Dynamic Changes of Betatrophin, Insulin and Glucose during STZ Induction of Diabetes in Rhesus Monkeys

Yongqiang Liu, Bingdi Wang, Guofeng Sun, Xunhua Ding, Xiaoli Wang, Keefe Chng, Francine Gregoire, Yi-Xin (Jim) Wang, Yong-Fu Xiao.

Crown Bioscience, Inc. Taicang, China and Crown Bioscience North Carolina Inc, NC USA

In life, for life

Abstract

Betatrophin, also called angiopoietin-like protein 8/lipasin, characterized from the liver can increase β-cells proliferation and enhance insulin secretion in several animal studies. However, its effect on human β-cells has not been clearly evidenced. This study was to investigate correlation of betatrophin and β-cell proliferation in nonhuman primates (NHPs, rhesus monkeys) with normal blood glucose (<80 mg/dL) before streptozocin (STZ) treatment. In the STZ-treated NHPs (n=7), both blood glucose and betatrophin increased dramatically. Blood insulin initially increased on day 1 after STZ injection and then decreased dramatically. Two NHPs with blood glucose back to ~80 mg/dL on day 7 after STZ injection showed the recovery of insulin secretion and responded ivGTT toward the pre-treatment level. In another 2 STZ-treated NHPs with blood glucose >250 mg/dL, proliferation of both β-cell and non-β-cell was observed in their islets. The rest 2 NHPs with more completed β-cell destruction after STZ treatment showed much higher blood glucose (>300 mg/dL) with no obvious β-cell regeneration and ivGTT showed almost no insulin secretion. Our data demonstrate that blood betatrophin rised parallelly with glucose elevation during STZ induction of diabetes in NHPs. Such increase in blood betatrophin could lead to β-cell regeneration in some animals, 5 out of 7, but not in those NHPs with severely destroyed islets, 2 out of 7. Our data may explain, at least partially, why circulating betatrophin level is elevated in patients with type 2 diabetes. Betatrophin may play an important role in the early stage of type 2 diabetes.

Introduction

Regenerating pancreatic islets/β-cells is a novel approach for diabetes therapy. Betatrophin, a polypeptide secreted by liver or adipose tissue, stimulates islet cell proliferation and enhancing insulin production in rodents reported by Dr. Melton's lab. In this study, we explored if this discovery in rodents could also be observed in nonhuman primates (NHPs). We evaluated the correlations among the levels of blood betatrophin, glucose and insulin after vehicle or STZ treatment. We also tested the β-cell proliferation in animals with or without the change of plasma betatrophin contents.

Methods

Blood sampling and assay: Blood samples were collected from overnight fasted monkeys under the conscious condition. Blood betatrophin levels were tested via commercial available betatrophin ELISA kits (Wuhan, China).
Tissue collection: Pancreas and liver tissues from 8 rhesus macaques with or without STZ treatment were used for evaluating β-cell proliferation and betatrophin transcription level. Ki67 was used for cell proliferation marker. Liver biopsy was conducted once before STZ treatment.
Betatrophin transcription levels: Total RNA was extracted from the liver of the NHPs and reverse transcribed into cDNA. Amplification of betatrophin and the internal control of GAPDH were conducted on Stratagene Mx3005p (Agilent Technologies) using the SYBR-green reaction system. Relative quantity of betatrophin was calculated by the ΔΔCT method.
Islet and insulin content evaluation: Serial sections of the pancreas were made through FFPE blocks for IHC for quantification of insulin contents.
Biochemistry test: Conducted in Taicang city hospital lab
Data analysis: Data are presented as mean ± SEM.

