Poster #: T2050

A Simplified Process to Develop "Fit-for-Purpose" PK/PD Assays in Support of Early Drug Discovery Programs

Rong Liu, Renuka Pillutla, Binodh Desilva, Yan Zhang

BAS-Biologics, Bristol Myers Squibb, Princeton, NJ

Rong Liu
Rt. 206 & Province Line Road
Lawrenceville, NJ 08543
Phone: 609-252-5909
E-mail: rong.liu@bms.com

Abstract

Purpose: During early drug discovery, the pharmacokinetic (PK) and pharmacodynamic (PD) data play key roles in proof-of-concept, lead selection, and lead optimization. With rapid expansion of biologic targets and modalities in biopharmaceutical industry, more and more novel biologic therapeutic programs are being initiated. Speedy development of ligand binding PK/PD assays to generate data enabling quick decisions in early discovery stages has become more important than ever. We took advantage of the capability of Gyros immunoassay platform in full automation and fast turnaround time to develop a simplified process in streamlining PK/PD assay development to support early drug discovery programs.

Methods: A 4-step process has been developed in our lab to quickly develop "fit-for-purpose" PK/PD assays.

Step 1: Prepare biotinylated -capture antibody/target and Alexa Fluor 647 labeled-detection antibody. It is recommended to label the potential capture and detection pair/pairs simultaneously with both biotin and Alexa 647.

Step 2: Perform feasibility run to identify best combination of capture and detection pair(s). This run may be performed at saturating concentration of capture (700nM recommended by Gyros) and a fixed concentration of detection testing multiple combinations within the same run.

Step 3: Fine tune standard curve range, optimize the detection concentration, and test the matrix interference. All of the above purpose in Steps 3 can be fulfilled in one run.

Step 4: Finalize standard curve range, and test QC recovery and dilution linearity.

Results: Six assays have been developed using the 4-step process. Development time is shortened to two to three days. All of the assays showed sufficient sensitivity with LLOQs ranging from 6-120 ng/mL and at least 2 logs of dynamic range to fit our study needs. Intra-assay and inter-assay precision of QCs are <25% with recovery between 75-125%.

Conclusion: A simplified 4-step process using the Gyros immunoassay platform has been successfully adopted to rapidly develop "fit-for-purpose" PK assays to support early discovery programs in timely manner. The process not only shortens assay development time, but generates assays with acceptable sensitivity, accuracy and dynamic range for study needs.

An Example To Demonstrate 4-Step Work Flow to Detect Lead1 in Program X

Step 1: labeled reagent generation:

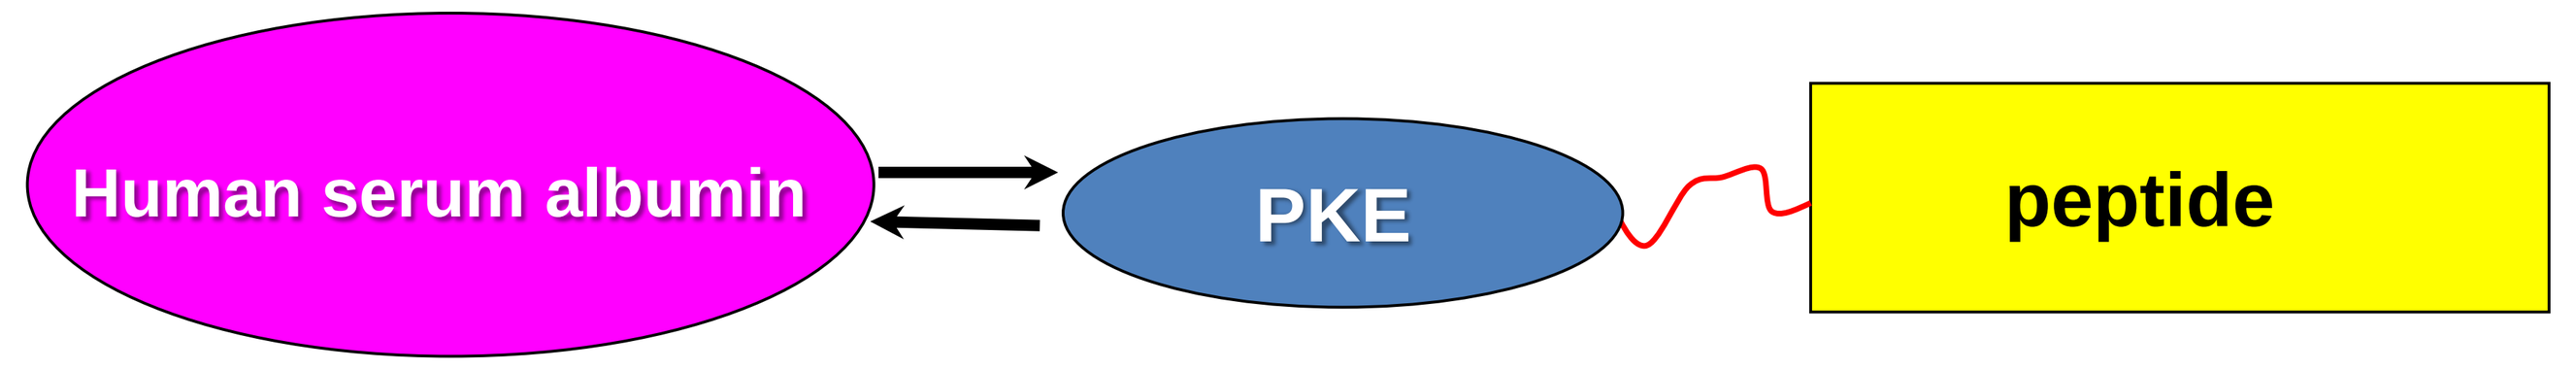

Human serum albumin
PKE
peptide

Lead structure in program X

- Biotinylated and Alexa Fluor 647 labeled anti-peptide polyclonal antibody
- Biotinylated and Alexa Fluor 647 labeled anti-PK enhancer (PKE) polyclonal antibody

