IN-DEPTH CHARACTERIZATION OF VARIABILITY IN APHERESIS COLLECTIONS FROM NORMAL DONOR POPULATIONS FOR ALLOGENEIC CELL THERAPY

Shawn P. Fahl, Ph.D

DISCOVERY LIFE SCIENCES™

© 2021 Discovery Life Sciences

ABSTRACT

The success of autologous CAR-T cell therapies has revolutionized and accelerated development in the cell therapy field. However, the requirement for patient-specific starting material for these therapies remains an impediment to establishing availability for all patients who could benefit, highlighting the need for a highly characterized normal donor pool to generate allogeneic cell therapy material.

We have established a network of >2800 normal donors that have been genotyped at the HLA loci (6 digits) and stratified by reactivity to common human viruses, such as cytomegalovirus (CMV) and Epstein Barr Virus (EBV). Furthermore, cell collections from 35 randomly selected donors have been screened by flow cytometry for major immune cell subsets, including T cells, B cells, NK cells, and monocytes. The T cell compartment was further characterized by expression of activation markers (CD25, PD1, CD69) and proliferative capacity in response to anti-CD3/CD28 stimulation.

There was substantial variability (%CV 14.52%-50.58%, see Table) in the percentage of each immune cell population across the donor pool, which would have effects on the relative success of downstream cell manufacturing. We are evaluating additional donors to identify specific sources of variability. Collectively, these data highlight the need for in-depth genotypic and phenotypic characterization of donor populations to ensure that the most robust material is selected for each type of cell therapy manufacturing.

CONCLUSIONS

- HLA genotyping revealed substantial heterogeneity at the MHC class I and class II loci
- The majority of normal donors displayed reactivity to EBV, while CMV reactivity was only observed in ~50% of normal donors.
- Variability was observed in the percentage of lymphocyte and monocyte populations among donors, with the largest donor-to-donor variability present in the B cell compartment.
- Analysis of activation receptor expression in T cells following anti-CD3/CD28 revealed donor-specific alteration in receptor expression following stimulation
- Collectively, these characterizations inform donor selection for downstream cell therapy manufacturing.