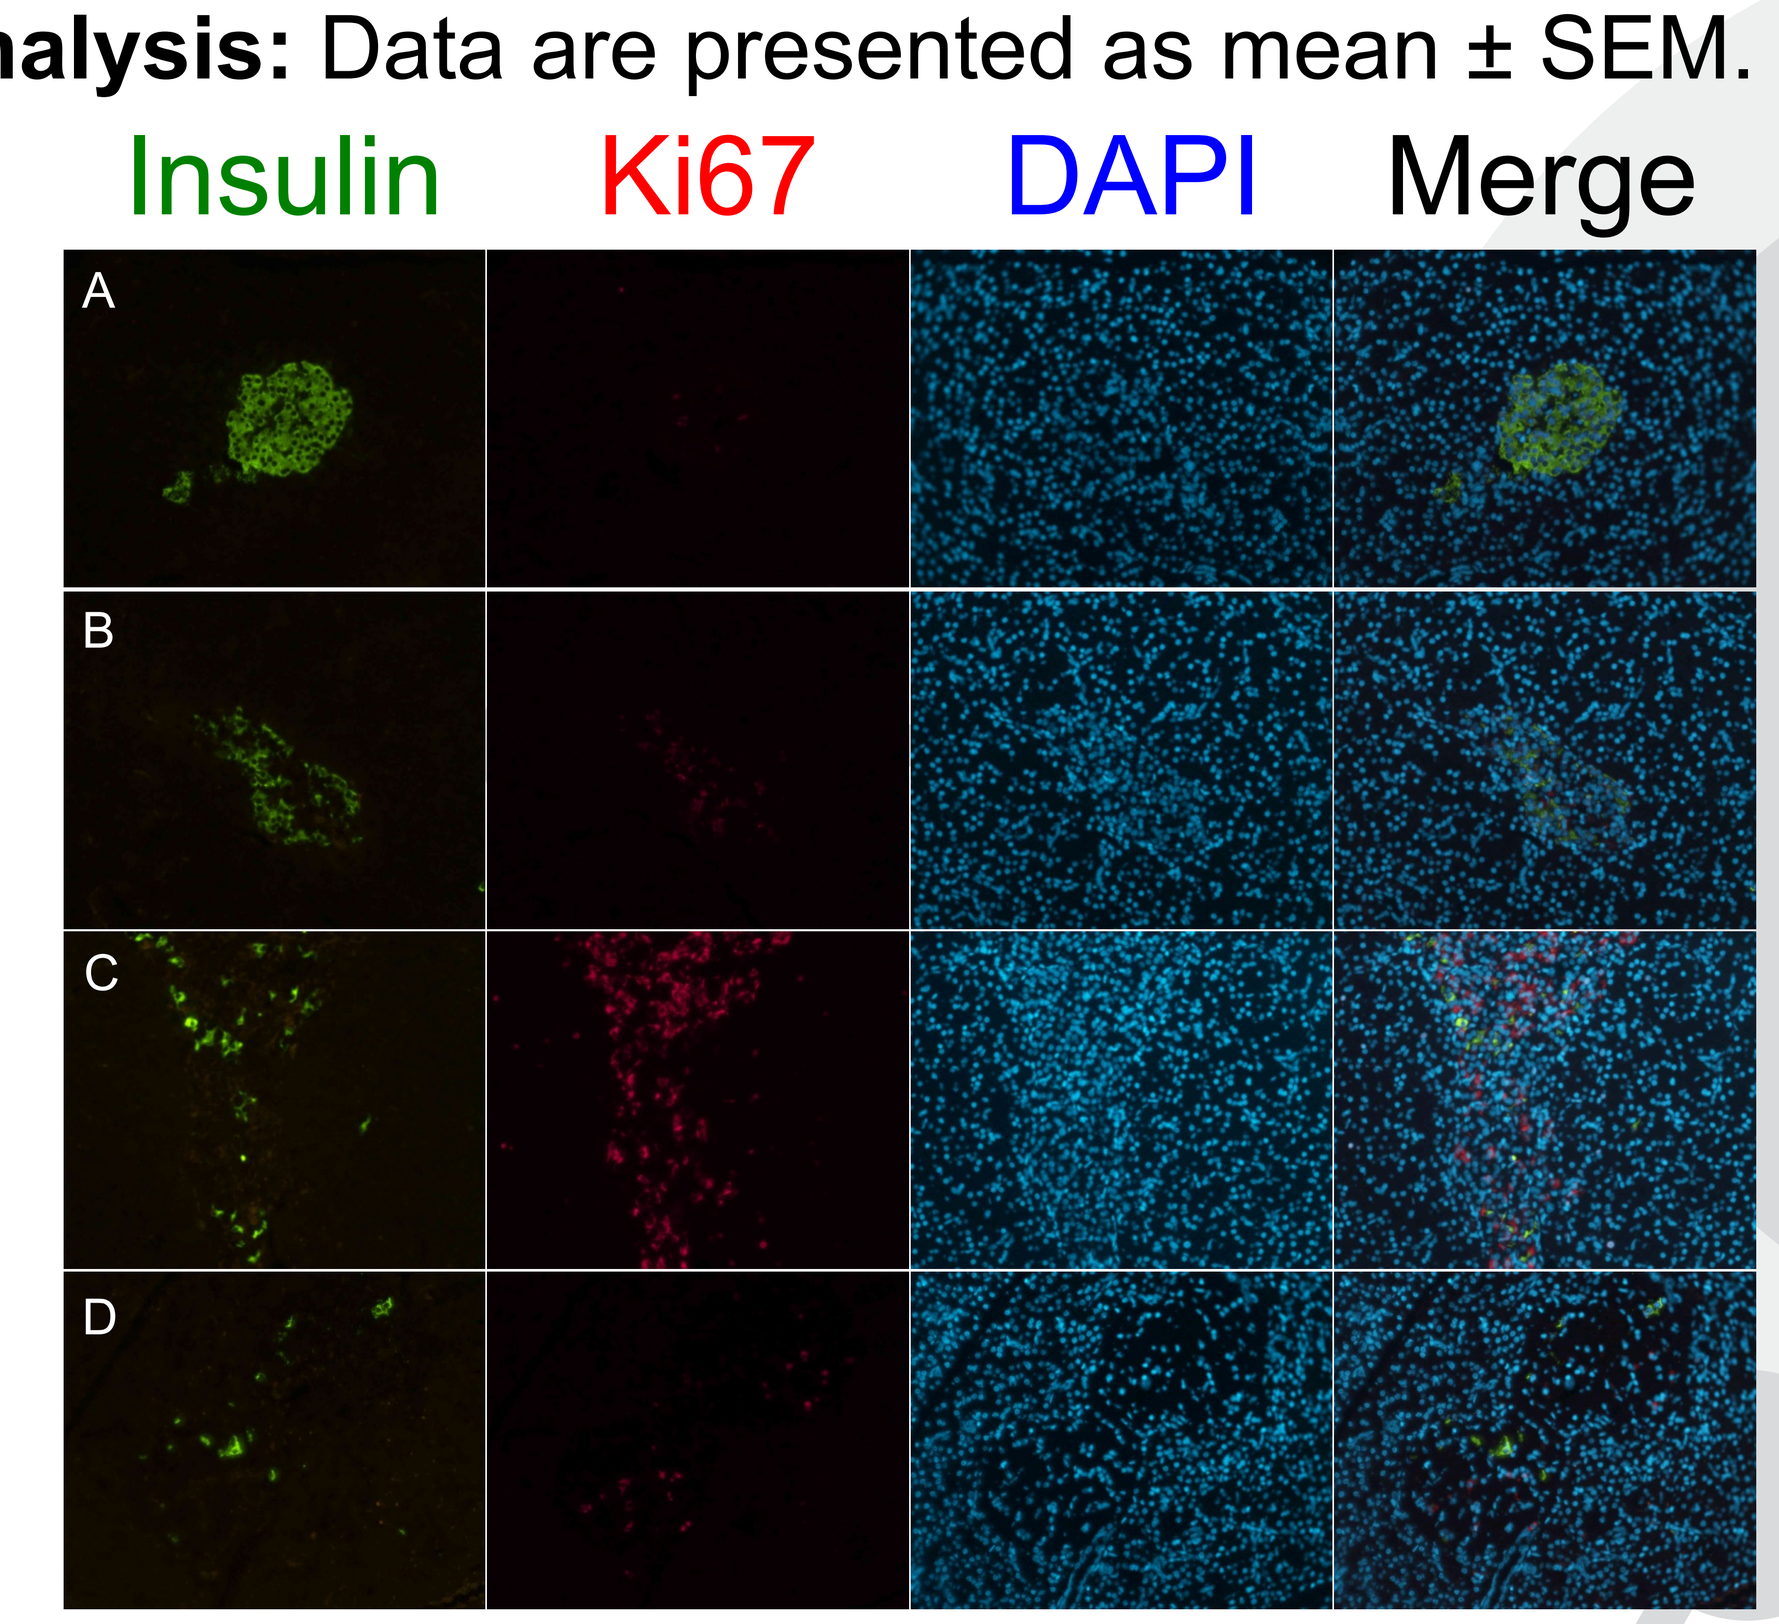
Insulin
Ki67
DAPI
Merge
A
B
C
D

Fig. 3. Immunohistological staining of islets against insulin, Ki67, DAPI. A. Islets of vehicle treated NHP models showed rare β-cell proliferation **B.