ponatinib/vandetanib resistant cases.

References

1. *Joseph M. Gozgit YS, Tzu-Hsiu Chen, Tim Clackson and Victor M. Rivera. RET fusions identified in colorectal cancer PDX models are sensitive to the potent RET inhibitor ponatinib. AACR-EORTC-NCI Annual Molecular Therapeutics Conference 2013.*
2. *Mengmeng Yang JC, Sheng Guo, Jean Pierre Wery, and Henry Q.X. Li. Rapid conversion to resistance, of a colon PDX with ret-fusion, by ponatinib treatment could potentially be attributed to the introduction of the gate keeper mutation, V804M. AACR-Annual Conference 2015*

Results

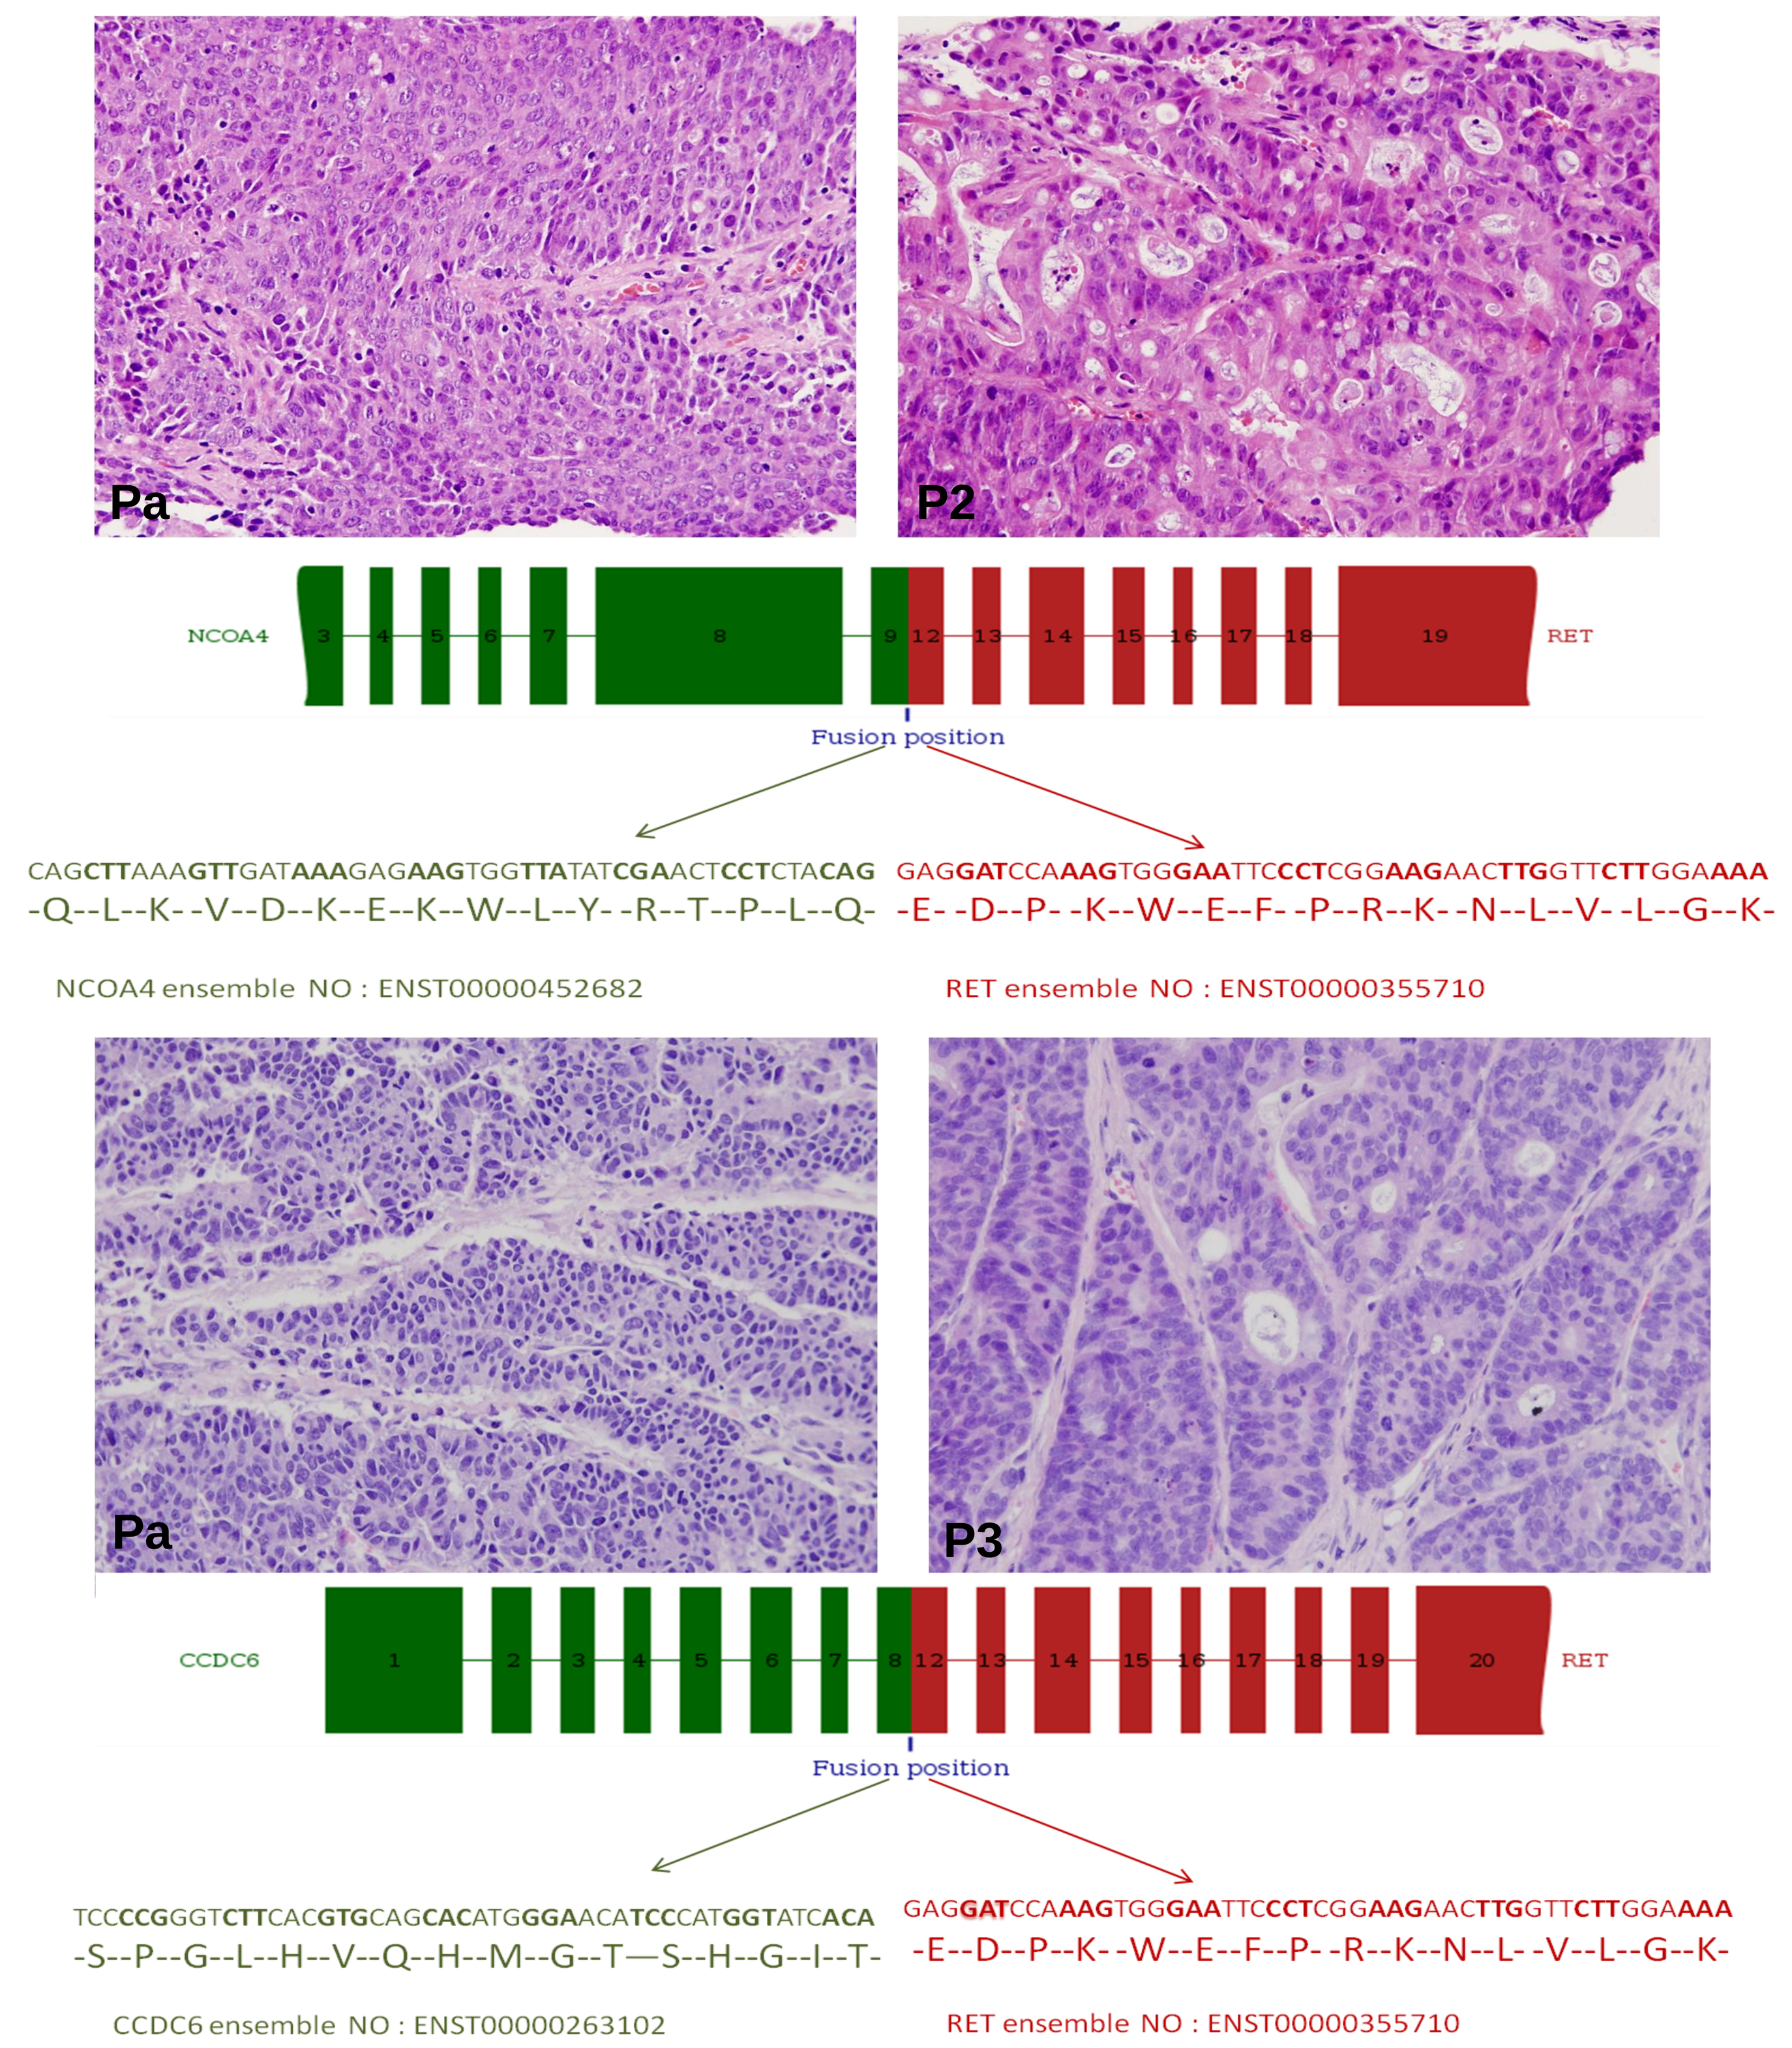
Pa
P2
NCOA4
RET
Fusion position
CAGCTTAAAGTTGATAAAGAGAAGTGGTTATATCGAACTCCTCTACAG
-Q--L--K--V--D--K--E--K--W--L--Y--R--T--P--L--Q-
GAGGATCCAAAGTGGGAATTCCCTCGGAAGAACTTGGTTCTTGGAAAA
-E--D--P--K--W--E--F--P--R--K--N--L--V--L--G--K-
NCOA4 ensemble NO : ENST00000452682
RET ensemble NO : ENST00000355710
Pa
P3
CCDC6
RET
Fusion position
TCCCCGGGTCTTCACGTGCAGCACATGGGAACATCCCATGGTATCACA
-S--P--G--L--H--V--Q--H--M--G--T—S--H--G--I--T-
GAGGATCCAAAGTGGGAATTCCCTCGGAAGAACTTGGTTCTTGGAAAA
-E--D--P--K--W--E--F--P--R--K--N--L--V--L--G--K-
CCDC6 ensemble NO : ENST00000263102
RET ensemble NO : ENST00000355710

