#M1230-06-41

Development and Validation of In Vitro Release Test (IVRT) Method for Ophthalmic Suspensions – A Walkthrough Exercising Regulatory Guidance

Amit Dahal, Ameya Deshpande, and Srinivasan Shanmugam

Avomeen, 4840 Venture Dr, Ann Arbor, MI, USA 48108

CONTACT INFORMATION: sshanmugam@avomeen.com.

aaps 360

Advancing Pharmaceutical Sciences, Careers, and Community

PURPOSE

- In vitro release test (IVRT) has proven to be a useful technique to facilitate semisolid and topical/ophthalmic formulation development and approval processes.
- Although Guidance for Industry "Nonsterile Semisolid Dosage Forms: Scale-Up and Postapproval Changes" has been available for the industry since 1997, resource on method development and validation of IVRT method for ophthalmic suspensions is scarce[1].
- Ophthalmic suspensions are a class of non-invasive ocular formulations that contain dissolved (limited) and undissolved drug of specific particle size range.
- The purpose of this poster is to focus on delineating the critical parameters from *discriminatory method development to validation* of an IVRT method for ophthalmic suspensions in light of available regulatory and compendial guidelines.

OBJECTIVE(S)

The main objective of this work is

- To provide a step by step explanation of the critical elements/attributes required for IVRT method development and method validation for ophthalmic suspensions.

METHOD(S)

- Essential components of IVRT Method includes,
1. Analytical method to quantitate release of drug
2. An IVRT apparatus or Vertical Diffusion Cell (VDC) system (Fig.1 and Fig.2 along with accessories Fig. 3)

