STOP STROKE©: Acute care coordination medical application; a brief report on post implementation performance at a primary stroke center.

Dickson, Robert MD[1], Sumathipala, Dineth MBBS[2], Reeves-Reed, Jennifer RN-MSN[3]
Baylor College of Medicine Houston Texas[1], Palmerston North Hospital NZ[2], Good Shepherd Medical Center Longview Texas[3]

Take Home Points

46%
REDUCTION IN DOOR TO NEEDLE TIME WITH STOP STROKE USE

3.7x
IMPROVEMENT IN DTN TIME <60 MINUTES

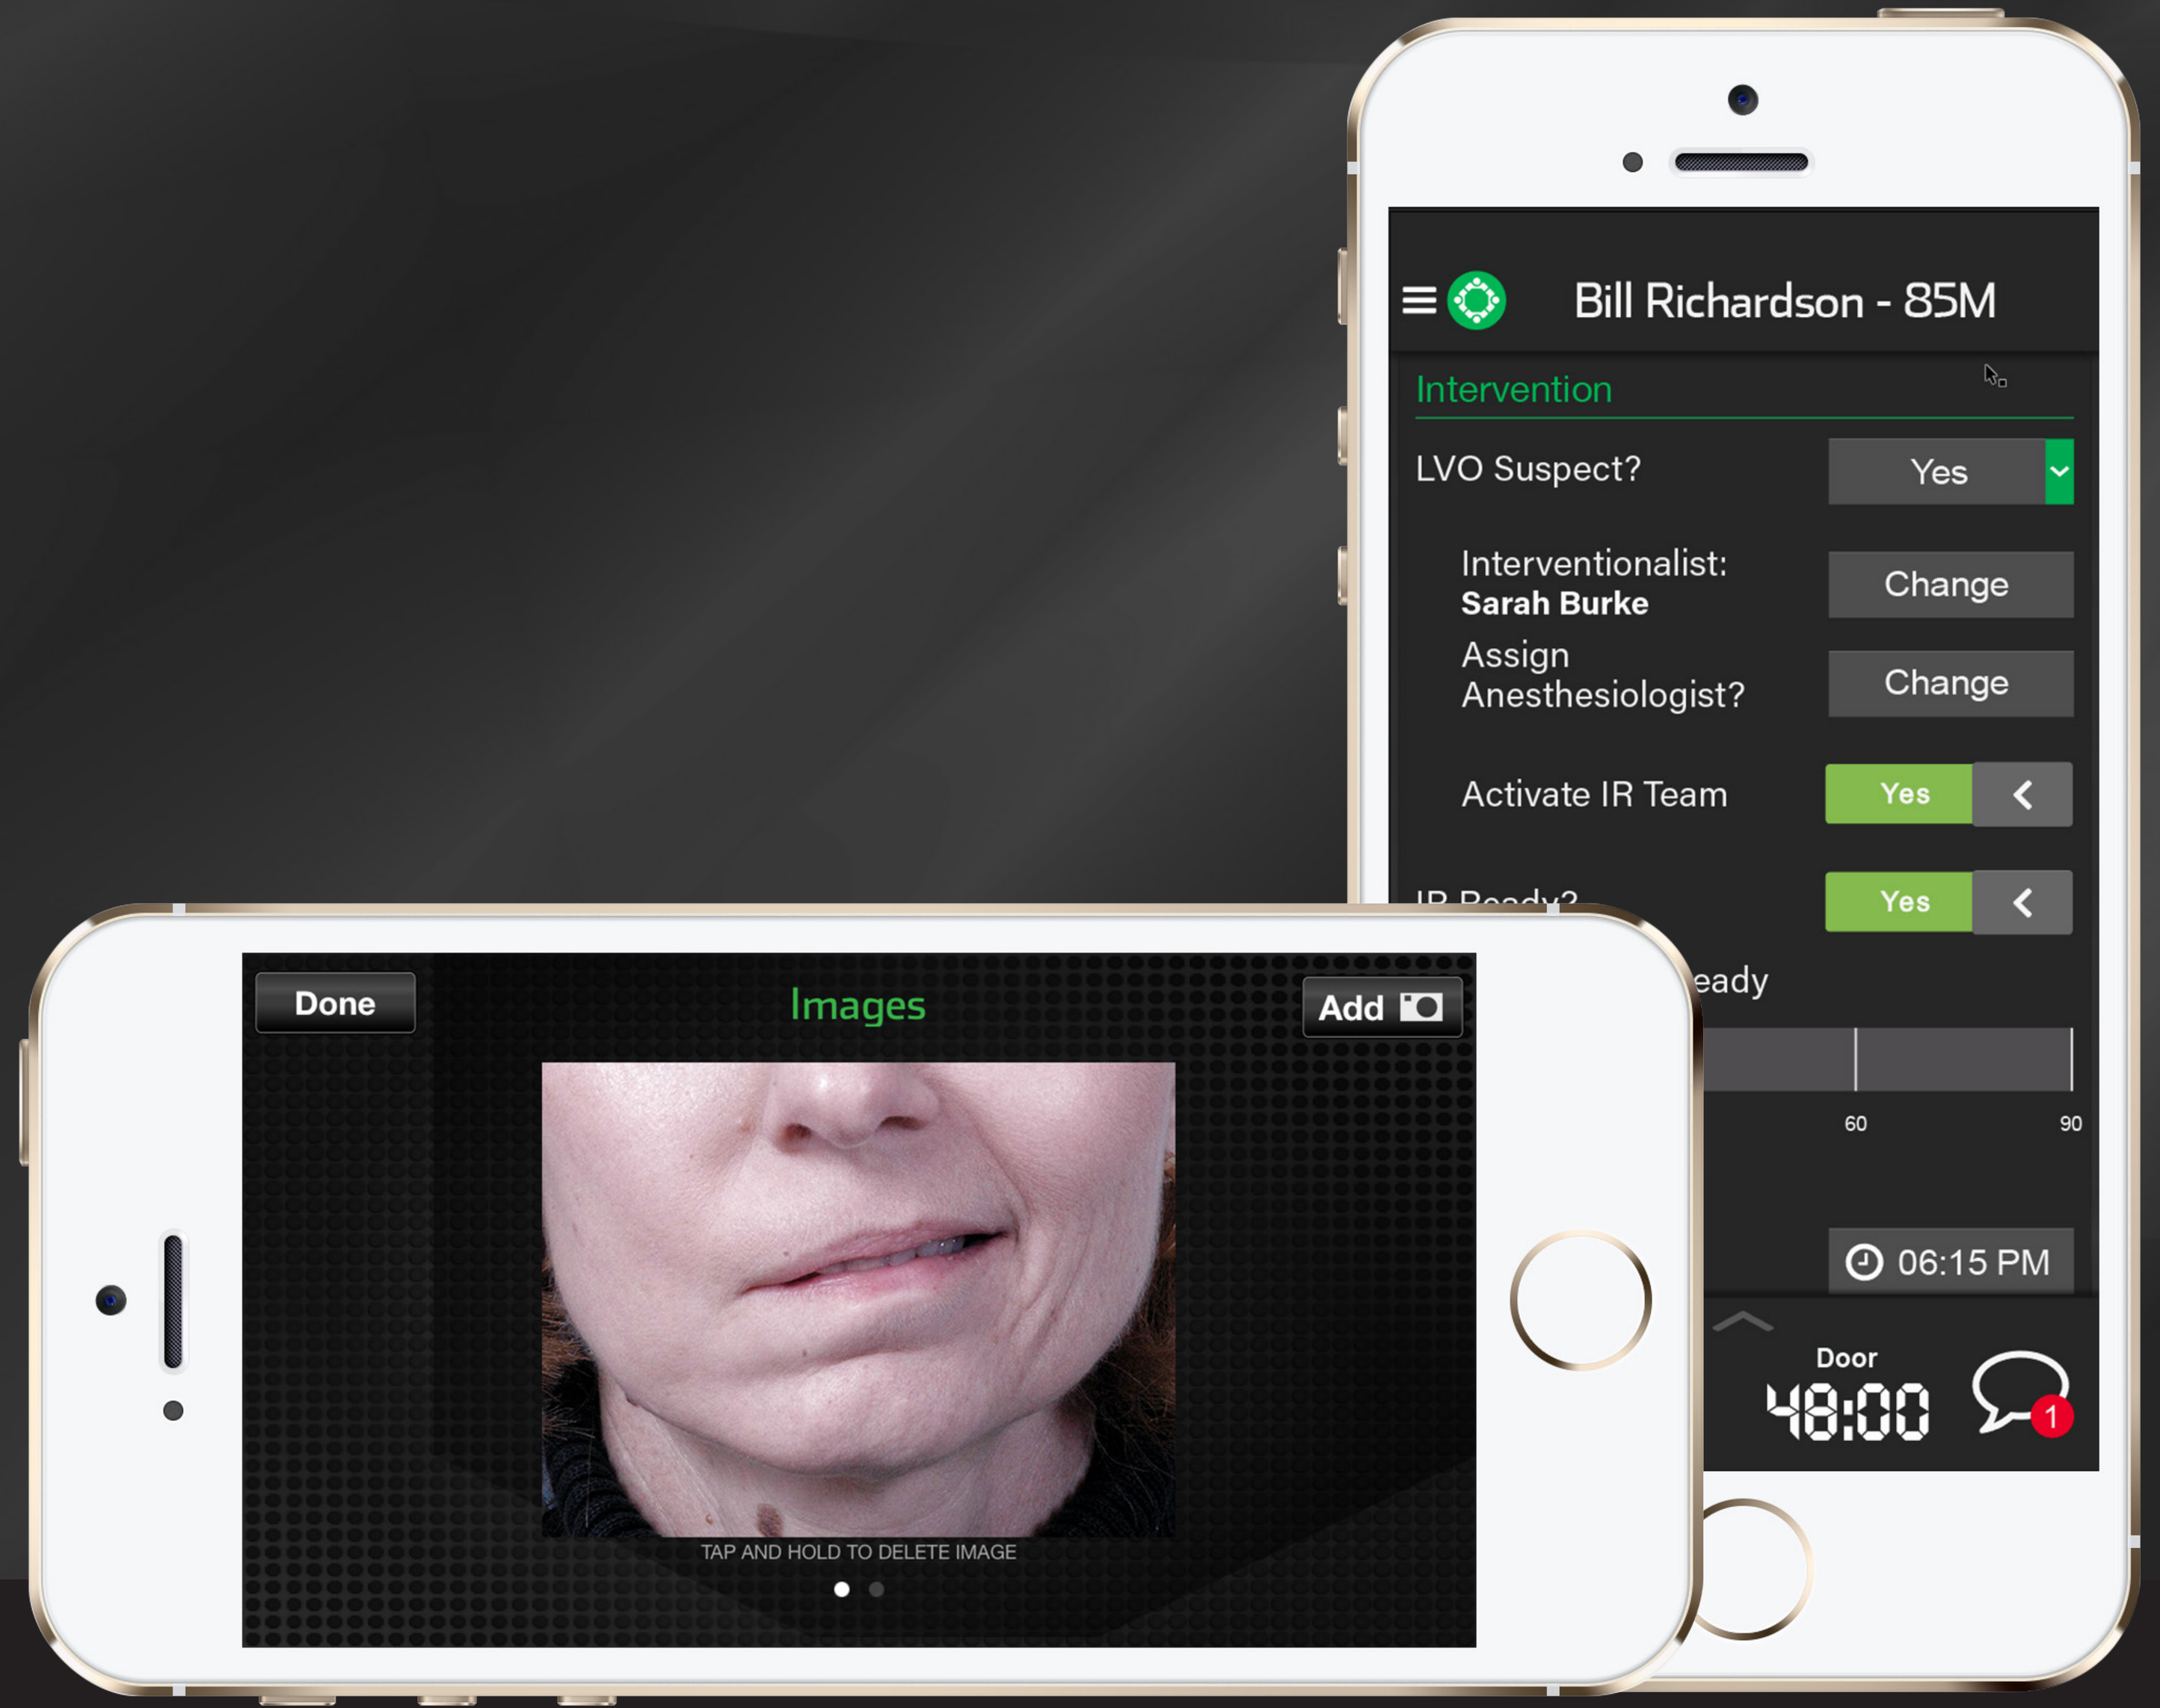
Bill Richardson - 85M
Intervention
LVO Suspect?
Yes
Interventionalist:
Sarah Burke
Change
Assign
Anesthesiologist?
Change
Activate IR Team
Yes
Yes
06:15 PM
Door
48:00
Done
Images
Add
TAP AND HOLD TO DELETE IMAGE