Step 2: Identification of best combination of capture and detection pair(s)

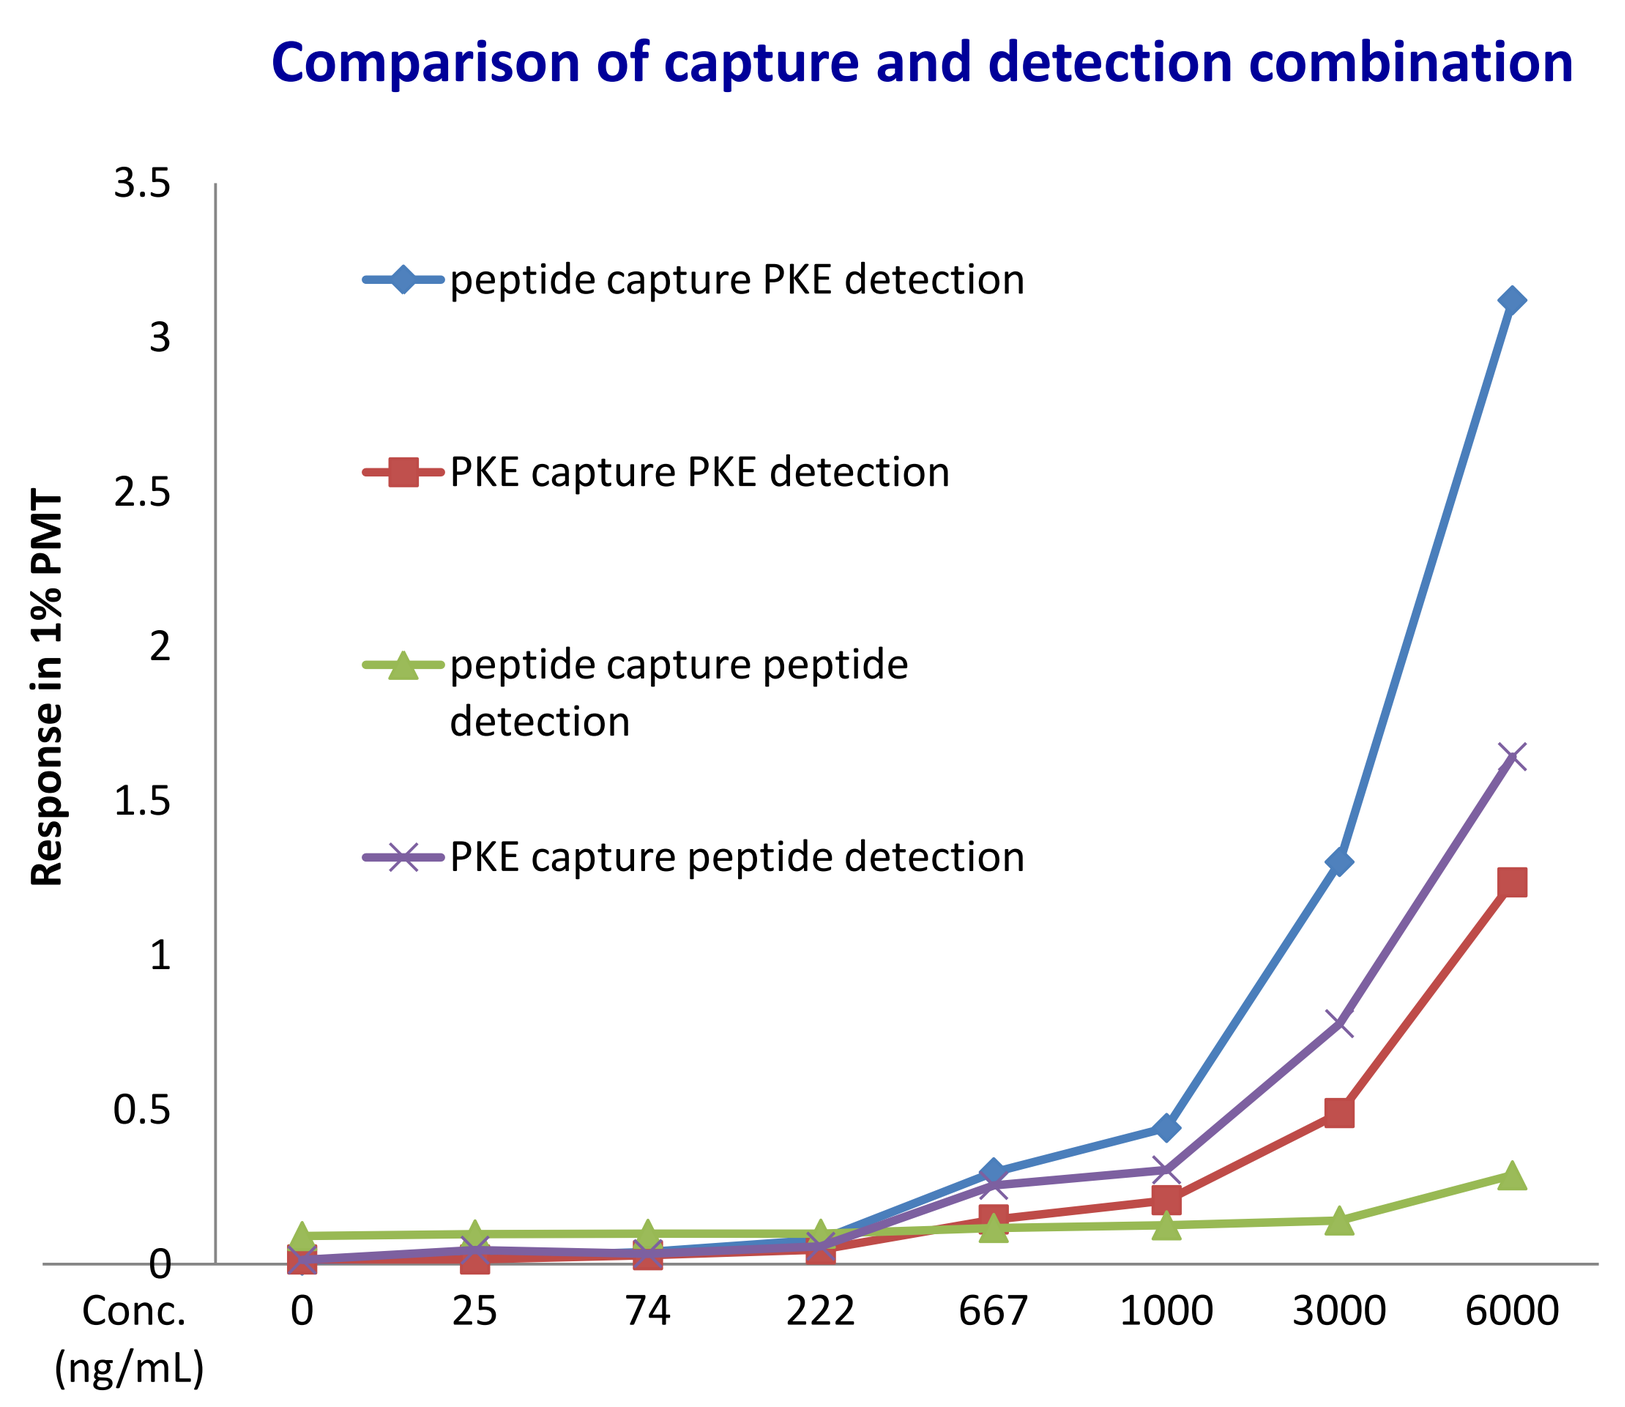

Comparison of capture and detection combination
Response in 1% PMT
peptide capture PKE detection
PKE capture PKE detection
peptide capture peptide detection
PKE capture peptide detection
Conc. (ng/mL)
0 25 74 222 667 1000 3000 6000

- The best combination is biotinylated anti-peptide pAb as capture and Alexa Fluor 647 labeled anti-PKE pAb as detection.
- The pair was selected for further assay optimization.

Step 3: Optimization of the assay:

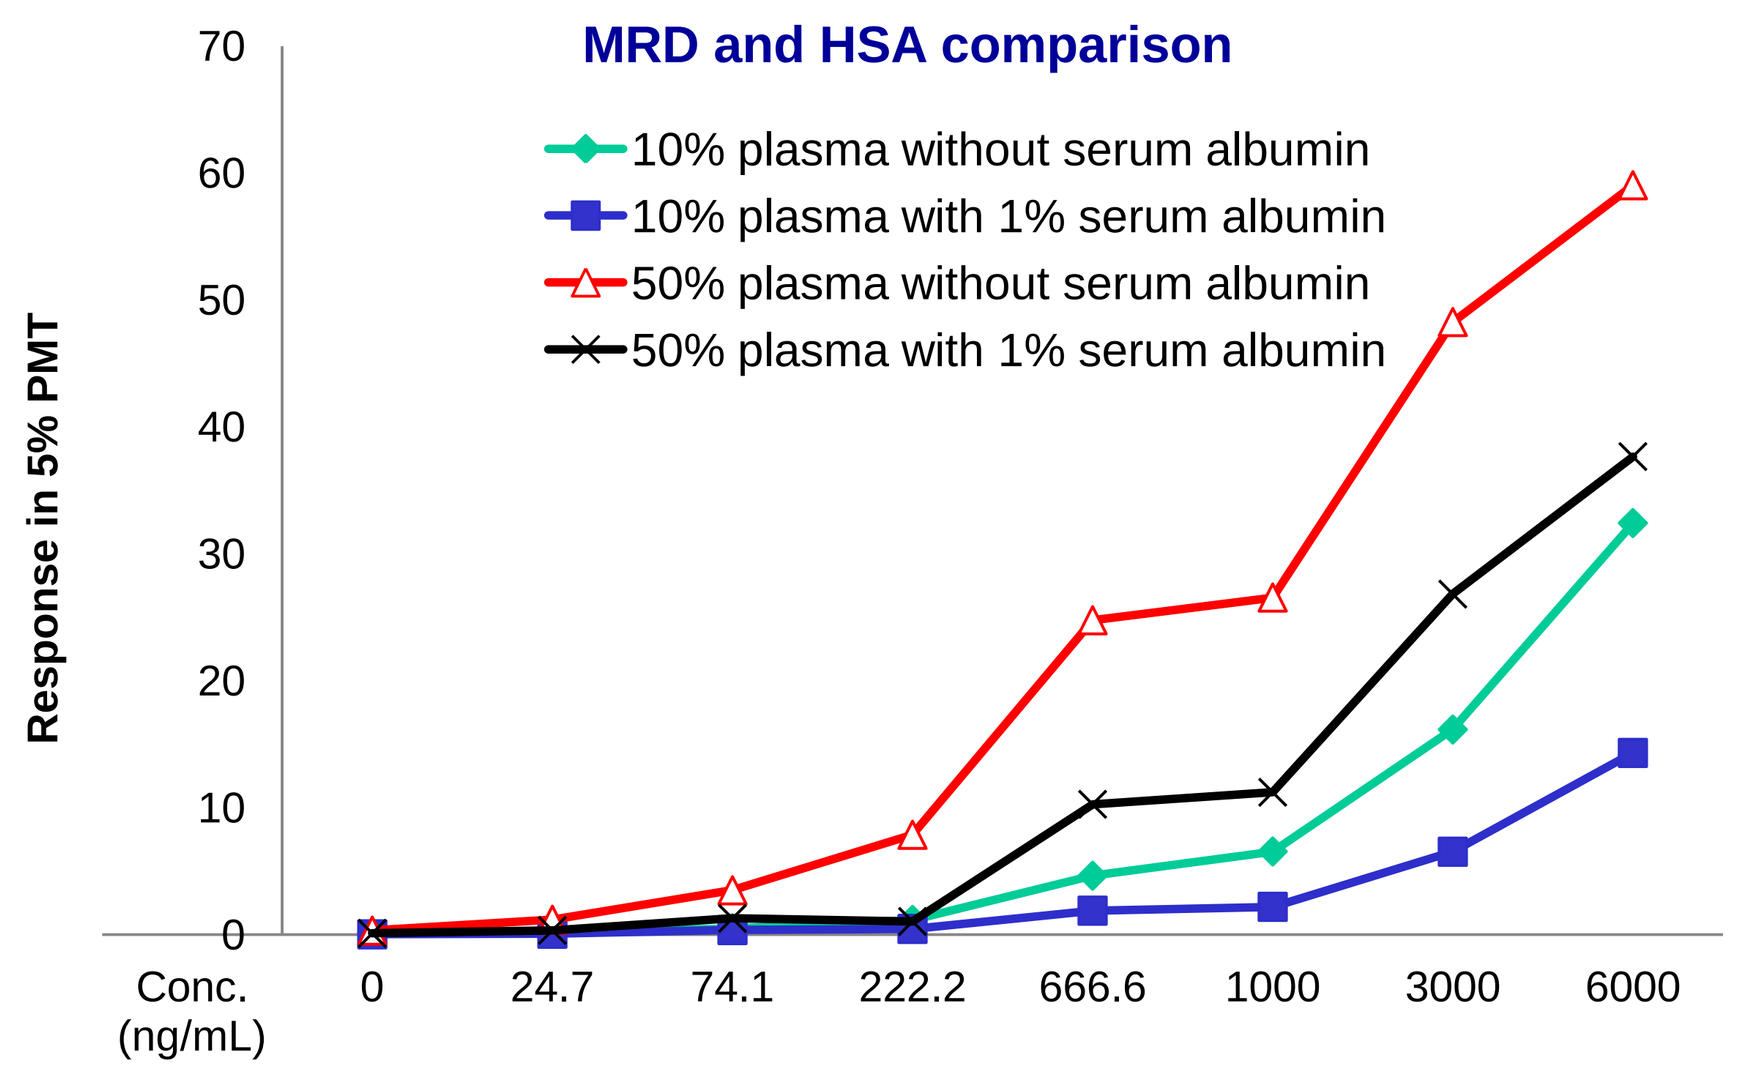

MRD and HSA comparison
Response in 5% PMT
10% plasma without serum albumin
10% plasma with 1% serum albumin
50% plasma without serum albumin
50% plasma with 1% serum albumin
Conc. (ng/mL)
0 24.7 74.1 222.2 666.6 1000 3000 6000

Response	10% plasma		50% plasma	
Conc. (ng/mL)	No Serum Albumin	1% Serum Albumin	No Serum Albumin	1% Serum Albumin
0	0.18	0.03	0.33	0.11
24.7	0.18	0.08	1.19	0.35
74.1	0.51	0.37	3.51	1.29
222.2	1.17	0.46	7.88	1.05
666.6	4.65	1.90	24.78	10.28
1000	6.56	2.20	26.57	11.23
3000	16.17	6.54	48.27	26.83
6000	32.43	14.33	59.04	37.67

- Assay sensitivity is better in 50% matrix than in 10% matrix.
- Human serum albumin interference and MRD can be evaluated in the same run.
- Assay optimization is rapid.