RESULTS

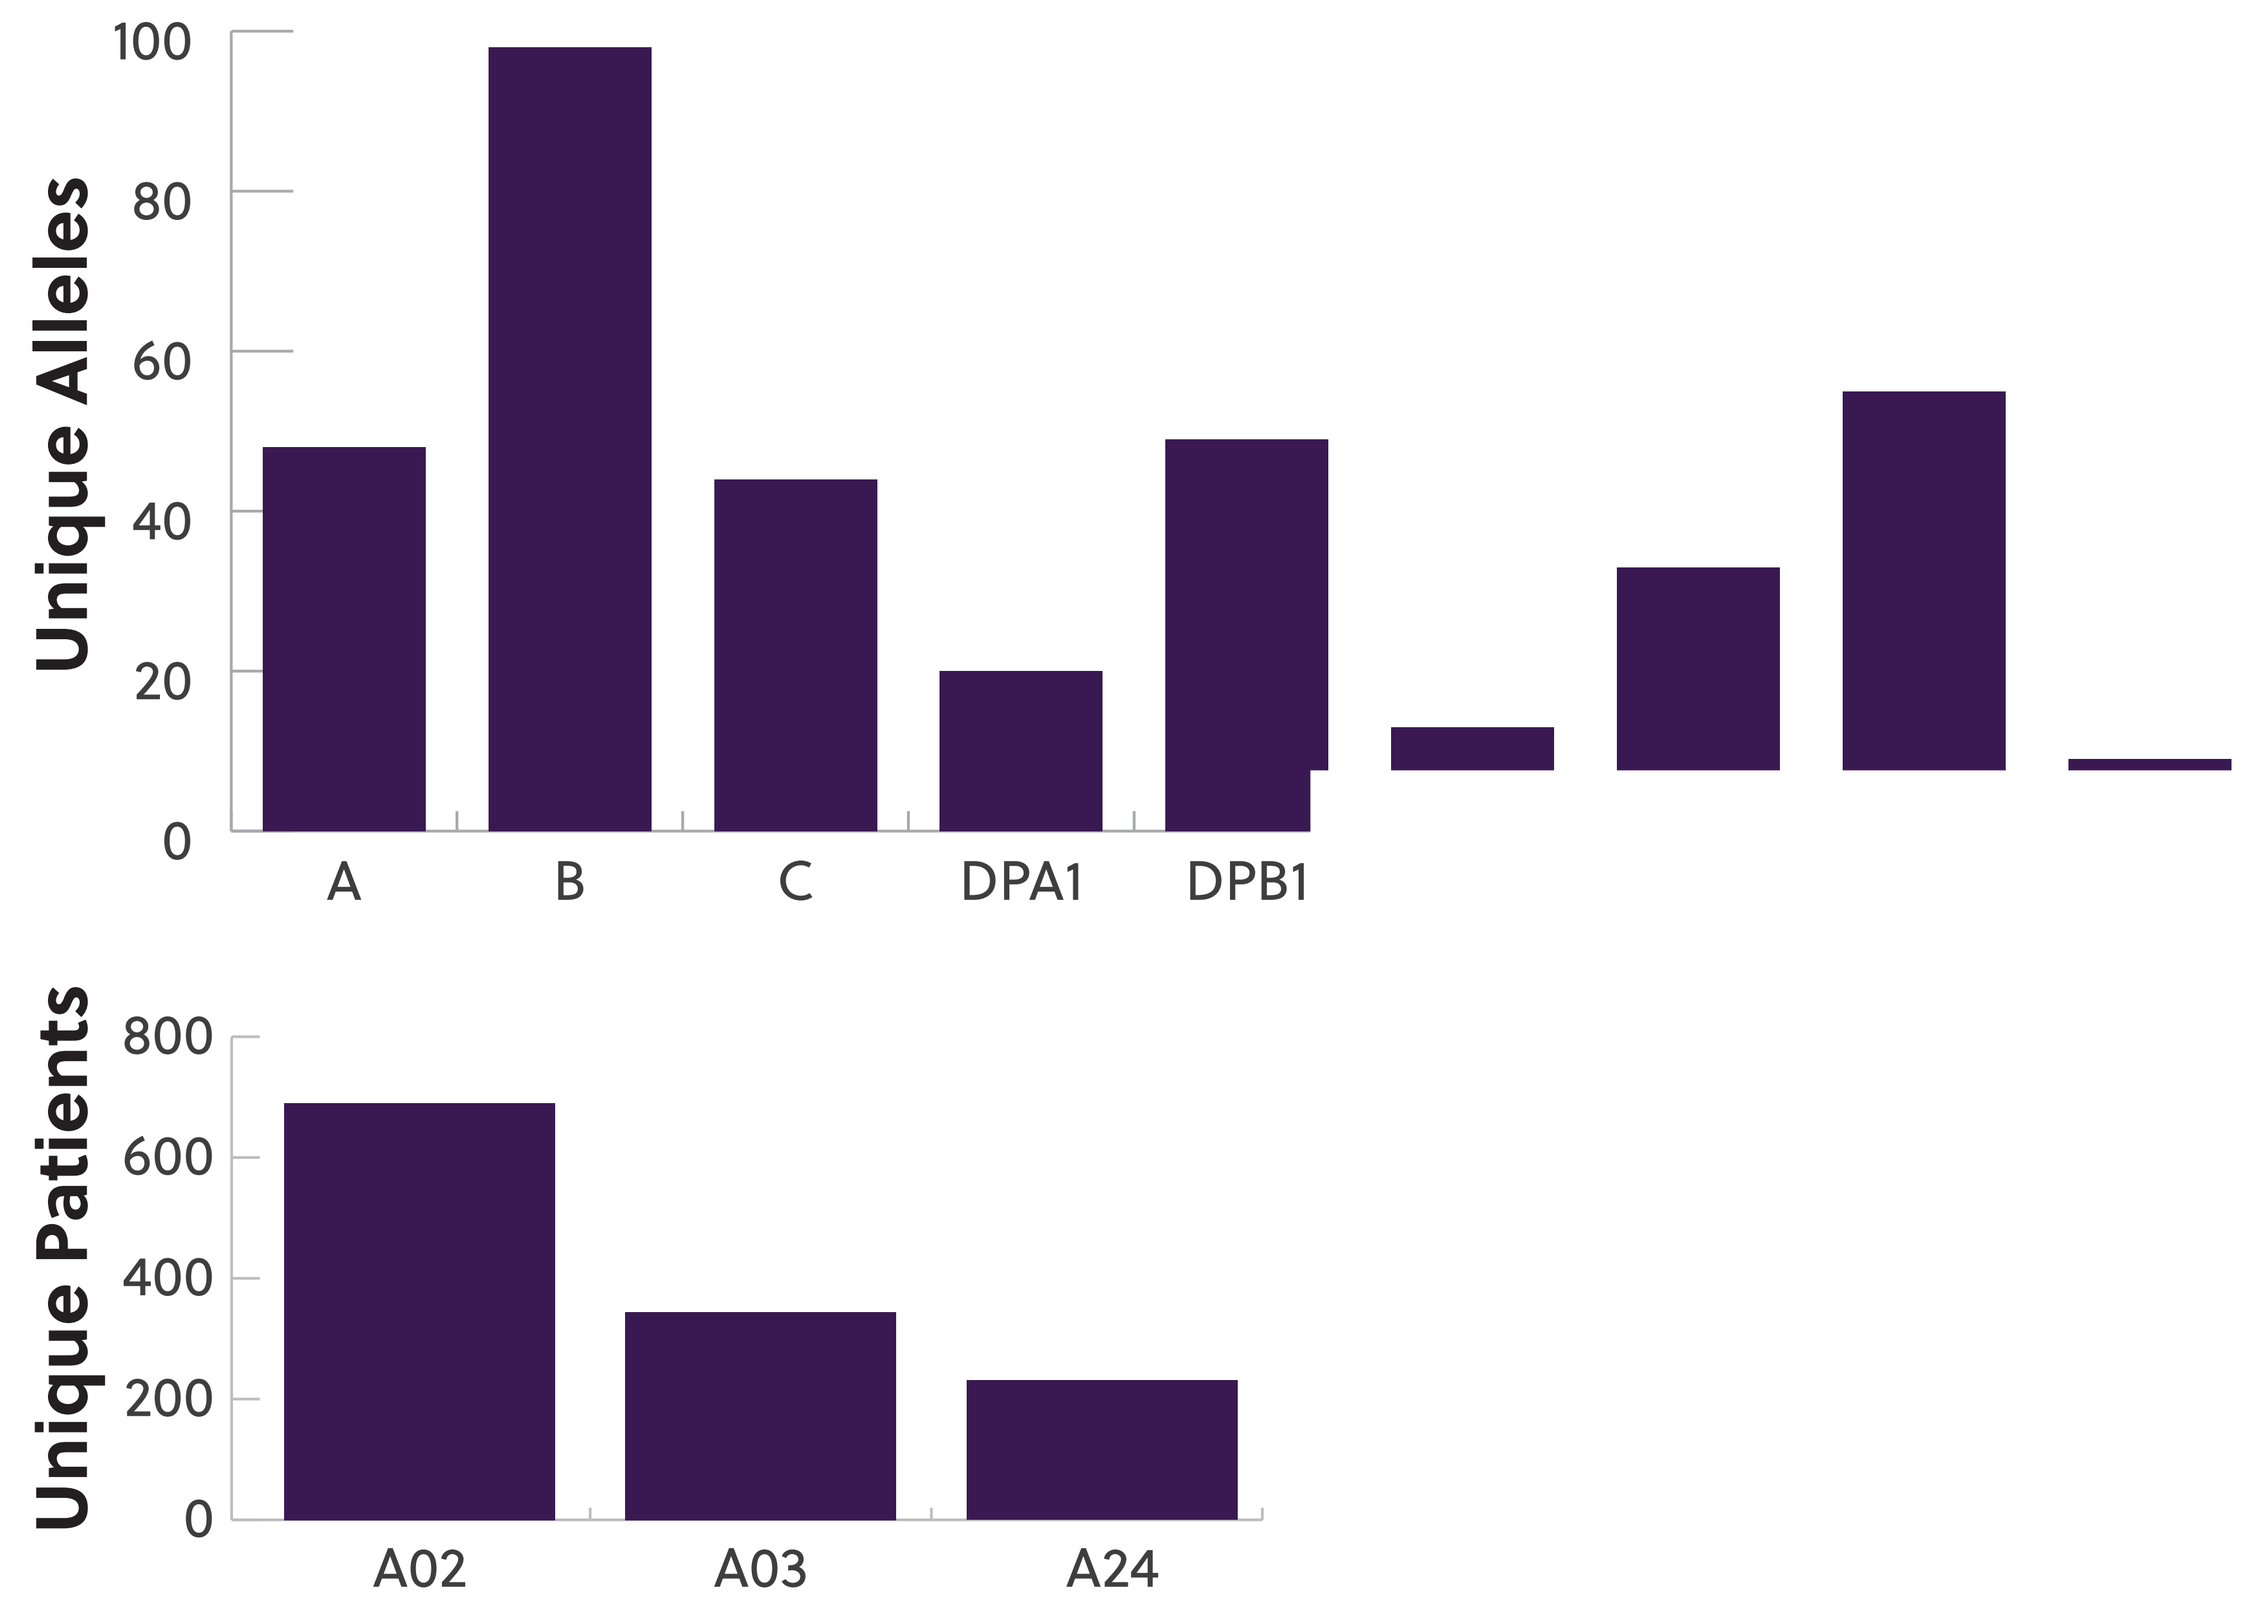
Unique Alleles
100
80
60
40
20
0
A
B
C
DPA1
DPB1
Unique Patients
800
600
400
200
0
A02
A03
A24

