Invicro A Konica Minolta Company

Voyager THERAPEUTICS

Multi-modality Imaging Study to Determine AAV Bio-Distribution Compared to AAV Transduction in Whole Animals

Invicro.com
hdimant@invicro.com

Hemi Dimant[1], Ankit Gandhi[1], Rob Holt[1], Sanjana Pannem[1], Ildiko Polyak[1], Stefan Collins[1], Jacob Hesterman[1], Holger Patzke[2], Jenna Soper[2], Adrian Kells[2]

[1]Invicro, A Konica Minolta Company, Boston MA; [2]Voyager Therapeutics, Cambridge MA

Introduction

AAVs are at the forefront of gene therapy, allowing for efficient gene delivery with a safe profile. Various AAV serotypes, either natural or genetically engineered, together with various promoters allow researchers to selectively target AAVs to diseased organs and cells. One approach to determine AAV selectivity is to compare the bio-distribution of the AAV capsid to its subsequent gene transduction in whole animals. Various imaging modalities provide insight into these scientific questions. Some *in vivo* modalities can provide information to the bio-distribution and PK of AAVs in whole animal, while others can provide information to gene transduction and cellular localization in single organs. However, no single modality can answer all these questions as they require a broad spectrum of sensitivity, imaging volume and resolution.

Objective

Image and compare AAV bio-distribution compared to AAV transduction in whole rats.

Approach

We designed an advanced multi-modality imaging approach that utilized both nuclear and fluorescence imaging, including SPECT, CT, cryofluorescence tomography (CFT) and microscopy, to investigate where the AAVs are going versus where they eventually transduce. Rats were administered a mixture of ^{125}I-labeled AAVs and non-labeled AAVs intrathecally that carried an iRFP gene. Radiolabeled AAVs enabled longitude visualization of the AAV capsid and its bio-distribution at several time point using SPECT/CT, while AAV-iRFP enabled high resolution 3D visualization of gene transduction in whole rats using CFT. Furthermore, sections were collected to confirm the organ and cellular localization of AAV-mediated iRFP expression.

PK	PE	Localization
SPECT / CT	CFT	Microscopy
^{125}I-AAV	AAV-mediated iRFP expression	AAV-mediated iRFP expression
Whole Rat	Rat vertebrae	Rat vertebrae
In vivo	*Ex vivo*	*Ex vivo*
Temporal & Spatial	Spatial 3D	Spatial 2D
~1mm	~ 50µm	~ 0.5µm