Step 4: Finalization of standard curve range, and test QC recovery:

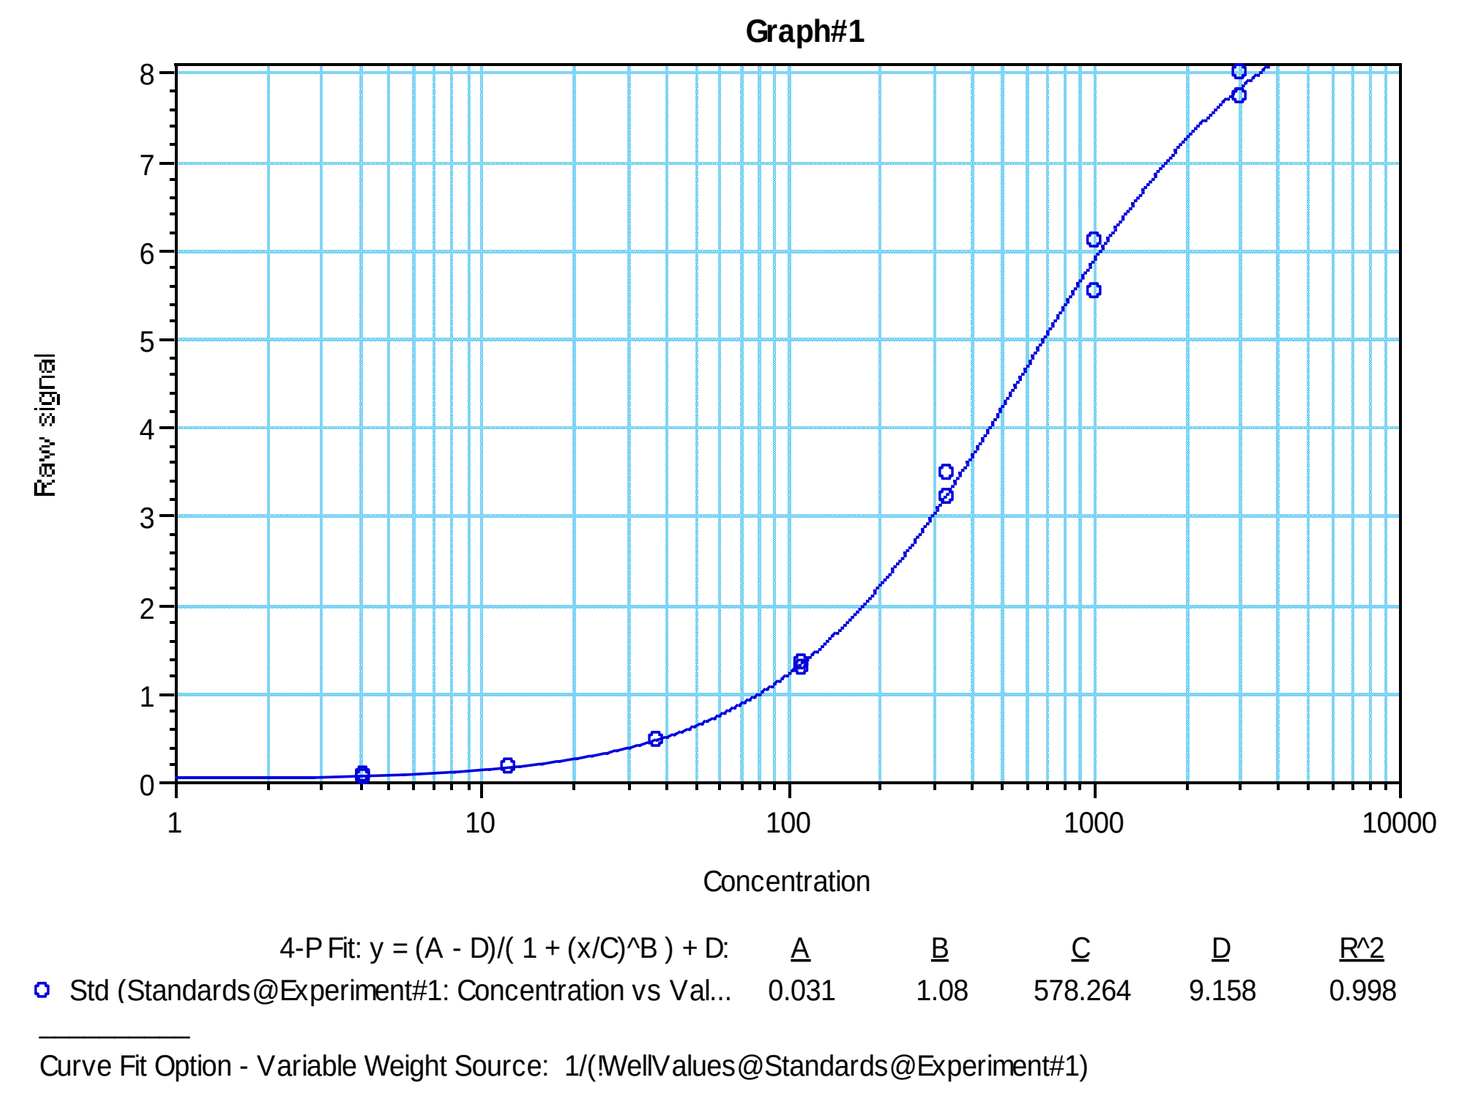

Graph#1
Raw signal
Concentration
4-P Fit: y = (A - D)/(1 + (x/C)^B) + D: A 0.031 B 1.08 C 578.264 D 9.158 R^2 0.998
Std (Standards@Experiment#1: Concentration vs Val...
Curve Fit Option - Variable Weight Source: 1/(WellValues@Standards@Experiment#1)

Expected std. Conc.(ng/mL)	Back Calculated Conc.(ng/mL)	Values	CV%	%Recovery
3000	3511.8	8.02	2.4	117.1
	2798.9	7.75		93.3
1000	855.0	5.54	6.8	85.5
	1092.1	6.10		109.2
333	366.9	3.50	5.4	110.1
	327.6	3.24		98.3
111	111.6	1.35	3.8	100.5
	105.1	1.28		94.6
37	36.5	0.47	2.4	98.4
	37.8	0.49		102.0
12	12.0	0.17	4.1	97.6
	12.9	0.18		104.3
4	5.3	0.09	22.5	129.7
	3.2	0.06		78.0

