Poster
W1230-03-20

Development of a Library of Plastics Additives for Identification of Extractables

Emma Leishman, Peipei Hu*, Alexis Langtry‡, Yizhong Wang, Andrew Kolbert
Avomeen Analytical Services, Ann Arbor, MI, USA

CONTACT INFORMATION: Emma Leishman, Ph.D., email: eleishman@avomeen.com, phone 855-631-1585
* Current affiliation = United States Pharmacopeia, ‡ Current affiliation = Michigan State University

aaps PharmSci 360®
Advancing Pharmaceutical Sciences, Careers, and Community

PURPOSE

It is important to identify extractables and leachables from pharmaceutical packaging and container closure systems for patient safety and submissions to regulatory authorities. Many of the substances reported in extractables and leachables studies are additives to the polymer formulations of the packaging and container closure systems.

To facilitate the identification of extractables for clients and employees, we developed a screening library of over 120 polymer additives.

METHODS: 7 CATEGORIES OF ADDITIVES

Bisphenol Analogs
- Used in asymmetric catalysis. Potential Endocrine disruptors.
- 7 samples. Example: Bis(4-hydroxyphenyl) sulfone

Accelerants
- Speed up chemical reactions
- 1 sample: 2-mercaptobenzothiazole

Antioxidants
- Scavenge free radicals to inhibit oxidation
- 79 samples. Example: 1,3-diphenyl-2-thiourea

Antidegradants
- Deter degeneration via oxidation, heat, UV light, weathering, ozone, and/or forms of dynamic fatigue
- 91 samples. Example: DL-α-Tocopherol

Plasticizers
- Reduce brittleness, promote flexibility and easier handling
- 48 samples. Example: Dibutyl phthalate

UV Stabilizers
- Protect the polymer product against UV light by absorbing light
- 12 samples. Example: 2,4-dihydroxybenzophenone

Cross-linkers
- Reagents for linking polymer chains together, which can improve elasticity, solidity, and electrical insulation
- 3 samples. Example: Stearic acid

Samples were diluted to 100 ppm in suitable solvents for analysis with HPLC, and to 10 ppm for analysis with LC-MS and GC-MS

RESULTS

Resolving Polymer Additives with HPLC and Obtaining Mass Spectra with LC-MS and GC-MS

Detection	Column	Mobile Phases	Gradient	Flow rate	Column Temp.	Injection volume	Number of analytes from each category						Total
							Antioxidant	Accelerant	Bisphenol analog	Cross-linker	Plasticizer	UV Stabilizer	
Diode array detector (DAD)	C18. 4.6 x 150 mm, 5 µm	A: H_2O + 0.1% formic acid B: ACN + 0.1% formic acid	5 min: 95% A, 5% B; 35 min: 5% A, 95% B; 40 min: 5% A, 95% B; 40.1 min: 95% A, 5% B; 45 min: 95% A, 5% B	1.0 mL/min	30 °C	20 µL	33	1	7	1	12	6	60
DAD	C8. 4.6 x 150 mm, 2.6 µm	A: H_2O + 0.1% formic acid B: ACN + 0.1% formic acid	Same as above	1.0 mL/min	30 °C	20 µL	2	0	0	0	7	0	9
DAD	Styragel. 4.6 x 300 mm, 5 µm	A: THF	Isocratic, 50 minutes	0.3 mL/min	40 °C	30 µL	2	0	0	0	0	0	2
ELSD	C8. 4.6 x 150 mm, 2.6 µm	A: ACN B: Chloroform	Same as row 1	1.25 mL/min	30 °C	20 µL	5	0	0	0	0	0	5
LC-MS (positive ESI)	Union	A: H_2O + 0.1% formic acid B: ACN + 0.1% formic acid	Isocratic, 5 minutes, 50% A 50% B	0.3 mL/min	N/A	5 µL	45	1	3	3	42	7	101
GC-MS	DB-1. 30 m x 0.35 mm, 0.25 µm	Wash solvent: IPA	Oven temperature ramp: 40 °C from 0-5 min, then to 300 °C at 10 °C/min, hold for at 300 °C for 10 min	He. 1.2 mL/min	Inlet temp 260 °C	1 µL	8	0	0	0	14	2	24

