Immunogenicity monitoring during preclinical development of Nanobodies®: comparing assay formats and species matrices

Sofie Poelmans, Ingrid Ottevaere, Sofie Priem, Marie-Paule Bouche, Judith Baumeister and Josefin-Beate Holz • Ablynx NV, Zwijnaarde, Belgium

Introduction

- Ablynx develops antibody-derived therapeutic Nanobodies for the treatment of cardiovascular, bone and inflammatory diseases

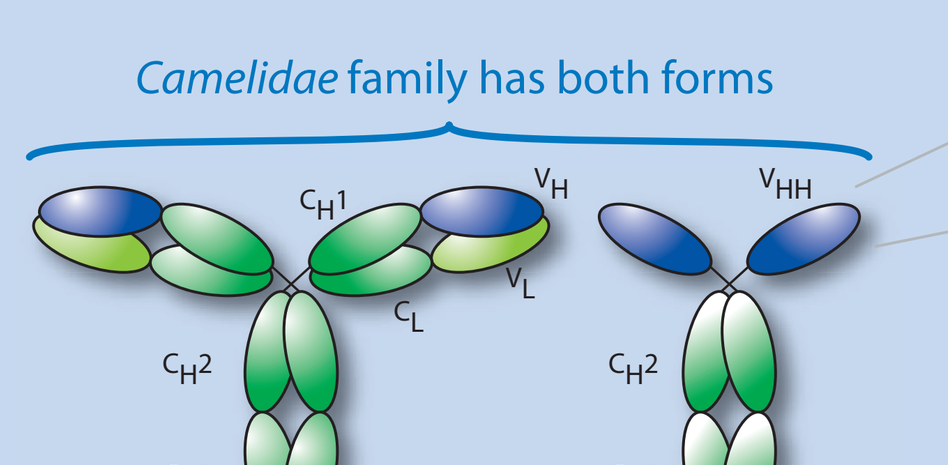
Camelidae family has both forms
C_H1
V_H
V_{HH}
C_L
V_L
C_H2
C_H3

Conventional Antibody	Heavy-Chain Antibody
Heavy and light chains	Only heavy chains
Both chains required for antigen binding and stability	Full antigen binding capacity and very stable

Ablynx's Nanobody®

Based on the smallest functional fragment of a naturally occurring heavy-chain antibody

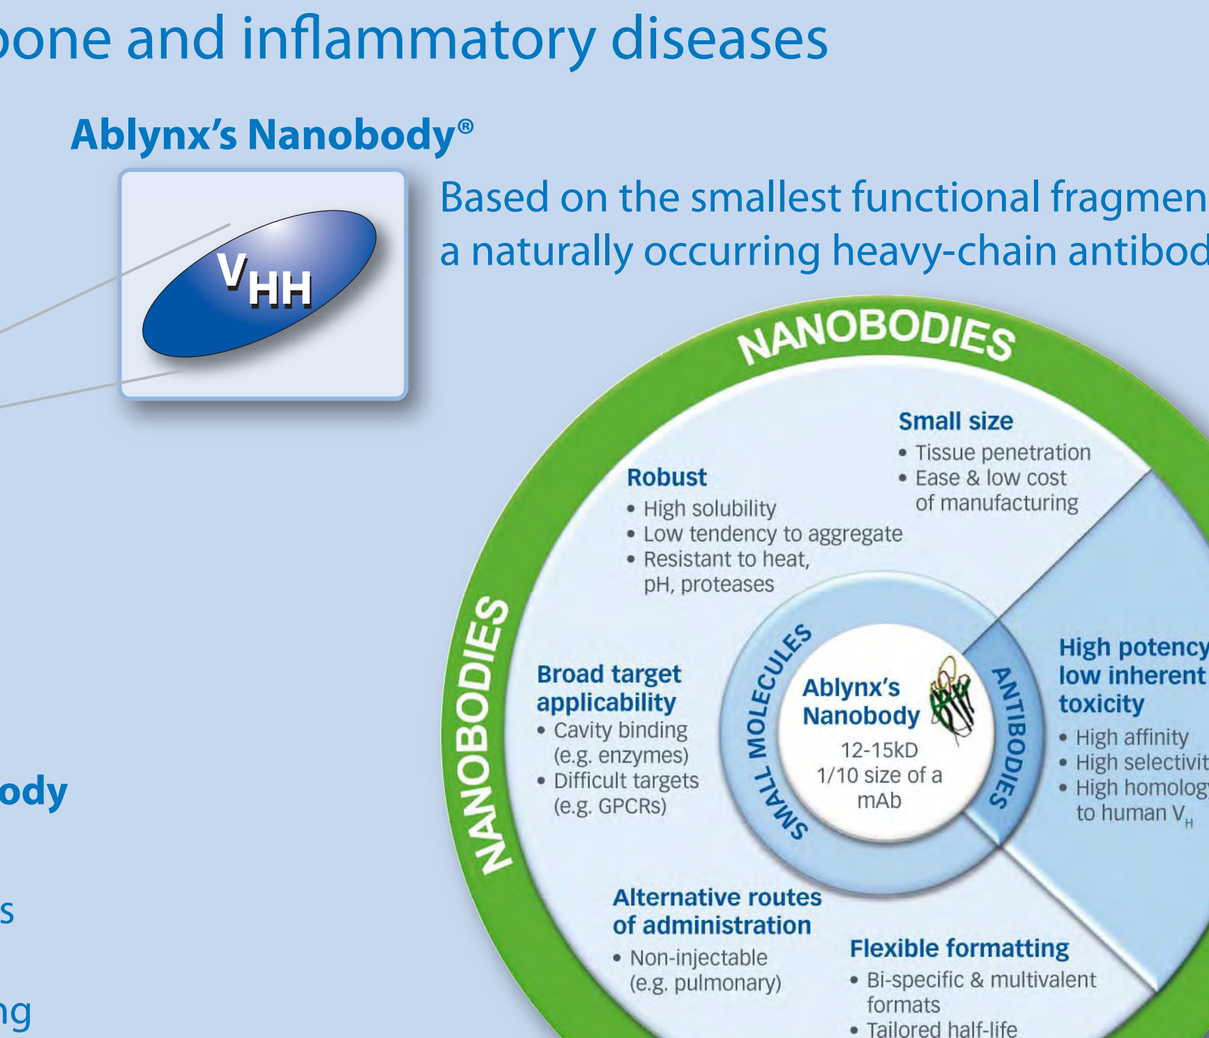
V_{HH}
NANOBODIES
Small size
• Tissue penetration
• Ease & low cost of manufacturing
Robust
• High solubility
• Low tendency to aggregate
• Resistant to heat, pH, proteases
Broad target applicability
• Cavity binding (e.g. enzymes)
• Difficult targets (e.g. GPCRs)
Ablynx's Nanobody
12-15kD
1/10 size of a mAb
SMALL MOLECULES
ANTIBODIES
High potency/ low inherent toxicity
• High affinity
• High selectivity
• High homology to human V_H
Alternative routes of administration
• Non-injectable (e.g. pulmonary)
Flexible formatting
• Bi-specific & multivalent formats
• Tailored half-life