QC recovery

Expected QC Conc.(ng/mL)	Back Calculated Conc.(ng/mL)	Values	CV%	%Recovery
2500	2172.2	7.40	10.0	86.9
	1876.3	7.16		75.1
80	85.1	1.05	2.0	106.3
	87.4	1.08		109.3
10	11.8	0.17	10.1	117.7
	12.8	0.18		127.6

Successful Development of Multiple Assays Using the Simplified Work Flow

3 Assays for Program X

- Program need:
 - Program X is in early drug discovery and progresses very fast. Bioanalytical methods measuring multiple leads with variants in the peptide, linker and PKE regions are needed for lead selection. The same work flow was applied to develop those assays to support the program.
- Assays:
 - Assay1 is to measure intact compounds biotinylated anti-peptide as capture, Alexa Fluor 647 labeled anti-PKE as detection
 - Assay2, a generic assay, is to measure PKE portion of the compound and used for compounds with different peptide variants biotinylated anti-PKE as capture, Alexa Fluor 647 labeled anti-PKE as detection
 - Assay3 is to measure peptide portion of the compound and used for comparator compounds with different PKE biotinylated anti-peptide as capture, Alexa Fluor 647 labeled anti-peptide as detection

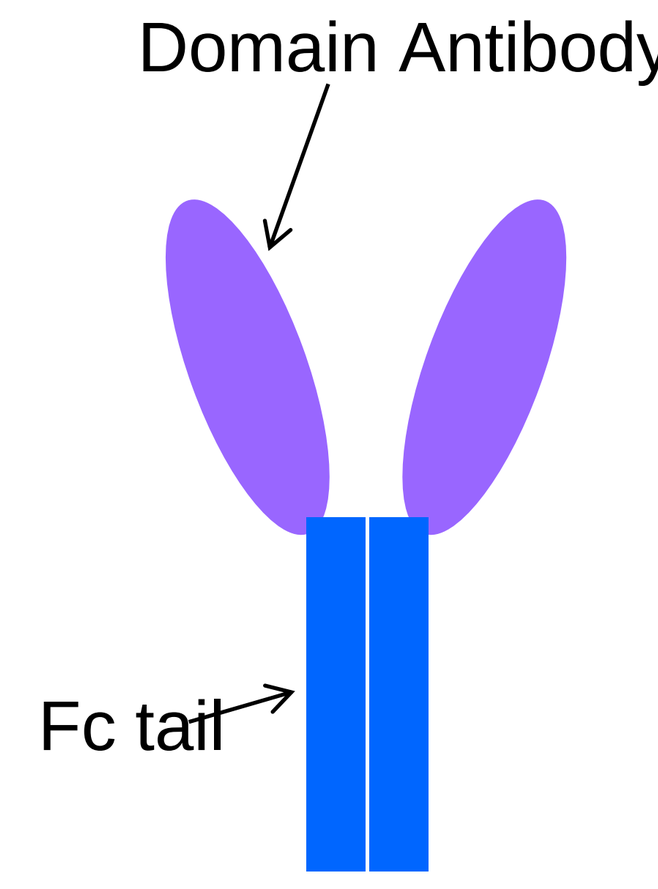

Domain Antibody
Fc tail

3 Assays for Program Y

Lead structure in program Y

- Program Y need:
 - The leads in program Y are fusion proteins binding either human or cyno Target and with different Fc tails (IgG4/IgG1). Assays are needed to enable selecting which Fc tail to move forward.
- Assays:
 - Assay4 is to detect cyno lead with different tails (assay 4-1 for IgG1 tail and assay 4-2 for IgG4 tail): biotinylated cyno Target as capture, Alexa Fluor 647 labeled anti-Vh domain antibody (dAb) pAb as detection
 - Assay5 is to detect human lead with different tails (assay 5-1 for IgG1 tail and assay 5-2 for IgG4 tail): biotinylated human Target as capture, Alexa Fluor 647 labeled anti-Vh domain antibody (dAb) pAb as detection
 - Assay6, a more sensitive assay to detect cyno lead with IgG4 tail: biotinylated cyno Target as capture, Alexa Fluor 647 labeled anti-human IgG4 mAb as detection