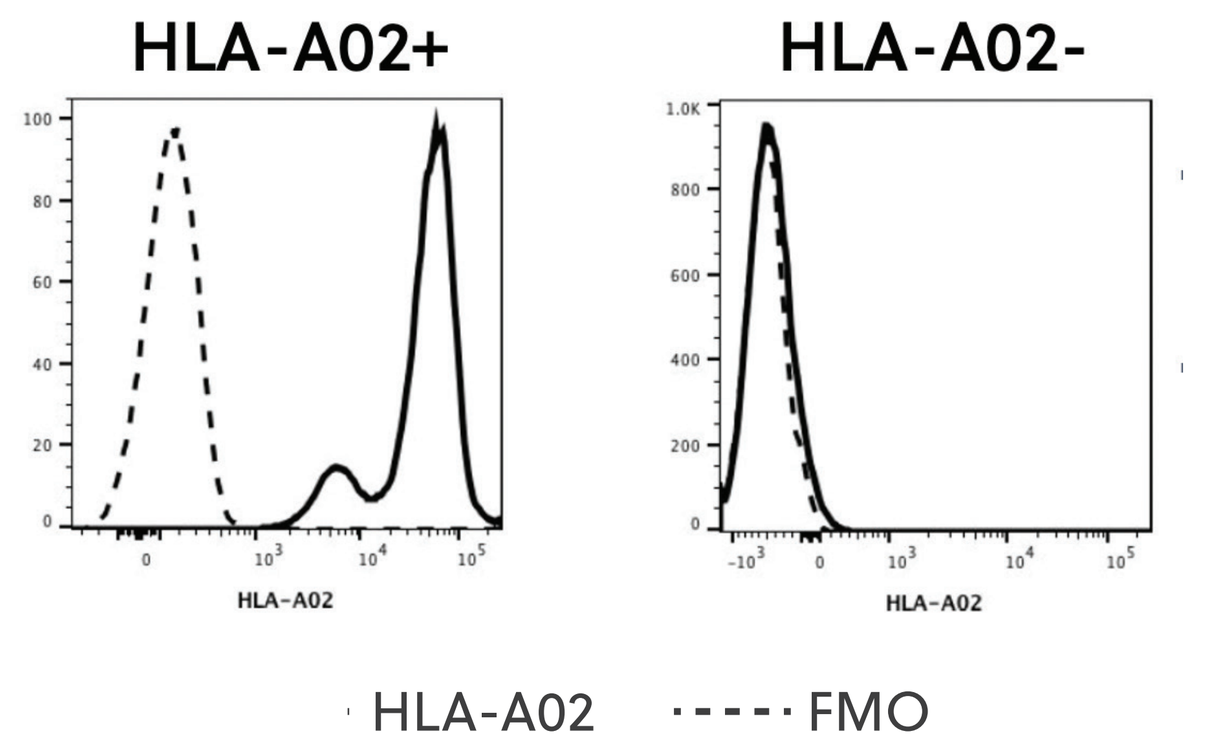
HLA-A02+
HLA-A02-
HLA-A02
FMO

iscovery Donor Clinic were
e number of unique alleles
nber of donors identified with
e. (C) PBMCs from HLA-A02+
sion by flow cytometry. FMO =