AAV QC

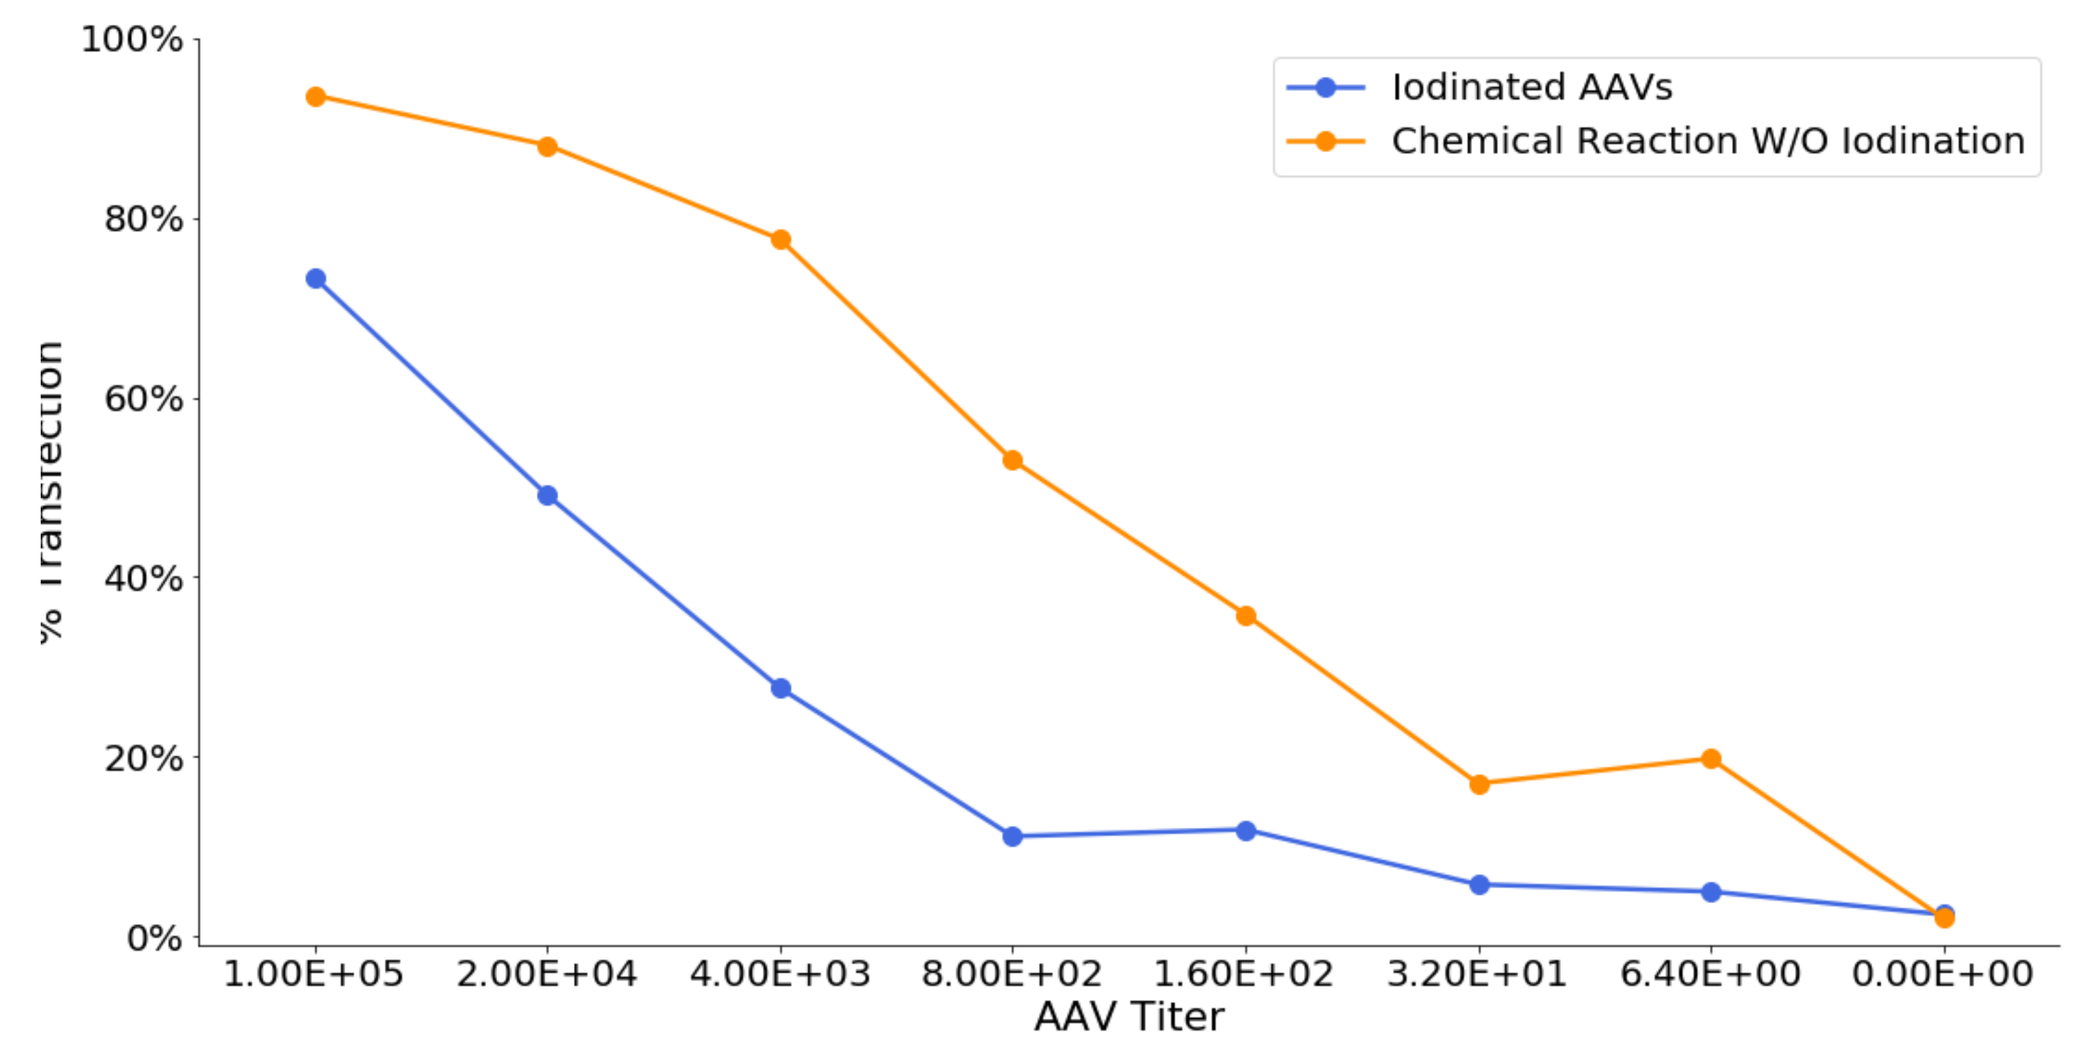
100%
80%
60%
40%
20%
0%
% Transfection
Iodinated AAVs
Chemical Reaction W/O Iodination
1.00E+05
2.00E+04
4.00E+03
8.00E+02
1.60E+02
3.20E+01
6.40E+00
0.00E+00
AAV Titer