Therapeutic Nanobodies

ALX-0081	ALX-0141	ALX-0061
Clinical programme in phase II • therapeutic Nanobody targeting von Willebrand Factor (vWF) • may reduce risk of thrombosis in patients with acute coronary syndrome (ACS) and thrombotic thrombocytopenic purpura (TTP)	**Clinical programme in phase I** • therapeutic Nanobody targeting von receptor activator of NF-Kappa B ligand (RANK-L) • may reduce risk of fractures through highly potent inhibition of key driver of bone resorption in post-meno-pausal osteoporosis, rheumatoid arthritis and bone metastasis/cancer	**Programme entering IND phase (pre-phase I)** • therapeutic Nanobody targeting IL-6 receptor (IL-6R) • may reduce inflammation in rheumatoid arthritis
Bivalent target binding Nanobody (28kDa) • binds with high affinity to the A1 domain of vWF • blocks the interactions between vWF, platelets and vascular collagen • selectively prevents thrombus formation under high shear conditions	**Bivalent target binding Nanobody (41kDa)** • binds with high affinity to RANK-L • blocks the RANK-RANK-L interaction • tailored half life	**Monovalent target binding Nanobody (26kDa)** • binds with high affinity to mIL-6R and sIL-6R • blocks the interactions between IL-6 and IL-6R without cross-linking • tailored half life

Purpose of immunogenicity monitoring

- Biopharmaceutical therapeutics are potentially immunogenic, which may
 - Influence pharmacokinetic behavior
 - Induce loss of efficacy and
 - Potentially lead to serious side effects
- Therefore immunogenicity of therapeutic proteins should be assessed during preclinical and clinical development
- Approach
 - Tiered approach
 - Risk based
 - Event driven
- Challenges
 - Assay formats and platforms
 - Drug interference
 - Acid dissociation
 - Validity of controls and established cut point
 - Clinical versus non clinical
 - Immunogenicity data interpretation

Assay formats

	Direct ELISA	Bridging ELISA
Setup	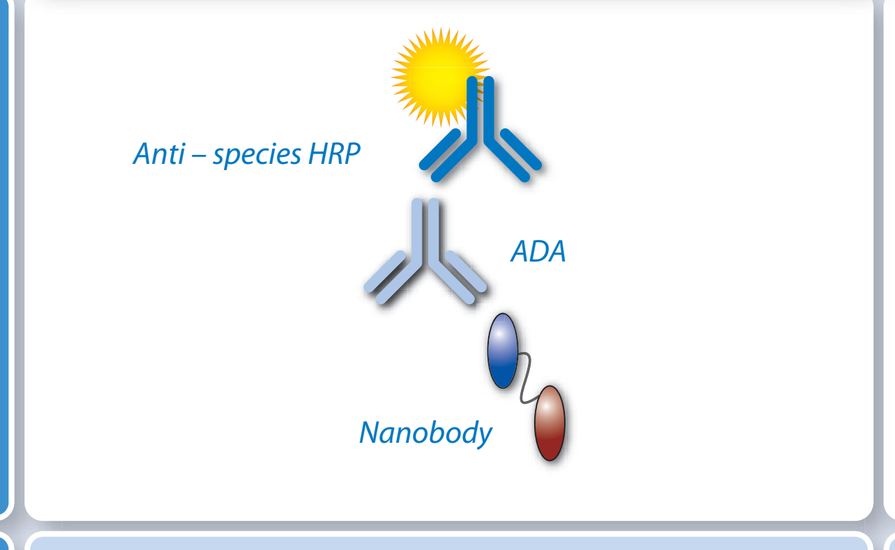	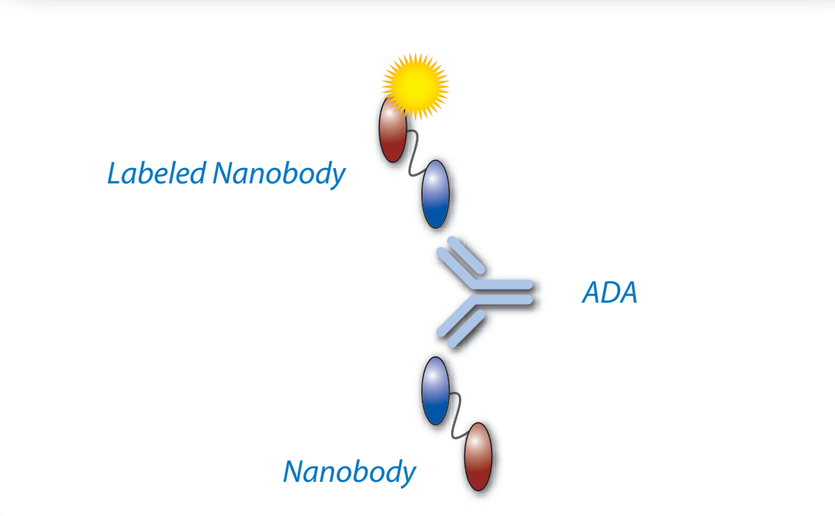
Advantages	• Sensitive, less drug interference • No labeling • Commercial detection Abs	• Species independent • Highly specific • All isotypes
Disadvantages	• Species dependent • High background • Isotype dependence of detector Ab • Altered epitopes due to immobilization • Restricted detection of low affinity Abs	• Labeling of drug • Drug interference • Possible limited detection of IgG4 • Altered epitopes due to immobilization • Restricted detection of low affinity Abs