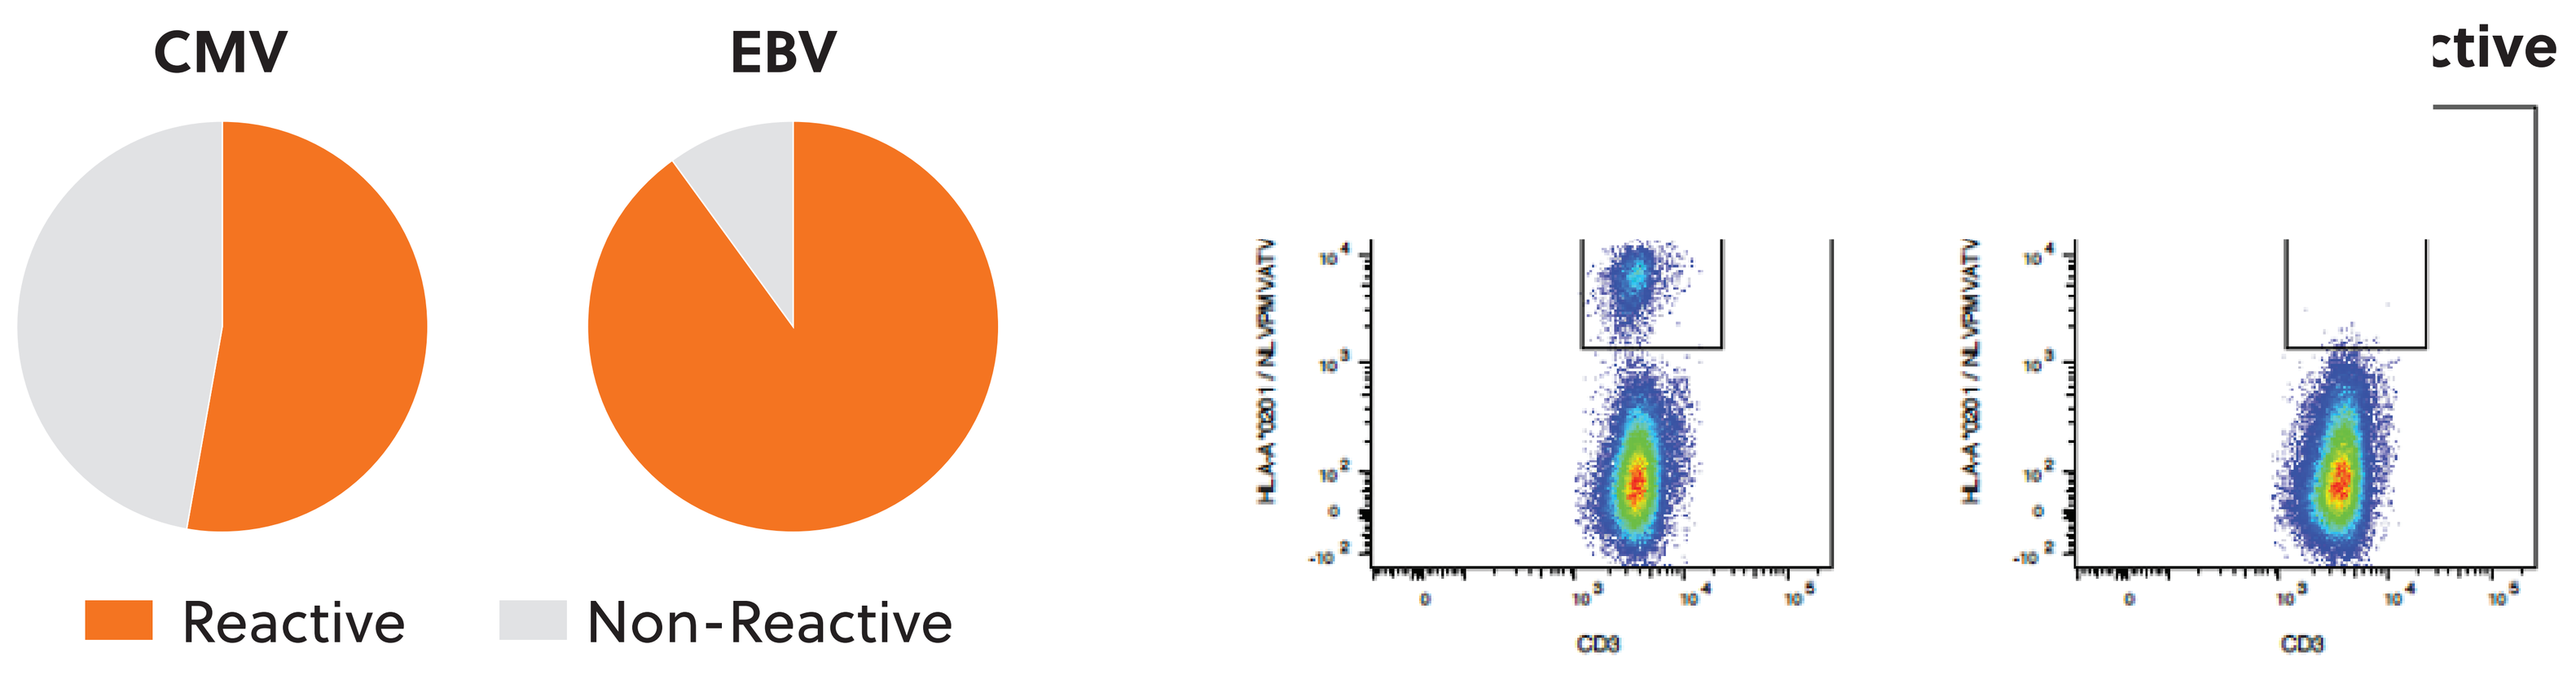
CMV
EBV
Reactive
Non-Reactive
ctive
CD3

Fig 2. CMV and EBV Reactivity of Normal Donor Pool. CMV and EBV serostatus of normal donors in the Discovery Donor Clinic was evaluated by ELISA. (A) Pie charts representing the number of CMV (top) or EBV (bottom) reactive