CrownBio
CONNECTING SCIENCE TO PATIENTS

Using HuTrial™ To Aid Predicting Response in a Patient Population

Jayant Thatte, Jill Ricono, Colleen Scott, and Thomas Broudy
Crown Bioscience, Inc., San Diego, 11011 Torreyana Road, Suite 200, San Diego, CA 92121, USA

Abstract No. 5186

INTRODUCTION

One of the biggest challenges in drug discovery is directing drug treatment to the correct population of patients. The CrownBio collection of patient-derived xenograft (PDX) models – HuPrime® – currently counts more that 2,500 models over more than 70 indications. These human surrogate mouse models, are reflective of the diversity of genetics, tumor heterogeneity, and response to therapy observed in the patient population. HuPrime models are searchable in HuBase™, CrownBio's curated database including gene expression, copy number, mutational analysis as well as patient clinical data comprising diagnosis, pathology, and IHC information. The vast number of models available allows researchers to perform surrogate Phase II-like trials, utilizing one patient sample per tumor bearing animal per treatment group (n=1 studies). In this study we show reproducibility of n=1 studies compared to traditional preclinical mouse studies consisting of multiple animals grafted with the same tumor and treated with the same test agent. Furthermore, we show that the use of one patient sample per treatment allows the expansion of the study, utilizing samples from multiple patients, thereby reducing the cost of preclinical studies. HuPrime models can be leveraged to discover biomarkers, identify patient responder and non-responder profiles to accelerate drug development and improve chances of success in the clinic.

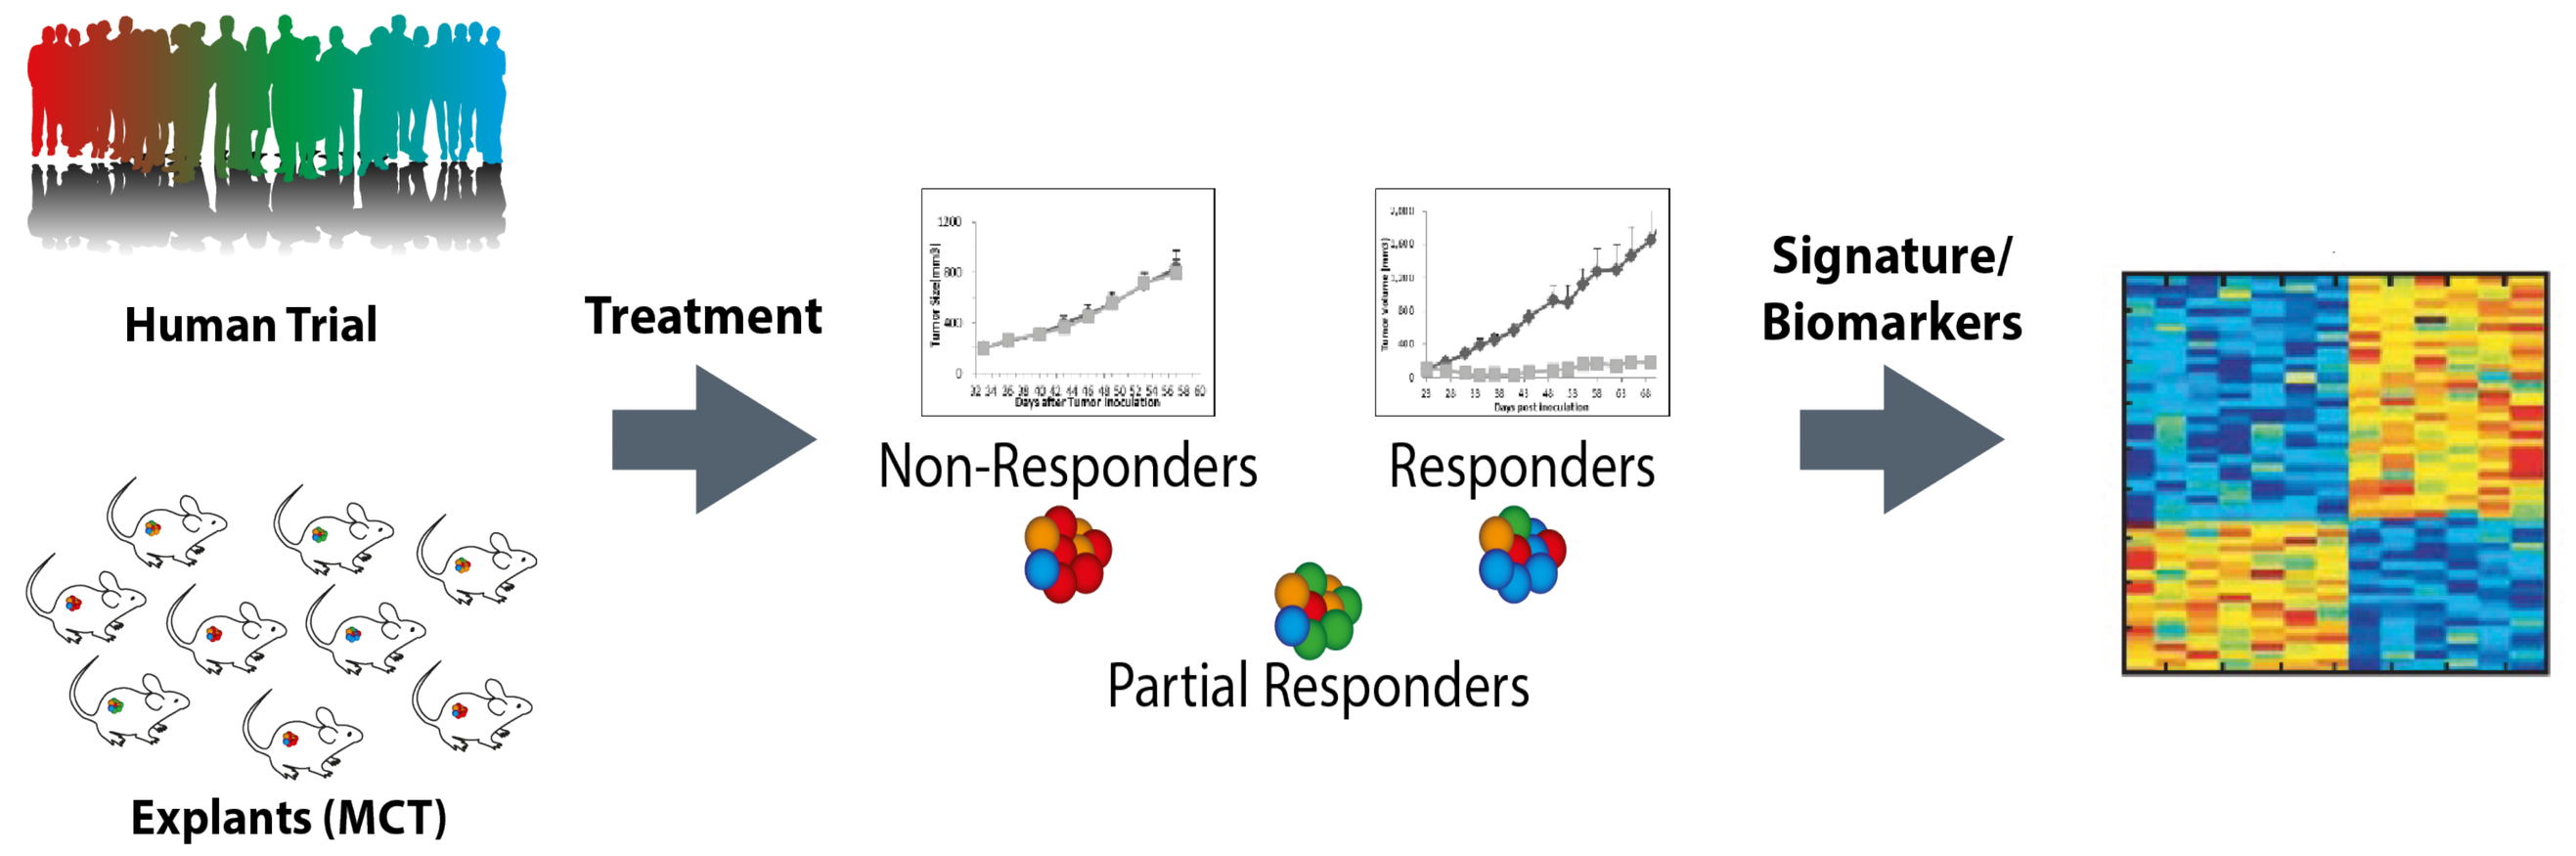
Human Trial
Explants (MCT)
Treatment
Non-Responders
Responders
Partial Responders
Signature/
Biomarkers