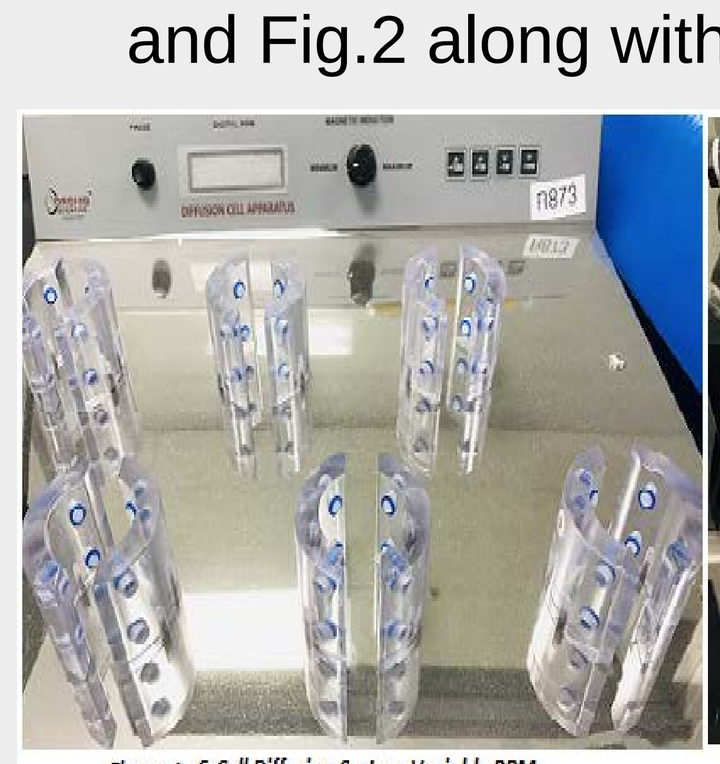

Figure 1. 6-Cell Diffusion System-Variable RPM

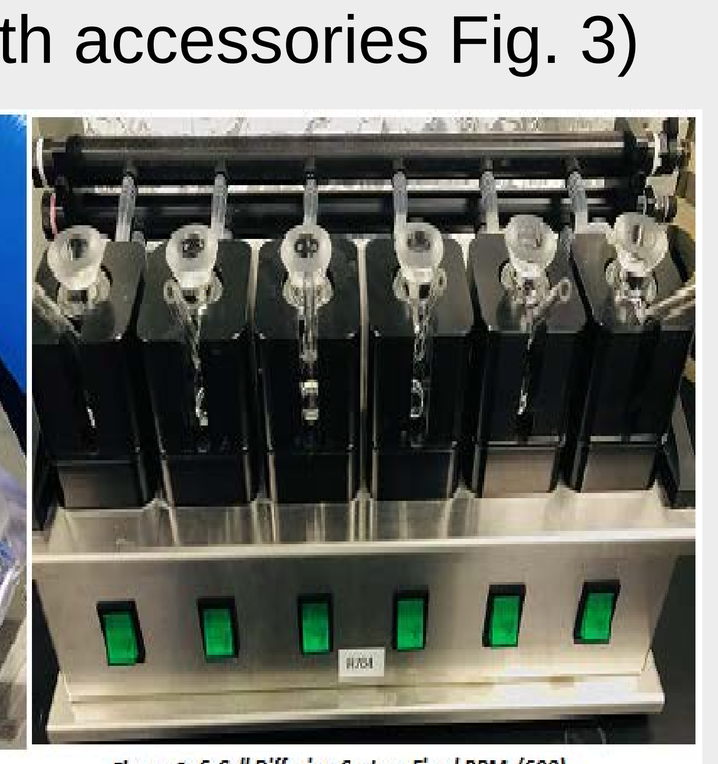

Figure 2. 6-Cell Diffusion System-Fixed RPM (600)

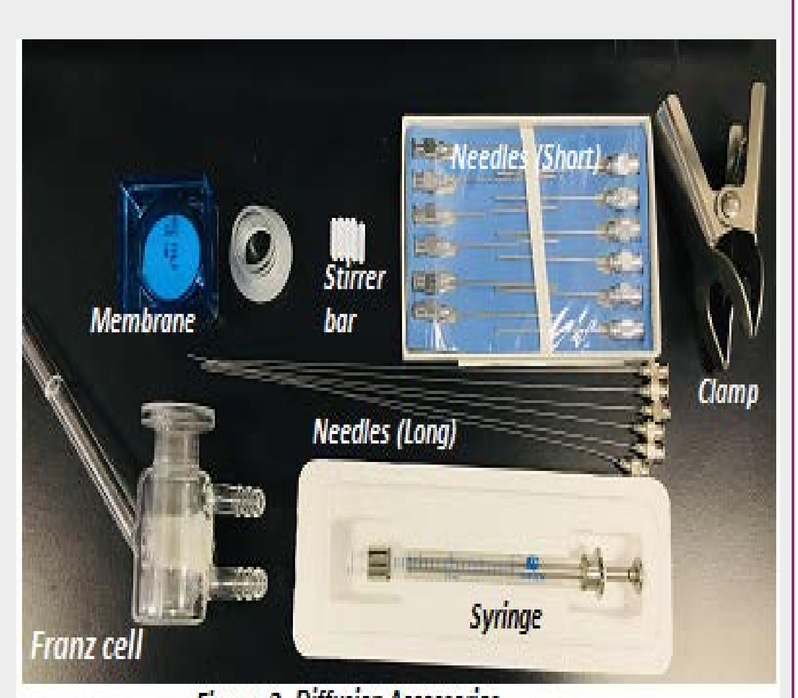

Membrane
Stirrer bar
Needles (Short)
Needles (Long)
Clamp
Franz cell
Syringe

Figure 3. Diffusion Accessories

Table 1. Critical Parameters of IVRT

Parameters	Value	
Diffusion System/Apparatus	Vertical diffusion cell (VDC) or Franz cell[1] (*Widely used*)	
Membrane	Hydrophilic or Hydrophobic	
Receptor medium	Physiologically based or Hydro-alcoholic	
Temperature for IVRT study	Ophthalmic application	37 ºC

- Primarily, for successful IVRT method development of ophthalmic suspensions, the following characteristics need to be evaluated and understood.
 - The physicochemical properties of the drug, specifically solubility, stability, and particle size.
 - Product characteristics such as pH, viscosity, dose strength, etc.