and non-reactive donors. (B) CMV-reactive and CMV-non-reactive HLA-A02+ donors were evaluated for CMV-specific CD8+ T cells by flow cytometry using HLA*0201 / NLVPMVATV dextramers.

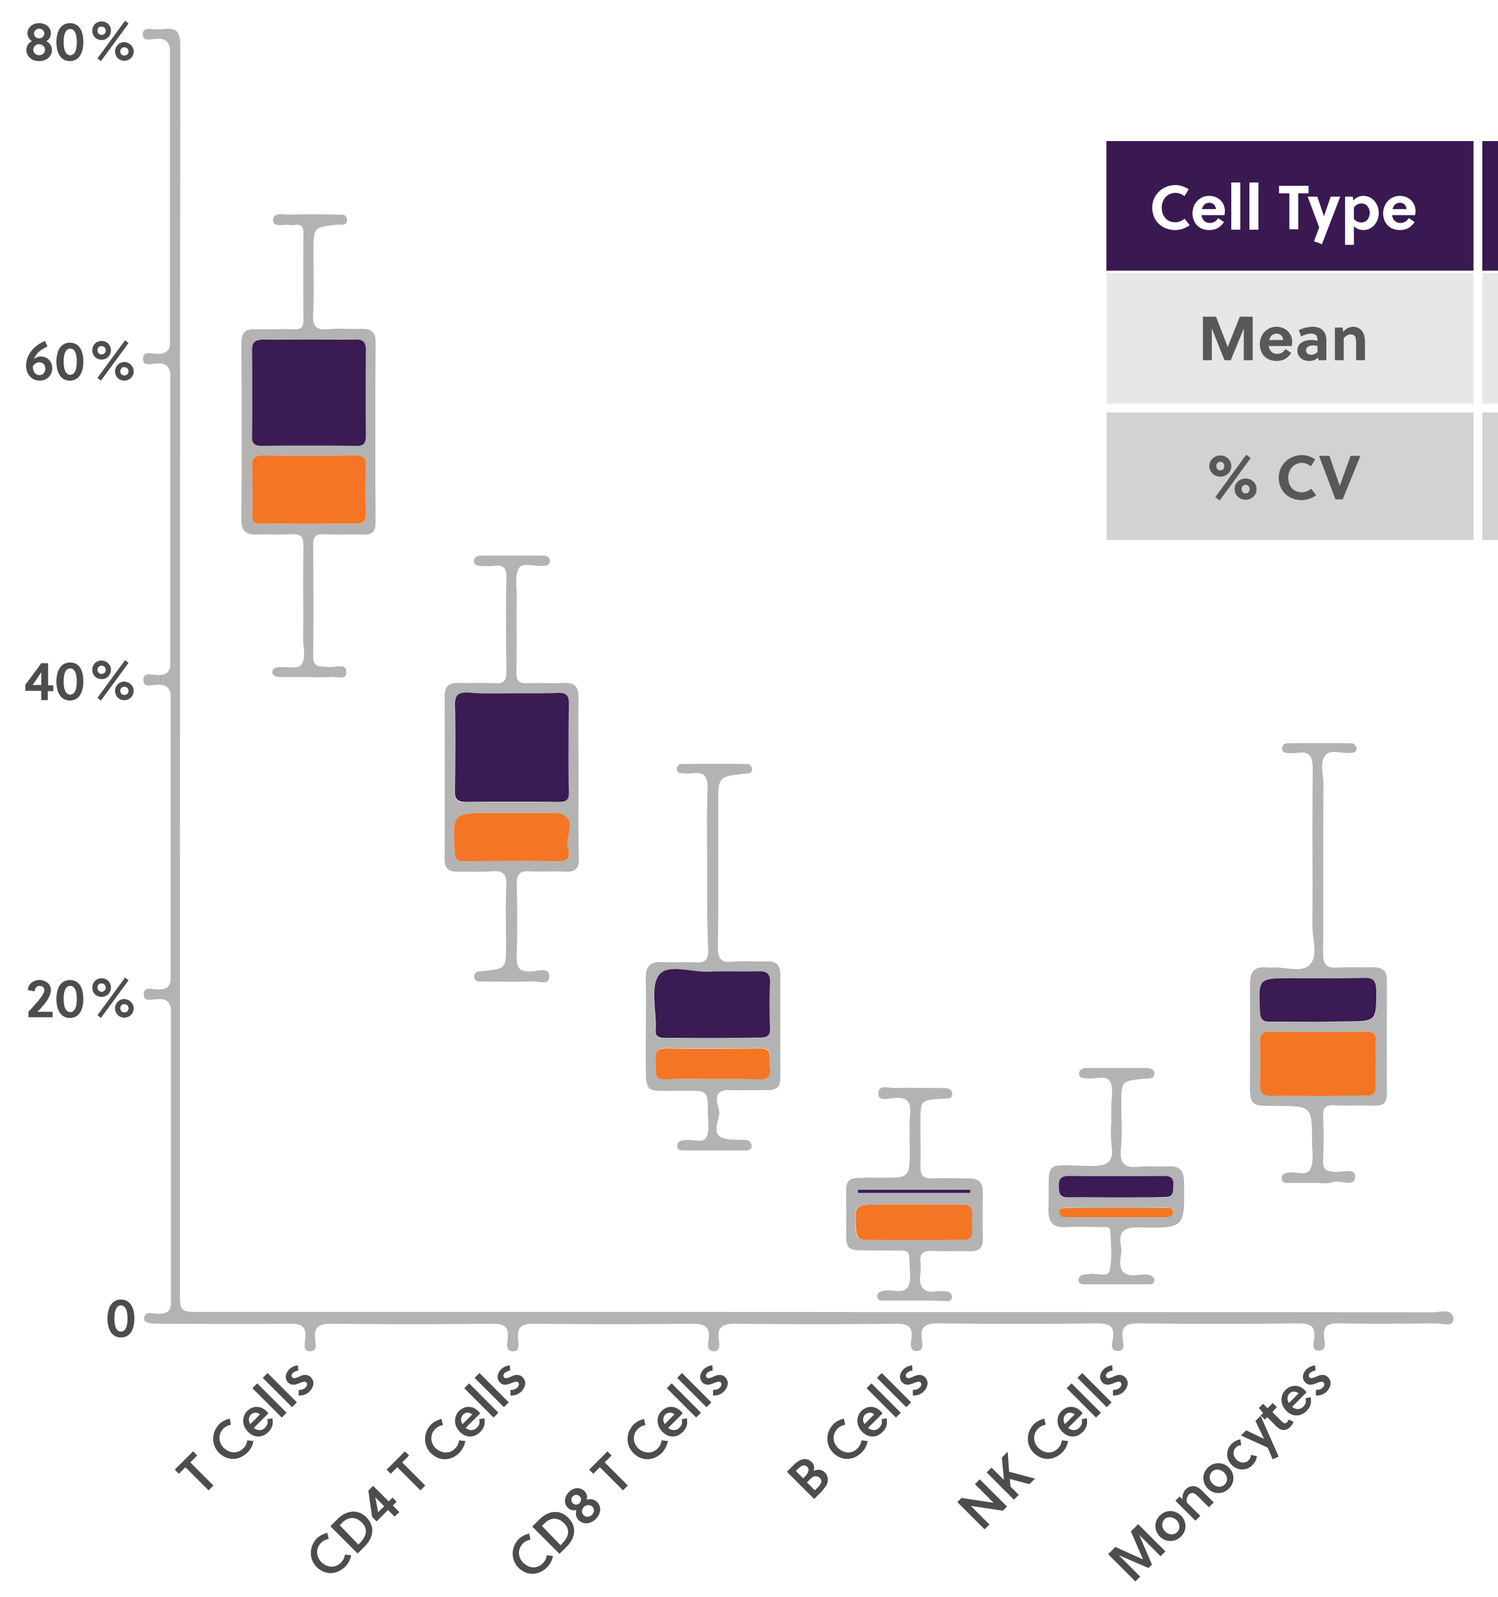
80%
60%
40%
20%
0
T Cells
CD4 T Cells
CD8 T Cells
B Cells
NK Cells
Monocytes