STUDY DESIGN

27 SCLC and 18 NSCLC PDX models were evaluated in this study. Tumors were inoculated subcutaneously in the rear flank of NOD-SCID mice. For each model, 1 animal served as control and 1 animal was treated with standard of care (SoC) agents. For validation of the n=1 SCLC study, we evaluated 4 models in a n=5 per group format. Tumor volume and body weight measurements were taken 3 times a week for all studies.

MODEL INFORMATION

SCLC

Model ID	Sex	Age	Biopsy Site	Prior Treatment
LU5166	M	--	Pleural fluid	Pretreated
LU5173	M	68.9	Right upper lobe lung	Naïve
LU5178	F	43.9	Right thoracentesis fluid	Naïve
LU5188	F	74.5	Left axillary mass	Naïve
LU5192	F	56.6	Chest wall	Pretreated
LU5195	M	60.9	Right pleural fluid	Pretreated
LU5202	F	69.8	Left lung nodule	Naïve
LU5203	F	46.7	Left neck mass	Pretreated
LU5206	F	77	Right temporal lobe	Naïve
LU5207	M	53.3	Paracentesis fluid	Pretreated
LU5215	F	59.1	Brain	Naïve
LU5218	M	36.7	Left upper quadrant skin nodule	Pretreated
LU5220	M	65	Left upper lobe lung	--
LU5222	F	59.8	Left ovary	Pretreated
LU5223	F	60.3	Right neck mass	--
LU5224	F	48.9	Omentum	--
LU5228	F	55.8	Left upper lobe lung	--
LU5235	F	75.1	R4 lymph node	Naïve
LU5236	F	73.6	Left upper lobe lung	Naïve
LU5242	F	68.3	Right paraspine	Pretreated
LU5249	F	72.3	Chest wall	Pretreated
LU5250	M	63.8	Right upper lobe lung	Naïve
LU5255	M	59.1	Right lower lobe lung	Naïve
LU5263	F	71.5	Level 7 lymph node	Naïve
LU5264	M	16.8	Left pleura	Pretreated
LU5265	M	60.6	Chest wall	Pretreated
LU5267	M	52.8	Left adrenal gland	Pretreated

NSCLC

Model ID	Sex	Age	Biopsy Site	Prior Treatment
LU5138	M	69	Lung	--
LU5139	F	71	Lung	--
LU5140	M	70	Lung	--
LU5141	M	53	Brain	--
LU5142	F	66	Lung	--
LU5143	M	78	Lung	--
LU5144	M	74	Lung	--
LU5146	M	70	Lung	--
LU5147	M	65	Lung	--
LU5148	M	59	Lung	--
LU5177	F	77.7	Left upper lobe lung	Pretreated
LU5185	F	47.3	Breast lymph nodes	Pretreated
LU5190	M	69.4	Arm nodule	Pretreated
LU5204	M	36.7	Ascites fluid	Pretreated
LU5231	M	69.7	Right upper lobe lung	Naïve
LU5246	F	62	Left chest wall	Pretreated
LU5251	M	71.7	Left groin	Pretreated
LU5355	M	66.1	Brain	--