Overlay HPLC-DAD chromatograms

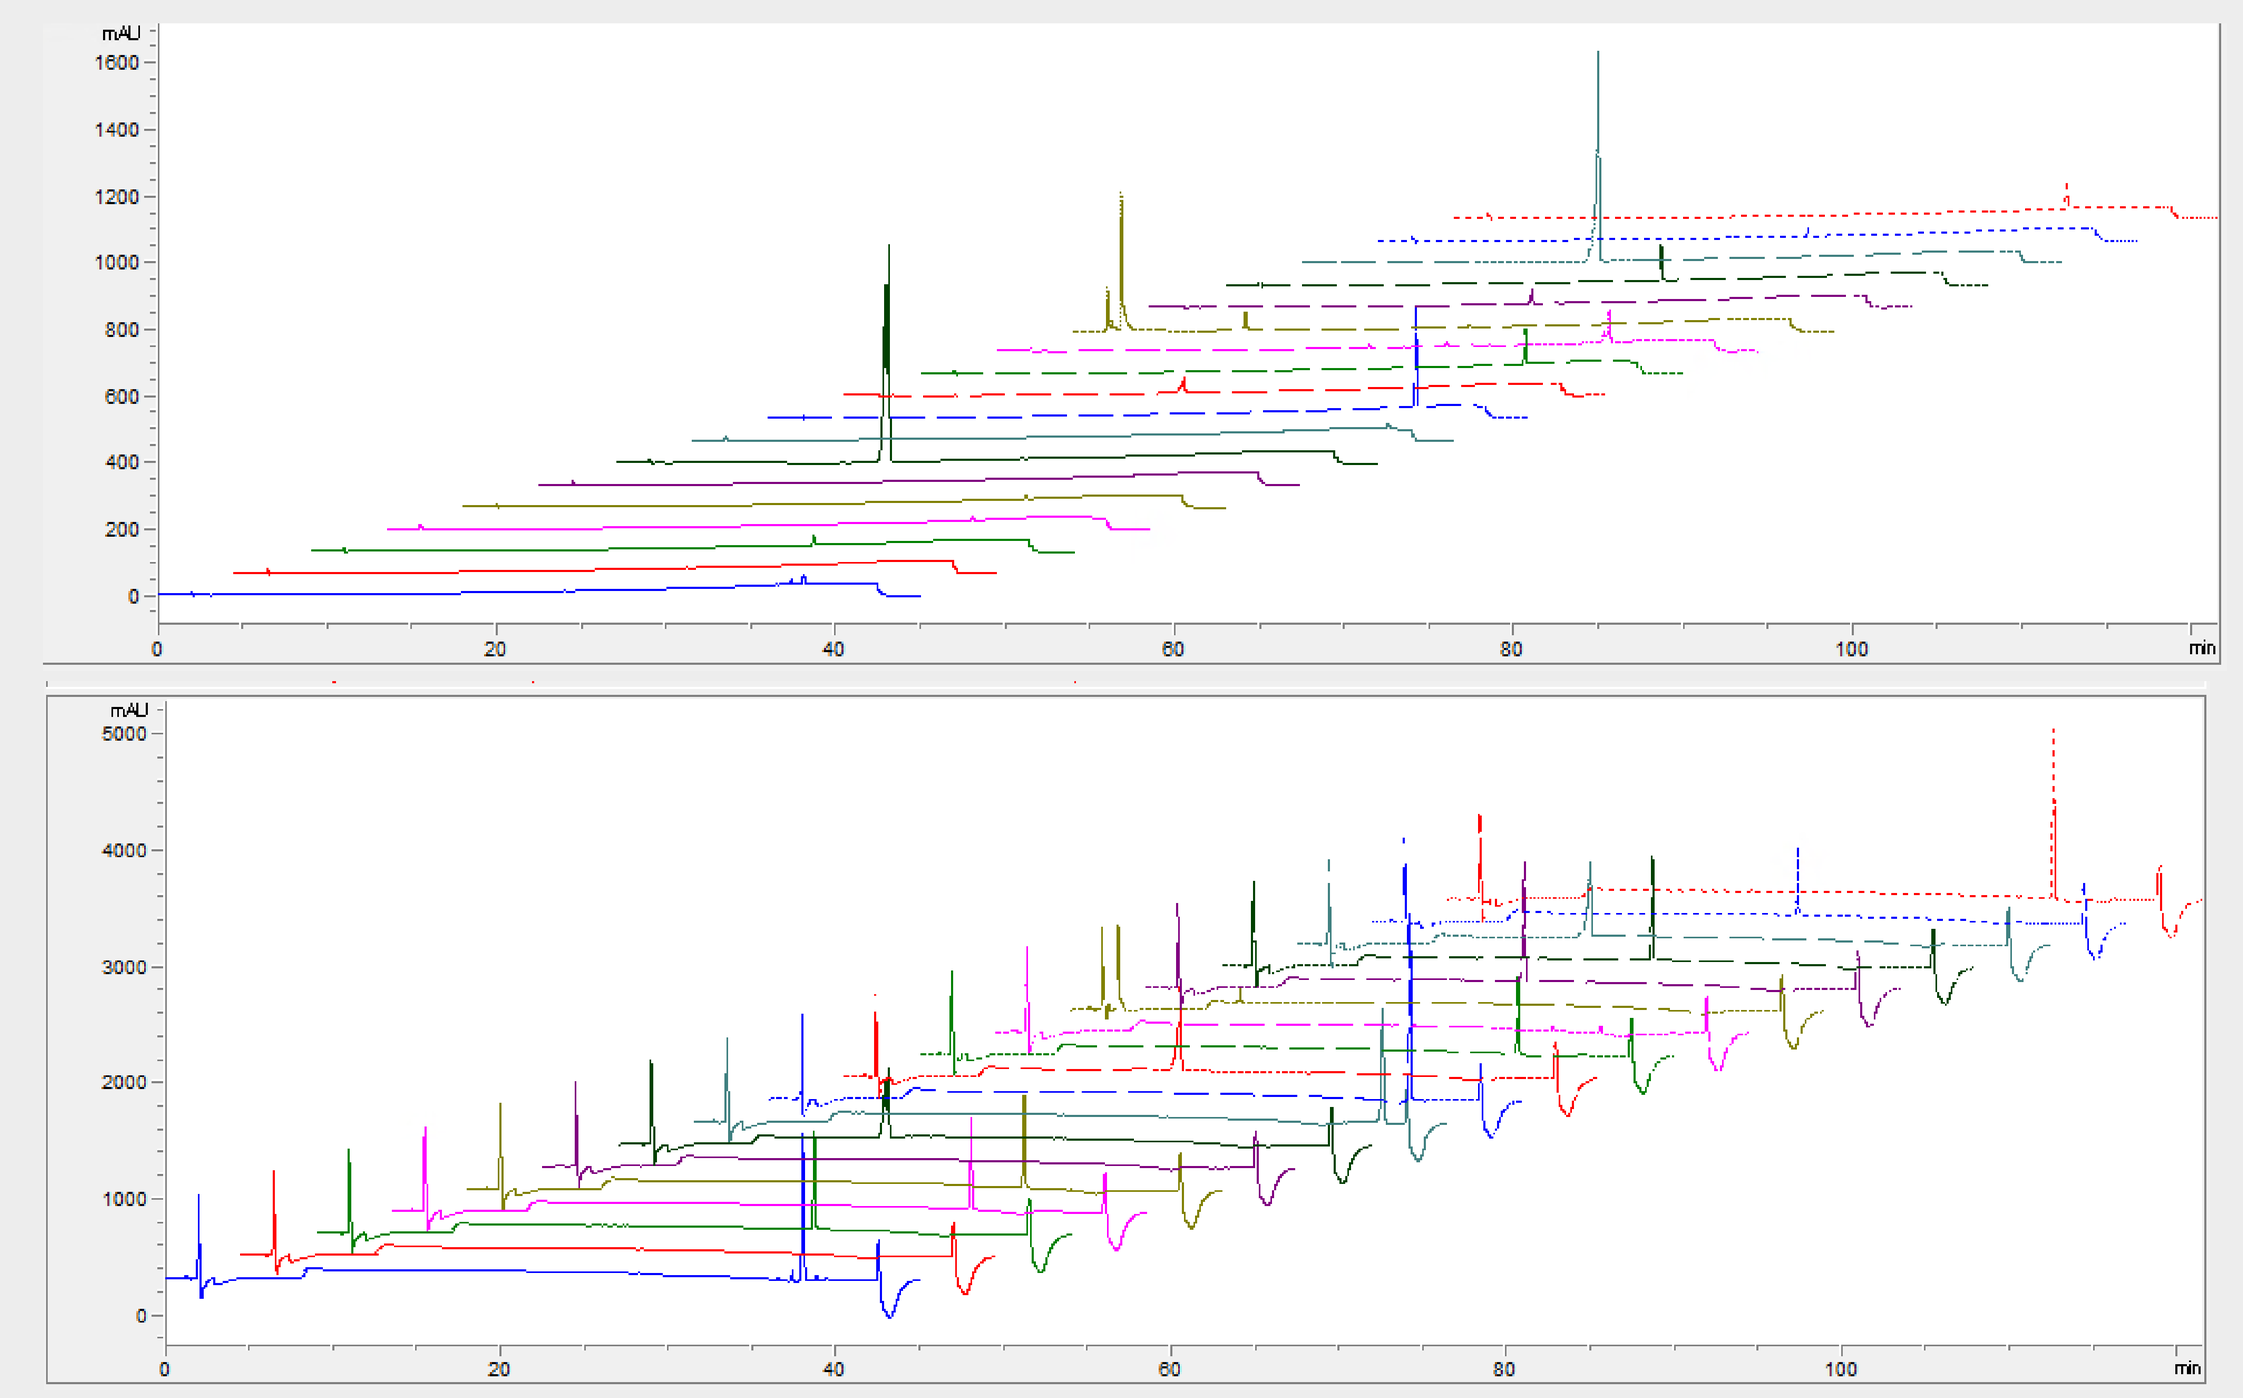

Application: **Extracts from everyday products will be analyzed using the same HPLC conditions. HPLC chromatograms (retention time and UV absorption) and LC-MS pattern will be compared to results generated in library for best match to find its identification.**