Figure 1. Histopathology and in-frame RET fusion: Top panel: NCOA4-RET fusion in the CR1520 model. Lower panel: CCDC6-RET fusion in the CR2518 model

treatments	Absolute IC_{50} (μM)	Max inhibition (%)
R101	0.561	99.51%
Ponatinib	0.468	99.90%

Comp ID	Absolute IC_{50} (μM)	Max inhibition (%)
R101	5.021	96.91%
Ponatinib	0.251	99.99%

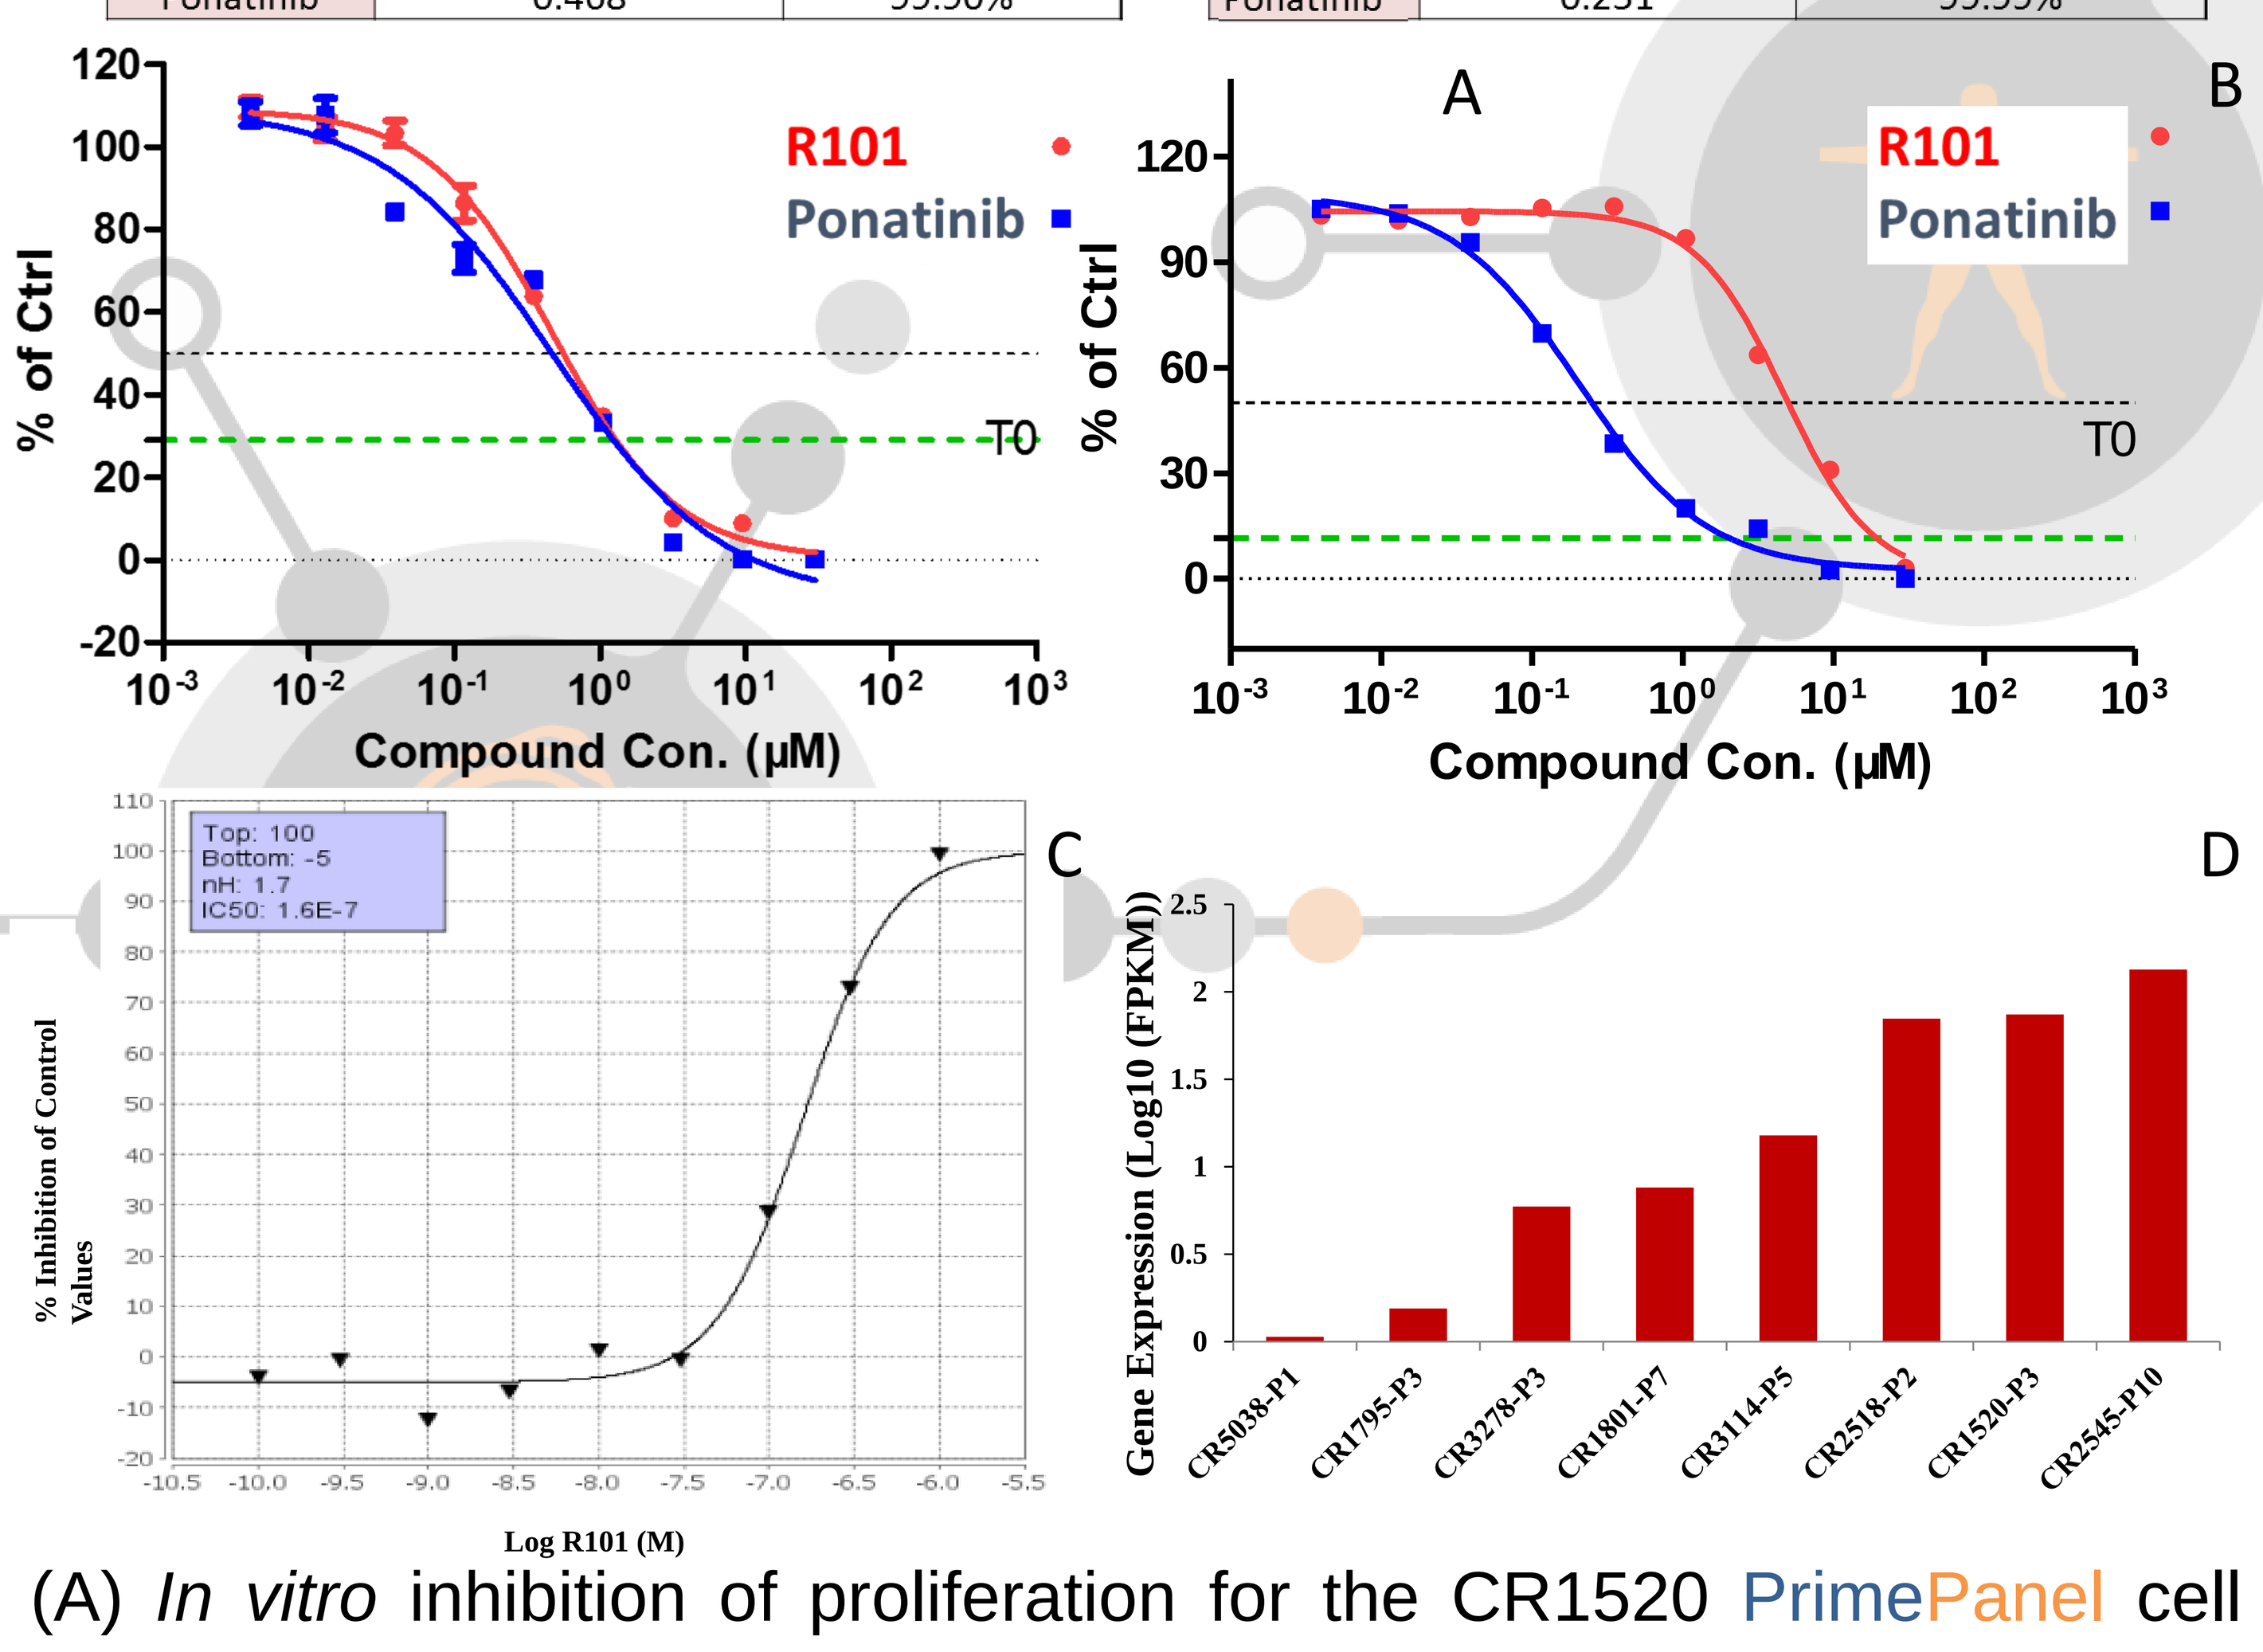
A
B
C
D
R101
Ponatinib
T0
% of Ctrl
Compound Con. (μM)
Top: 100
Bottom: -5
nH: 1.7
IC50: 1.6E-7
% Inhibition of Control Values
Log R101 (M)
Gene Expression (Log10 (FPKM))
CR5038-P1
CR1795-P3
CR3278-P3
CR1801-P7
CR3114-P5
CR2518-P2
CR1520-P3
CR2545-P10