Figure 1: Reduced functionality was observed following direct Iodination (blue), attributed to the directly conjugated iodine itself, compared to AAVs that undergone the chemical reaction without the iodine conjugation (orange).
#1- Iodinated AAVs
#2- Chemical reaction w/o iodination

Summary

- We present an advanced multi-modality imaging approach to allow imaging across a wide-spectrum of resolution, sensitivity and volume from the same subject
- We demonstrate successful radiolabeling of AAV, stable for up to 8 hours
- Labeled AAVs demonstrate reduced functionality indicated by lower transfections rate
- Rapid AAV bio-distribution was observed along the neuroaxis within the first hour
- High discrepancy observed between AAV bio-distribution and AAV transduction indicated by overlaid SPECT scan with CFT 3D data set.
- High resolution imaging of spine section confirmed the cellular localization of iRFP and demonstrated lack of transfection it the spinal cord.

AAV Pharmacokinetics

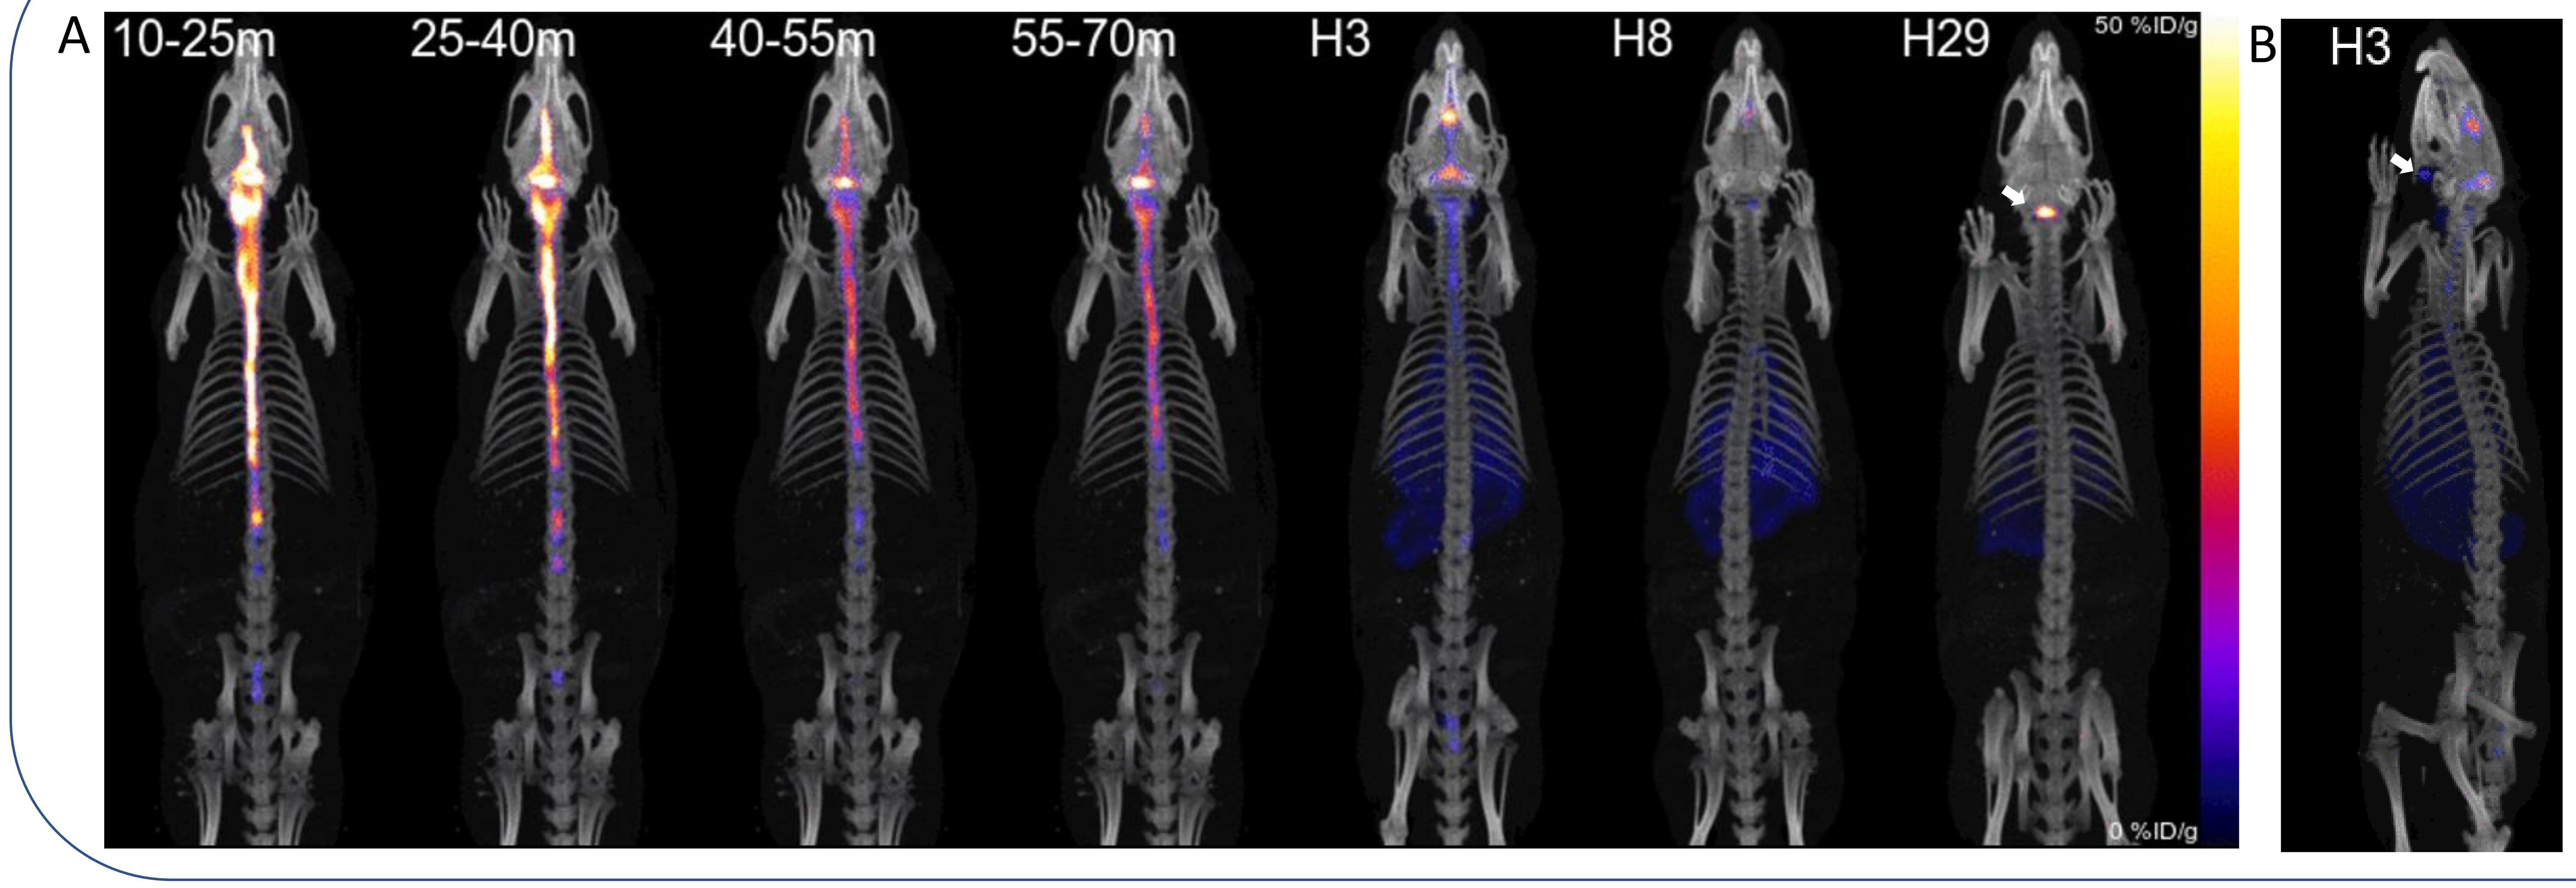
A
10-25m
25-40m
40-55m
55-70m
H3
H8
H29
50 %ID/g
0 %ID/g
B
H3