** Islets of two STZ induced NHP models with a renormalized blood glucose level, from 120mg/dL on day 1 STZ treatment to 80mg/dL 7 days and 65mg/dL 14 days after STZ treatment. **C.** Islets of another 2 STZ treated animals with higher blood glucose, >300mg/dL, more cell proliferation was observed in the islets but without blood glucose recovery. **D.** Islets of the rest 2 STZ treated animals with >300 mg/dL blood glucose whose β-cells are severely destroyed showed little cell proliferation.

Results

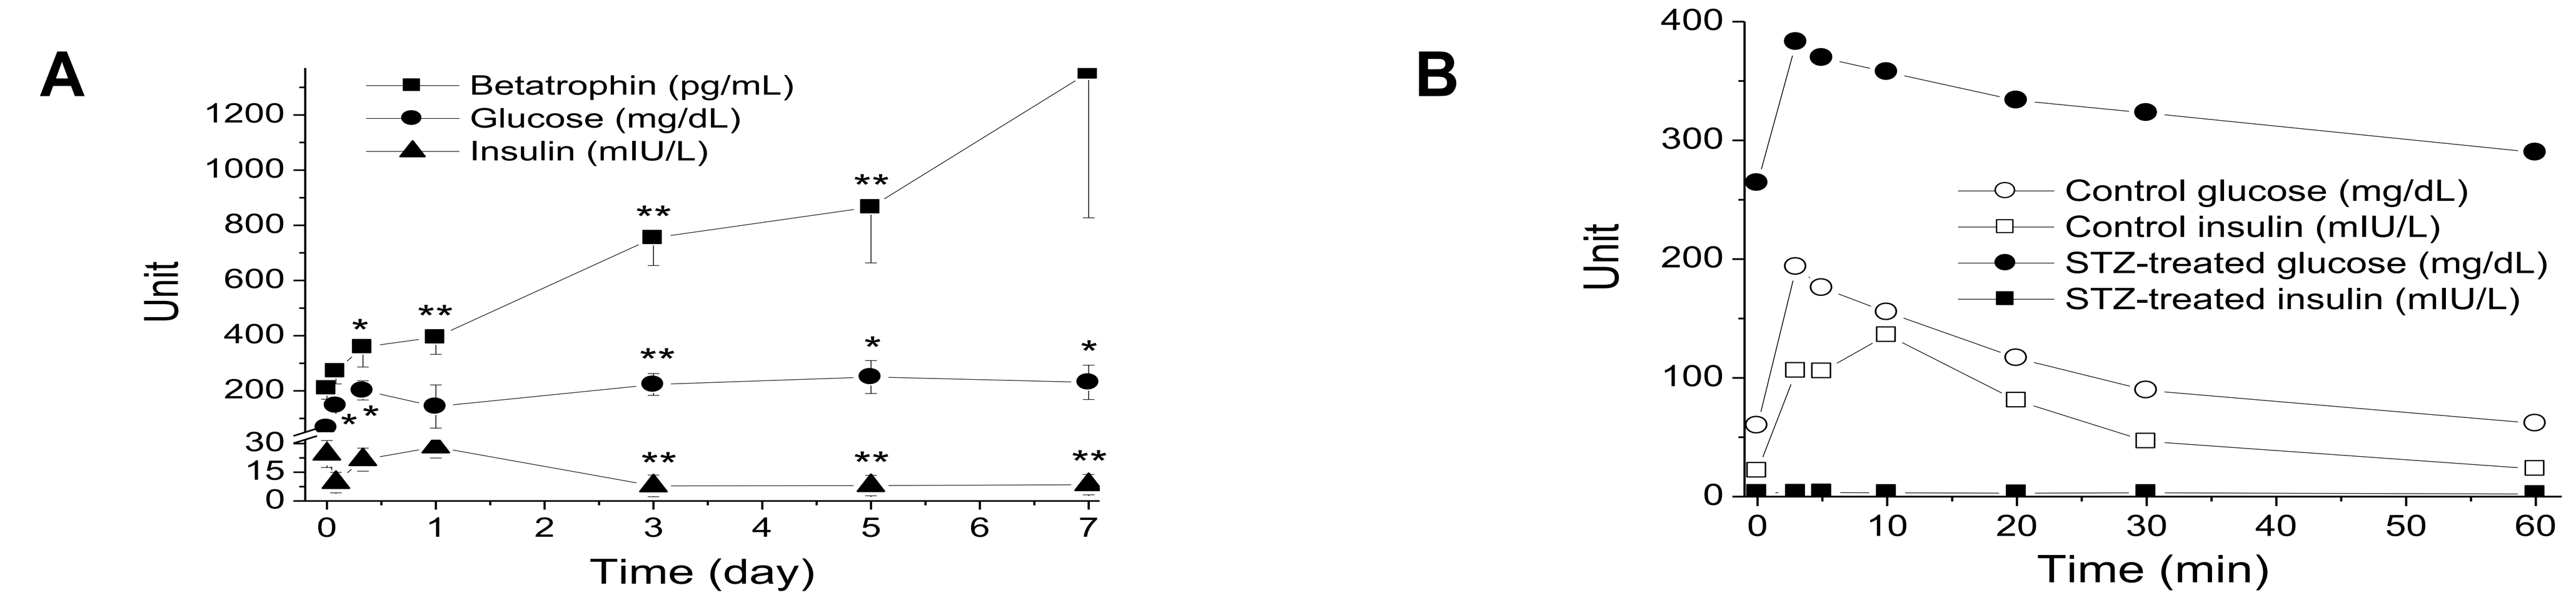
A
Betatrophin (pg/mL)
Glucose (mg/dL)
Insulin (mIU/L)
Unit
Time (day)
B
Control glucose (mg/dL)
Control insulin (mIU/L)
STZ-treated glucose (mg/dL)
STZ-treated insulin (mIU/L)
Unit
Time (min)