RESULT(S)

- Critical parameters associated with method development/validation of the ophthalmic suspension are summarized below:

IVRT METHOD DEVELOPMENT & VALIDATION

ANALYTICAL METHOD

- DEVELOPMENT: Sensitivity, Specificity, Solution stability, LOQ establishment
- VALIDATION: Linearity and Range, Specificity, Accuracy, Precision, Solution stability, Filter compatibility

IVRT METHOD

- DEVELOPMENT: Membrane Selection, Membrane compatibility, Sampling Volume, Dose Volume/amount, Sampling time, Sampling Volume
- VALIDATION: Membrane Compatib., Sensitivity, Specificity, Precision, Accuracy, Robustness

Table 2. Analytical Method Development Attributes

Sensitivity	• Method should be sensitive enough to detect the lowest concentration at predetermined time points for quantification
Specificity	• No interference from the diluent/medium and placebo peak at the retention time of API peak for quantification
Analytical Solution Stability*	• Degradation of NMT 2% for 24 hr. or the duration of Analysis is established
LOQ	• Method should be able to quantify the drug as low as possible to measure the release from formulations

** Product Specific*

Table 3. IVRT Method Development Attributes

Membrane Selection*	• Evaluate the inertness of membrane to drug; should be no/low drug binding • Evaluate the resistance to diffusion; should be minimal resistance • Membrane compatibility to medium (integrity issues) • Discriminatory power (***based on particles sizes/formulation variables***) of suspensions • Fit into the diffusion cell diameter (e.g., 25 mm standard)
Receptor solution Selection	• Drug solubility in the medium at experimental temperature • Drug solubility in the formulation • Physiological based (ex- Phosphate buffer pH 7.4) with or without surfactants • Hydro alcoholic based (aqueous ethanol or suitable solvent) • Sink condition establishment-at least 3X • Degassing the medium
Sampling Time	• Achieve steady state diffusion w/o lag and plateau effect • Multiple sampling times (at least 5 times) over an appropriate time period • Generate adequate release profile for analysis/comparison • Typically up to 6 hours
Sample/Dose Volume	• Typically ≤1 g or equivalent volume • Infinite dose condition • Occluded
Sampling Volume/Sample withdrawn	• ≤ 0.5 mL is recommended • More the sample volume more intra/inter cell variation • Air bubble removal consideration • Filter the sample and analyzed using developed analytical method
Robustness	• <u>Temperature:</u> 100% reference ±2 °C for 6 cells for standard formulation • <u>Stirring rate:</u> ± 50-200 RPM from the standard RPM to ensure homogeneity w/o release profile with minimal or low membrane/medium interface perturbation • <u>pH variation</u>: ± 0.2 units of receptor medium

Table 4. Commercially Available Synthetic Membranes

Name/Brand		Composition/Material	Pore size (µm)	Source/Manufacturer
Polymer Based	Supor 450	Polyethersulfone (PES)	0.1 – 0.8	Pall
	Versapor 450	Acrylic Copolymer	0.45 – 3	Pall
	Fluropore	Polytetrafluroethylene (PTFE)	0.2	Millipore
	Isopore	Polycarbonate (PC)	0.2	Millipore
	Nulcepore	Polycarbonate (PC)	0.10	Whatman
	Cyclopore	Polycarbonate (PC)	0.10	Whatman
	Tuffryn	Polysulfone (PC)	0.45	Pall
	Biodyne	Polyamide (Nylon)	0.45	Pall
	Silicone	Polydimethysiloxane (PDMS)	-	SAMCO
Cellulose Based	Advantee	Cellulose acetate	0.2	Advantee
	Metricel	Mixed Cellulose ester	0.45	Pall
	MF	Cellulose acetate/ nitrate mixtures	0.45	Milipore
	Cuprophan	Regenerated Cellulose	0.45	Medicell
	Visking	Regenerated cellulose	0.45	Medicell