Total Ion Chromatograms and Mass Spectra for Tributyl Citrate

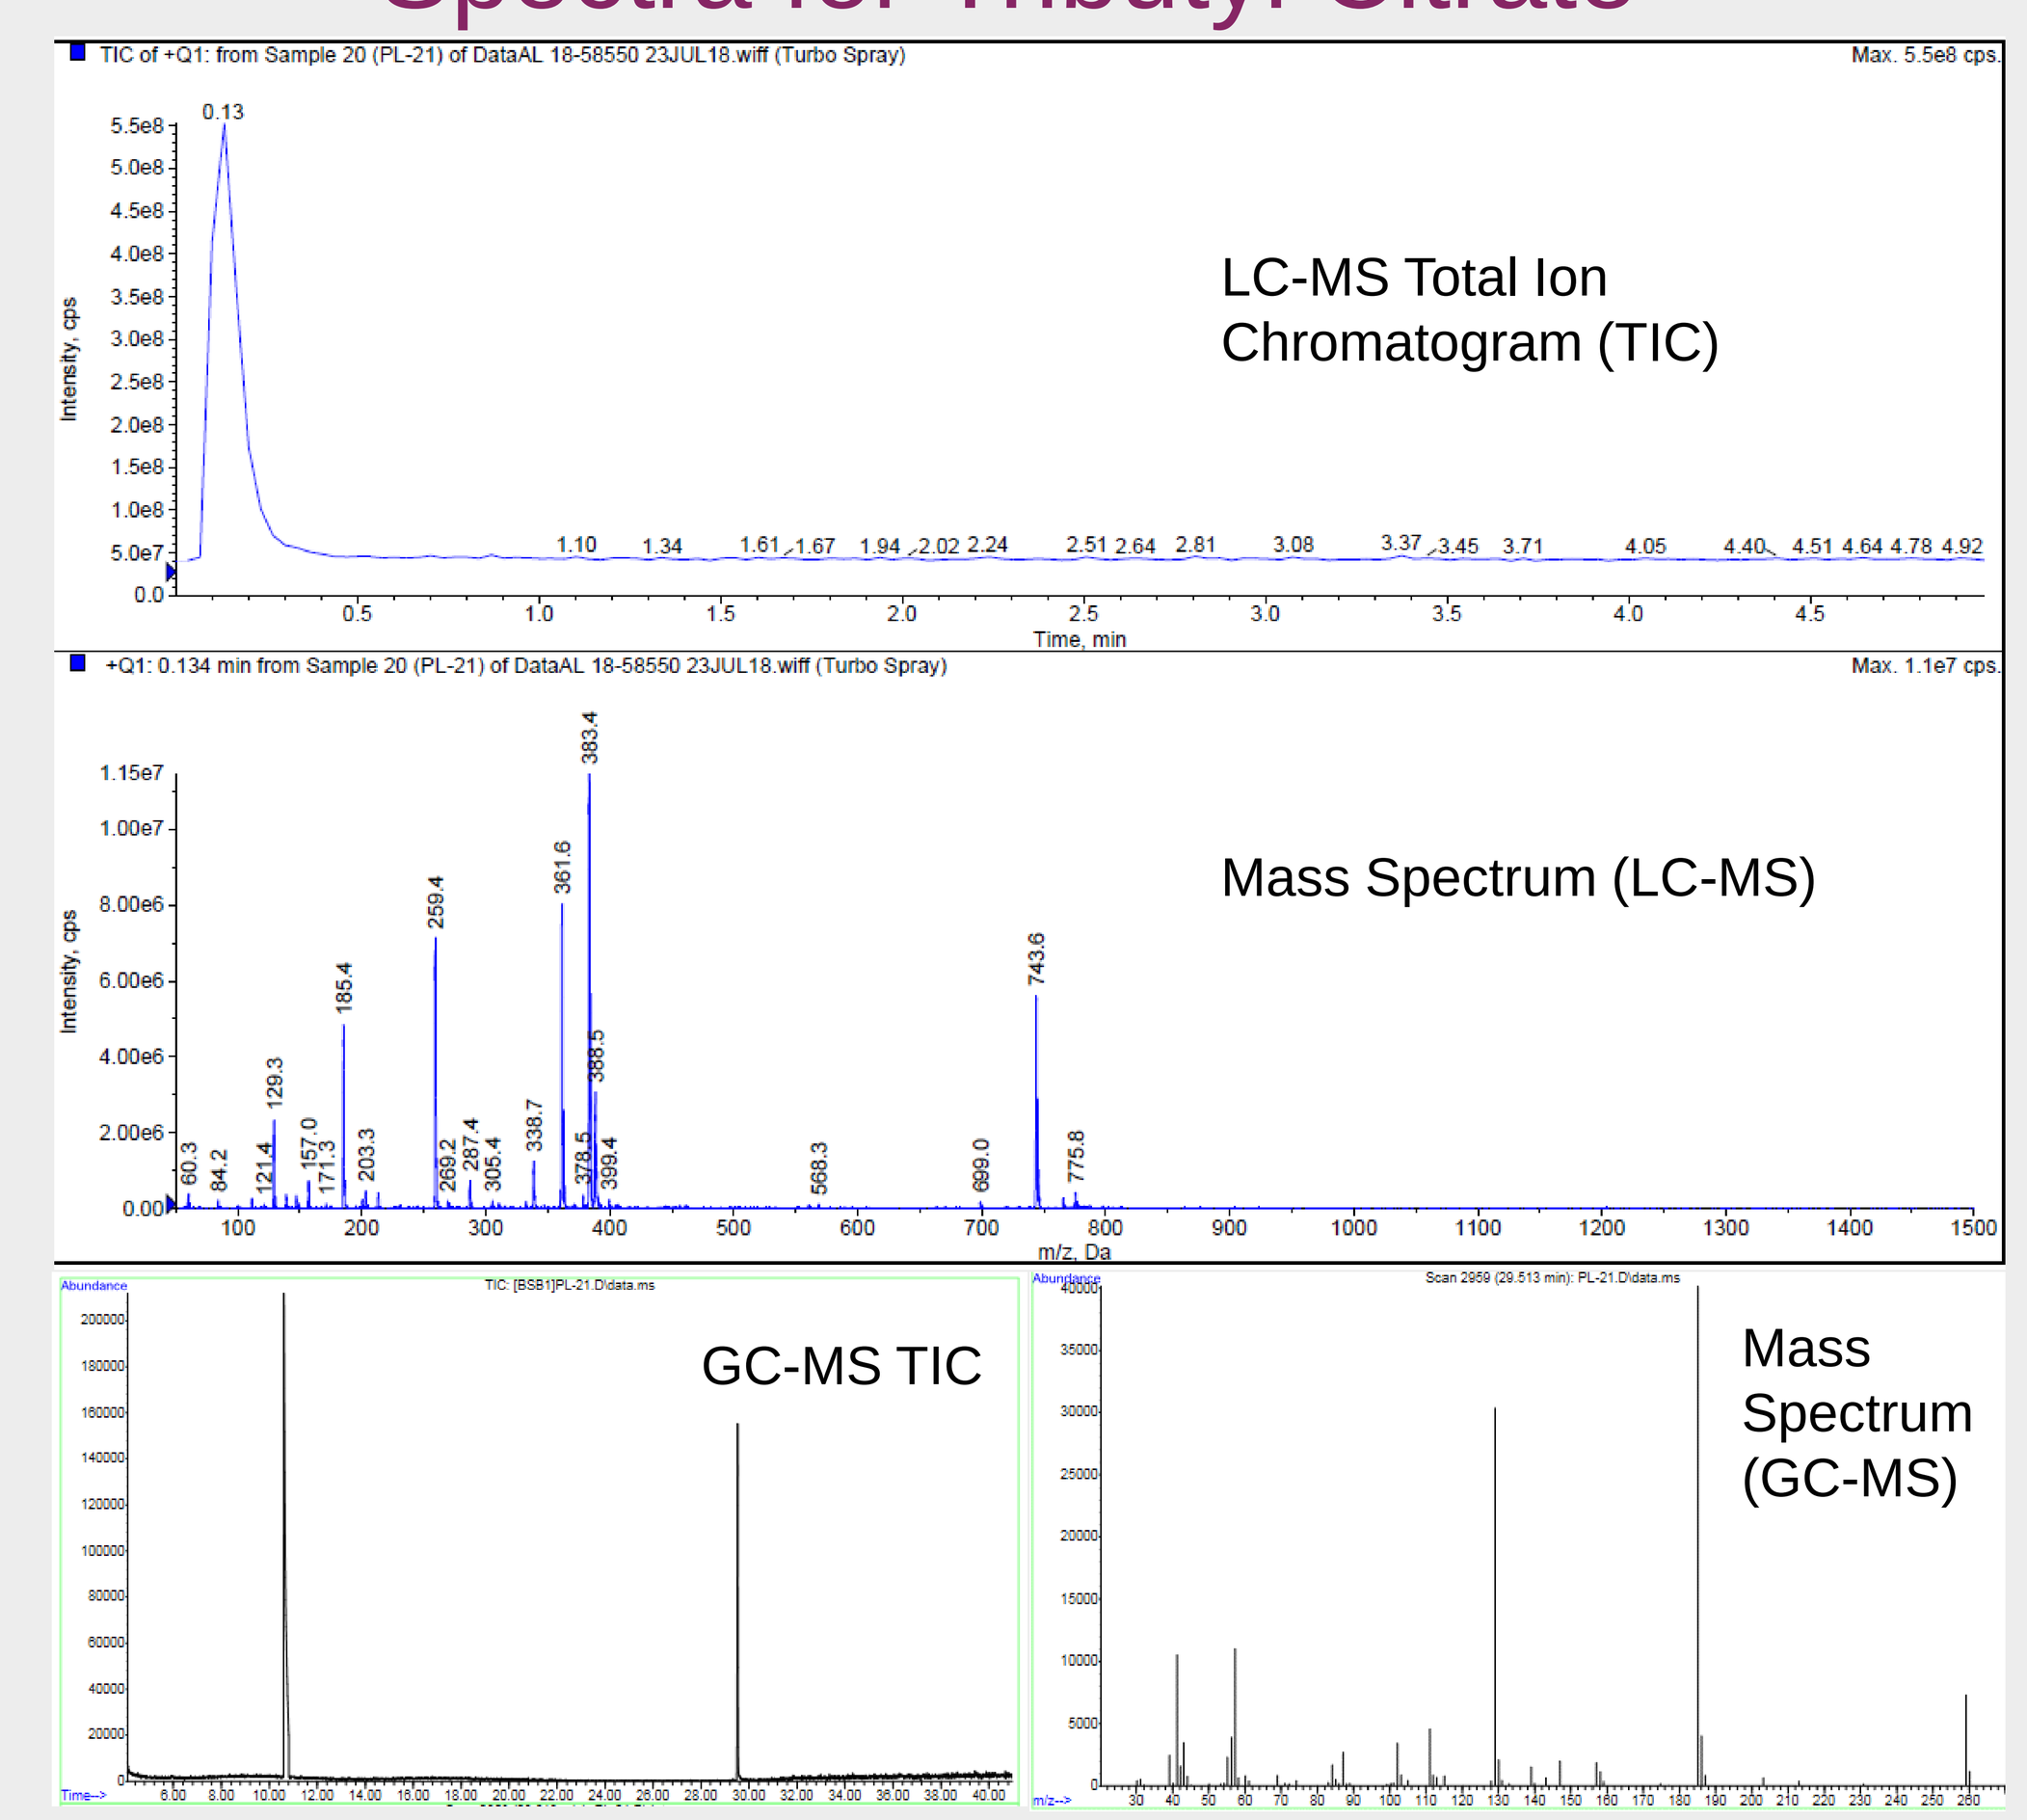

LC-MS Total Ion Chromatogram (TIC)
Mass Spectrum (LC-MS)
GC-MS TIC
Mass Spectrum (GC-MS)

- Tributyl citrate is an example of a bio-based plasticizer

Example: Benzyl Butyl Phthalate (BBP)