Cell Type	T Cells	CD4+ T Cells	CD8+ T Cells	B Cells	NK Cells	Monocytes
Mean	55.06%	33.47%	18.70%	6.88%	7.59%	18.59%
% CV	14.52%	21.51%	32.68%	50.58%	38.47%	32.31%

Figure 3. Immunophenotyping of Normal Donor Pool. PBMCs were analyzed for T cells (CD3+), CD4 T cells (CD3+ CD4+), CD8 T cells (CD3+ CD8+), B cells (CD19+), NK cells (CD56+ CD3-), and monocytes (CD14+) by flow cytometry. Data was generated from 35 random normal donors in the Discovery Donor Clinic.

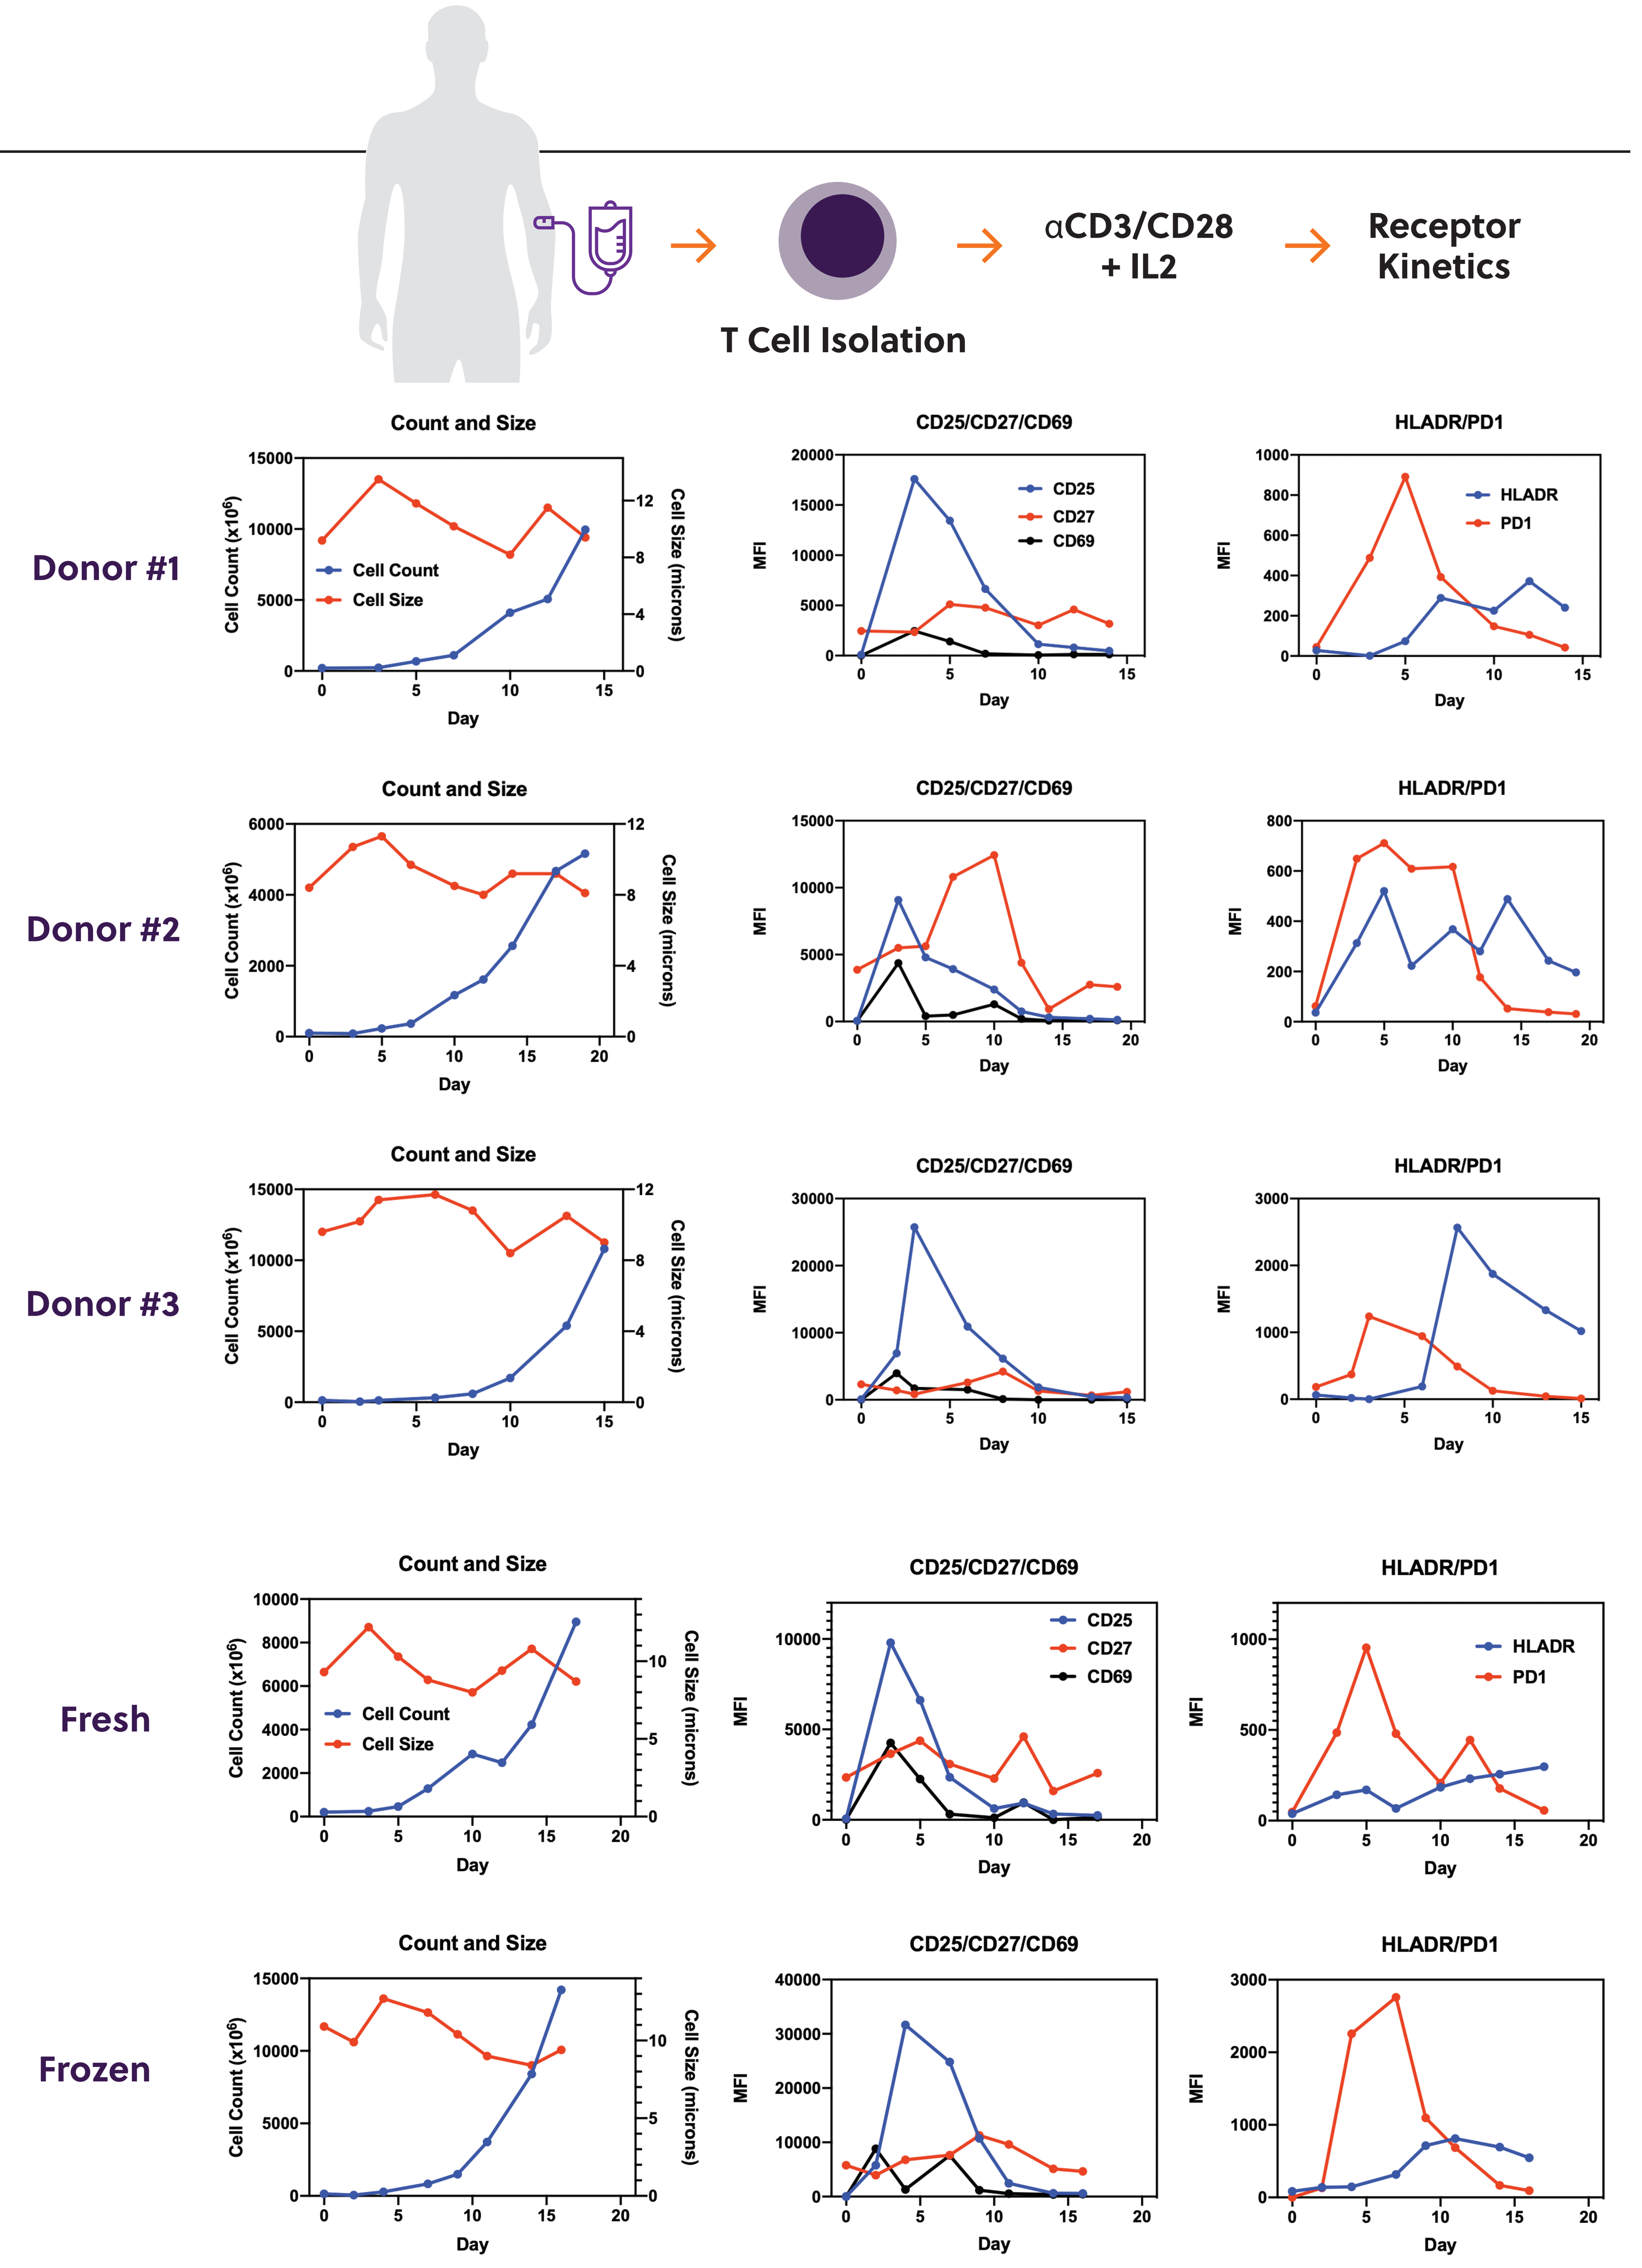
T Cell Isolation
αCD3/CD28
+ IL2
Receptor
Kinetics
Donor #1
Donor #2
Donor #3
Fresh
Frozen
Count and Size
CD25/CD27/CD69
HLADR/PD1
Cell Count
Cell Size
CD25
CD27
CD69
HLADR
PD1
Cell Count (x10⁶)
Cell Size (microns)
MFI
Day

Figure 4. T Cell Expansion of Fresh vs Frozen Normal T Cells. T cells were immunomagnetically isolated from apheresis material from normal donors. T cells were subsequently stimulated with anti-CD3/CD28 (T Cell TransActTM) in TexMACSTM media supplemented with IL2 and cultured for 14-17 days. Cells were analyzed every 2-3 days for cell counts, cell size, and surface receptor expression. Cell counts and cell size were determined using acridine orange/propidium iodide staining. Surface expression of CD25, CD27, CD69, HLADR, and PD1 were determined by flow cytometry