Anti – species HRP
ADA
Nanobody
Labeled Nanobody
ADA
Nanobody

Assay platforms

	ECL bridging assay on MSD platform	Gyrolab™ BioAffy System
Setup	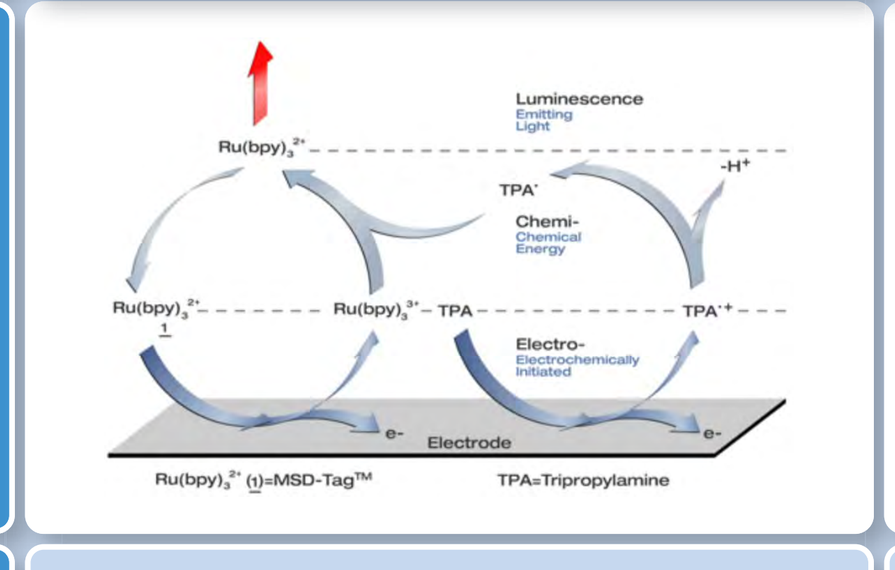	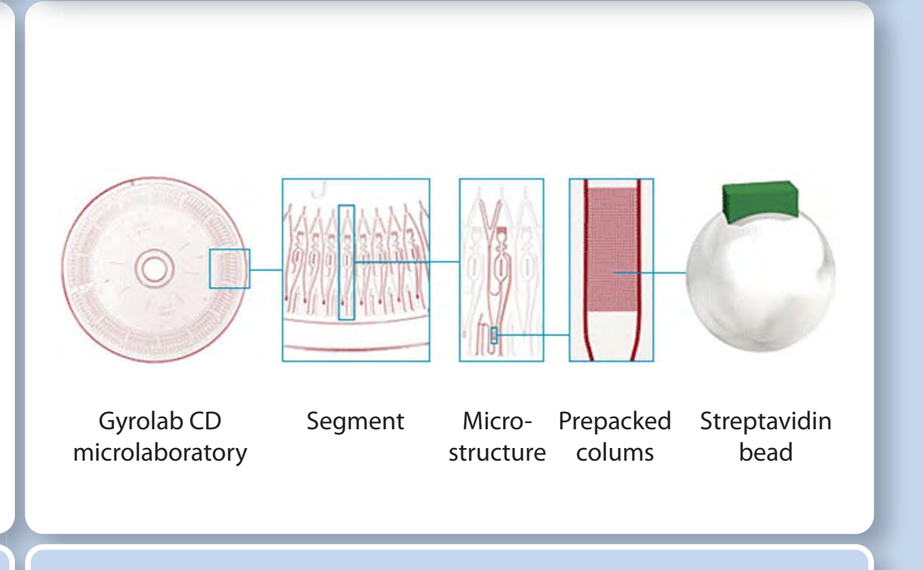
Advantages	• Sensitive • Broad dynamic range • In solution phase kinetics/less washing steps ➔ detection of low affinity Ab • High drug tolerance	• Small sample and reagent consumption • High throughput • Broad dynamic range • High drug tolerance
Disadvantages	• Single vendor • 2 labeled reagents • Expensive consumables	• Single vendor • 2 labeled reagents • Expensive consumables

Luminescence
Emitting Light
Chemi- Chemical Energy
Electro- Electrochemically Initiated
TPA
Electrode
TPA=Tripropylamine
Gyrolab CD microlaboratory
Segment
Micro-structure
Prepacked colums
Streptavidin bead

Drug interference

- Established drug interference levels will depend on
 - Assay format/assay platform
 - Positive control
- Acceptable drug interference levels will depend on
 - Dosing scheme
 - PK/PD parameters
- Drug interference can be tackled by
 - Drug removal step
 - Drug dissociation step
 - Affinity capture elution

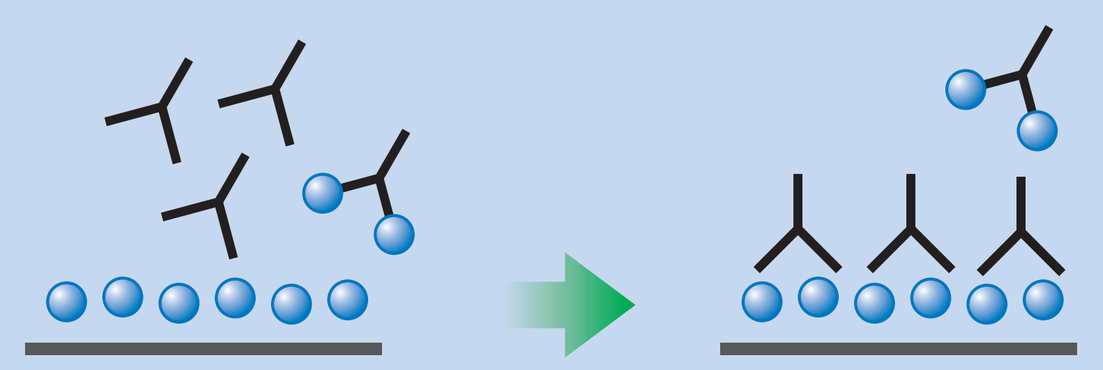

Low drug levels

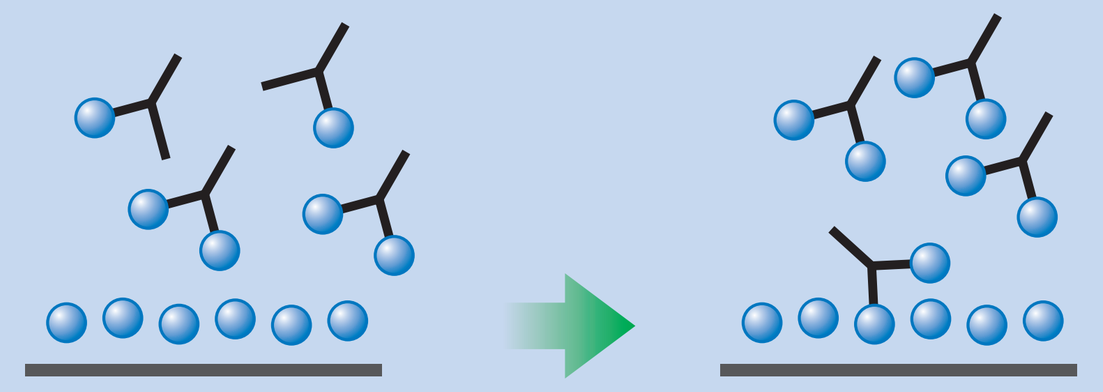

High drug levels

Assay performance I

	ALX-0081	
Drug concentrations in samples	Low drug concentrations in wash out samples due to short half life and dosing schedule	
Assay format	Bridging ELISA	ECL bridging
Species	human	human
MRD	1/4	1/6
LOD [ng/mL]	50	125
Drug tolerance level [µg/mL]	< 0.5	< 0.5
Acid treatment	No	Yes
Positive control	mAb	mAb

MRD: minimum required dilution • LOD: limit of detection

Assay performance II

	ALX-0141			ALX-0061	
Drug concentrations in samples	Higher drug concentrations for longer period of time due to long half life ***Need for assay optimization • different assay formats and platforms***				
Assay format	Bridging ELISA	Direct ELISA	ECL bridging	Direct ELISA	Bridging ELISA
Species	NHP	NHP	NHP	NHP	NHP
MRD	1/2	1/25	1/30	1/20	1/2
LOD [ng/mL]	385	506	662	970	385
Drug tolerance level [µg/mL]	0.005 - 0.05	0.5 - 5	20 - 50	0 - 3	0.005 - 0.05
Acid treatment	No	No	Yes	No	No
Positive control	pAb	pAb	mAb	pAb	pAb