Calibration Curves of 6 Fit-for-Purpose Assays

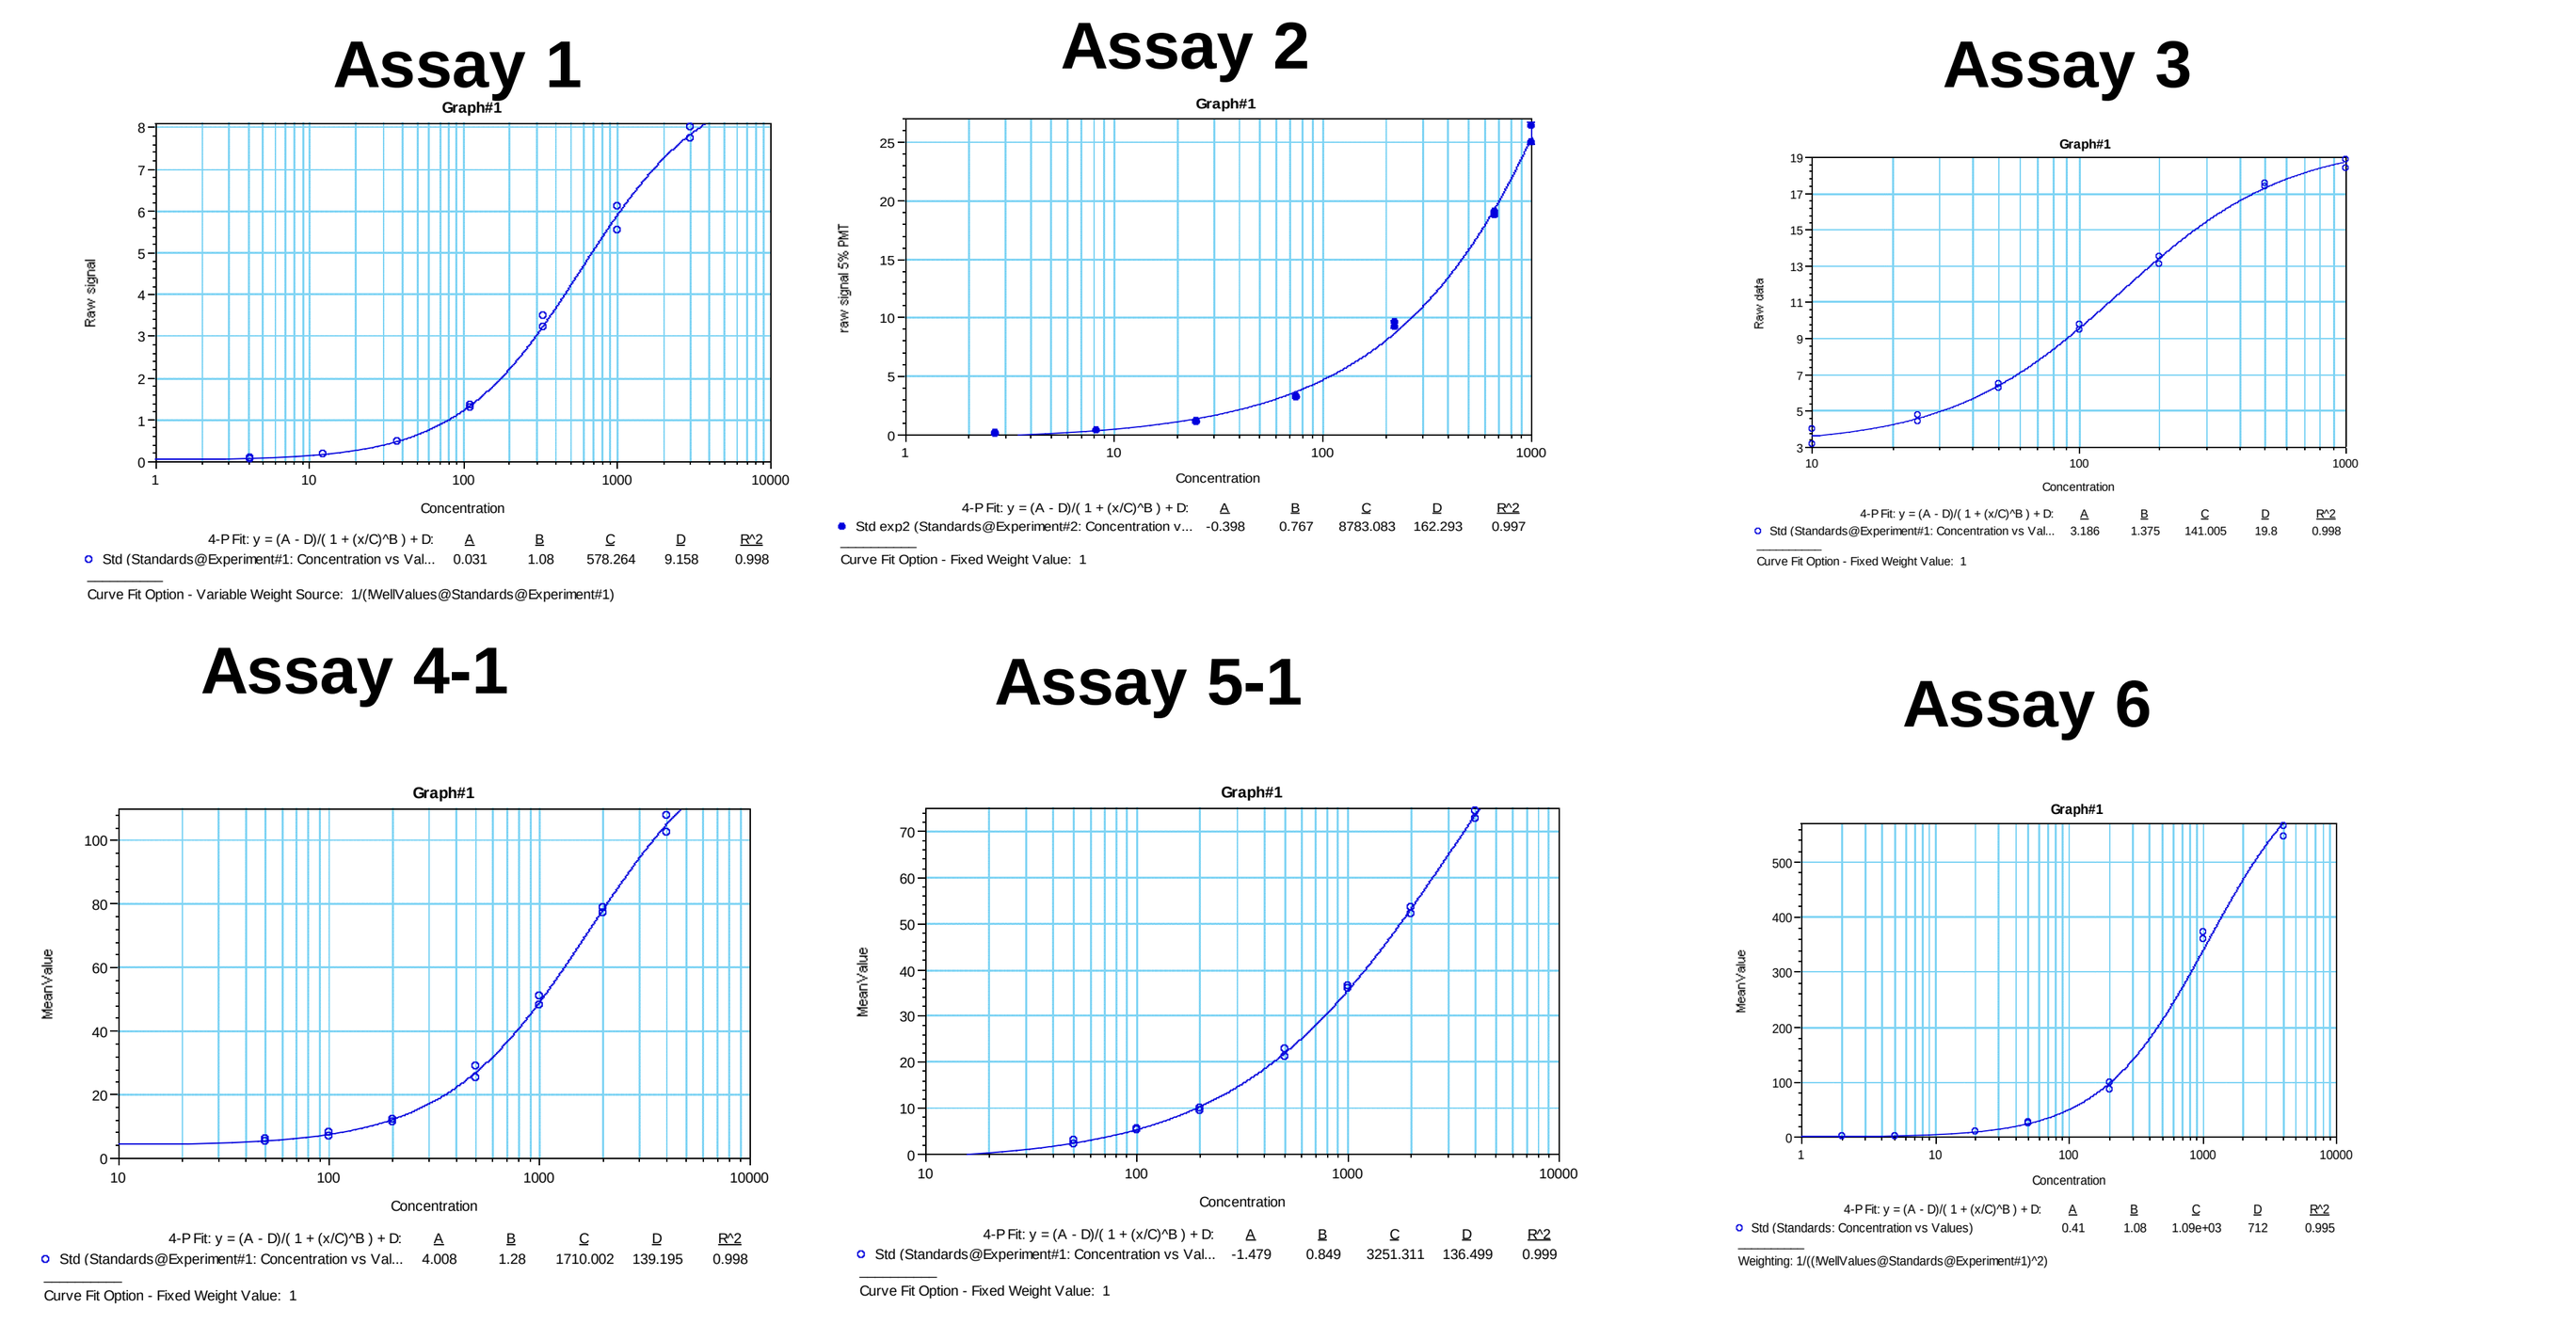

Assay 1
Assay 2
Assay 3
Assay 4-1
Assay 5-1
Assay 6

Assay Performance of the Six Fit-for-Purpose Assays

Assay #	Expected QC Conc.(ng/mL)	CV%*	%Recovery*
1	2500	10.0	75-87
	80	2.0	106-109
	10	10.1	118-128
2	800	2.8	99 - 106
	100	4.6	108 - 115
	15	5.4	88 - 97
3	800	8.0	95-107
	100	5.5	83-90
	20	17	82-104
4-1	3000	3.1	98-109
	600	3.8	96-102
	120	3.4	96-103
5-1	3000	2.6	89-97
	300	1.5	85-88
	60	6.0	105-113
6	3000	1.4	101-110
	200	0.5	81
	6	0.1	106

*n=4

Conclusion

A simplified 4-step process using the Gyros immunoassay platform has been successfully adopted to rapidly develop "fit-for-purpose" PK assays to support early discovery programs in timely manner. The process not only shortens assay development time to 2 to 3 days with minimal hands-on time, but generates assays with acceptable sensitivity and dynamic range for study needs.

RLiu_AAPS_2012