- BBP is a plasticizer added to various materials to promote flexibility

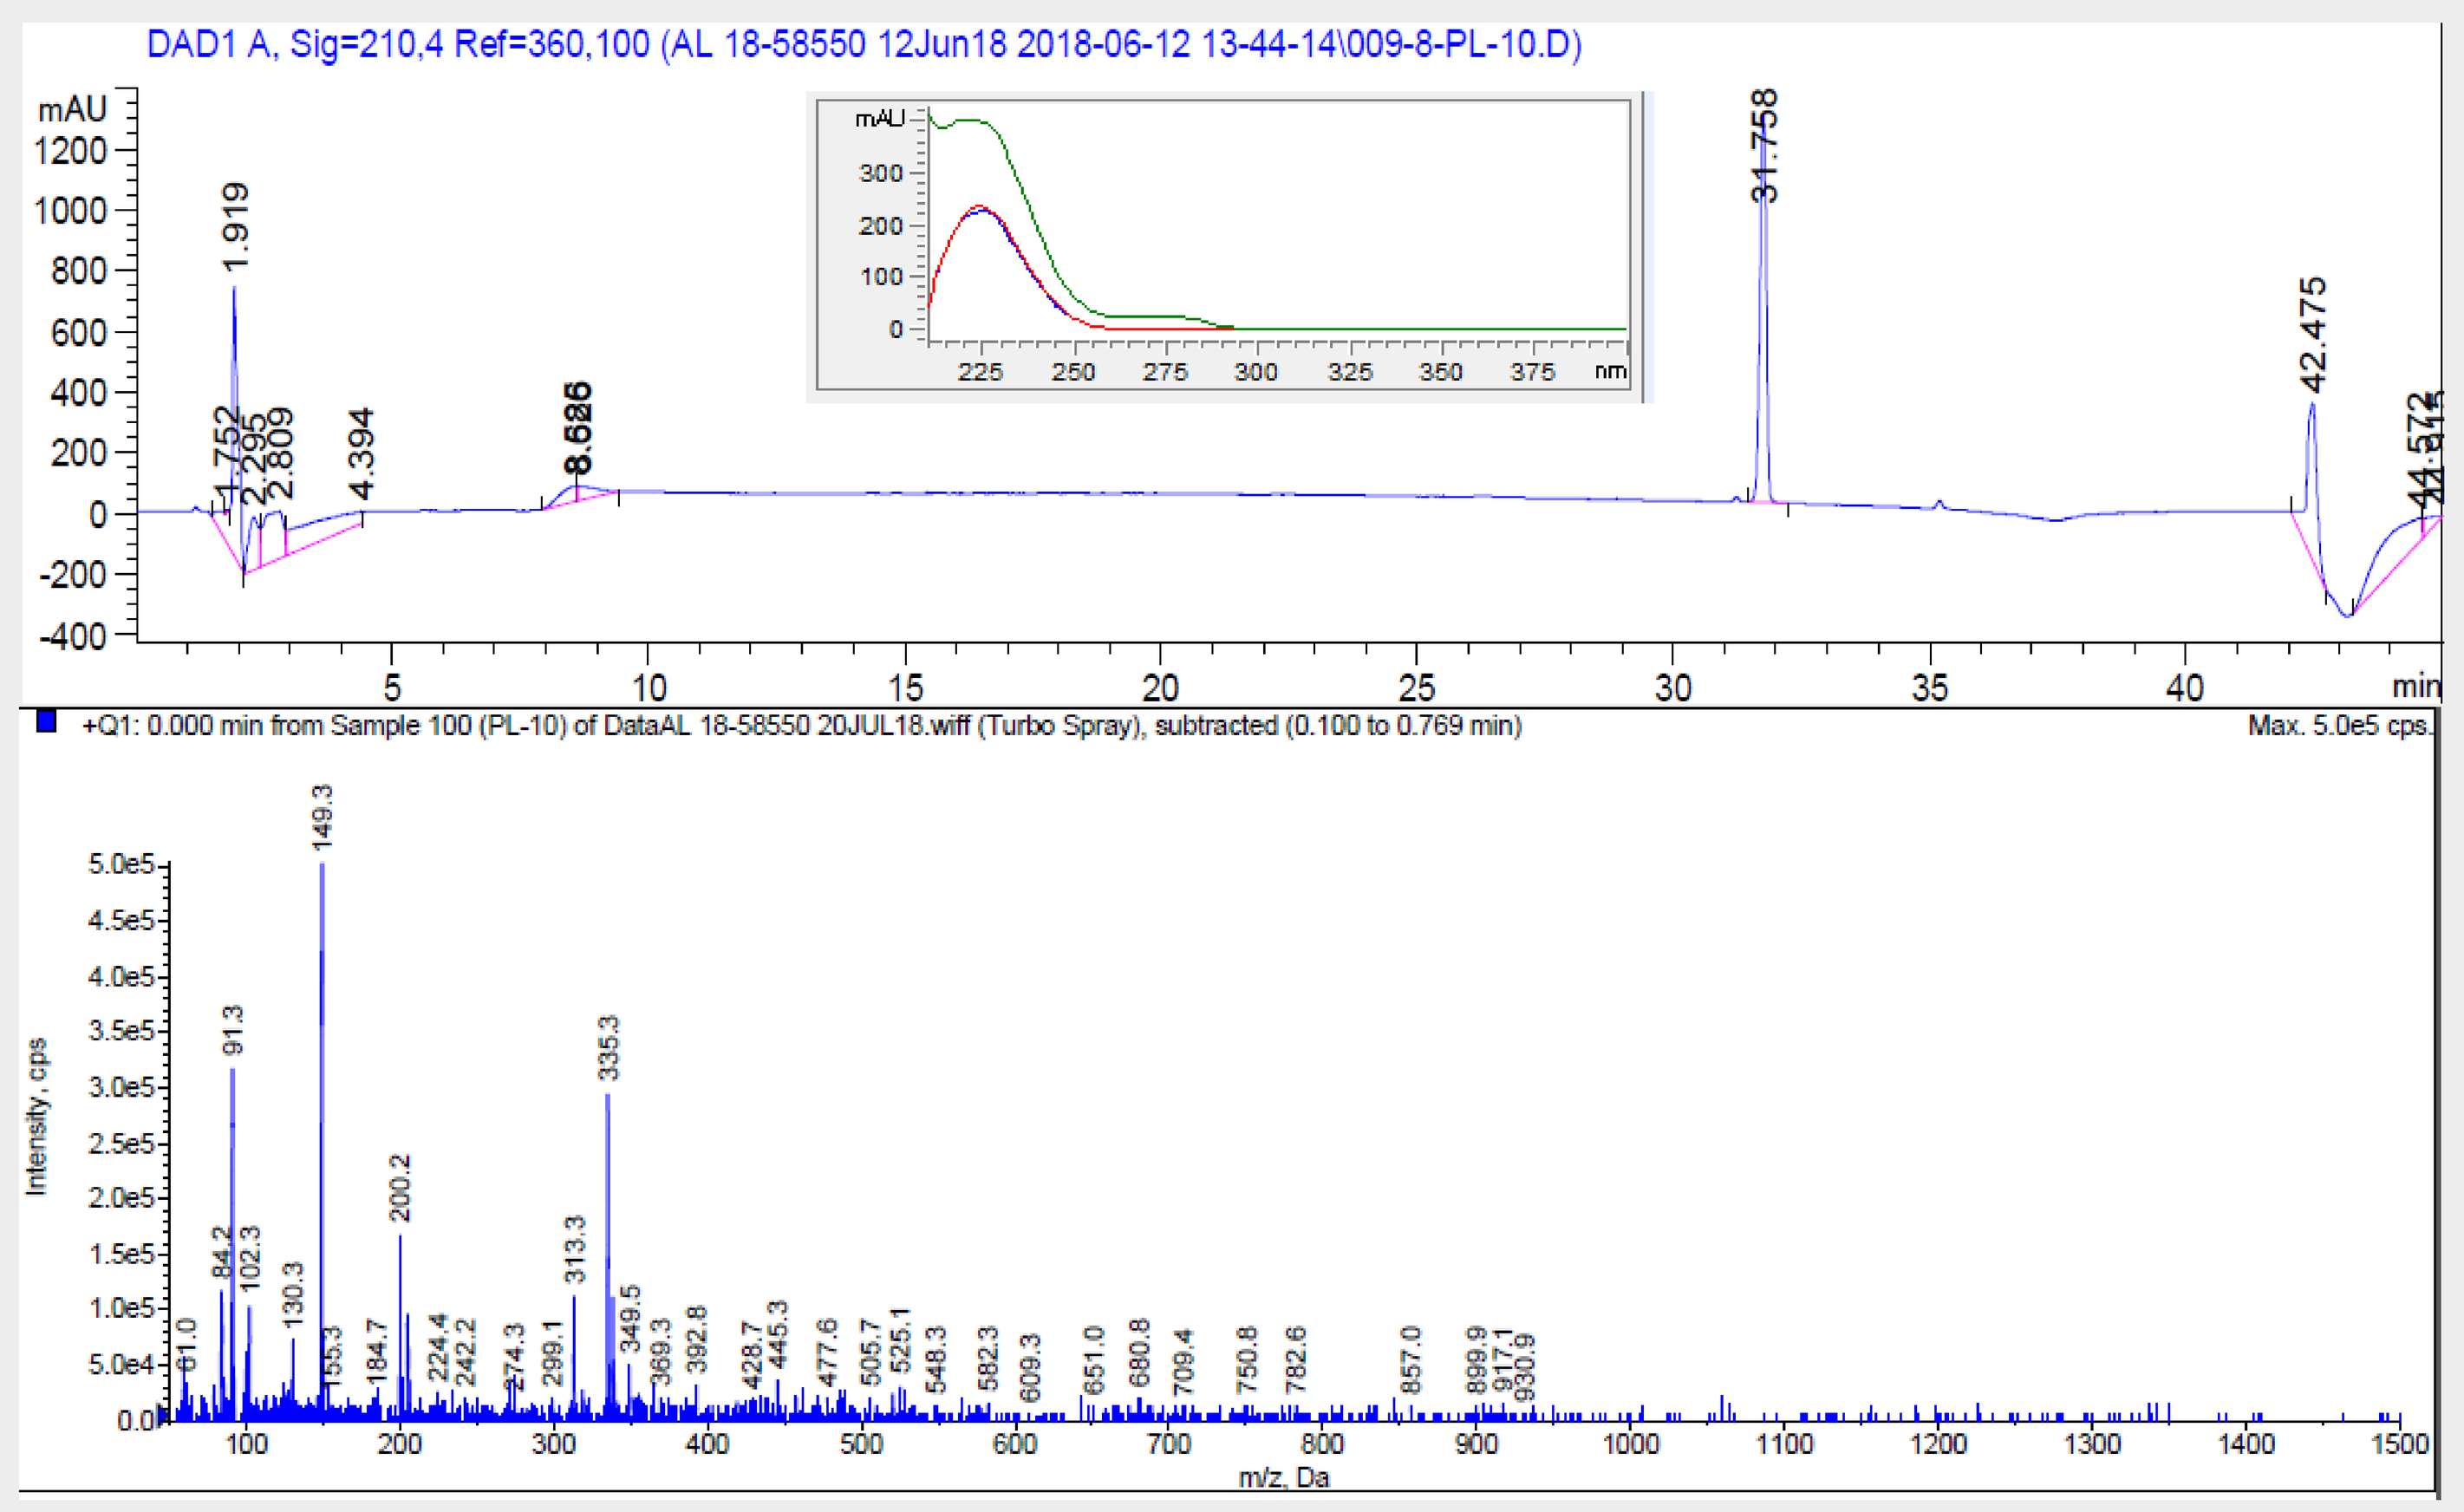

DAD1 A, Sig=210,4 Ref=360,100 (AL 18-58550 12Jun18 2018-06-12 13-44-14\009-8-PL-10.D)
+Q1: 0.000 min from Sample 100 (PL-10) of DataAL 18-58550 20JUL18.wiff (Turbo Spray), subtracted (0.100 to 0.769 min)
Max. 5.0e5 cps.

- The top panel shows the DAD signal, with UV absorbance data in the inset
- The bottom panel shows the mass spectra of BBP

CONCLUSION

The development of a polymer additive library of prevalent compounds observed during the analysis of everyday products facilitates rapid identification of polymer additives in extractables and leachables screenings.

AVOMEEN ANALYTICAL SERVICES

FDA-Registered, DEA-Licensed, GMP/GLP-Compliant, REACH/RoHS Compliant, ISO 17025-Accredited

Analytical Testing • Product Development • Pharma R&D • Problem Solving • Litigation Support

www.avomeen.com