Objective

The objective of our study is to evaluate the effect of the Pulsara Stop Stroke© medical application on door-to-needle (DTN) time in patients presenting to our emergency department with acute ischemic stroke (AIS). The secondary objective was to evaluate the DTN performance of dedicated neurohospitalists vs. private practice neurologists covering emergency department stroke call, with and without Stop Stroke© app usage.

Methods

We conducted a retrospective cohort study of the Good Shepherd Health System stroke quality improvement dashboard for an 18-month period between February 2014 and August 2015. The dashboard tracks metrics on stroke cases receiving Tissue Plasminogen Activator (TPA) at our primary stroke center with annual emergency department volume of 90k. We analyzed all data from cases receiving TPA for AIS during the study period. The primary outcome was comparison of mean door-to-needle (DTN) time performance in cases with and without app usage. Secondary outcome was the comparison of mean DTN time between neurohospitalist and private neurologists with and without use of Stop Stroke©.

Results

During the study period, there were 85 stroke activations receiving TPA (63 with Stop Stroke©, 22 without). In cases where the app was used, we observed a reduction in mean DTN time of 40 minutes, 87 minutes to 47 minutes, a 46% reduction (p=<0.001). There was no significant difference in DTN time observed between the neurohospitalist and private neurologist performance independent of app usage, with a mean DTN of 47 minutes seen for both groups using the app (p=0.87) and 88 minutes and 85 minutes respectively without app use (p=0.81). Mean DTN <60 minutes improved with app use from 18% to 85% with Stop Stroke©.