Acid dissociation consequences ECL bridging I

No acid dissociation versus short acid dissociation treatment

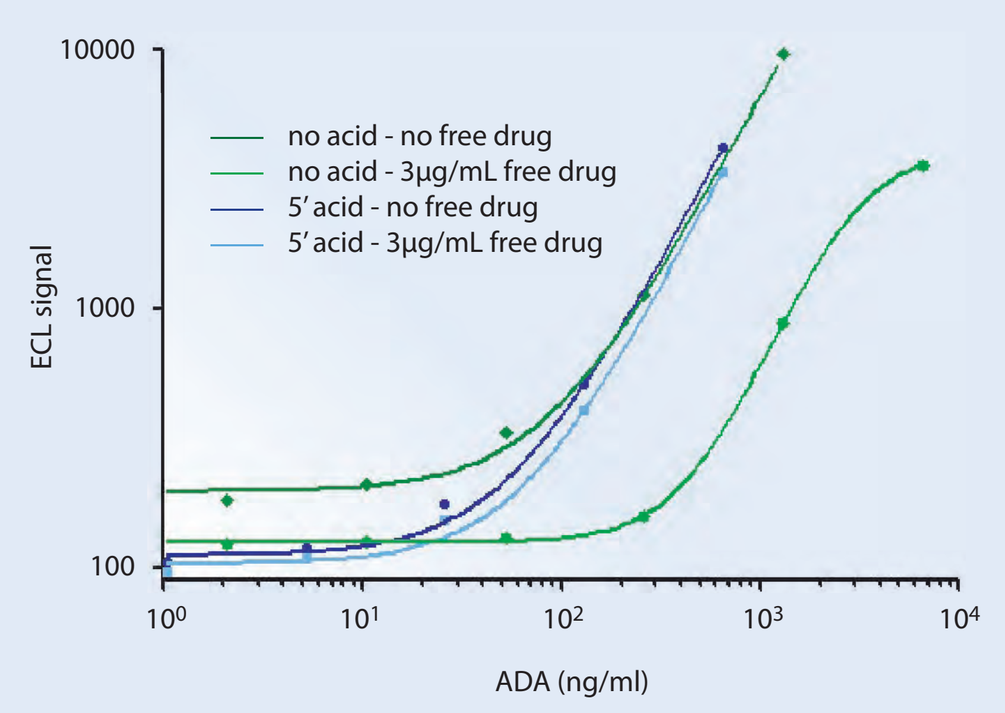
ALX-0141
10000
1000
100
ECL signal
no acid - no free drug
no acid - 3µg/mL free drug
5' acid - no free drug
5' acid - 3µg/mL free drug
ADA (ng/ml)

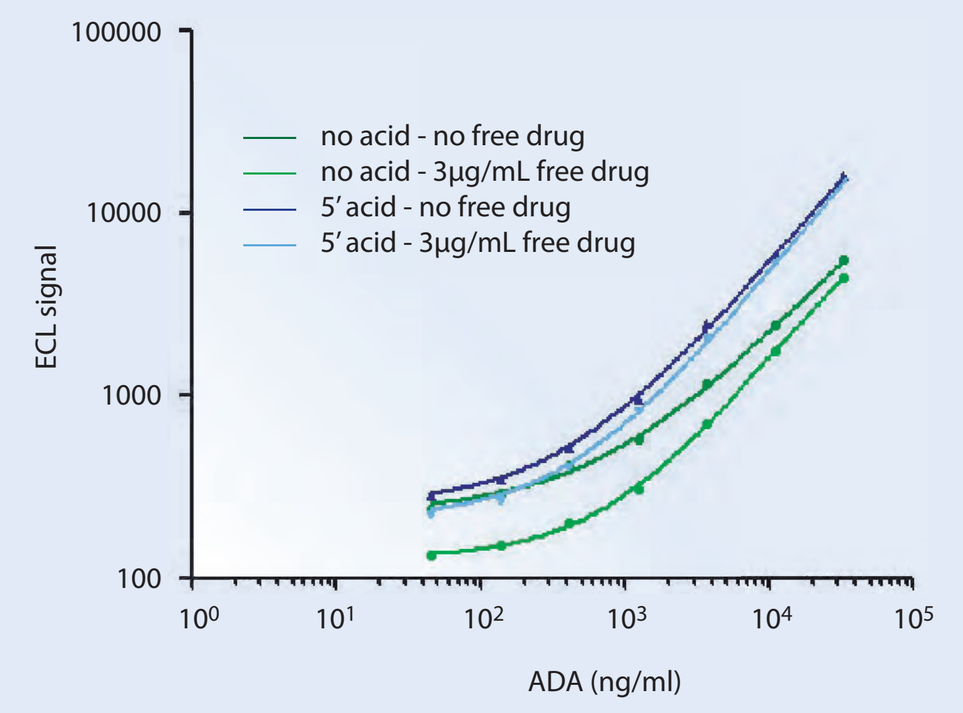
ALX-0061
100000
10000
1000
100
ECL signal
no acid - no free drug
no acid - 3µg/mL free drug
5' acid - no free drug
5' acid - 3µg/mL free drug
ADA (ng/ml)

Acid dissociation step clearly improved drug tolerance levels without affecting the positive control

Acid dissociation consequences ECL bridging II

Short versus long acid dissociation treatment

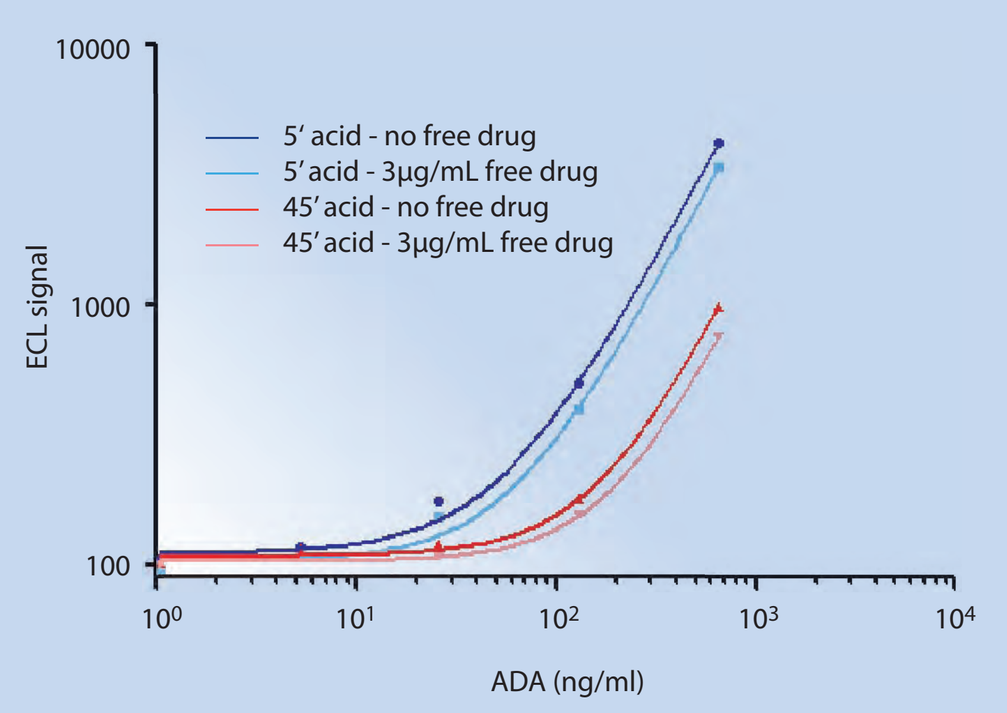
ALX-0141
10000
1000
100
ECL signal
5' acid - no free drug
5' acid - 3µg/mL free drug
45' acid - no free drug
45' acid - 3µg/mL free drug
ADA (ng/ml)