Figure 2. (A) *In vitro* inhibition of proliferation for the CR1520 PrimePanel cell line model. (B) Inhibition of HUVEC by R101/ponatinib in 3 day 2D assays. (C) RET enzymatic inhibition by R101. (D) RET mRNA expression in CRC-PDXs.

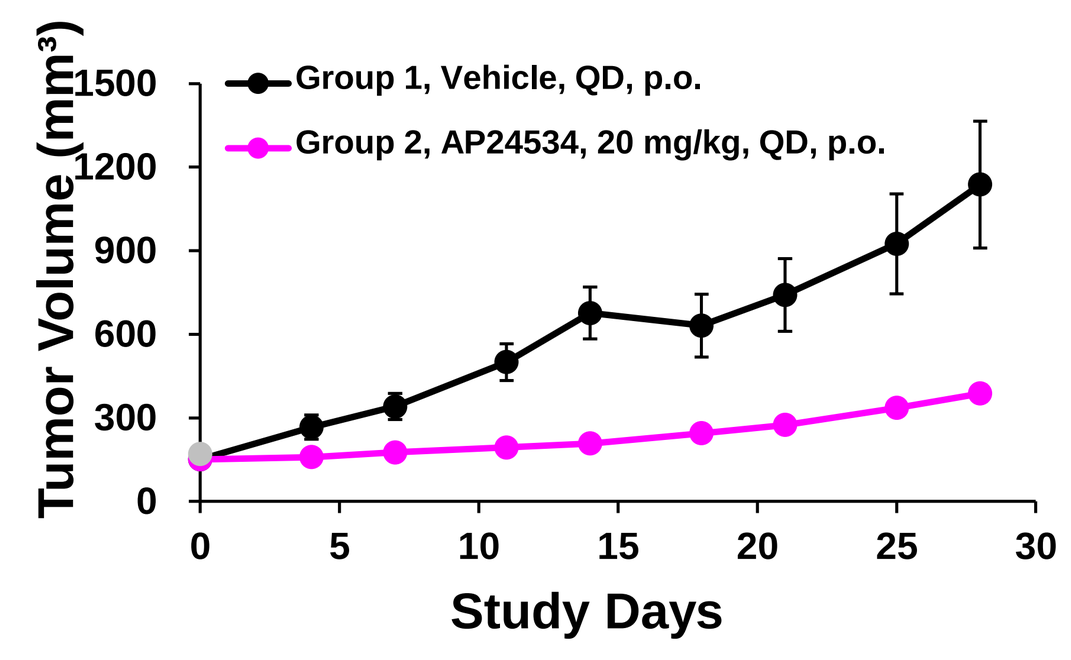
Group 1, Vehicle, QD, p.o.
Group 2, AP24534, 20 mg/kg, QD, p.o.
Tumor Volume (mm3)
Study Days