Fig. 1. Parameter changes after STZ treatment
A. Elevation of blood glucose and betatrophin during the diabetic induction by intravenous injection of STZ in NHPs (n=6).
B. Changes of blood glucose and insulin during ivGTT conducted before (Control) and day 7 after STZ treatment (STZ-treated) in one representative monkey.

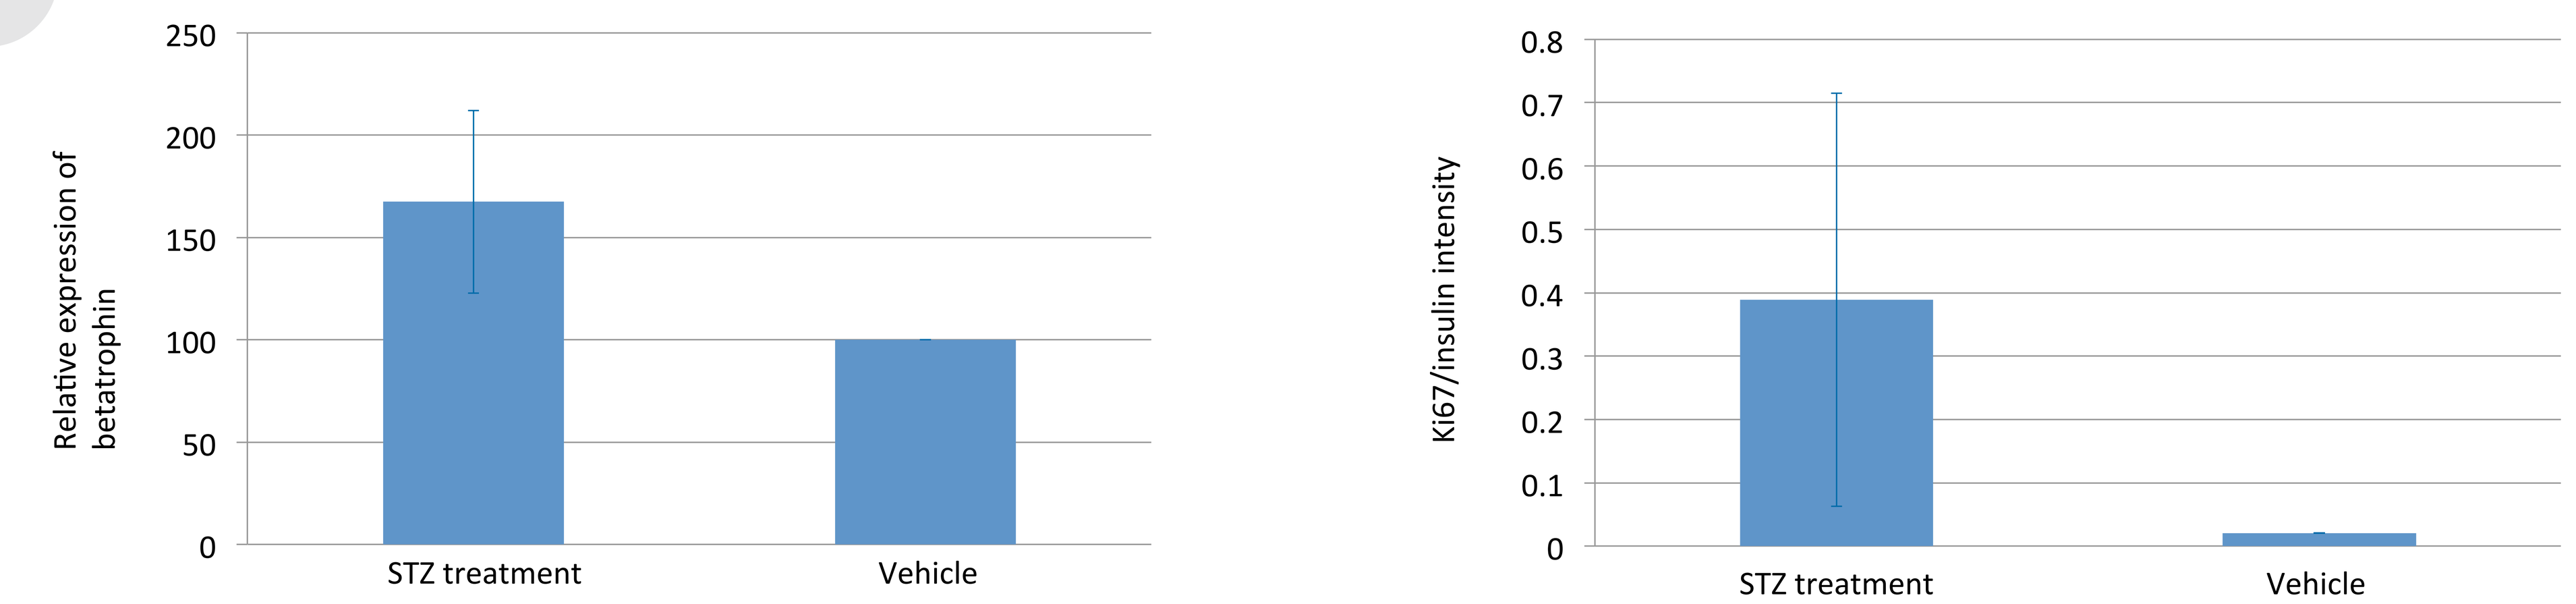
Relative expression of betatrophin
STZ treatment
Vehicle
Ki67/insulin intensity
STZ treatment
Vehicle

Fig. 2. Betatrophin expression and β-cell proliferation after STZ treatment.
A. Relative betatrophin expression in the livers of vehicle and STZ treated animals.
B. β-cell proliferation rates in the vehicle and STZ-treated animals