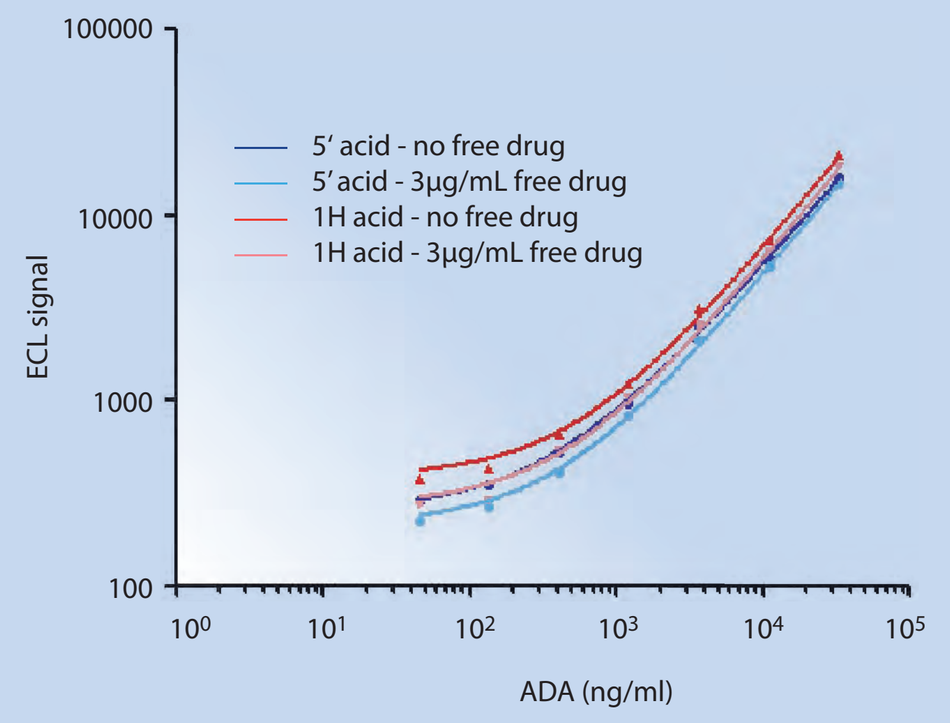
ALX-0061
100000
10000
1000
100
ECL signal
5' acid - no free drug
5' acid - 3µg/mL free drug
1H acid - no free drug
1H acid - 3µg/mL free drug
ADA (ng/ml)

Implementation of an acid dissociation step can have a deleterious effect on the study sample/positive control

Validity of control and validation samples

- Relevance of mouse monoclonal as positive control
 - Stability in human serum
 - Species specific control not always available
 - Limited availability of polyclonal control samples
- mAbs and pAbs can behave different in the assay
 - Affinity of mouse monoclonal will determine assay performance

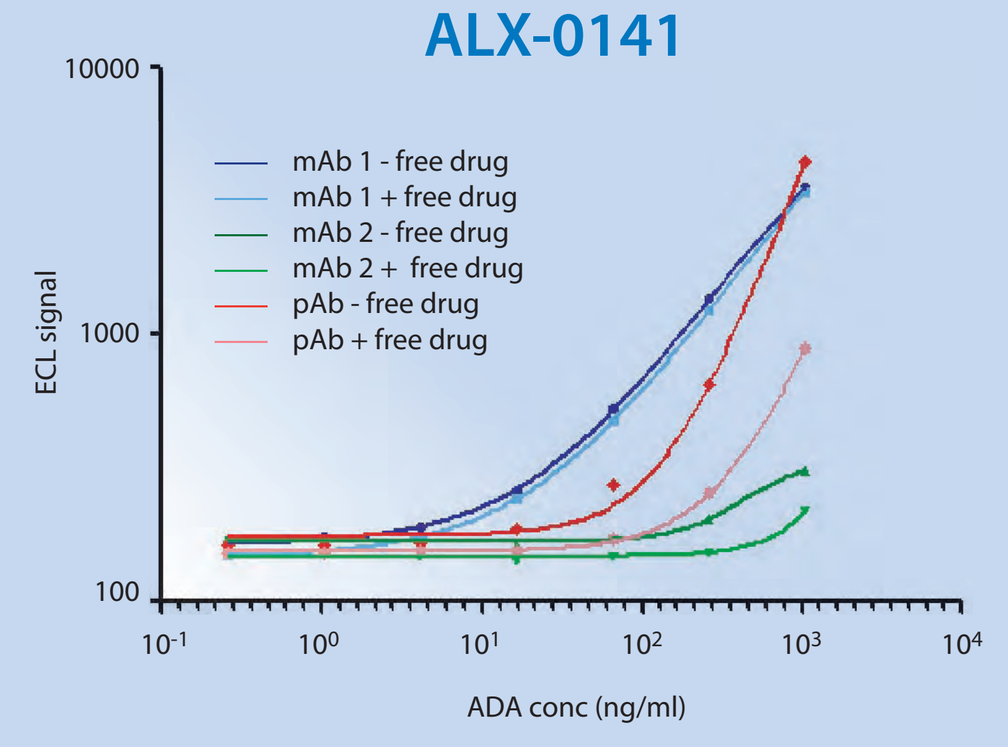
ALX-0141
10000
1000
100
ECL signal
mAb 1 - free drug
mAb 1 + free drug
mAb 2 - free drug
mAb 2 + free drug
pAb - free drug
pAb + free drug
ADA conc (ng/ml)