Figure 3. CR1520 is sensitive to ponatinib treatment

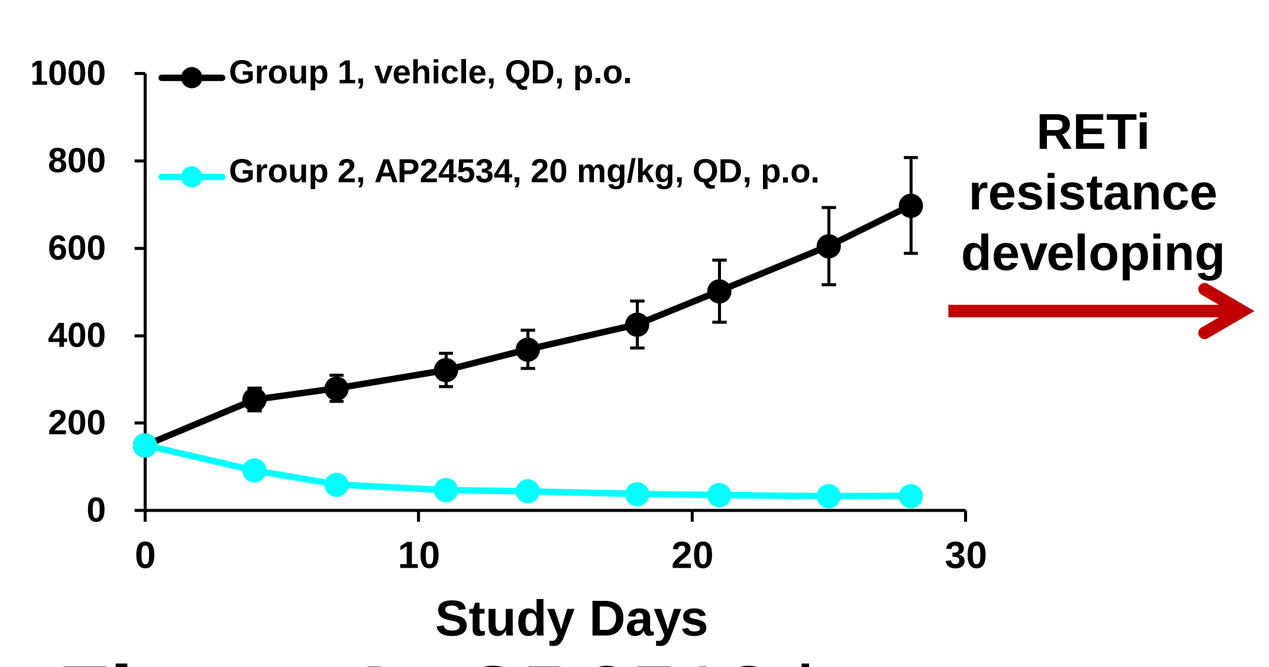
Group 1, vehicle, QD, p.o.
Group 2, AP24534, 20 mg/kg, QD, p.o.
Study Days
RETi resistance developing

Figure 4. CR2518 is sensitive to ponatinib treatment

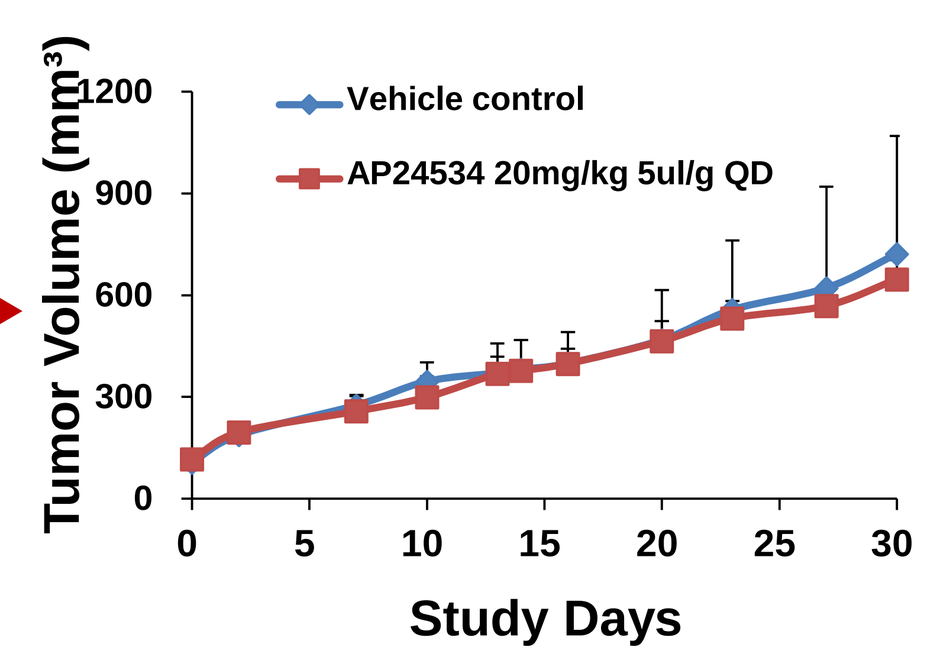
Vehicle control
AP24534 20mg/kg 5ul/g QD
Tumor Volume (mm3)
Study Days

Figure 5. CR2545 is resistant to ponatinib treatment

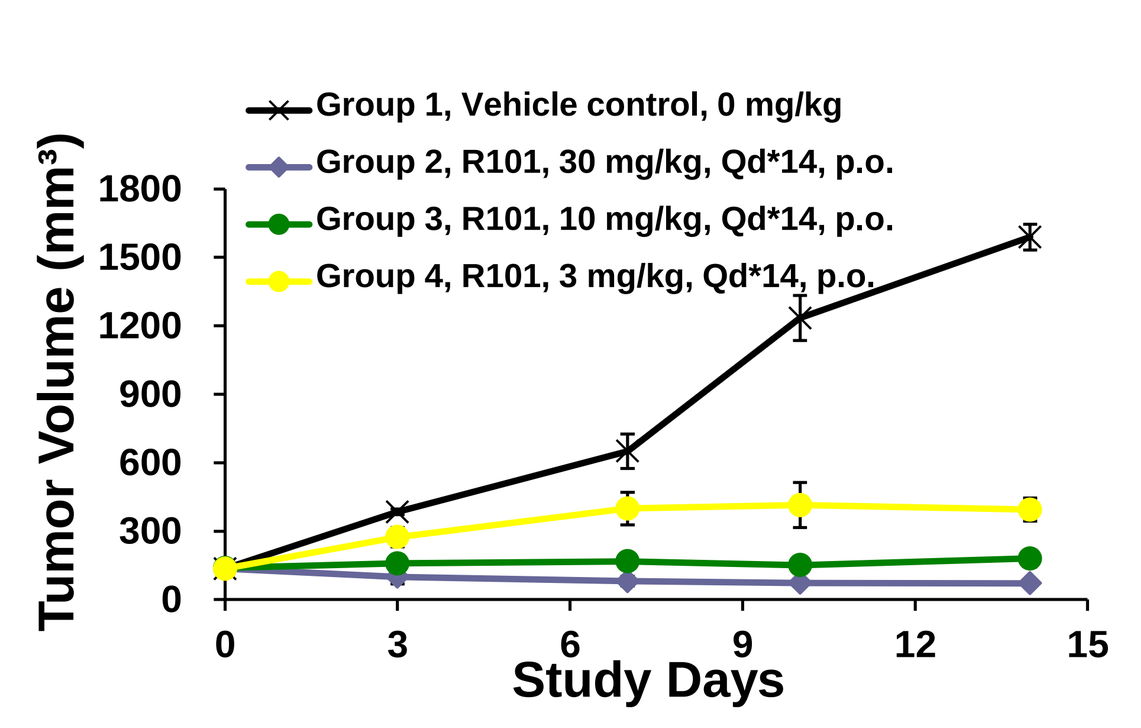
Group 1, Vehicle control, 0 mg/kg
Group 2, R101, 30 mg/kg, Qd*14, p.o.
Group 3, R101, 10 mg/kg, Qd*14, p.o.
Group 4, R101, 3 mg/kg, Qd*14, p.o.
Tumor Volume (mm3)
Study Days