Systematic Development Approach for IVRT Method for Ophthalmic Suspension[1]

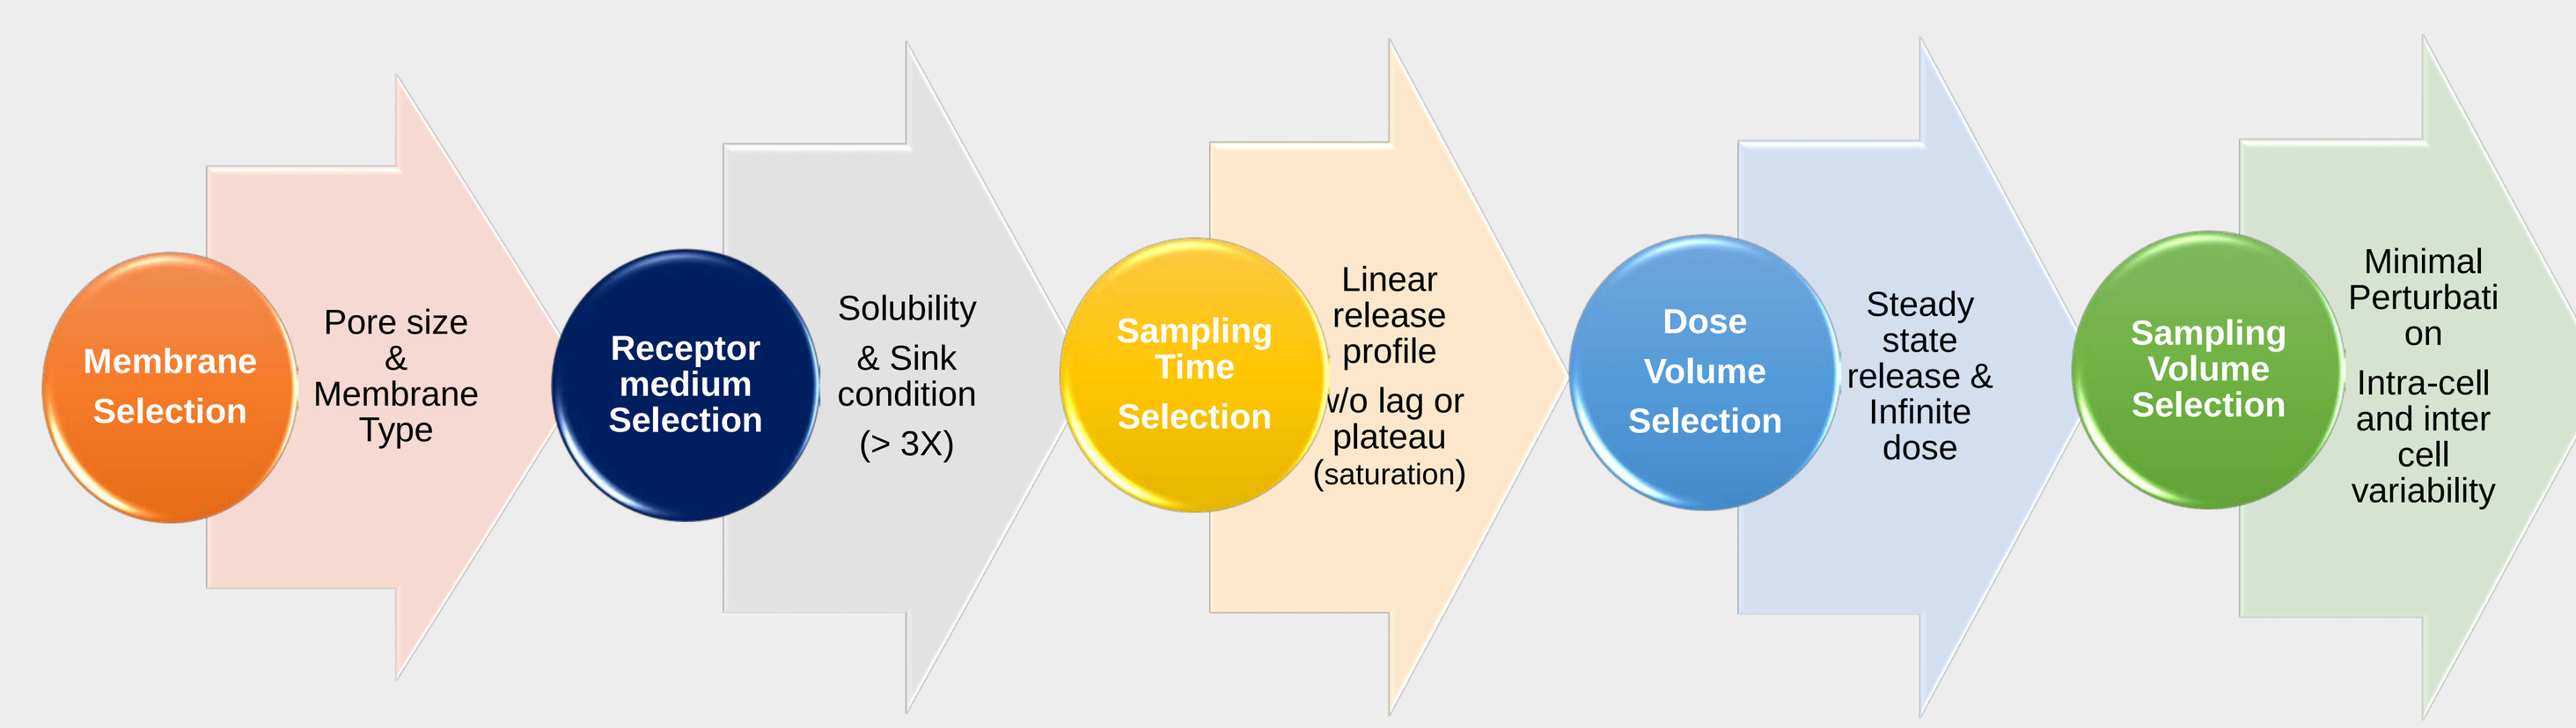

Membrane Selection
Pore size & Membrane Type
Receptor medium Selection
Solubility & Sink condition (> 3X)
Sampling Time Selection
Linear release profile w/o lag or plateau (saturation)
Dose Volume Selection
Steady state release & Infinite dose
Sampling Volume Selection
Minimal Perturbation on Intra-cell and inter cell variability

Table 5. Analytical Method Validation

Validation Parameters		Study	General Criteria*
Linearity		Range (min. five concentration levels)	Correlation coefficient ≥ 0.98
System Precision/Suitability		% RSD of 5 OR 6 injections of standard	%RSD ≤ 2 OR 3%
		USP Tailing factor	Tailing factor ≤ 2
Accuracy	***Lower % level***	Recovery performance with product placebo matrix spiked with lower% (50-80), 100 % and higher % (120-150) levels of standard solution	Recovery 90-110% in each level
	100% level		
	Higher % level		
Method Precision	***Precision (Repeatability)***	Calculate the % RSD for 100 % level for 6 standard replicates	% RSD ≤ 2
	Reproducibility (Intermediate)	Calculate the % RSD for 100 % level for 12 (6 standard replicates x 2 days) diff. analyst/system/column	% RSD ≤ 2
LOD		Calculate the S/N ratio for 0.5% of standard	S/N ≥ 3 OR Establish concentration with S/N ≥ 3
LOQ		Calculate the S/N ratio for 1% of standard	S/N ≥ 10 OR Establish concentration with S/N ≥ 10
		Calculate the % RSD for the 6 replicate injections of LOQ samples	%RSD ≤ 20
Specificity		Observe the chromatograms for Placebo, Diluent and receptor media	No interference of the diluent or placebo matrix at the retention time of API peak
Analytical Solution Stability		Standard solution will be tested for 24 hr.	Solution is stable if degradation is NMT 2%
Filter Compatibility		Compare recovery of filtered and unfiltered standard solution	% Recovery NLT 95%