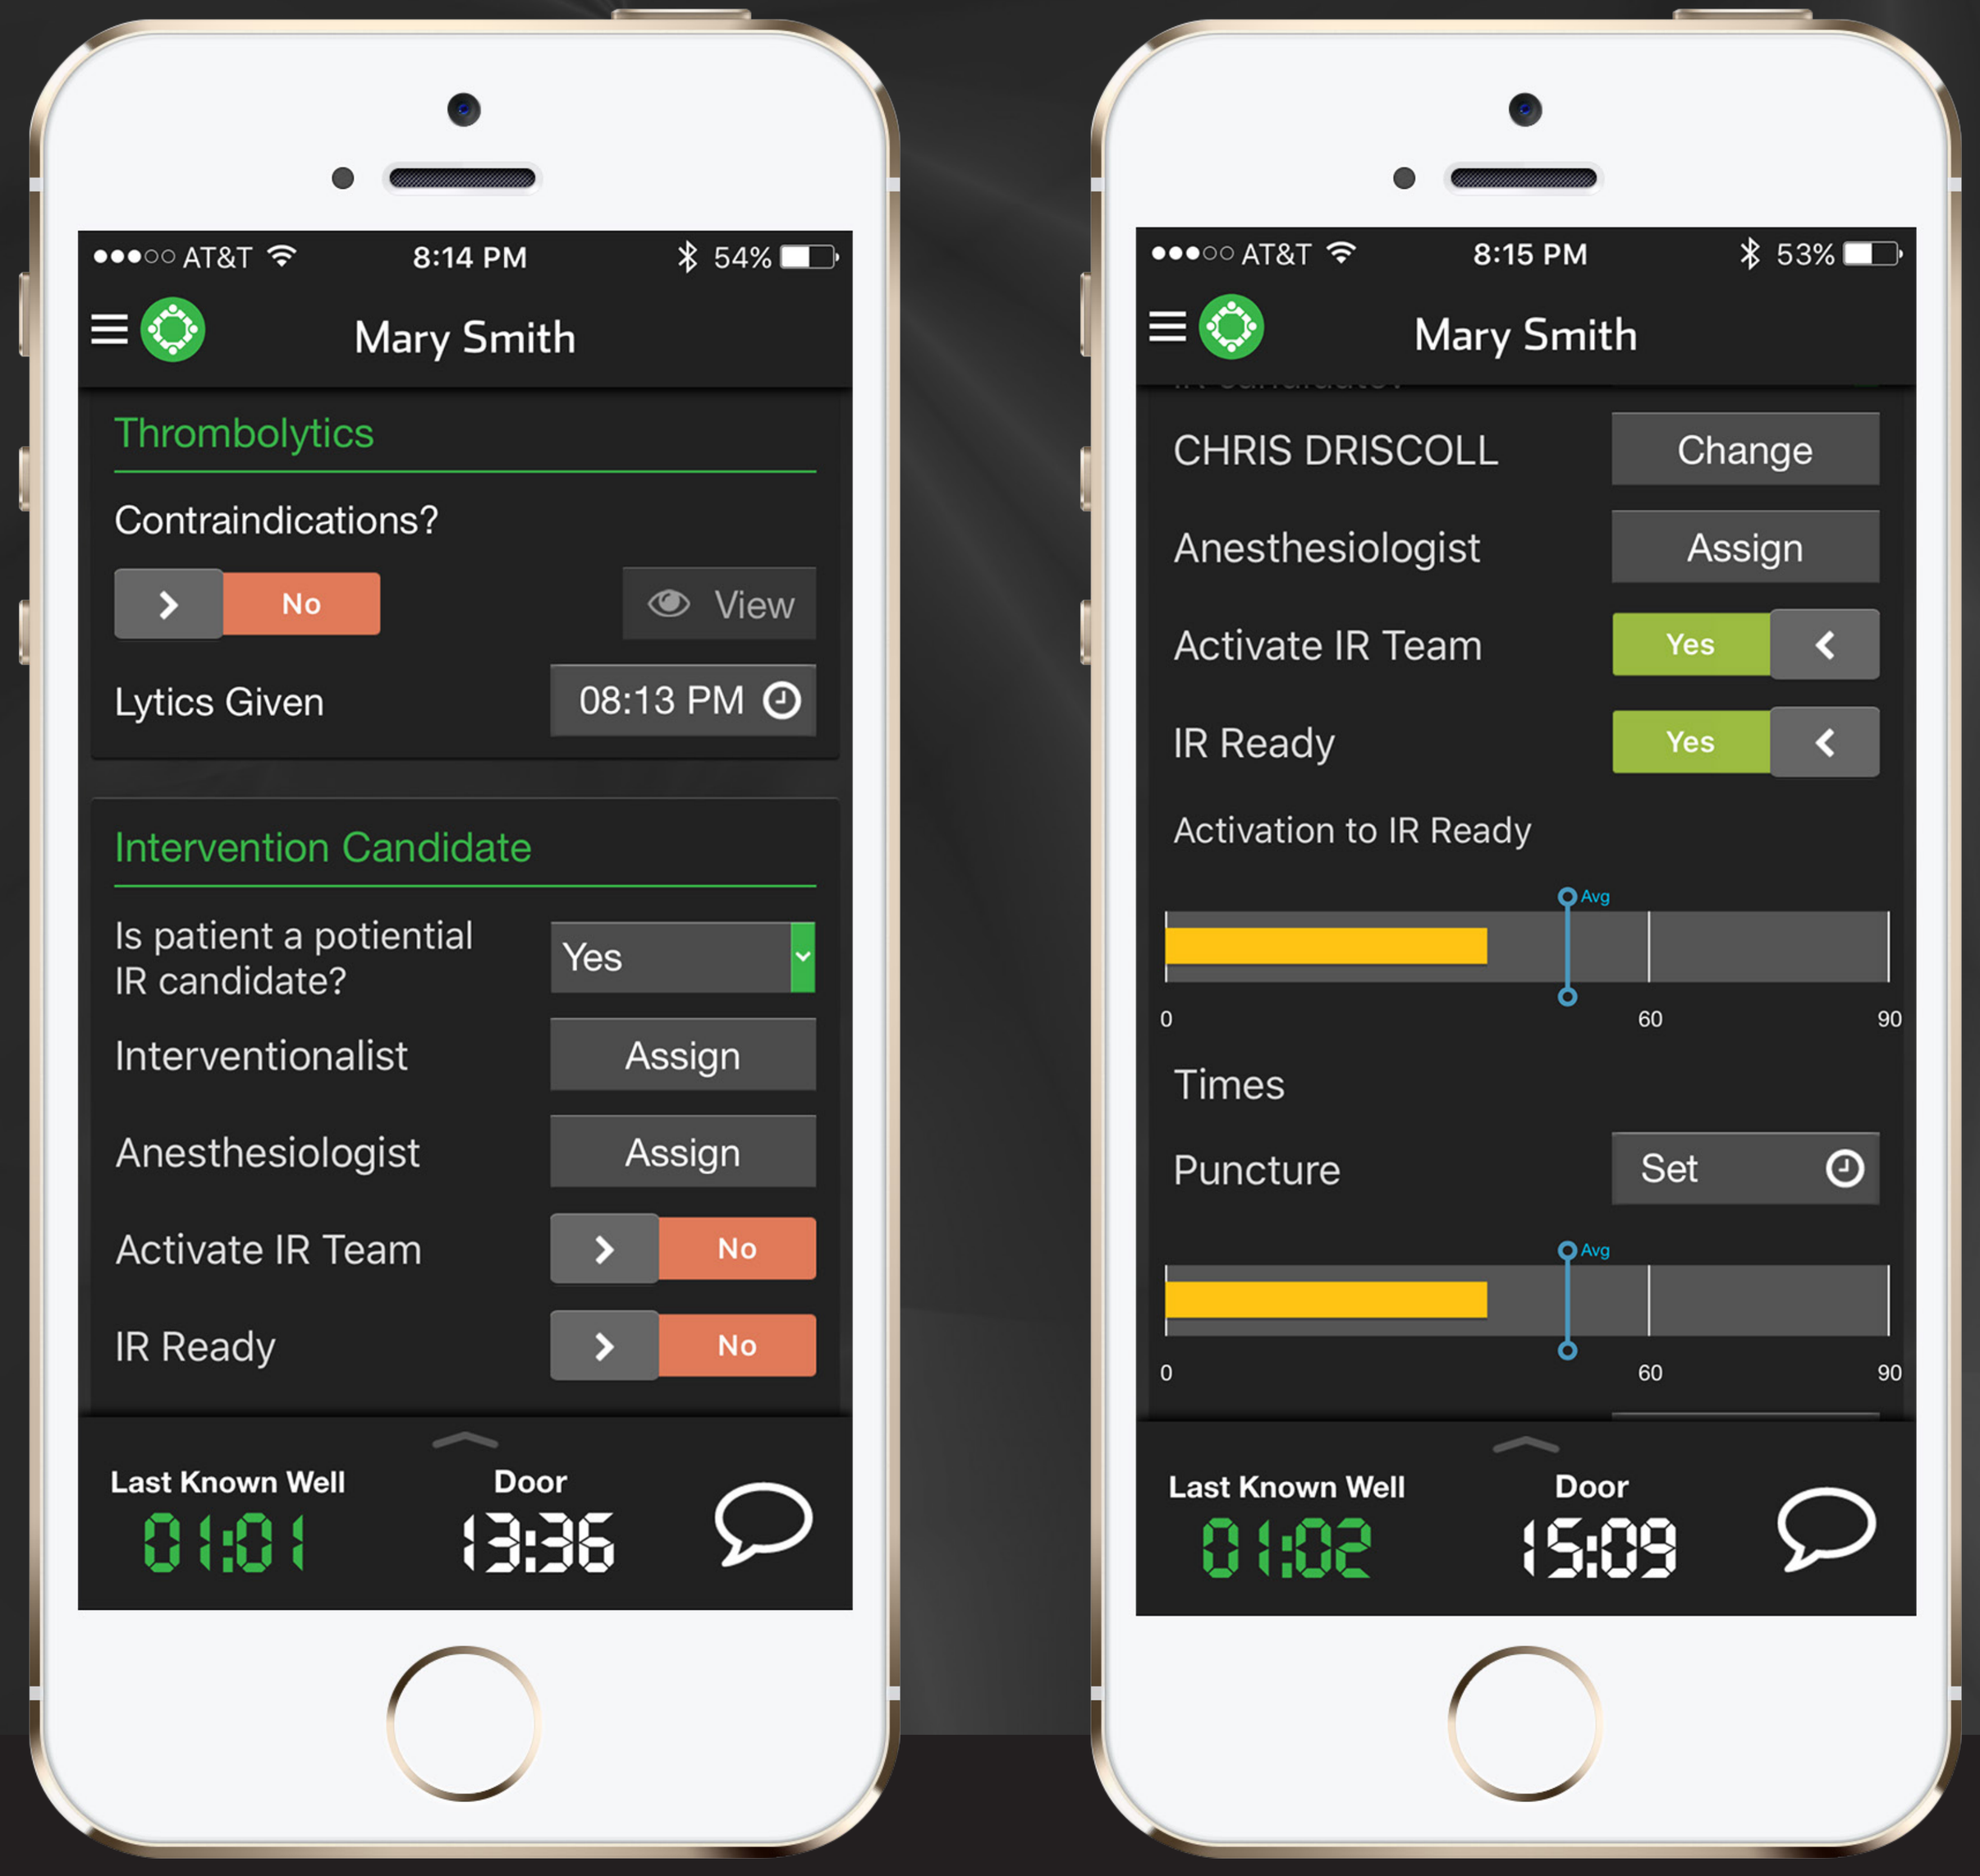
AT&T 8:14 PM 54%
Mary Smith
Thrombolytics
Contraindications?
No
View
Lytics Given
08:13 PM
Intervention Candidate
Is patient a potential IR candidate?
Yes
Interventionalist
Assign
Anesthesiologist
Assign
Activate IR Team
No
IR Ready
No
Last Known Well
01:01
Door
13:36
AT&T 8:15 PM 53%
Mary Smith
CHRIS DRISCOLL
Change
Anesthesiologist
Assign
Activate IR Team
Yes
IR Ready
Yes
Activation to IR Ready
Times
Puncture
Set
Last Known Well
01:02
Door
15:09

Conclusions

In patients arriving to our primary stroke center with AIS, use of Pulsara Stop Stroke© acute care coordination app decreased mean DTN time by 40 minutes. This time reduction represents a significant 46% improvement in this metric and is consistent with the effect seen in our original analysis after implementing the app at our institution. We further observed a 3.7x improvement in DTN <60 minutes with use of the app.

Institutions

BCM® Baylor College of Medicine
GOOD SHEPHERD HEALTH SYSTEM
MIDCENTRAL DISTRICT HEALTH BOARD Te Pae Hauora o Ruahine o Tararua
Tameside Hospital NHS Foundation Trust NHS

pulsara