Figure 6. R101 showed strong antitumor activity in CR1520

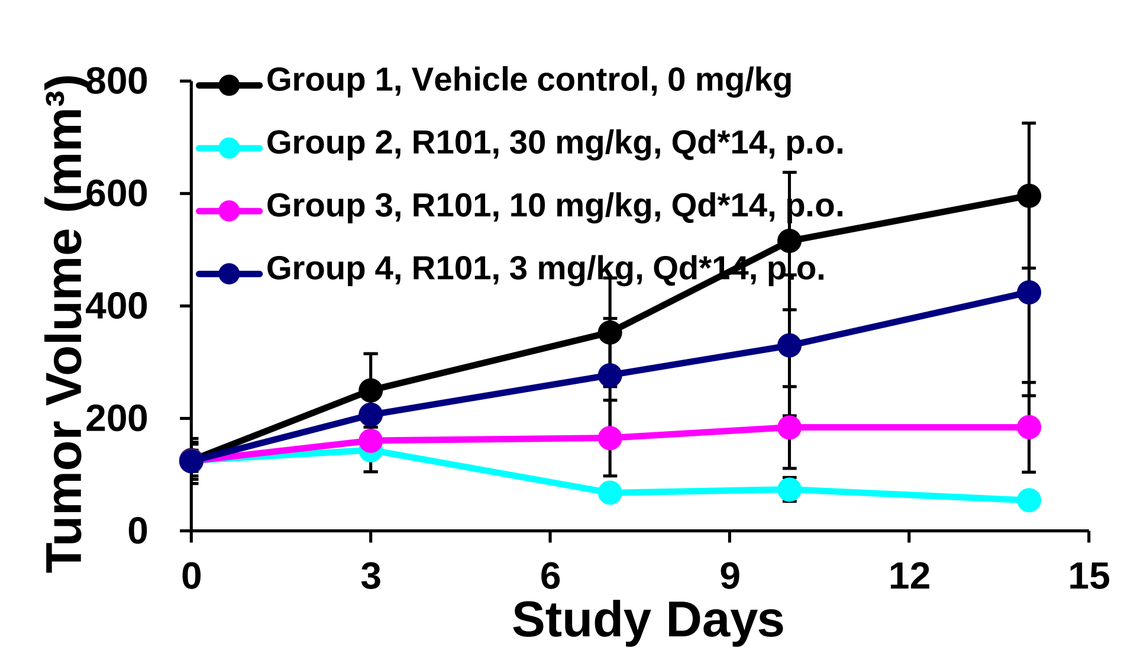
Group 1, Vehicle control, 0 mg/kg
Group 2, R101, 30 mg/kg, Qd*14, p.o.
Group 3, R101, 10 mg/kg, Qd*14, p.o.
Group 4, R101, 3 mg/kg, Qd*14, p.o.
Tumor Volume (mm3)
Study Days

Figure 7. R101 showed strong antitumor activity in CR2518

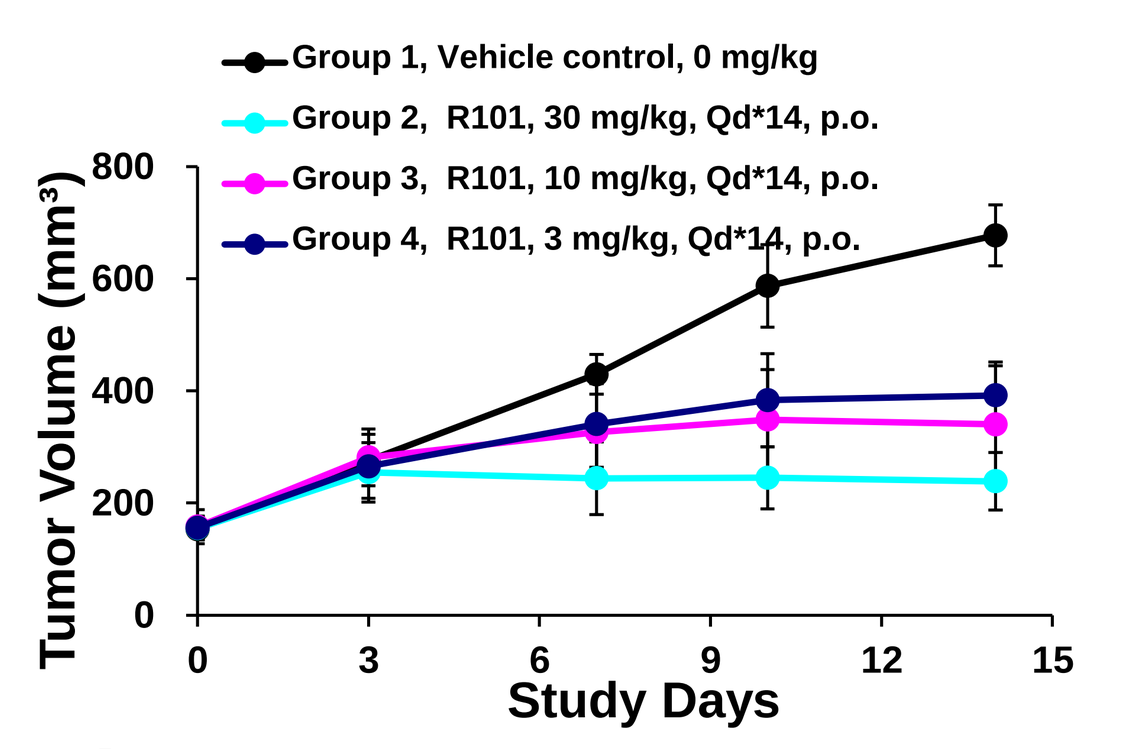
Group 1, Vehicle control, 0 mg/kg
Group 2, R101, 30 mg/kg, Qd*14, p.o.
Group 3, R101, 10 mg/kg, Qd*14, p.o.
Group 4, R101, 3 mg/kg, Qd*14, p.o.
Tumor Volume (mm3)
Study Days

Figure 8. R101 overcame ponatinib induced resistance (CR2545)

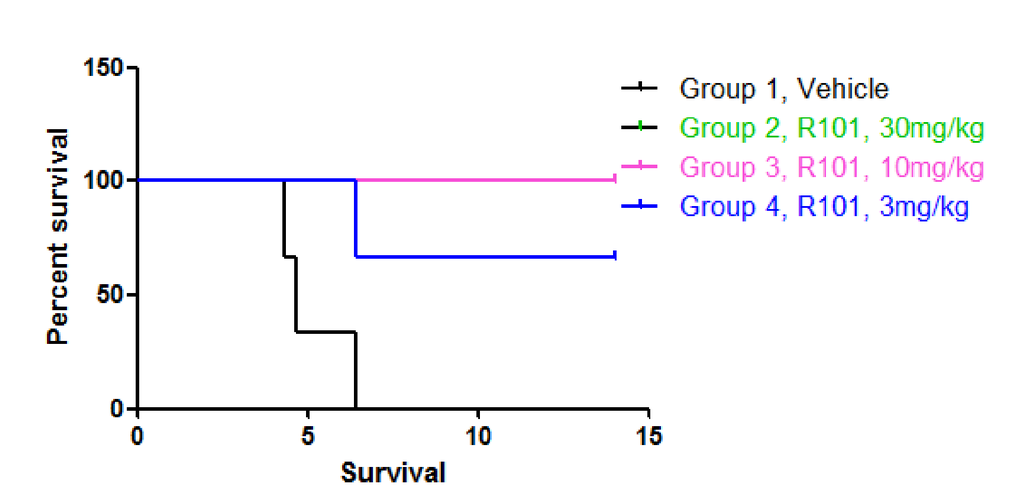
Group 1, Vehicle
Group 2, R101, 30mg/kg
Group 3, R101, 10mg/kg
Group 4, R101, 3mg/kg
Percent survival
Survival