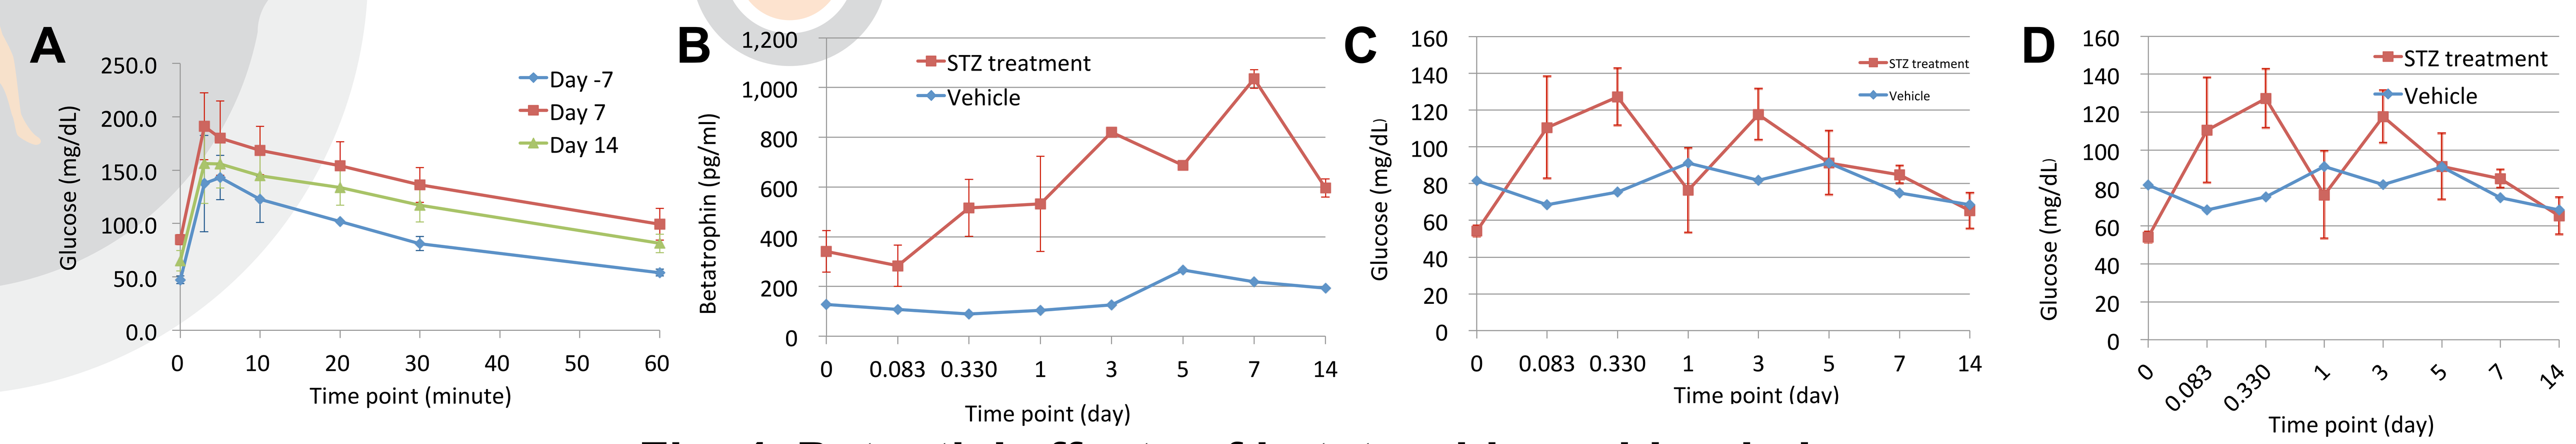
A
Day -7
Day 7
Day 14
Glucose (mg/dL)
Time point (minute)
B
STZ treatment
Vehicle
Betatrophin (pg/ml)
Time point (day)
C
STZ treatment
Vehicle
Glucose (mg/dL)
Time point (day)
D
STZ treatment
Vehicle
Glucose (mg/dL)
Time point (day)

Fig. 4. Potential effects of betatrophin on blood glucose
A.IVGTT of vehicle treated animal. **B.** IVGTT of 2 STZ-treated animals.
C. Betatraophin plasma level of 3 STZ/ vehicle treated animals **D.** Blood glucose of 3 STZ/vehicle treated animals.

Conclusions

1. **Blood betatrophin was detected in NHPs and its mRNA level in liver and protein level in plasma increased after STZ treatment.**
2. **Increased betatrophin level in plasma after STZ treatment shows a high correlation between the blood glucose level and beta cell proliferation.**
3. **Beta-cell regeneration enhanced by betatrophin seems to require a minimal base level of pancreatic β-cell or islets.**
4. **Betatrophin may work better at pre-diabetes and the early stage of diabetes.**