Figure 2: SPECT imaging of AAV capsids conjugated to ^{125}I conducted at several time points following intrathecal (IT) administration. Rapid bio-distribution along the neuroaxis was observed within the first hour of administration (A). Radiopharmaceutical uptake is observed at 3 hours, immediately to the right of midline at the level of the angle of the mandibles. The uptake is in the region of the mandibular salivary glands and lymph nodes, but could not be definitively localized to a specific tissue (B, arrow). ^{125}I conjugation was stable for up to 8 hours as indicated by signal localization to the thyroid (A, arrow).

AAV-Mediated Gene Transduction and Cellular Localization

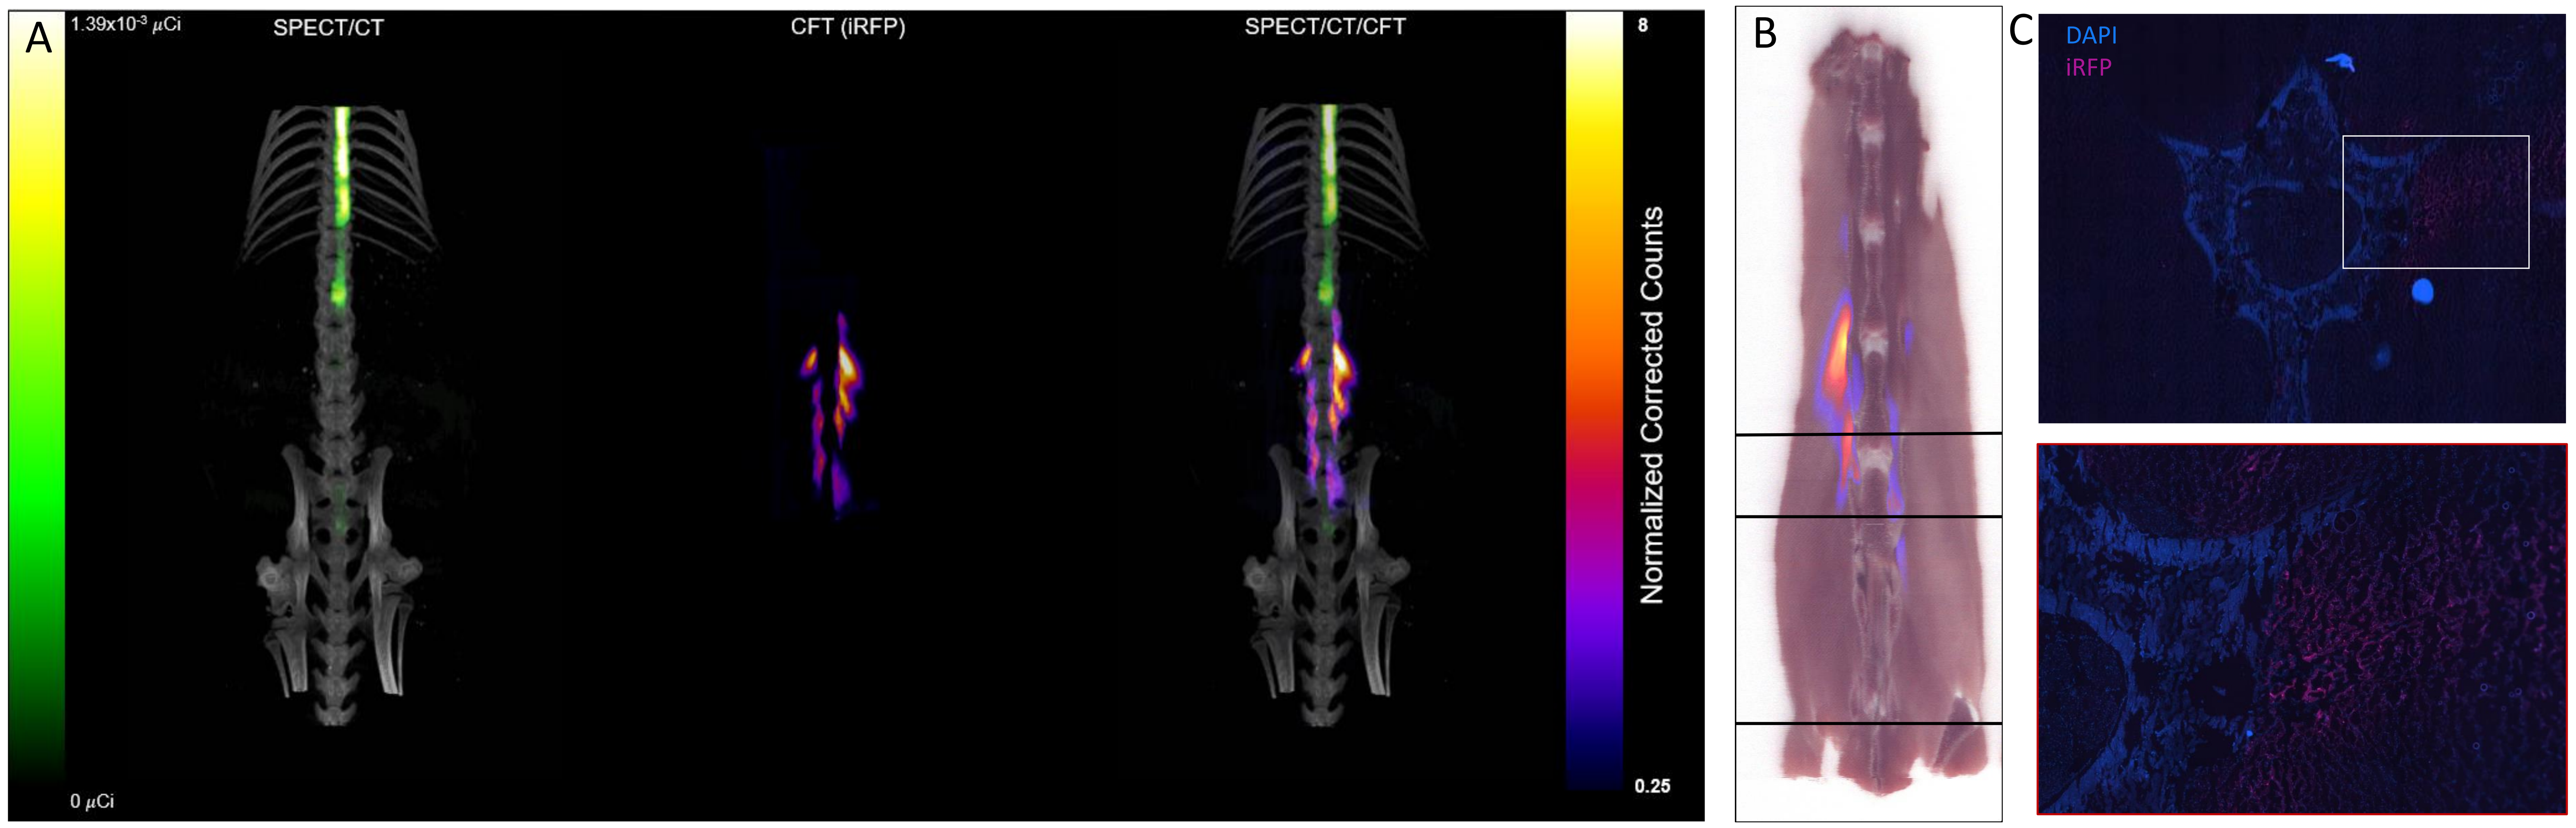
A
1.39x10^{-3} µCi
SPECT/CT
CFT (iRFP)
SPECT/CT/CFT
8
Normalized Corrected Counts
0.25
0 µCi
B
C
DAPI
iRFP

Figure 3: Comparing AAV-mediated iRFP expression to AAV capsid distribution (A). Four weeks following IT administration, once iRFP was sufficiently expressed, animals were taken down and the entire vertebrae resected and frozen whole. Whole Rat vertebrae was embedded in an OCT block for 3D image acquisition in the white light and fluorescent channels using CFT. Maximum intensity projection (MIP) of iRFP (A, middle) was overlaid onto the SPECT/CT scan (A, left) to compare AAV capsid bio-distribution to AAV-mediated transduction (A, right). Overlaid datasets demonstrate high discrepancy between the two. Further investigation to the CFT dataset, suggested that iRFP expression occurred outside the vertebrae (B), sections collected from the same sample were imaged in a microscope and confirmed iRFP expression in muscle tissue outside the vertebrae, likely the result of AAV leakage through the IT catheter.