Figure 9. CR1520 survival curve (cutoff: $500mm^3$)

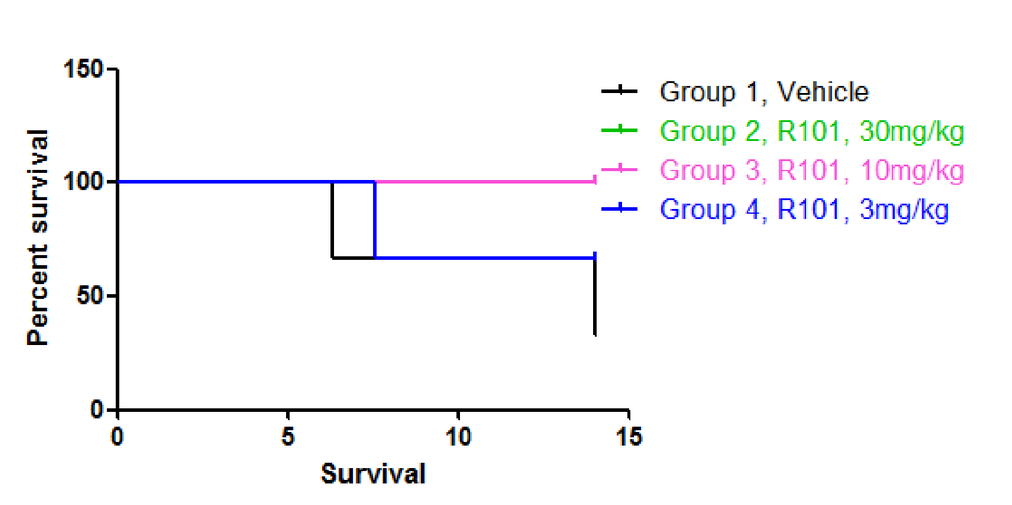
Group 1, Vehicle
Group 2, R101, 30mg/kg
Group 3, R101, 10mg/kg
Group 4, R101, 3mg/kg
Percent survival
Survival

Figure 10. CR2518 survival curve (cutoff: $500mm^3$)

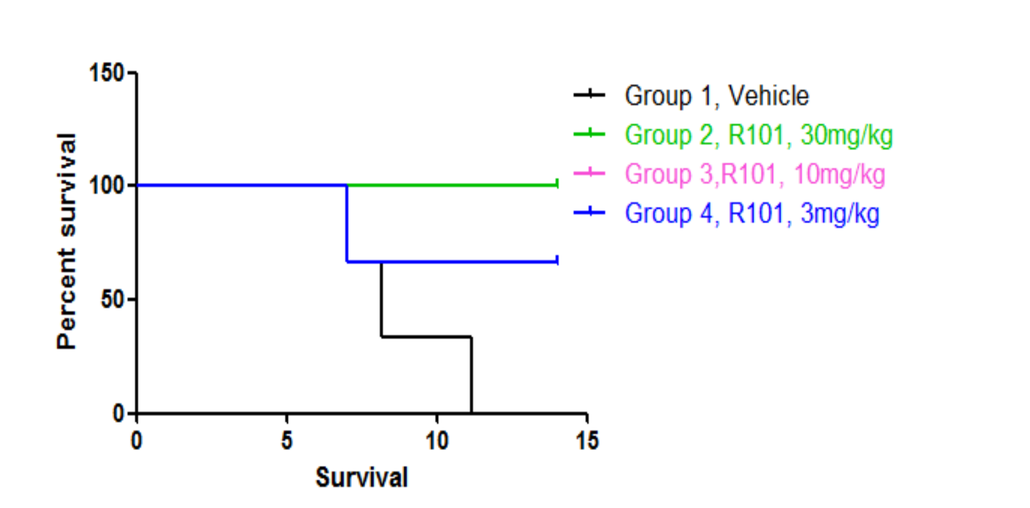
Group 1, Vehicle
Group 2, R101, 30mg/kg
Group 3,R101, 10mg/kg
Group 4, R101, 3mg/kg
Percent survival
Survival

Figure 11. CR2545 survival curve (cutoff: $500mm^3$)

Table 1. Patients information

ID	Gender	Age	Pathology diagnosis
CR1520	M	82	Moderately-poorly differentiated adenocarcinoma (Pa, P2)
CR2518	F	82	Poorly differentiated adenocarcinoma (Pa, P3).