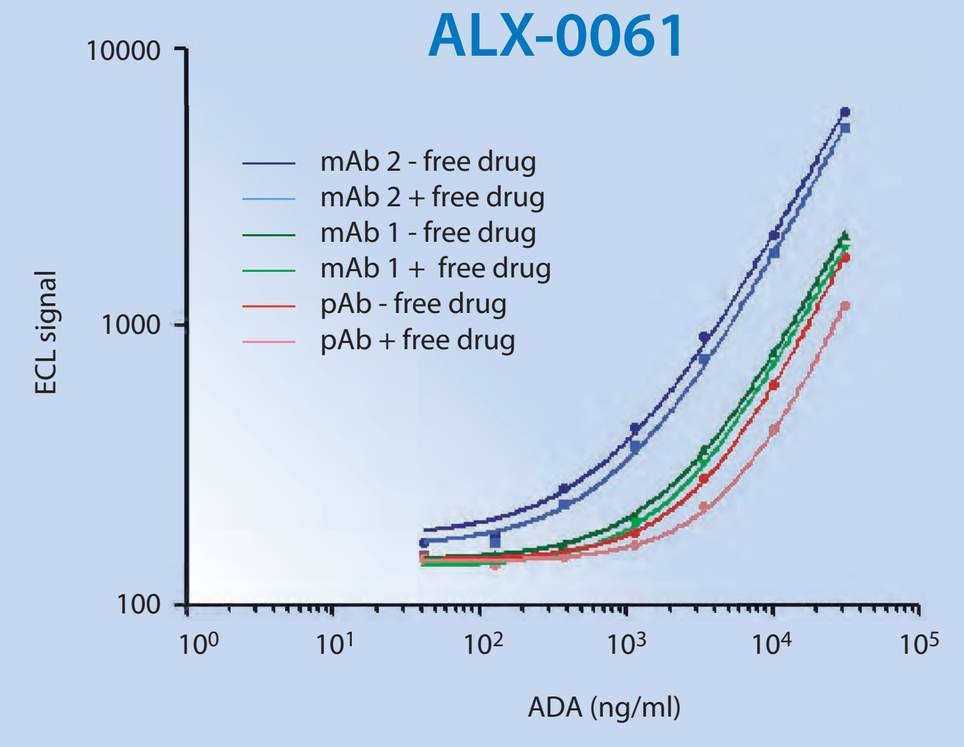
ALX-0061
10000
1000
100
ECL signal
mAb 2 - free drug
mAb 2 + free drug
mAb 1 - free drug
mAb 1 + free drug
pAb - free drug
pAb + free drug
ADA (ng/ml)

Preclinical versus clinical outcome

- Limited predictive power of animal models for immunogenicity of drug in clinical situation
 - Drugs are sequence optimized for human targets, not for animal targets
 - Differences in the immune system function
- Incidence of ADAs in preclinical studies does not necessarily increase the risk of antibody related clinical sequelae

Then why monitor immunogenicity in preclinical studies?

Preclinical immunogenicity

- Immunogenicity data are needed for correct interpretation of preclinical studies
 - To demonstrate exposure and/or explain altered PK/PD profiles
 - To demonstrate biological activity is not neutralized
- Correlate ADA/PK/PD/toxicology data

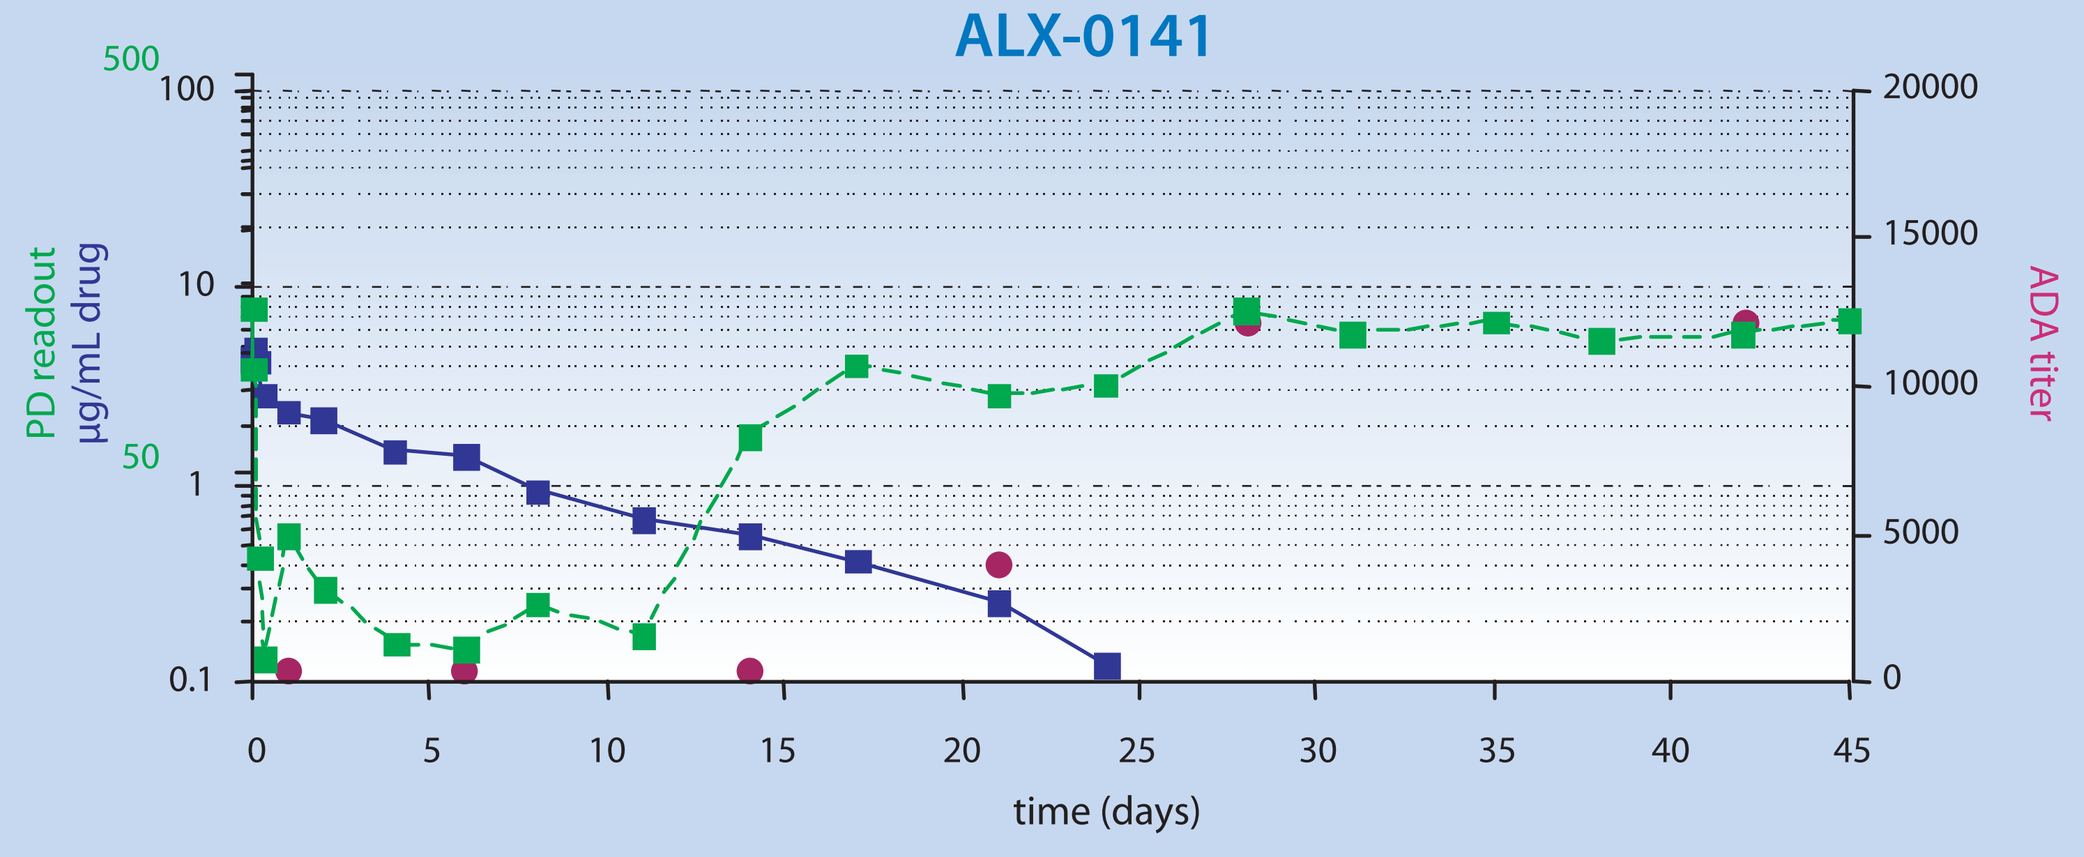
ALX-0141
500
50
PD readout
µg/mL drug
100
10
1
0.1
20000
15000
10000
5000
0
ADA titer
0
5
10
15
20
25
30
35
40
45
time (days)