RESULTS

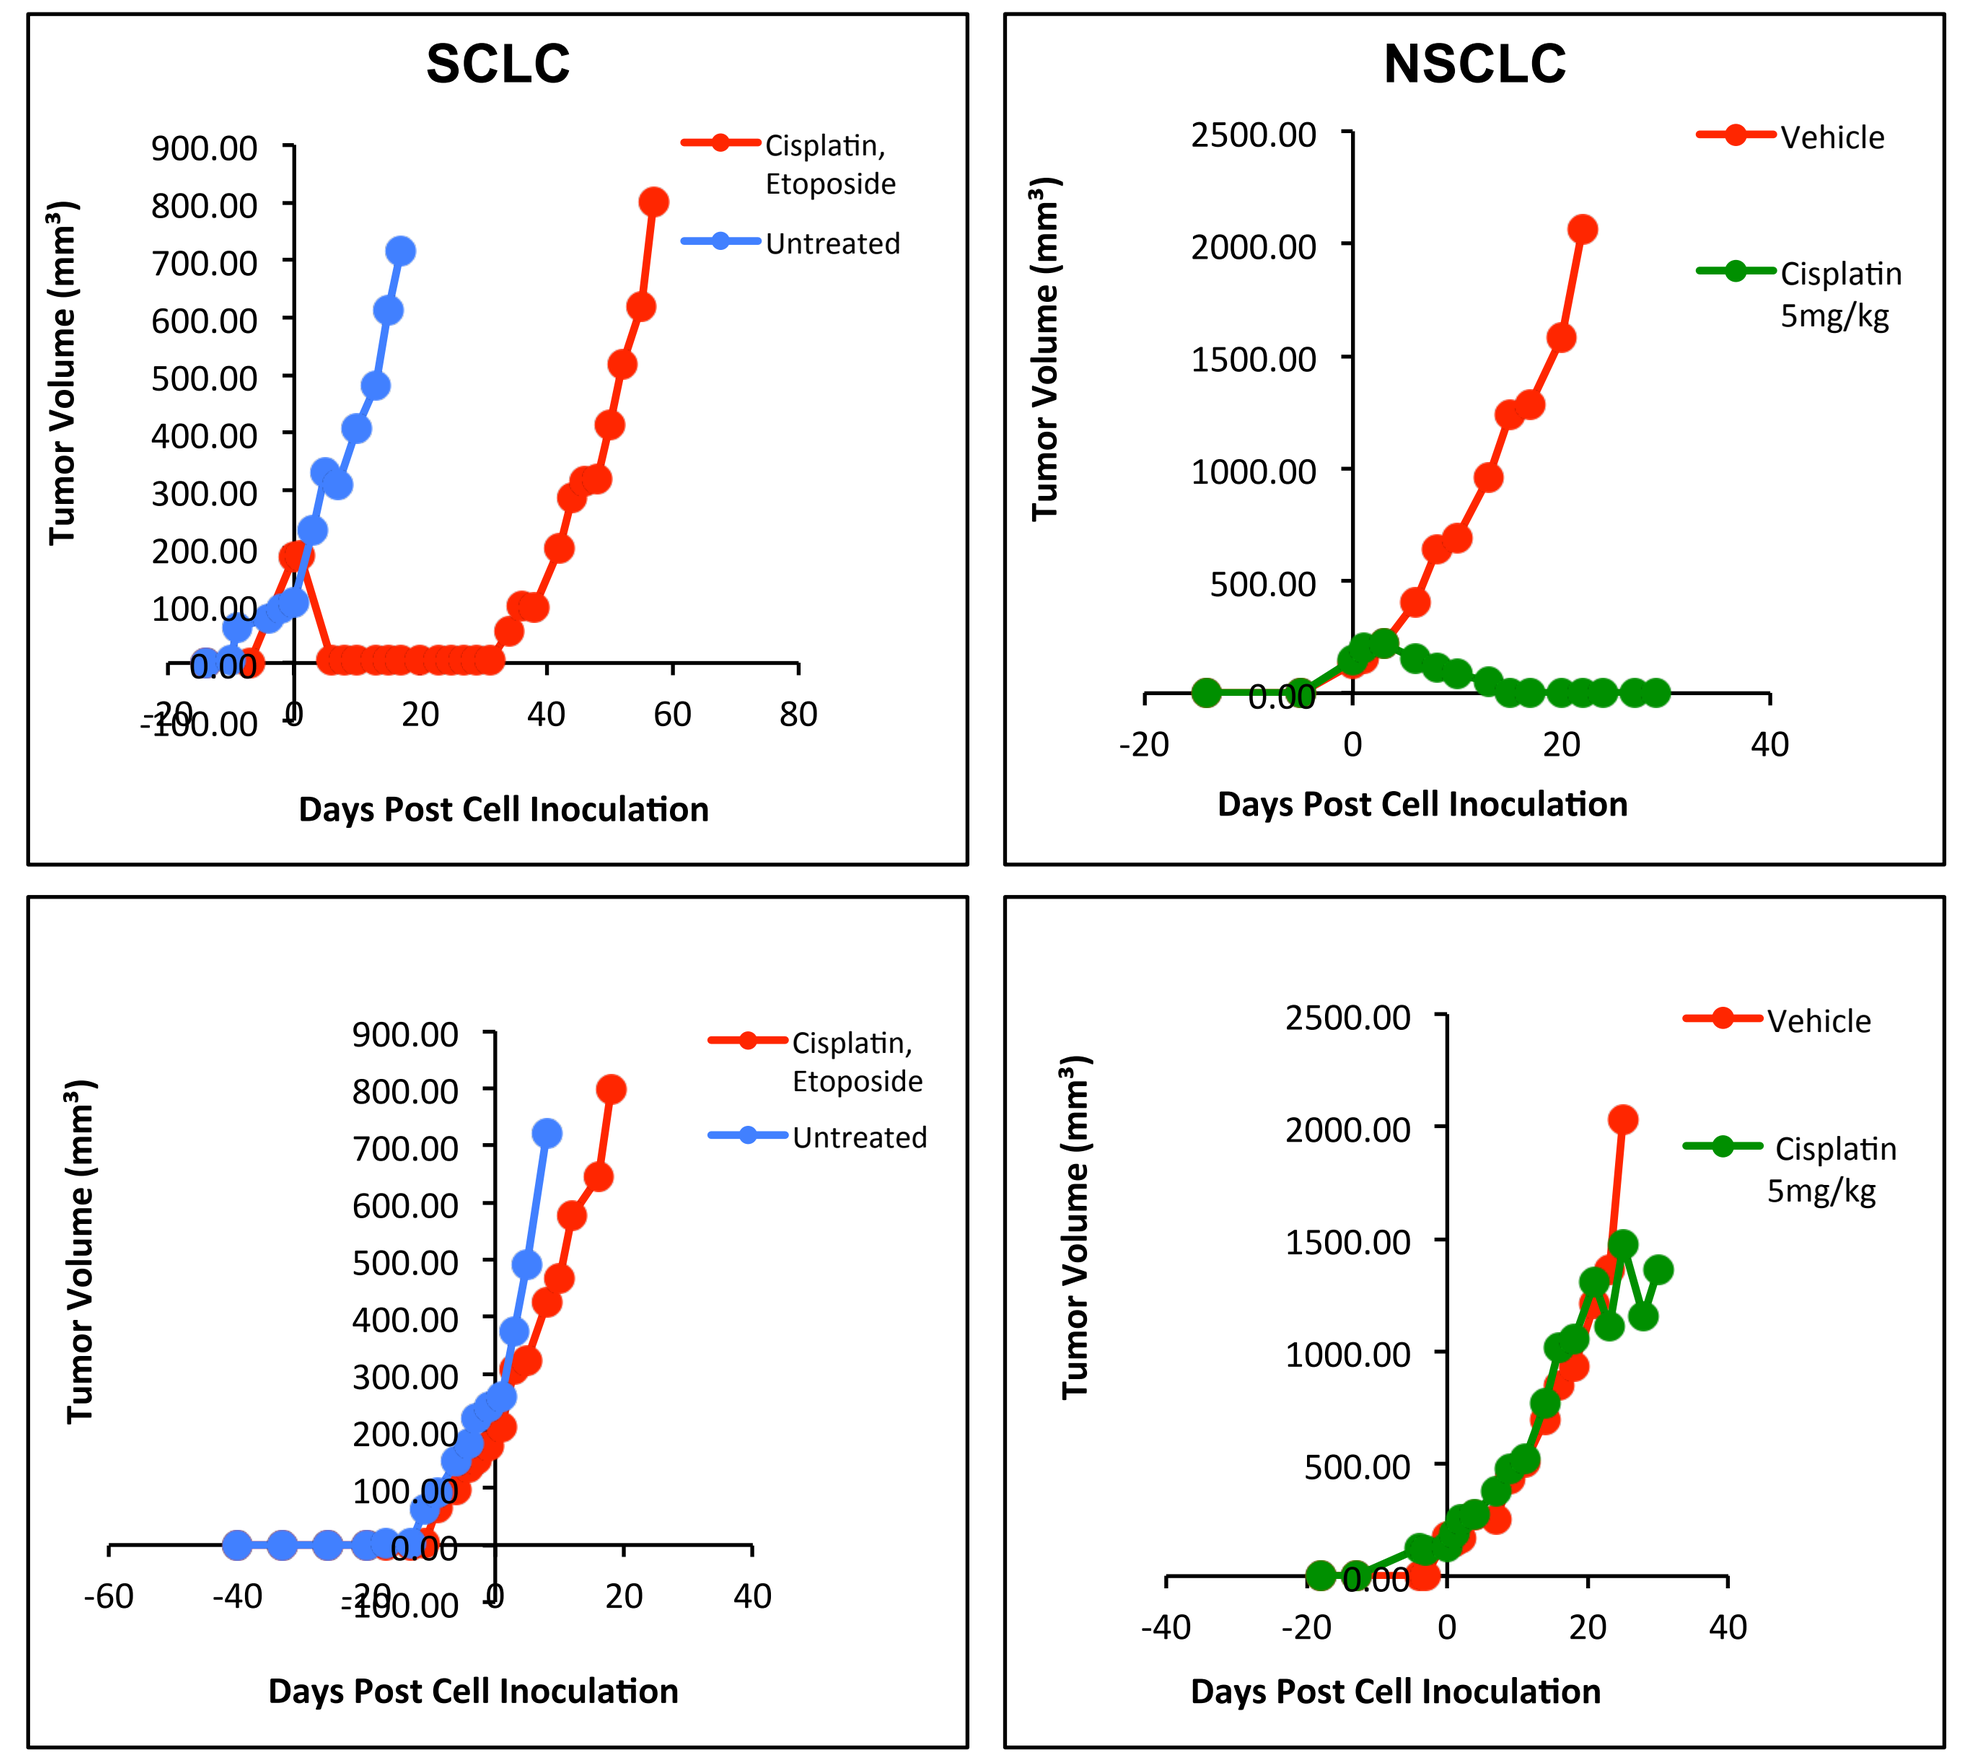
SCLC
NSCLC
Tumor Volume (mm³)
Days Post Cell Inoculation
Cisplatin, Etoposide
Untreated
Vehicle
Cisplatin 5mg/kg

Figure 1 (left): Representative SoC-sensitive (left panel) and SoC-resistant (right panel) models for SCLC and NSCLC indications. Cisplatin was administered on Day 1 and etoposide on Days 1,2,3.