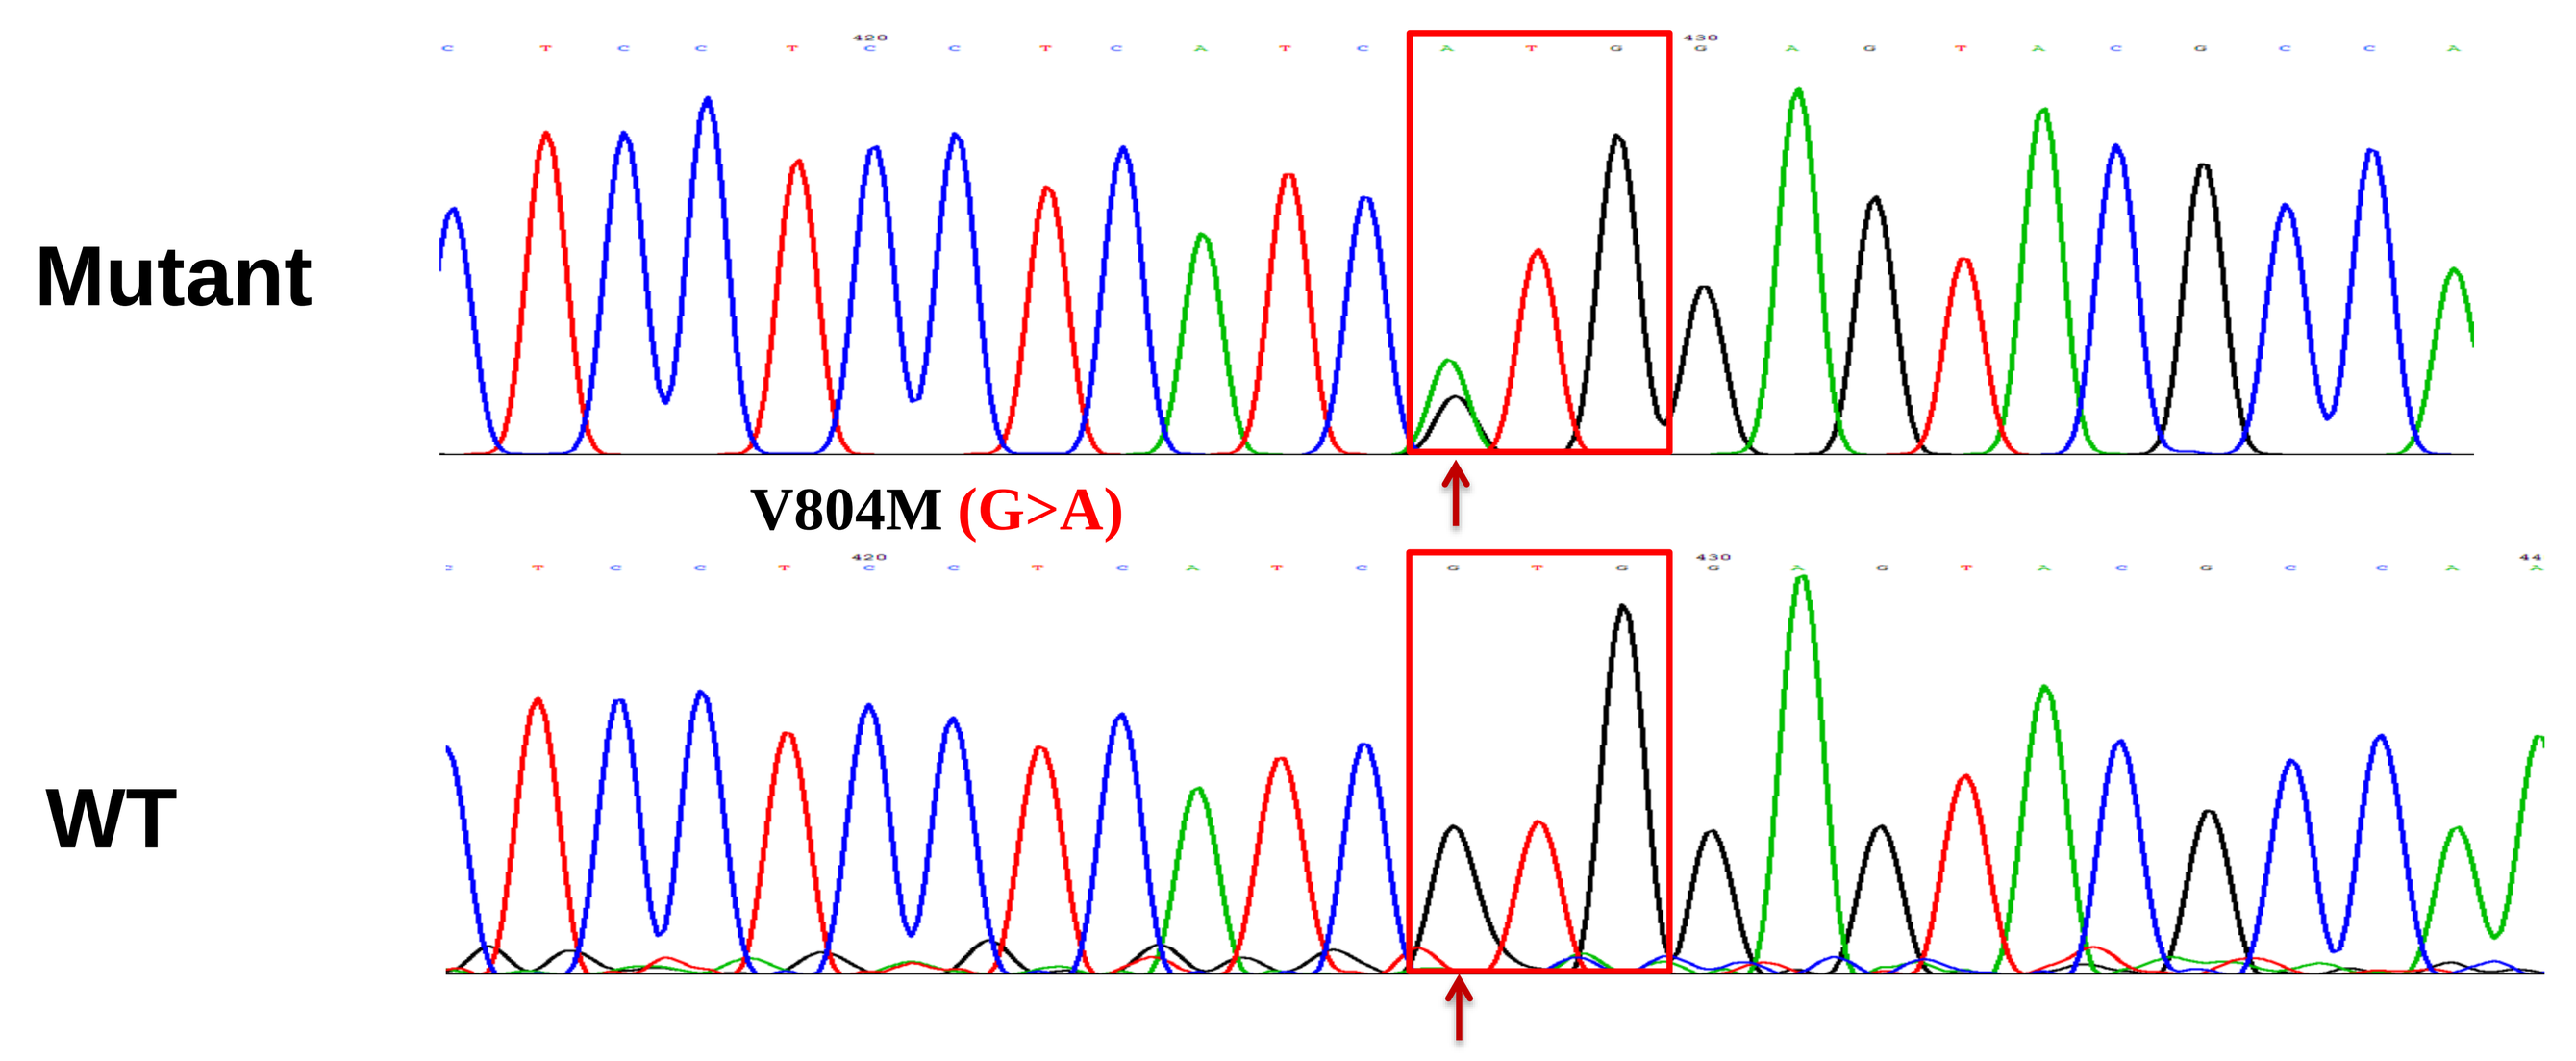
Mutant
V804M (G>A)
WT

Figure 12. RET kinase gate keeper V804M mutation is introduced in the ponatinib resistant CR2545

Conclusions

1. CRC-PDXs carry RET oncogene fusions: CR1520 (NCOA4-RET) and CR2518 (CCDC6-RET) and overexpress RET.
2. Both models are initially sensitive to the RET inhibitor ponatinib.
3. A Novel RET-TKI, R101, showed strong antitumor activity in both models.
4. R101 overcame ponatinib (AP24534) induced resistance in model CR2545.
5. R101 inhibits the wild-type RET enzyme with IC_{50} of 1.6e-7 M. R101 inhibits RET at IC_{50} of 5.6e-7M (similar to ponatinib) in 2D assay on the PrimePanel CR1520. R101 inhibits HUVEC at IC_{50} of 5e-6M (20x less toxic than ponatinib).
6. RNAseq reveals that a classic gate keeper mutation is introduced into the ponatinib resistant CR2545 model after ponatinib exposure.

For more information contact HenryLi@crownbio.com