Conclusions

- Immunogenicity assay performance was determined in ***different species matrices*** and ***different formats*** were compared. Assay performance can vary significantly in different species matrices, even if a species independent assay format is used
- Acceptable ***drug tolerance levels*** are determined on a case-by-case basis
- ***Drug tolerance can be improved*** by implementing an acid dissociation step, using in solution phase kinetics or by switching assay format or platform
- The ***choice of positive control*** influences the outcome of validation, therefore it is advised to confirm the validation cut points using real study samples
- Although immunogenicity observed in preclinical studies cannot predict the immunogenicity potential of a drug candidate in the clinic, it is an ***essential tool*** in the interpretation of preclinical results

Acknowledgements

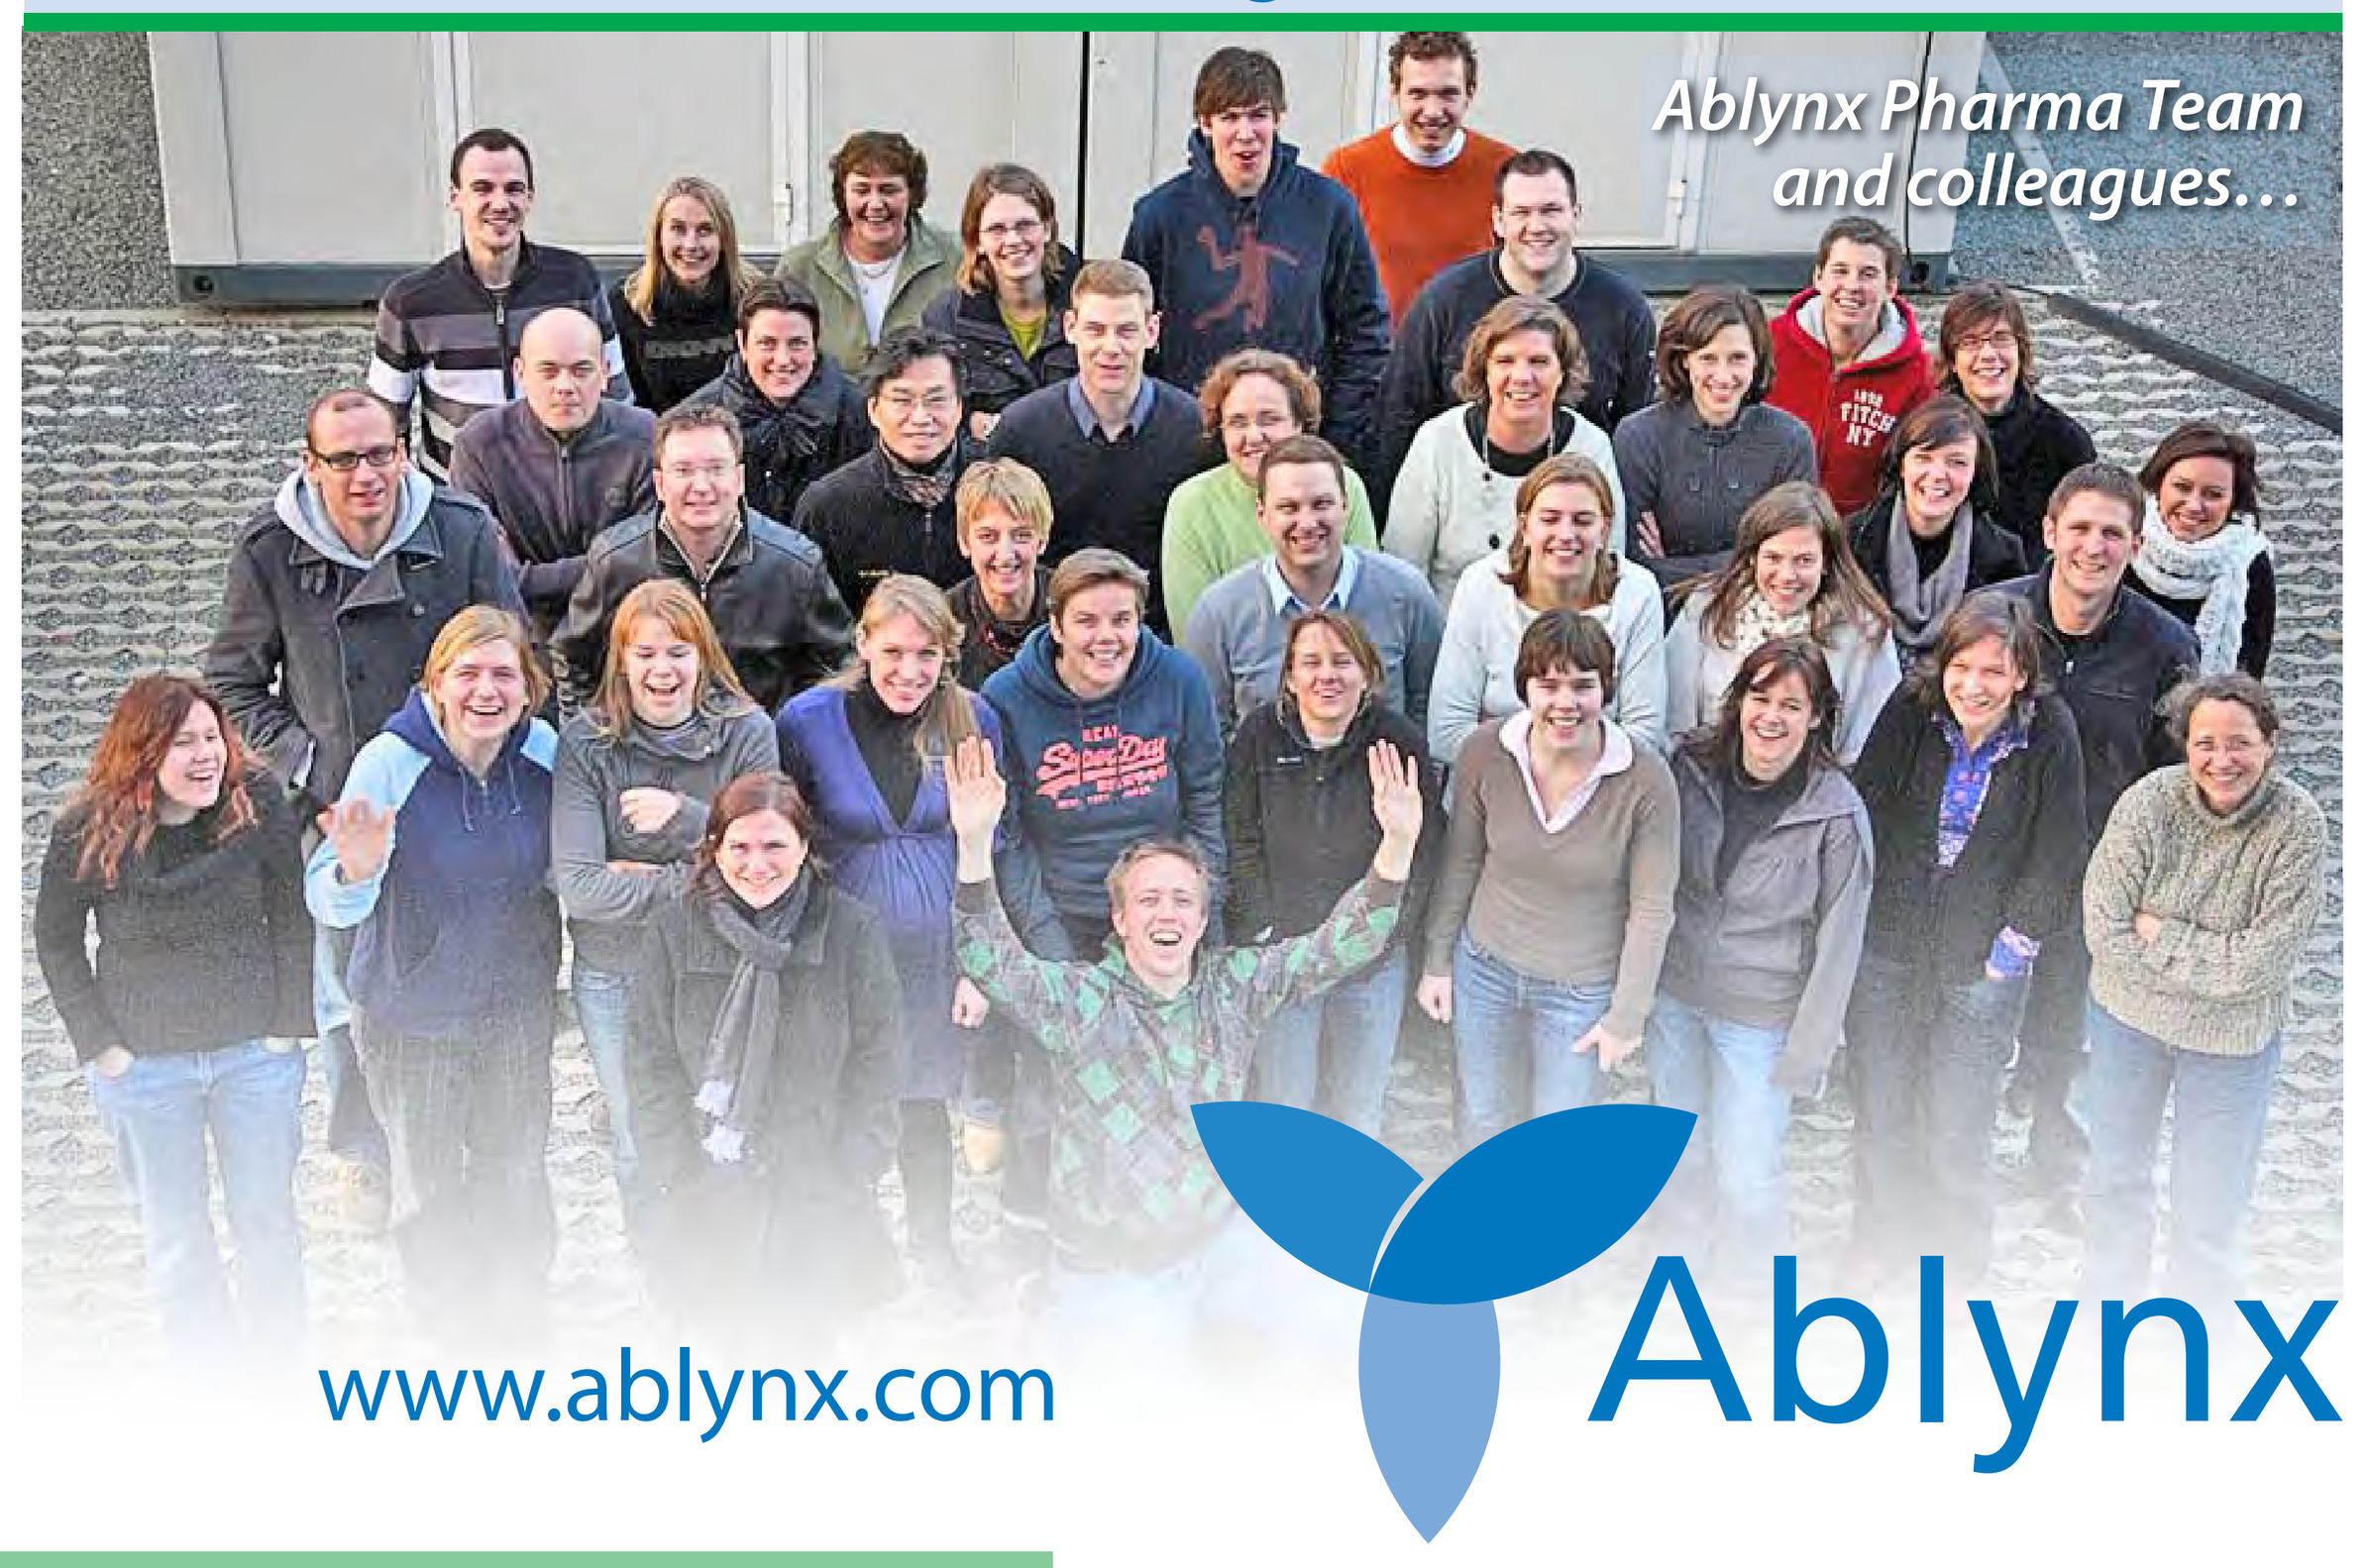
Ablynx Pharma Team
and colleagues…

www.ablynx.com

Ablynx

Nanobody is a registered trademark of Ablynx NV.