**Recommended criteria, and is product specific*

Table 6. IVRT Method Validation Elements

Validation[1] parameters	Study	Criteria*
Apparatus[2,3] Qualification	<u>Franz cell (VDC)</u> Receptor chamber volume (12 mL), Orifice diameter (Standard-25 mm) Stirrer rate (Stanard-600 RPM), Temperature of heater/circulator	
Membrane[2] Compatibility	Soak membrane in the standard solution prepared in the receptor media @ 37 °C for the duration of study	% Recovery in between 90-110 %
Sensitivity	Perform IVRT study for 50 %, 100 % and 150 % of the test formulation in 6 cells in three different runs/days	Mean cumulative release rate (flux, µg/cm²/hr$^{1/2}$) should not be overlapping (different) i.e. 150 % > 100 % > 50 %
Specificity	Linearity of release rate (flux, µg/cm²/hr$^{1/2}$) from sensitivity run of 50 %, 100 %, and 150 %	Proportionality R^2 ≥ 0.9
Selectivity	Sameness testing (non-parametric analysis) of 50 %, 100 %, and 150 %	Fails sameness testing (outside of 75-133.33%)
Precision	Calculate the % RSD in between the intra-day run inter-day run from sensitivity runs	% RSD for mean (flux, µg/cm²/hr$^{1/2}$) intra-run, one run, n=6 should be NMT 15 %
		% RSD for mean (flux, µg/cm²/hr$^{1/2}$) for final pull point for 3 different runs(days), n=3 should be NMT 20 %
Accuracy	Mass balance and dose depletion calculated from sensitivity studies for 100% level	Report results
Robustness	Perform the IVRT study with the temperature change for 100 % reference ±2 °C for 6 cells for standard formulation	% RSD for the cumulative release should be NMT 20%

**Recommended criteria, and is product specific*

CONCLUSION(S)

- The IVRT method has been serving as an excellent tool to demonstrate the performance of topical semi-solid preparations, and has applicability for suspension performance too.
- The IVRT not only requires technical expertise but also regulatory understanding given its applicability in quality control and regulatory filings including bio-waivers.
- This work reiterates the key components for the discriminatory IVRT method development and validation for the purpose of performance evaluations of ophthalmic suspension formulations that is required as part of various regulatory filings.

REFERENCE(S)

1. Nonsterile Semisolid Dosage Forms; Scale-Up and Postapproval Changes:Chemistry, Manufacturing, and Controls;In Vitro Release Testing and In Vivo Bioequivalence Documentation (SUPAC-SS)
2. Tiffner, Katrin I., et al. "A comprehensive approach to qualify and validate the essential parameters of an in vitro release test (IVRT) method for acyclovir cream, 5%."
3. USP chapter <1724> Semisolid Drug Products - Performance Tests

Avomeen's IVRT/IVPT expertise include,

- ✓ *Systematic/comprehensive discriminatory method development and validation*
- ✓ *Bioequivalence waiver and sameness testing for all topical products (RLD vs. Test Product)*
- ✓ *Product release testing and SUPAC analysis*
- ✓ *Q3 (microstructure/performance) testing*
- ✓ *Responses to FDA deficiencies & regulatory dossier preparation*

For any inquiries or service related information, please visit https://www.avomeen.com/the-use-of-in-vitro-release-testing-ivrt-and-in-vitro-permeation-test-ivpt-for-semi-solid-topical-formulations/

AVOMEEN