Figure 2 (right): Waterfall plot for n=1 SoC R_x in SCLC and NSCLC models. Data plotted as %ΔT/ΔC. A wide response range was observed in both indications.

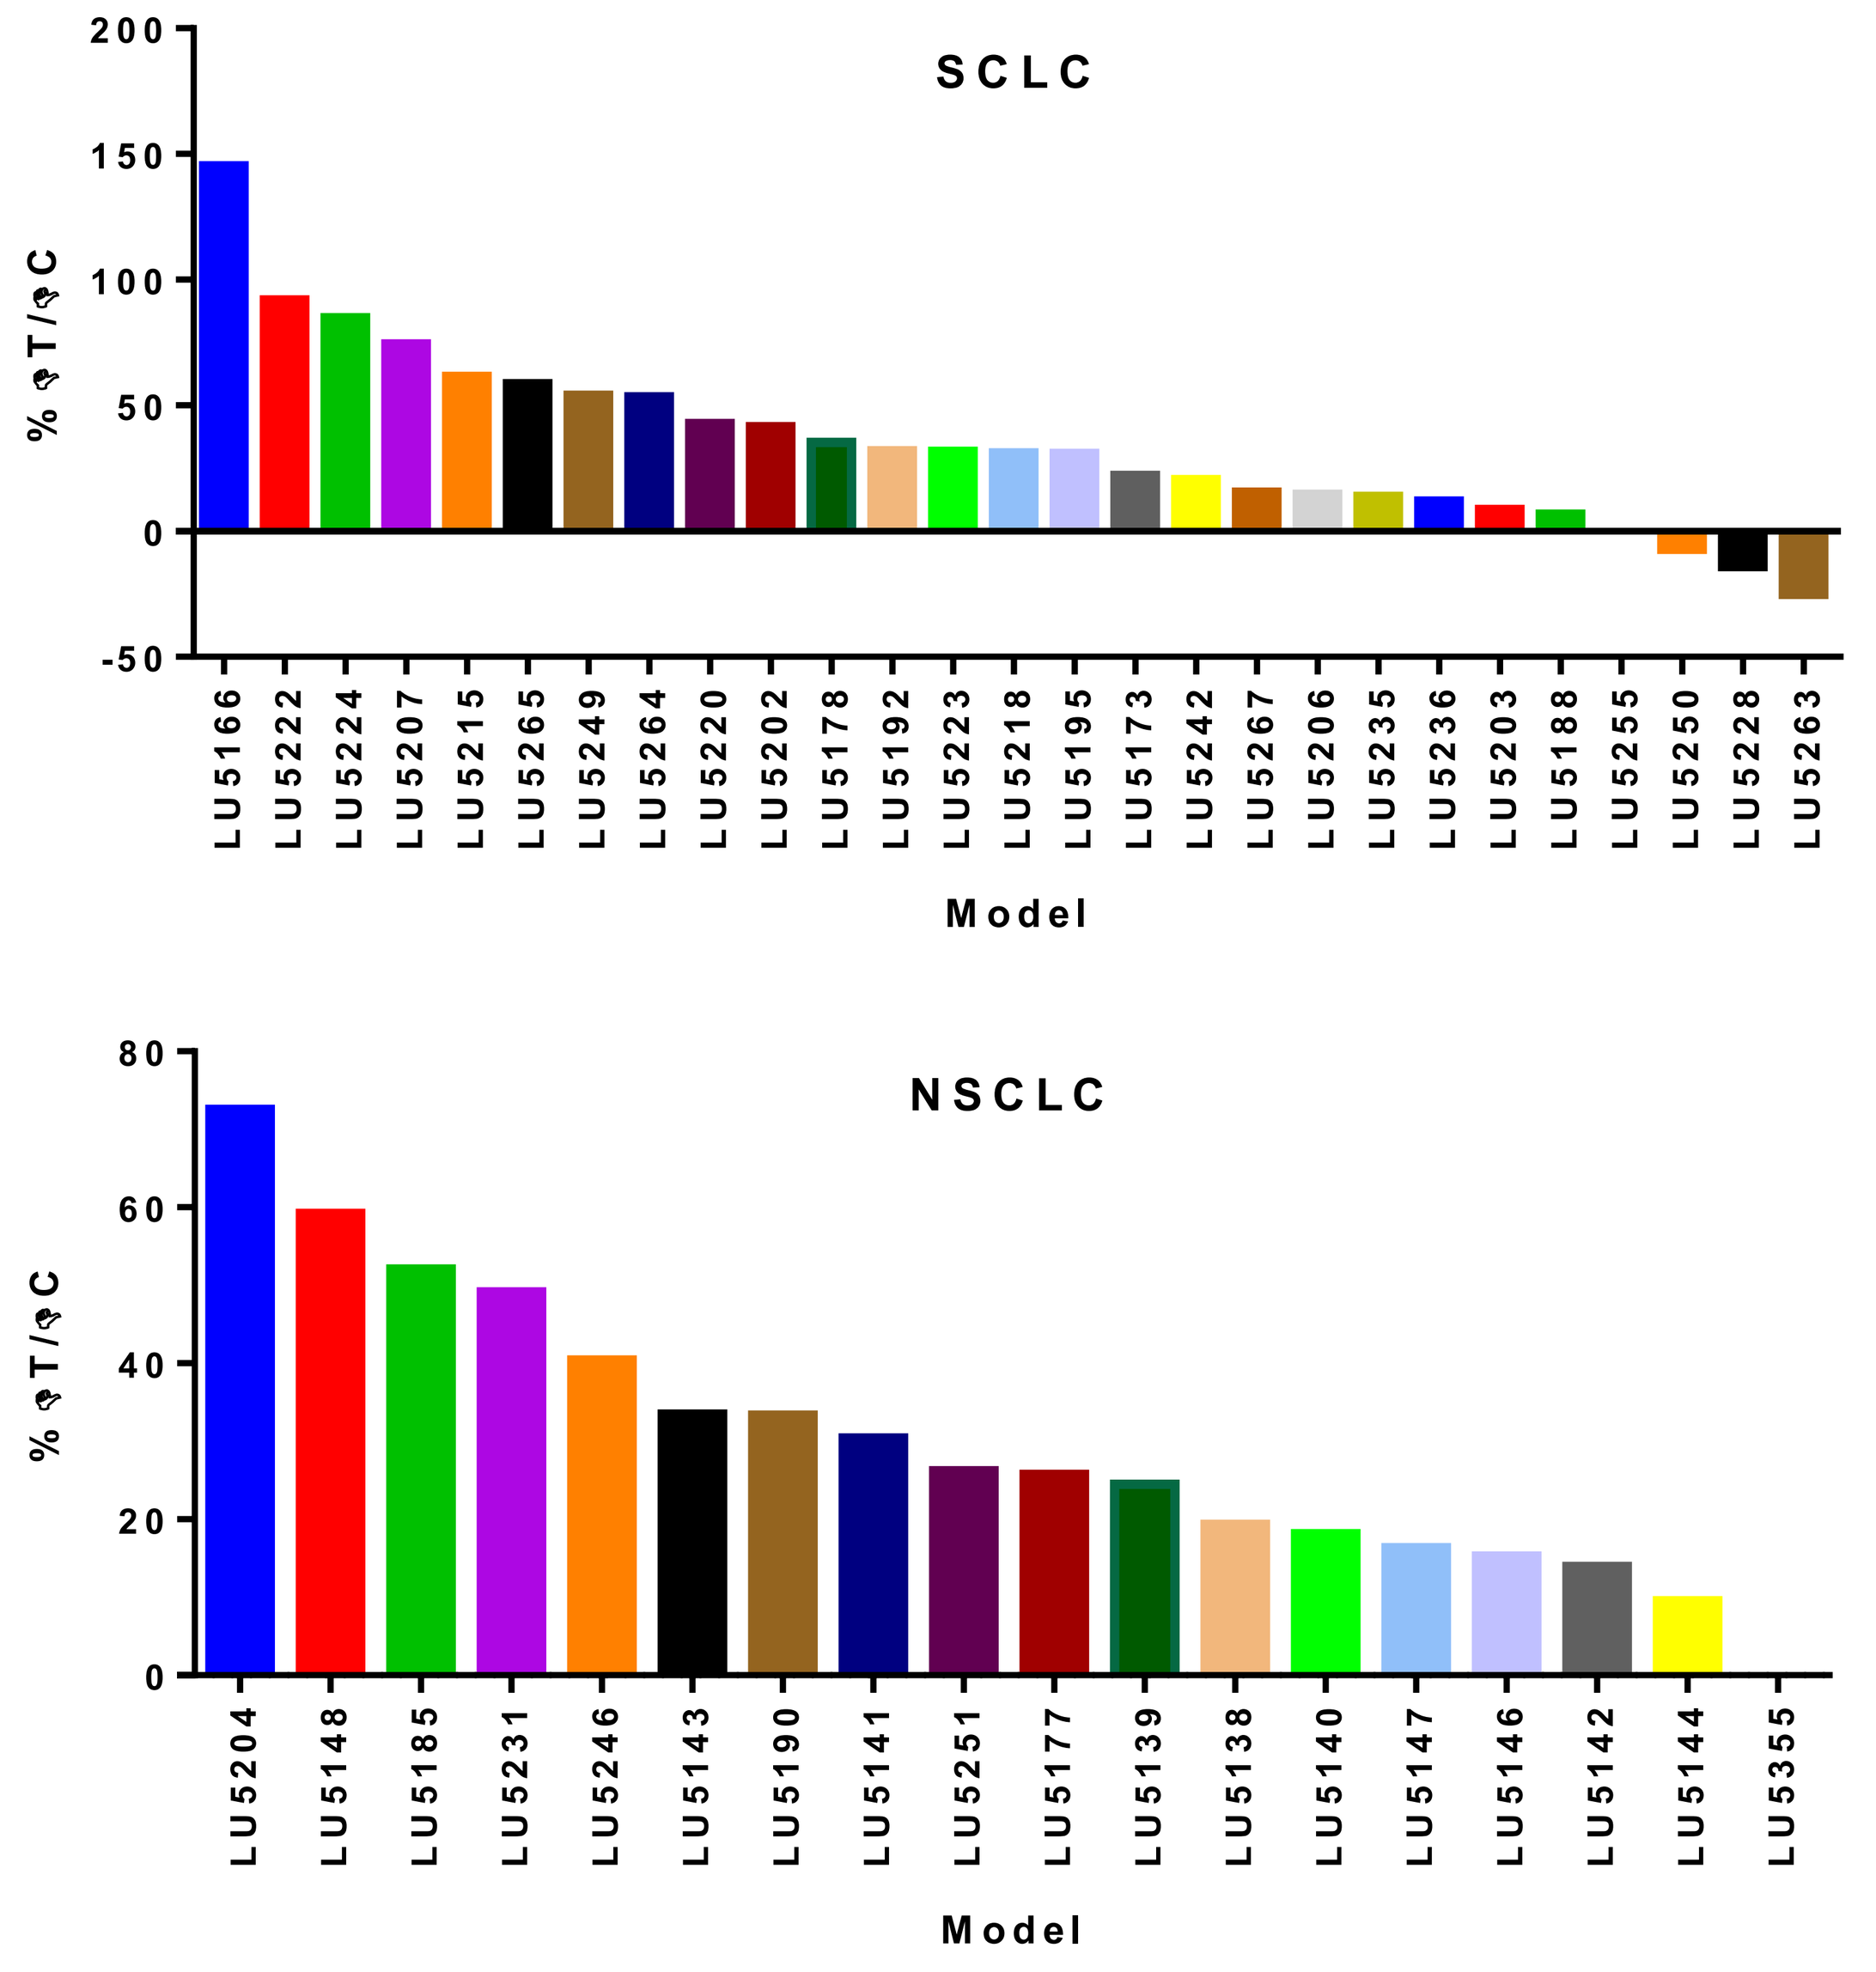
SCLC
NSCLC
Model

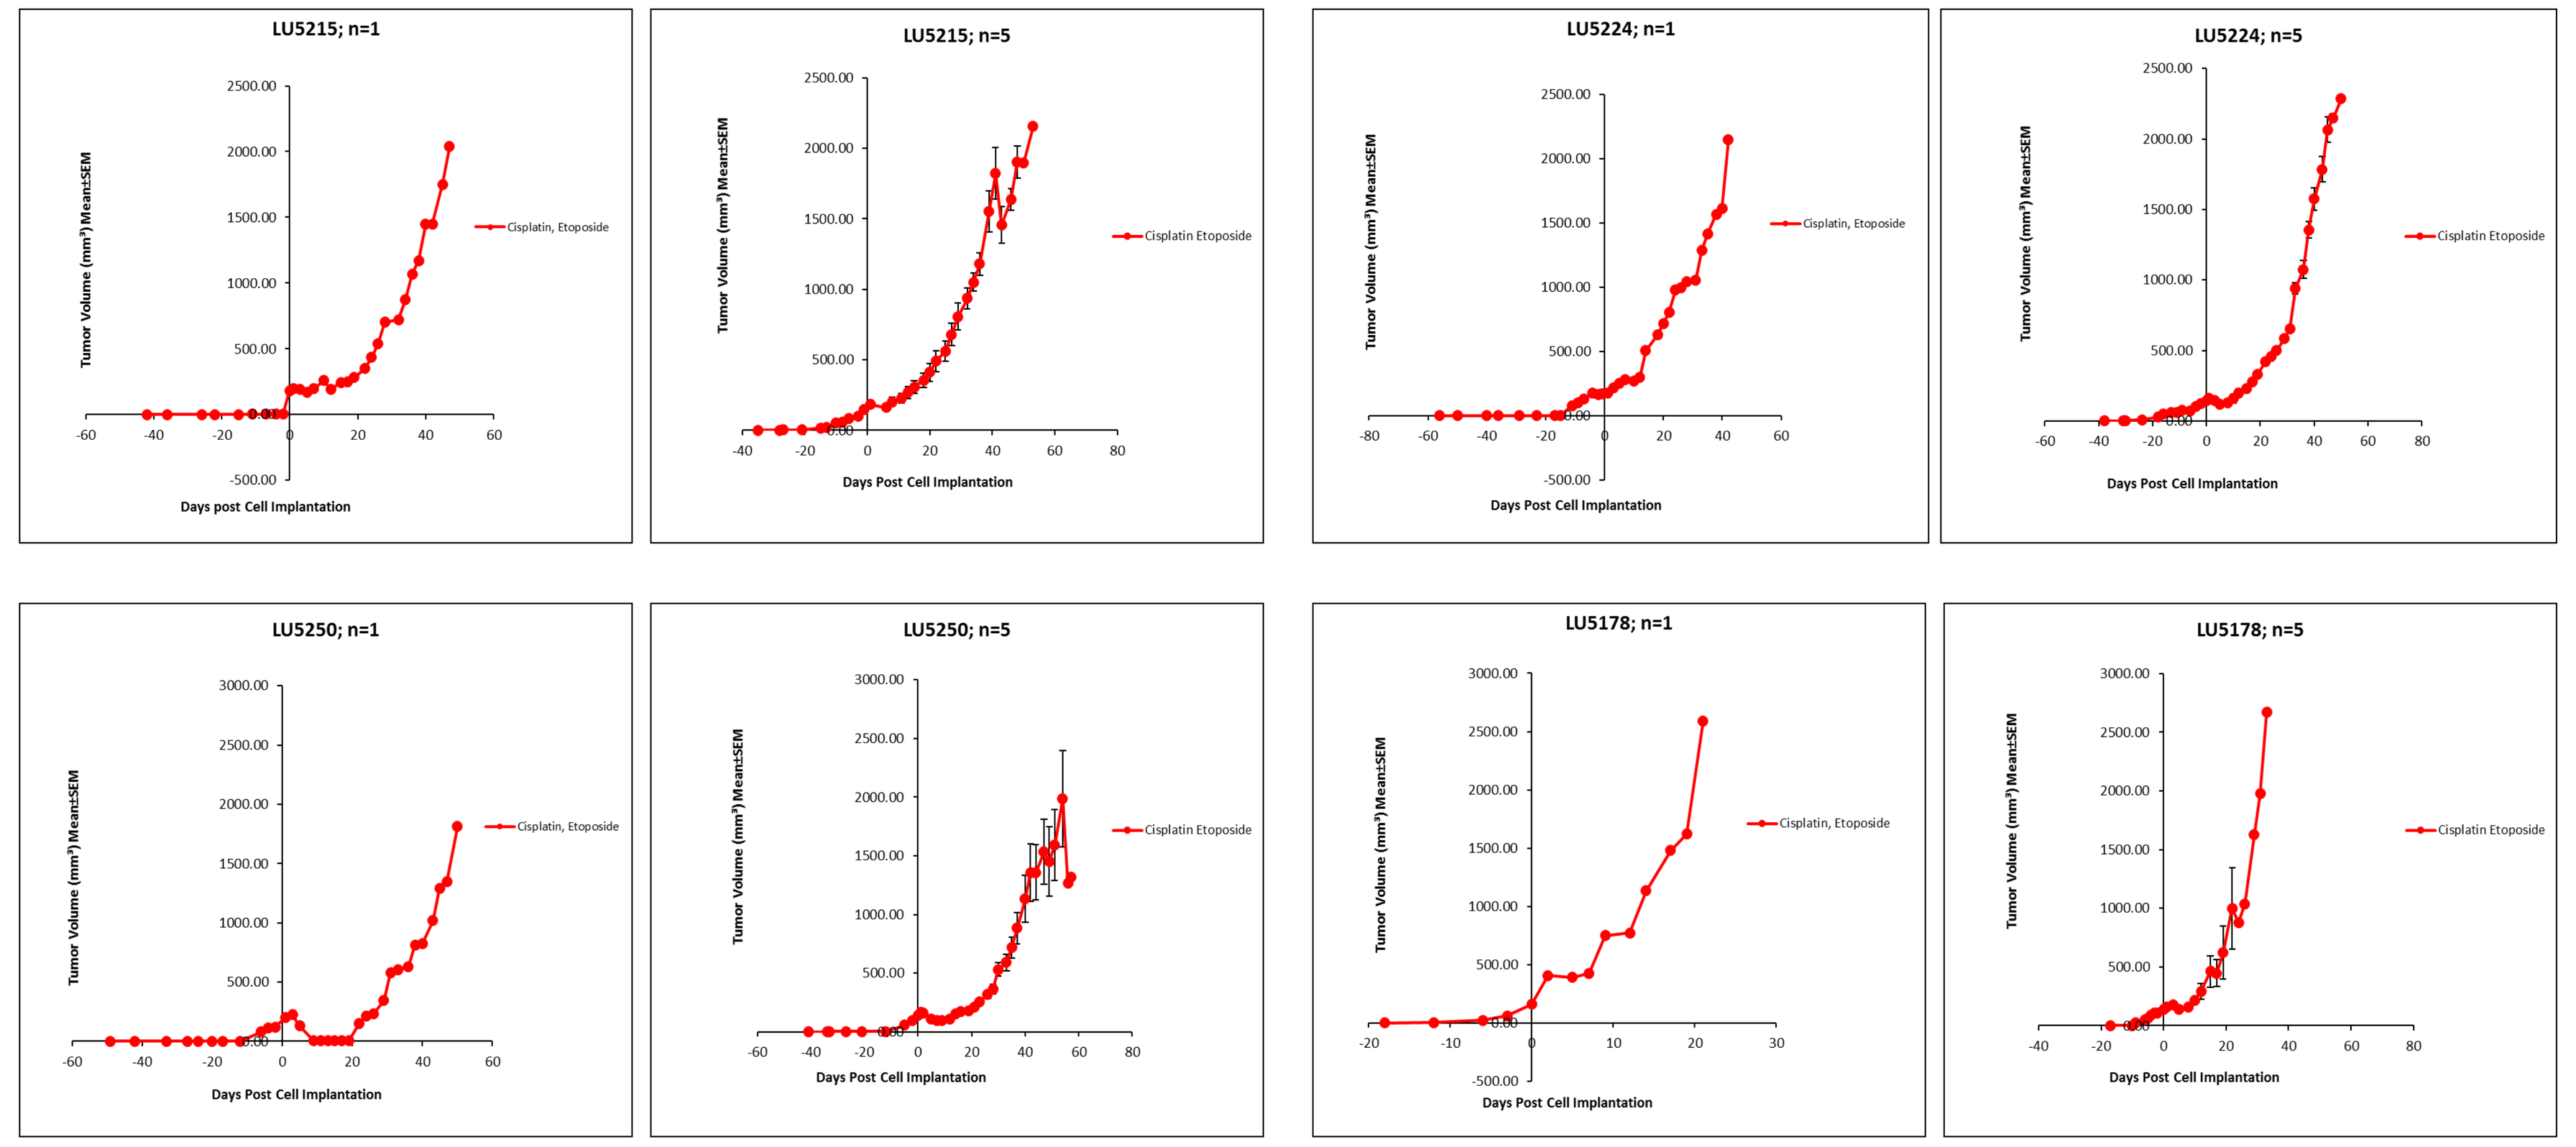
LU5215; n=1
LU5215; n=5
LU5224; n=1
LU5224; n=5
LU5250; n=1
LU5250; n=5
LU5178; n=1
LU5178; n=5
Tumor Volume (mm³) Mean±SEM
Days Post Cell Implantation
Cisplatin, Etoposide

Figure 3 (left): Model validation by comparing n=1 (left panels of the pair) versus n=5 (right panels of the pair) study in 4 SCLC models. Response observed in n=1 screen is confirmed in n=5 validation study.

SUMMARY

- **HuTrial study designs utilizing an n=1 approach as shown here are a translational means of evaluating therapeutics in HuPrime SCLC and NSCLC models before entering the clinic**
- **These study designs offer an efficient method to provide confident preclinical rationales for clinical development strategies that include selection of appropriate patient population, identification of appropriate biomarkers and combination therapies.**